P0339

ECCMID
Amsterdam, Netherlands
9-12 April 2016

Antimicrobial Activity of Ceftolozane/Tazobactam from the PACTS Programme Tested against Enterobacteriaceae Isolated from Patients Hospitalised in Intensive Care Units from 19 European Countries, Turkey and Israel (2013-2014)

Farrell DJ; Flamm RK; Castanheira M; Jones RN
JMI Laboratories, North Liberty, IA, USA

David J. Farrell, PhD
JMI Laboratories
345 Beaver Kreek Center, Suite A
North Liberty, IA 52317, USA
Phone: (319) 665-3370
Fax: (319) 665-3371
david-farrell@jmilabs.com

Introduction and Purpose

- Ceftolozane/tazobactam is an antibacterial consisting of ceftolozane, a novel antipseudomonal cephalosporin, and tazobactam, a well-established β-lactamase inhibitor[1]
- Ceftolozane exerts its bactericidal activity by inhibiting essential penicillin-binding proteins, resulting in inhibition of cell wall synthesis and subsequent cell death[1]
- Compared with ceftazidime and cefepime, ceftolozane demonstrates greater activity against *Pseudomonas aeruginosa*[2-4]
- Tazobactam is a potent inhibitor of most common class A and some class C β-lactamases; binding to the active site of these enzymes protects ceftolozane from hydrolysis and broadens coverage to include most extended-spectrum β-lactamase (ESBL)–producing Enterobacteriaceae and some AmpC–de-repressed Enterobacteriaceae[2,5]
- Ceftolozane/tazobactam has been approved by the US Food and Drug Administration for the treatment of complicated urinary tract infections and, in combination with metronidazole, for the treatment of complicated intra-abdominal infections[1]; a Phase 3 trial for the treatment of nosocomial pneumonia is under way
- In this study, we evaluated the *in vitro* activities of ceftolozane/tazobactam, ceftazidime, piperacillin/tazobactam, meropenem and other comparator agents against clinical Enterobacteriaceae isolates obtained from intensive care unit (ICU) patients in Europe, Turkey and Israel during 2013 and 2014

Methods

Organism collection

- 1,258 Enterobacteriaceae isolates were consecutively collected from ICU patients from 39 medical centres located in 21 countries (number of centres) during 2013 and 2014: Belgium (1), Czech Republic (1), Denmark (1), Finland (1), France (4), Germany (5), Greece (1), Ireland (2), Israel (1), Italy (4), Netherlands (1), Norway (1), Poland (1), Portugal (1), Russia (3), Spain (3), Sweden (1), Switzerland (1), Turkey (2), Ukraine (1) and United Kingdom (3)

Antimicrobial susceptibility testing

- Isolates were tested for susceptibility to multiple antimicrobial agents at a reference laboratory (JMI Laboratories) by standardised broth microdilution methods as described in Clinical and Laboratory Standards Institute (CLSI) M07-A10[6]
- Validated minimum inhibitory concentration (MIC) panels were manufactured by ThermoFisher Scientific Inc. (Oakwood Village, OH, USA)
- MIC results were interpreted according to CLSI criteria in M100-S26[7] (2016) and European Committee on Antimicrobial Susceptibility Testing (EUCAST) breakpoint tables (version 6.0, January 2016)[8]
- *Escherichia coli* and *Klebsiella* spp. isolates with MIC ≥2 mg/L for ceftazidime, ceftriaxone or aztreonam were categorised as ESBL phenotypes
- Multidrug-resistant (MDR), extensively drug-resistant (XDR) and pandrug-resistant (PDR) Enterobacteriaceae strains were classified according to recent guidelines using non-susceptibility (EUCAST breakpoints)[8] to ceftriaxone, meropenem, piperacillin/tazobactam, levofloxacin, gentamicin, tigecycline and colistin
 - Classifications were based on the following recommended parameters: MDR = non-susceptible to ≥3 antimicrobial classes; XDR = susceptible to ≤2 antimicrobial classes; PDR = non-susceptible to all antimicrobial classes[9]
- Concurrent quality control (QC) testing was performed to ensure proper test conditions and procedures
 - QC strains included *E. coli* ATCC 25922 and 35218 and *P. aeruginosa* ATCC 27853
 - QC ranges and interpretive criteria for comparator compounds used the CLSI M100-S26[7] guidelines, and all QC results were within published ranges

