P0341

In Vitro Activity of Plazomicin tested against Contemporary Clinical Isolates Collected in Asia-Pacific, Europe and Latin America

M CASTANHEIRA, TB DOYLE, DJ FARRELL, RN JONES
JMI Laboratories, North Liberty, IA, USA

ECCMID 2016
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370, fax 319.665.3371
mariana-castanheira@jmilabs.com

ABSTRACT

Background: Plazomicin is a next generation aminoglycoside that is stable against common aminoglycoside modifying enzymes and displays activity against Enterobacteriaceae, *S. aureus* (SA), including methicillin-resistant isolates, and some *P. aeruginosa* (PSA). Like other aminoglycosides, this agent is not active against isolates producing 16S rRNA methylases (RNAmet). We evaluated the activity of plazomicin and comparators tested against 3,660 clinical isolates collected in hospitals from the Asia-Pacific (APAC), Europe and Latin America (LATAM) during 2014.

Materials/methods: 3,224 Enterobacteriaceae, 236 Gram-positive cocci, 100 PSA and 100 *Acinetobacter* spp. were collected in hospitals in APAC (n=789), Europe (n=2315) and LATAM (n=556). Isolates were susceptibility (S) tested using the reference broth microdilution method. CLSI and EUCAST interpretative criteria were applied. Enterobacteriaceae displaying plazomicin MICs ≥128 mg/L were screened for the presence of RNAmet-encoding genes using PCR and sequencing.

Results: Overall, plazomicin ($MIC_{50/90}$, 0.5/2 mg/L) inhibited 89.4 and 96.2% of Enterobacteriaceae at ≤1 and ≤2 mg/L, respectively; the number of isolates inhibited at these values were 91.8 and 96.8% in APAC, 88.7 and 96.1% in Europe and 88.8 and 95.6% in LATAM. Plazomicin displayed good activity against *E. coli* ($MIC_{50/90}$, 0.5/