Results

- Overall, 82.3% of 1,258 Enterobacteriaceae isolates tested were susceptible to ceftolozane/tazobactam (minimum inhibitory concentration required to inhibit the growth of 50% and 90% of isolates [$MIC_{50/90}$], 0.25/8 mg/L) using EUCAST breakpoints (**Table 1**)
- Meropenem was the most active agent against Enterobacteriaceae ($MIC_{50/90}$, ≤0.06/0.12 mg/L; 95.2% susceptible), followed by tigecycline (94.6%) and gentamicin (84.2%; **Table 2**) using EUCAST breakpoints
- EUCAST susceptibility rates for other agents were lower: ceftazidime: 70.8%; cefepime: 77.1%; ceftriaxone: 68.4%; piperacillin/tazobactam: 73.8%; levofloxacin: 77.5%; and colistin: 78.6% (**Table 2**)
- 317 isolates of Enterobacteriaceae (25.2%) were MDR
 - Ceftolozane/tazobactam retained activity ($MIC_{50/90}$, 2/>32 mg/L) against 46.1% of strains at the EUCAST breakpoint of ≤1 mg/L (**Table 1**)
 - Ceftolozane/tazobactam was up to 16-fold more potent than ceftazidime (MIC_{50}, 32 mg/L) and piperacillin/tazobactam (MIC_{50}, 32 mg/L) and at least 4- and 8-fold more potent than ceftriaxone (MIC_{50}, >8 mg/L) and cefepime (MIC_{50}, >16 mg/L), respectively, against the MDR strains (**Table 2**)
 - Meropenem was the most potent agent ($MIC_{50/90}$, ≤0.06/>8 mg/L; 81.0% susceptible by EUCAST criteria) against the MDR subset, followed by tigecycline ($MIC_{50/90}$, 0.5/2 mg/L; 86.1% susceptible by EUCAST criteria; **Table 2**)
 - Colistin susceptibility was 68.7% against the MDR subset (**Table 2**)
- Overall, 87 isolates of Enterobacteriaceae (6.9%) were classified as XDR
 - Ceftolozane/tazobactam exhibited limited activity against XDR Enterobacteriaceae strains (**Table 1**)
 - Resistance rates (by EUCAST criteria) for other agents ranged from 3.4% (tigecycline) to 96.6% (ceftriaxone) against XDR strains (**Table 2**)
- 10 isolates were PDR (Russia 3, Greece 2, Turkey 2, Italy 2, Poland 1); all 10 were resistant to ceftolozane/tazobactam (**Table 1**), piperacillin/tazobactam, cefepime and ceftazidime (data not shown)
- Ceftolozane/tazobactam demonstrated potent activity against 386 *E. coli* ($MIC_{50/90}$, 0.25/0.5 mg/L; 96.4% susceptible), including 91 ESBL-phenotype strains ($MIC_{50/90}$, 0.5/2 mg/L; 84.6% susceptible; **Table 1**) using EUCAST criteria
 - All non–ESBL-phenotype strains were inhibited at a ceftolozane/tazobactam MIC ≤1 mg/L (**Table 1**)
- Ceftolozane/tazobactam showed potent activity against non–ESBL-phenotype strains of *Klebsiella pneumoniae* ($MIC_{50/90}$, 0.25/0.5 mg/L; 99.4% susceptible; highest MIC, 2 mg/L) and retained activity against some ESBL-positive strains ($MIC_{50/90}$, 4/>32 mg/L; 36.5% susceptible; **Table 1**) using EUCAST criteria
 - In general, ceftolozane/tazobactam was inactive against meropenem–non-susceptible *K. pneumoniae* ($MIC_{50/90}$ >32/>32 mg/L; **Table 1**)
 - Tigecycline ($MIC_{50/90}$, 0.25/1 mg/L; 91.2% susceptible [EUCAST]) and colistin ($MIC_{50/90}$, ≤0.5/>8 mg/L; 78.0% susceptible [EUCAST]) exhibited the highest *in vitro* activity against ESBL-phenotype *K. pneumoniae* (**Table 2**)
- Ceftolozane/tazobactam also demonstrated good activity against *Klebsiella oxytoca* ($MIC_{50/90}$, 0.25/1 mg/L; 92.4% susceptible), *Serratia* spp. ($MIC_{50/90}$, 0.5/2 mg/L; 89.2% susceptible), *Proteus mirabilis* ($MIC_{50/90}$, 0.5/0.5 mg/L; 98.2% susceptible), *Citrobacter* spp. ($MIC_{50/90}$, 0.25/8 mg/L; 80.0% susceptible) and indole-positive *Proteae* ($MIC_{50/90}$, 0.5/1 mg/L; 95.9% susceptible; **Table 2**) using EUCAST criteria
- Ceftolozane/tazobactam showed good activity against *Enterobacter* spp. ($MIC_{50/90}$, 0.5/8 mg/L; 66.7% susceptible; **Table 3**) using EUCAST criteria
 - *Enterobacter cloacae* (n = 139) and *Enterobacter aerogenes* (n = 67) were 71.2% and 58.2% susceptible to ceftolozane/tazobactam, respectively (**Tables 1 and 3**)
 - Ceftolozane/tazobactam activity against presumptive AmpC phenotype (ceftazidime resistant and cefepime susceptible) was low (16.0%-18.8%) but higher than with piperacillin/tazobactam (0.0%-4.0%), with all isolates susceptible to meropenem (**Table 3**)
- Non-susceptibility (by EUCAST criteria) to ceftazidime was very high in Israel (48.4%), Poland (87.2%), Russia (55.7%) and Turkey (48.0%); ceftolozane/tazobactam showed lower activity against Enterobacteriaceae from these countries as well (**Table 4**)
- Susceptibility to meropenem was high overall (95.1%) and was >95% in most countries, but it was lower in Poland (46.2%), Israel (87.1%) and Russia (83.6%; **Table 4**)
- The overall MDR rate was 25.2% and it varied widely (in countries with >10 isolates), ranging from 5.6% in Switzerland to 87.2% in Poland; the overall XDR rate was 6.9% and it also varied widely, ranging from 0.0% in several countries to 56.4% in Poland (**Table 4**)

Table 1. Cumulative MIC distributions of ceftolozane/tazobactam tested against Enterobacteriaceae, including various resistant subsets

Organism / resistant subset (n)	Number of isolates (cumulative %) inhibited at ceftolozane/tazobactam MIC, mg/L										MIC_{50}	MIC_{90}
	≤0.12	0.25	0.5	1	2	4	8	16	32	>32		
All Enterobacteriaceae (1,258)	180 (14.3)	454 (50.4)	272 (72.0)	129 (82.3)†	51 (86.3)	37 (89.3)	26 (91.3)	22 (93.1)	21 (94.8)	66 (100.0)	0.25	8
MDR (317)	2 (0.6)	29 (9.8)	62 (29.3)	53 (46.1)	30 (55.5)	25 (63.4)	16 (68.5)	18 (74.1)	18 (79.8)	64 (100.0)	2	>32
XDR (87)	0 (0.0)	0 (0.0)	2 (2.3)	6 (9.2)	8 (18.4)	6 (25.3)	7 (33.3)	2 (35.6)	10 (47.1)	46 (100.0)	>32	>32
PDR (10)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	10 (100.0)	>32	>32
CAZ-S (890)	179 (20.1)	417 (67.0)	206 (90.1)	72 (98.2)	15 (99.9)	1 (100.0)					0.25	0.5
CAZ-NS (368)	1 (0.5)	37 (16.4)	66 (46.1)	57 (68.0)	36 (80.4)	36 (90.4)	26 (95.4)	22 (96.8)	21 (98.2)	66 (100.0)	1	4
Escherichia coli (386)	103 (26.7)	194 (76.9)	53 (90.7)	22 (96.4)	5 (97.7)	2 (98.2)	3 (99.0)	1 (99.2)	0 (99.2)	3 (100.0)	0.25	0.5
Non-ESBL-phenotype (295)	101 (34.2)	170 (91.9)	21 (99.0)	3 (100.0)							0.25	0.25
ESBL-phenotype (91)	2 (2.2)	24 (28.6)	32 (63.7)	19 (84.6)	5 (90.1)	2 (92.3)	3 (95.6)	1 (96.7)	0 (96.7)	3 (100.0)	0.5	2
Klebsiella pneumoniae (329)	30 (9.1)	108 (42.0)	51 (57.5)	31 (66.9)	14 (71.1)	12 (74.8)	3 (75.7)	9 (78.4)	14 (82.7)	57 (100.0)	0.5	>32
Non-ESBL phenotype (159)	30 (18.9)	93 (77.4)	27 (94.3)	8 (99.4)	1 (100.0)						0.25	0.5
ESBL phenotype (170)	0 (0.0)	15 (8.8)	24 (22.9)	23 (36.5)	13 (44.1)	12 (51.2)	3 (52.9)	9 (58.2)	14 (60.5)	57 (100.0)	4	>32
ESBL phenotype-MEM-S (116)	0 (0.0)	15 (12.9)	24 (33.6)	22 (52.6)	13 (63.8)	12 (74.1)	3 (76.7)	6 (81.9)	7 (87.9)	14 (100.0)	1	>32
MEM-S (275)	30 (10.9)	108 (50.2)	51 (68.7)	30 (79.6)	14 (84.7)	12 (89.1)	3 (90.2)	6 (92.4)	7 (94.9)	14 (100.0)	0.25	8
MEM-NS (52)	0 (0.0)	0 (0.0)	0 (0.0)	1 (1.9)	0 (1.9)	0 (1.9)	0 (1.9)	1 (3.9)	7 (17.3)	43 (100.0)	>32	>32
Klebsiella oxytoca (166)	27 (40.9)	19 (69.7)	13 (89.4)	2 (92.4)	2 (95.5)	1 (97.0)	1 (98.5)	1 (100.0)			0.25	1
Enterobacter spp. (210)	9 (4.3)	74 (39.5)	32 (54.8)	25 (66.7)	18 (75.2)	19 (84.3)	15 (91.4)	10 (96.2)	5 (98.6)	3 (100.0)	0.5	8
Enterobacter cloacae (139)	4 (2.9)	51 (39.6)	25 (57.6)	19 (71.2)	6 (75.5)	10 (82.7)	13 (92.1)	6 (96.4)	3 (98.6)	2 (100.0)	0.5	8
Enterobacter aerogenes (67)	5 (7.5)	22 (40.3)	6 (49.3)	6 (58.2)	11 (74.6)	9 (88.1)	2 (91.0)	3 (95.5)	2 (98.5)	1 (100.0)	1	8
Serratia spp. (102)	0 (0.0)	2 (2.0)	55 (55.9)	34 (89.2)	9 (98.0)	1 (99.0)	0 (99.0)	0 (99.0)	0 (99.0)	1 (100.0)	0.5	2
Proteus mirabilis (57)	0 (0.0)	13 (22.8)	39 (91.2)	4 (98.3)	1 (100.0)						0.5	0.5
Citrobacter spp. (55)	10 (18.2)	28 (69.1)	5 (78.2)	1 (80.0)	2 (83.6)	2 (87.3)	4 (94.6)	1 (96.4)	2 (100.0)		0.25	8
Indole-positive *Proteae* (49)	0 (0.0)	16 (32.7)	22 (77.6)	9 (95.9)	0 (95.9)	0 (95.9)	0 (95.9)	0 (95.9)	0 (95.9)	2 (100.0)	0.5	1

CAZ = ceftazidime; ESBL = extended-spectrum β-lactamase; EUCAST = European Committee on Antimicrobial Susceptibility Testing; MDR = multidrug-resistant; MEM = meropenem; MIC = minimum inhibitory concentration; MIC_{50} = minimum inhibitory concentration required to inhibit growth of 50% of isolates; MIC_{90} = minimum inhibitory concentration required to inhibit growth of 90% of isolates; NS = non-susceptible; PDR = pandrug-resistant; R = resistant; S = susceptible; XDR = extensively drug-resistant.
†Underlined results based on the EUCAST susceptibility breakpoints for ceftolozane/tazobactam.[8]

Table 2. Activity of ceftolozane/tazobactam and comparator antimicrobial agents when tested against Enterobacteriaceae from ICU patients in European, Turkish and Israeli hospitals (2013-2014)

Organism (n) / antimicrobial agent	MIC, mg/L		%S / %I / %R†	
	MIC_{50}	MIC_{90}	CLSI	EUCAST
Enterobacteriaceae (1,258)				
Ceftolozane/tazobactam	0.25	8	86.3 / 2.9 / 10.8	82.3 / –‡ / 17.7
Ceftazidime	0.25	>32	74.2 / 2.7 / 23.1	70.8 / 3.4 / 25.8
Cefepime	≤0.5	>16	79.2 / 3.8 / 17.0	77.1 / 3.6 / 19.3
Ceftriaxone	0.12	>8	68.4 / 2.1 / 29.5	68.4 / 2.1 / 29.5
Meropenem	≤0.06	0.12	95.1 / 0.2 / 4.8	95.2 / 2.1 / 2.7
Piperacillin/tazobactam	2	>64	79.4 / 7.7 / 13.0	73.8 / 5.6 / 20.6
Levofloxacin	≤0.12	>4	79.2 / 2.4 / 18.4	77.5 / 1.8 / 20.8
Gentamicin	≤1	>8	85.7 / 0.5 / 13.9	84.2 / 1.5 / 14.3
Tigecycline§	0.25	1	98.8 / 1.2 / 0.0	94.6 / 4.2 / 1.2
Colistin	≤0.5	>8	– / – / –	78.6 / – / 21.4
MDR (317)				
Ceftolozane/tazobactam	2	>32	55.5 / 7.9 / 36.6	46.1 / – / 53.9
Ceftazidime	32	>32	25.2 / 5.7 / 69.1	20.5 / 4.7 / 74.8
Cefepime	>16	>16	31.2 / 10.4 / 58.4	26.8 / 7.9 / 65.3
Ceftriaxone	>8	>8	14.2 / 3.5 / 82.3	14.2 / 3.5 / 82.3
Meropenem	≤0.06	>8	80.3 / 0.6 / 19.0	81.0 / 8.3 / 10.8
Piperacillin/tazobactam	32	>64	40.4 / 19.6 / 40.1	28.4 / 12.0 / 59.6
Levofloxacin	>4	>4	37.9 / 7.6 / 54.6	32.8 / 5.0 / 62.1
Gentamicin	4	>8	50.3 / 1.9 / 47.8	45.6 / 4.7 / 49.7
Tigecycline§	0.5	2	96.8 / 3.2 / 0.0	86.1 / 10.7 / 3.2
Colistin	≤0.5	>8	– / – / –	68.7 / – / 31.3
XDR (87)				
Ceftolozane/tazobactam	>32	>32	18.4 / 6.9 / 74.7	9.2 / – / 90.8
Ceftazidime	>32	>32	8.0 / 2.3 / 89.7	4.6 / 3.4 / 92.0
Cefepime	>16	>16	11.5 / 9.2 / 79.3	6.9 / 8.0 / 85.1
Ceftriaxone	>8	>8	1.1 / 2.3 / 96.6	1.1 / 2.3 / 96.6
Meropenem	4	>8	40.7 / 1.2 / 58.1	41.9 / 22.1 / 36.0
Piperacillin/tazobactam	>64	>64	8.0 / 17.2 / 74.7	5.7 / 2.3 / 92.0
Levofloxacin	>4	>4	9.2 / 6.9 / 83.9	5.7 / 3.4 / 90.8
Gentamicin	>8	>8	29.9 / 3.4 / 66.7	18.4 / 11.5 / 70.1
Tigecycline§	1	2	96.6 / 3.4 / 0.0	72.4 / 24.1 / 3.4
Colistin	4	>8	– / – / –	49.4 / – / 50.6
***Escherichia coli* (386)**				
Ceftolozane/tazobactam	0.25	0.5	97.7 / 0.5 / 1.8	96.4 / – / 3.6
Ceftazidime	0.25	16	82.6 / 1.8 / 15.5	78.8 / 3.9 / 17.4
Cefepime	≤0.5	>16	81.6 / 2.3 / 16.1	80.6 / 2.1 / 17.4
Ceftriaxone	≤0.06	>8	78.0 / 0.0 / 22.0	78.0 / 0.0 / 22.0
Meropenem	≤0.06	≤0.06	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Piperacillin/tazobactam	2	32	89.3 / 4.2 / 6.5	83.6 / 5.7 / 10.7
Levofloxacin	≤0.12	>4	71.9 / 1.8 / 26.2	71.4 / 0.5 / 28.1
Gentamicin	≤1	>8	86.8 / 0.0 / 13.2	85.5 / 1.3 / 13.2
Tigecycline§	0.12	0.25	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Colistin	≤0.5	≤0.5	– / – / –	98.4 / – / 1.6
ESBL phenotype (91)				
Ceftolozane/tazobactam	0.5	2	90.1 / 2.2 / 7.7	84.6 / – / 15.4
Ceftazidime	16	>32	26.4 / 7.7 / 65.9	9.9 / 16.5 / 73.6
Cefepime	>16	>16	22.0 / 9.9 / 68.1	18.7 / 7.7 / 73.6
Ceftriaxone	>8	>8	6.6 / 0.0 / 93.4	6.6 / 0.0 / 93.4
Meropenem	≤0.06	≤0.06	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Piperacillin/tazobactam	8	>64	73.6 / 11.0 / 15.4	60.4 / 13.2 / 26.4
Levofloxacin	>4	>4	26.4 / 5.5 / 68.1	26.4 / 0.0 / 73.6
Gentamicin	≤1	>8	63.3 / 0.0 / 36.7	62.2 / 1.1 / 36.7
Tigecycline§	0.12	0.25	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Colistin	≤0.5	≤0.5	– / – / –	97.8 / – / 2.2
***Klebsiella pneumoniae* (329)**				
Ceftolozane/tazobactam	0.5	>32	71.1 / 3.6 / 25.2	66.9 / – / 33.1
Ceftazidime	1	>32	53.2 / 5.5 / 41.3	50.2 / 3.0 / 46.8
Cefepime	≤0.5	>16	55.6 / 4.3 / 40.1	52.9 / 3.3 / 43.8
Ceftriaxone	1	>8	50.5 / 1.2 / 48.3	50.5 / 1.2 / 48.3
Meropenem	≤0.06	8	83.5 / 0.6 / 15.9	84.1 / 6.1 / 9.8
Piperacillin/tazobactam	8	>64	63.9 / 7.0 / 29.1	57.2 / 6.7 / 36.1
Levofloxacin	0.5	>4	66.5 / 3.7 / 29.9	64.6 / 1.8 / 33.5
Gentamicin	≤1	>8	71.3 / 0.3 / 28.0	69.6 / 2.1 / 28.3
Tigecycline§	0.25	1	99.1 / 0.9 / 0.0	93.3 / 5.8 / 0.9
Colistin	≤0.5	8	– / – / –	87.5 / – / 12.5
ESBL phenotype (170)				
Ceftolozane/tazobactam	4	>32	44.1 / 7.1 / 48.8	36.5 / – / 63.5
Ceftazidime	>32	>32	9.4 / 10.6 / 80.0	3.5 / 5.9 / 90.6
Cefepime	>16	>16	14.1 / 8.2 / 77.6	8.8 / 6.5 / 84.7
Ceftriaxone	>8	>8	4.1 / 2.4 / 93.5	4.1 / 2.4 / 93.5
Meropenem	≤0.06	>8	67.9 / 1.2 / 31.0	69.0 / 11.9 / 19.0
Piperacillin/tazobactam	>64	>64	32.0 / 13.0 / 55.0	21.9 / 10.1 / 68.0
Levofloxacin	>4	>4	39.4 / 5.9 / 54.7	36.5 / 2.9 / 60.6
Gentamicin	>8	>8	45.3 / 0.6 / 54.1	41.2 / 4.1 / 54.7
Tigecycline§	0.25	1	99.4 / 0.6 / 0.0	91.2 / 8.2 / 0.6
Colistin	≤0.5	>8	– / – / –	78.0 / – / 22.0
***Klebsiella oxytoca* (66)**				
Ceftolozane/tazobactam	0.25	1	95.5 / 1.5 / 3.0	92.4 / – / 7.6
Ceftazidime	0.12	0.5	95.5 / 0.0 / 4.5	94.0 / 1.5 / 4.5
Cefepime	≤0.5	1	97.0 / 3.0 / 0.0	93.9 / 4.5 / 1.5
Ceftriaxone	≤0.06	4	84.8 / 4.5 / 10.6	84.8 / 4.5 / 10.6
Meropenem	≤0.06	≤0.06	100.0 / 0 / 0.0	100.0 / 0 / 0.0
Piperacillin/tazobactam	2	>64	84.6 / 3.1 / 12.3	83.1 / 1.5 / 15.4
Levofloxacin	≤0.12	0.5	95.5 / 3.0 / 1.5	92.4 / 3.0 / 4.5
Gentamicin	≤1	≤1	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Tigecycline§	0.12	0.5	100.0 / 0.0 / 0.0	98.5 / 1.5 / 0.0
Colistin	≤0.5	≤0.5	– / – / –	98.5 / – / 1.5
***Enterobacter* spp.‖ (210)**				
Ceftolozane/tazobactam	0.5	8	75.2 / 9.0 / 15.7	66.7 / – / 33.3
Ceftazidime	0.5	>32	61.4 / 2.9 / 35.7	58.1 / 3.3 / 38.6
Cefepime	≤0.5	8	84.3 / 9.0 / 6.7	80.5 / 8.6 / 11.0
Ceftriaxone	0.5	>8	56.5 / 1.4 / 42.1	56.5 / 1.4 / 42.1
Meropenem	≤0.06	0.12	97.1 / 0.0 / 2.9	97.1 / 2.4 / 0.5
Piperacillin/tazobactam	4	>64	67.0 / 21.5 / 11.5	60.8 / 6.2 / 33.0
Levofloxacin	≤0.12	1	92.4 / 0.5 / 7.1	90.0 / 2.4 / 7.6
Gentamicin	≤1	2	92.4 / 1.0 / 6.7	90.0 / 2.4 / 7.6
Tigecycline§	0.25	0.5	99.5 / 0.5 / 0.0	96.7 / 2.9 / 0.5
Colistin	≤0.5	2	– / – / –	92.8 / – / 7.2
***Serratia* spp.¶ (102)**				
Ceftolozane/tazobactam	0.5	2	98.0 / 1.0 / 1.0	89.2 / – / 10.8
Ceftazidime	0.25	0.5	99.0 / 1.0 / 0.0	98.0 / 1.0 / 1.0
Cefepime	≤0.5	≤0.5	99.0 / 1.0 / 0.0	98.0 / 2.0 / 0.0
Ceftriaxone	0.25	2	82.4 / 7.8 / 9.8	82.4 / 7.8 / 9.8
Meropenem	≤0.06	≤0.06	98.0 / 0.0 / 2.0	98.0 / 1.0 / 1.0
Piperacillin/tazobactam	2	16	92.2 / 4.9 / 2.9	87.3 / 4.9 / 7.8
Levofloxacin	≤0.12	0.5	96.1 / 1.0 / 2.9	96.1 / 0.0 / 3.9
Gentamicin	≤1	≤1	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Tigecycline§	0.5	1	99.0 / 1.0 / 0.0	99.0 / 0.0 / 1.0
Colistin	>8	>8	– / – / –	3.9 / – / 96.1
***Proteus mirabilis* (57)**				
Ceftolozane/tazobactam	0.5	0.5	100.0 / 0.0 / 0.0	98.2 / – / 1.8
Ceftazidime	0.06	0.12	100.0 / 0.0 / 0.0	98.2 / 1.8 / 0.0
Cefepime	≤0.5	≤0.5	96.5 / 0.0 / 3.5	96.5 / 0.0 / 3.5
Ceftriaxone	≤0.06	≤0.06	94.7 / 0.0 / 5.3	94.7 / 0.0 / 5.3
Meropenem	≤0.06	0.12	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Piperacillin/tazobactam	≤0.5	1	100.0 / 0.0 / 0.0	98.2 / 1.8 / 0.0
Levofloxacin	≤0.12	>4	87.7 / 1.8 / 10.5	78.9 / 8.8 / 12.3
Gentamicin	≤1	>8	86.0 / 1.8 / 12.3	84.2 / 1.8 / 14.0
Tigecycline§	2	4	86.0 / 14.0 / 0.0	47.4 / 38.6 / 14.0
Colistin	>8	>8	– / – / –	0.0 / – / 100.0
***Citrobacter* spp.†† (55)**				
Ceftolozane/tazobactam	0.25	8	83.6 / 3.7 / 12.7	80.0 / – / 20.0
Ceftazidime	0.25	>32	80.0 / 0.0 / 20.0	78.2 / 1.8 / 20.0
Cefepime	≤0.5	1	94.5 / 3.6 / 1.8	90.9 / 5.5 / 3.6
Ceftriaxone	0.12	>8	75.9 / 1.9 / 22.2	75.9 / 1.9 / 22.2
Meropenem	≤0.06	≤0.06	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Piperacillin/tazobactam	2	64	85.5 / 5.5 / 9.1	74.5 / 10.9 / 14.5
Levofloxacin	≤0.12	1	92.7 / 3.6 / 3.6	90.9 / 1.8 / 7.3
Gentamicin	≤1	≤1	94.4 / 0.0 / 5.6	94.4 / 0.0 / 5.6
Tigecycline§	0.12	0.25	100.0 / 0.0 / 0.0	100.0 / 0.0 / 0.0
Colistin	≤0.5	1	– / – / –	100.0 / – / 0.0
Indole-positive *Proteae*‡‡ (49)				
Ceftolozane/tazobactam	0.5	1	95.9 / 0. / 4.1	95.9 / – / 4.1
Ceftazidime	0.12	16	83.7 / 4.1 / 12.2	69.4 / 14.3 / 16.3
Cefepime	≤0.5	≤0.5	91.8 / 2.0 / 6.1	91.8 / 0.0 / 8.2
Ceftriaxone	0.25	4	71.4 / 14.3 / 14.3	71.4 / 14.3 / 14.3
Meropenem	≤0.06	0.25	100.0 / 0 / 0.0	100.0 / 0.0 / 0.0
Piperacillin/tazobactam	≤0.5	4	93.9 / 4.1 / 2.0	93.9 / 0.0 / 6.1
Levofloxacin	≤0.12	>4	83.7 / 6.1 / 10.2	81.6 / 2.0 / 16.3
Gentamicin	≤1	>8	81.6 / 4.1 / 14.3	79.6 / 2.0 / 18.4
Tigecycline§	0.5	2	98.0 / 2.0 / 0.0	87.8 / 10.2 / 2.0
Colistin	>8	>8	– / – / –	0.0 / – / 100.0

CLSI = Clinical and Laboratory Standards Institute; ESBL = extended-spectrum β-lactamase; EUCAST = European Committee on Antimicrobial Susceptibility Testing; MIC = minimum inhibitory concentration; MIC_{50} = minimum inhibitory concentration required to inhibit 50% of isolates; MIC_{90} = minimum inhibitory concentration required to inhibit 90% of isolates; R = resistant; I = intermediate; S = susceptible.
†Criteria as published by the CLSI (2016)[7] and EUCAST (2016).[8]
‡"–" no breakpoint available for interpretation.
§In the absence of a CLSI breakpoint, US Food and Drug Administration breakpoints were applied when available.[10]
‖Includes *E. aerogenes* (67), *E. amnigenus* (1), *E. asburiae* (2), *E. cloacae* (126), *E. cloacae* species complex (13), *E. kobei* (1).
¶Includes *S. liquefaciens* (2), *S. marcescens* (98), *S. odorifera* (1), *S. plymuthica* (1).
††Includes *C. braakii* (3), *C. freundii* (18), *C. freundii* species complex (3), *C. koseri* (31).
‡‡Includes *Morganella morganii* (36), *Proteus vulgaris* (10), *Providencia rettgeri* (1), *Providencia stuartii* (2).

Table 3. Antimicrobial activity of ceftolozane/tazobactam, piperacillin/tazobactam, ceftazidime and meropenem against Enterobacteriaceae isolates stratified by phenotype (2013-2014)

Organism / phenotype (n)	TOL/TAZ	PIP/TAZ	CAZ	MEM
	$MIC_{50/90}$ (%S)†	$MIC_{50/90}$ (%S)†	$MIC_{50/90}$ (%S)†	$MIC_{50/90}$ (%S)†
Escherichia coli (386)	0.25 / 0.5 (96.4)	2 / 32 (83.6)	0.25 / 16 (78.8)	≤0.06 / ≤0.06 (100.0)
ESBL phenotype (91)	0.5 / 2 (84.6)	8 / >64 (60.4)	16 / >32 (9.9)	≤0.06 / ≤0.06 (100.0)
Klebsiella pneumoniae (329)	0.5 / >32 (66.9)	8 / >64 (57.2)	1 / >32 (50.2)	≤0.06 / 8 (84.1)
ESBL phenotype (170)	4 / >32 (36.5)	>64 / >64 (21.9)	>32 / >32 (3.5)	0.06 / >8 (69.1)
ESBL phenotype-MEM-S (116)	1 / >32 (52.6)	32 / >64 (32.2)	16 / >32 (4.3)	≤0.06 / 0.5 (100.0)
Enterobacter spp. (210)	0.5 / 8 (66.7)	4 / >64 (60.8)	0.5 / >32 (58.1)	≤0.06 / 0.12 (97.1)
E. cloacae (139)	0.5 / 8 (71.2)	4 / >64 (64.5)	0.5 / >32 (61.9)	≤0.06 / 0.12 (97.8)
CAZ-R and FEP-S‡ (16)	4 / 8 (18.8)	32 / 64 (0.0)	–§	≤0.06 / 0.12 (100.0)
E. aerogenes (67)	1 / 8 (58.2)	8 / 64 (53.7)	1 / >32 (50.8)	≤0.06 / 0.12 (95.5)
CAZ-R and FEP-S‡ (25)	2 / 4 (16.0)	32 / 64 (4.0)	–§	≤0.06 / 0.12 (100.0)

CAZ = ceftazidime; ESBL = extended-spectrum β-lactamase; EUCAST = European Committee on Antimicrobial Susceptibility Testing; FEP = cefepime; MEM = meropenem; $MIC_{50/90}$ = minimum inhibitory concentration required to inhibit growth of 50% and 90% of isolates; PIP/TAZ = piperacillin/tazobactam; S = susceptible; TOL/TAZ = ceftolozane/tazobactam.
MIC is expressed as mg/L.
†Susceptible breakpoints established by EUCAST (2016).[8]
‡Presumptive AmpC phenotype.
§Values not displayed because all isolates are resistant to ceftazidime.

Table 4. Antimicrobial activity of ceftolozane/tazobactam, ceftazidime and meropenem against Enterobacteriaceae strains stratified by country and MDR status (2013-2014)

Country, (n)	Ceftolozane/Tazobactam	Ceftazidime	Meropenem	%MDR / %XDR / %PDR‡
	$MIC_{50/90}$ (%S)†	$MIC_{50/90}$ (%S)†	$MIC_{50/90}$ (%S)†	
Belgium (66)	0.25 / 4 (80.3)	0.25 / 32 (74.2)	≤0.06 / 0.12 (97.0)	19.7 / 1.5 / 0.0
Czech Republic (27)	0.25 / 4 (85.2)	0.25 / >32 (74.1)	≤0.06 / 0.06 (100.0)	25.9 / 11.1 / 0.0
Denmark (6)	0.25 / – (–)	0.25 / – (–)	≤0.06 / – (–)	33.3 / 0.0 / 0.0
Finland (1)	– / –§	– / –	– / –	0.0 / 0.0 / 0.0
France (213)	0.25 / 1 (90.1)	0.25 / 32 (74.2)	≤0.06 / 0.06 (99.1)	20.2 / 3.8 / 0.0
Germany (146)	0.5 / 4 (84.9)	0.25 / 32 (72.6)	≤0.06 / ≤0.06 (98.6)	17.8 / 3.4 / 0.0
Greece (69)	0.5 / >32 (78.3)	0.25 / >32 (65.2)	≤0.06 / 1 (91.3)	31.9 / 8.7 / 2.9
Ireland (91)	0.5 / 2 (81.3)	0.25 / 32 (79.1)	≤0.06 / 0.06 (100.0)	20.9 / 2.2 / 0.0
Israel (31)	0.5 / >32 (71.0)	1 / >32 (51.6)	≤0.06 / >8 (87.1)	41.9 / 22.6 / 0.0
Italy (113)	0.5 / 4 (85.0)	0.25 / >32 (71.7)	≤0.06 / 0.06 (94.7)	24.8 / 8.0 / 1.8
Netherlands (10)	0.5 / 0.5 (–)	0.12 / 16 (–)	≤0.06 / 0.12 (–)	30.0 / 0.0 / 0.0
Norway (5)	0.25 / – (–)	0.25 / – (–)	0.03 / – (–)	20.0 / 0.0 / 0.0
Poland (39)	32 / >32 (18.0)	>32 / >32 (12.8)	4 / >8 (46.2)	87.2 / 56.4 / 2.6
Portugal (15)	0.25 / 0.5 (93.3)	0.12 / 0.25 (93.3)	≤0.06 / 0.06 (100.0)	0.0 / 0.0 / 0.0
Russia (61)	1 / >32 (55.7)	16 / >32 (44.3)	≤0.06 / >8 (83.6)	57.4 / 16.4 / 4.9
Spain (188)	0.25 / 4 (89.9)	0.25 / 16 (79.8)	≤0.06 / ≤0.06 (99.5)	14.9 / 2.1 / 0.0
Sweden (5)	0.25 / – (–)	0.25 / – (–)	≤0.06 / – (–)	0.0 / 0.0 / 0.0
Switzerland (18)	0.25 / 2 (88.9)	0.25 / 16 (88.9)	≤0.06 / 0.12 (94.4)	5.6 / 0.0 / 0.0
Turkey (75)	0.5 / >32 (76.0)	1 / >32 (52.0)	≤0.06 / 1 (92.0)	46.7 / 10.7 / 2.7
Ukraine (3)	>32 / – (–)	>32 / – (–)	0.25 / – (–)	66.7 / 33.3 / 0.0
United Kingdom (76)	0.25 / 0.5 (97.4)	0.12 / 4 (86.8)	≤0.06 / ≤0.06 (100.0)	6.7 / 1.3 / 0.0
Overall (1,258)	0.25 / 8 (82.3)	0.25 / >32 (70.8)	≤0.06 / 0.12 (95.1)	25.2 / 6.9 / 0.8

EUCAST = European Committee on Antimicrobial Susceptibility Testing; MDR = multidrug-resistant; $MIC_{50/90}$ = minimum inhibitory concentration required to inhibit growth of 50% and 90% of isolates; PDR = pandrug-resistant; S = susceptible; XDR = extensively drug-resistant.
MIC is expressed as mg/L.
†Susceptible breakpoint established by EUCAST (2016),[8] and % susceptibility is provided only for countries with more than 10 isolates.
‡MDR, XDR and PDR bacteria were classified according to Magiorakos et al.[9]
§"–" = data not available because of the limited number of isolates.

Conclusions

- Antimicrobial susceptibility and MDR and XDR rates for Enterobacteriaceae varied widely in Europe, Turkey and Israel; MDR, XDR and resistance rates to ceftazidime were generally elevated and particularly high in some countries
- At the EUCAST susceptibility breakpoint, ceftolozane/tazobactam provided greater coverage than β-lactams (except for carbapenems) against MDR and XDR Enterobacteriaceae (including *E. coli*, *K. pneumoniae*, *E. cloacae* and *E. aerogenes*) isolated from ICU patients in Europe, Turkey and Israel during 2013 and 2014

References

1. Zerbaxa [prescribing information]. Whitehouse Station, NJ: Merck & Co., Inc.; 2015.
2. Sader HS, Rhomberg PR, Farrell DJ, Jones RN. Antimicrobial activity of CXA-101, a novel cephalosporin tested in combination with tazobactam against Enterobacteriaceae, *Pseudomonas aeruginosa*, and *Bacteroides fragilis* strains having various resistance phenotypes. *Antimicrob Agents Chemother.* 2011;55:2390-2394.
3. Farrell DJ, Flamm RK, Sader HS, Jones RN. Antimicrobial activity of ceftolozane/tazobactam tested against Enterobacteriaceae and *Pseudomonas aeruginosa* with various resistance patterns isolated in U.S. hospitals (2011-2012). *Antimicrob Agents Chemother.* 2013;57:6305-6310.
4. Sader HS, Farrell DJ, Flamm RK, Jones RN. Ceftolozane/tazobactam activity tested against aerobic Gram-negative organisms isolated from intra-abdominal and urinary tract infections in European and United States hospitals (2012). *J Infect.* 2014;69:266-277.
5. Livermore DM, Mushtaq S, Ge Y, Warner M. Activity of cephalosporin CXA-101 (FR264205) against *Pseudomonas aeruginosa* and *Burkholderia cepacia* group strains and isolates. *Int J Antimicrob Agents.* 2009;34:402-406.
6. Clinical and Laboratory Standards Institute. M07-A10: Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: tenth edition. Wayne, PA: CLSI; 2015.
7. Clinical and Laboratory Standards Institute. M100-S26. Performance standards for antimicrobial susceptibility testing: 26th informational supplement. Wayne, PA: CLSI; 2016.
8. European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters. Version 6.0, January 2016. http://www.eucast.org/clinical_breakpoints/. Accessed March 18, 2016.
9. Magiorakos AP, Srinivasan A, Carey RB, et al. Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clin Microbiol Infect.* 2012;18:268-281.
10. Tygacil [prescribing information]. Philadelphia, PA: Wyeth Pharmaceuticals Inc.; January 2016.

Acknowledgements

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ, USA. Medical writing and editorial assistance was provided by Tracy Cao, PhD, of ApotheCom, Yardley, PA, USA. This assistance was funded by Merck & Co., Inc., Kenilworth, NJ, USA.

http://bit.ly/1RhwIsm

Copyright © 2016 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.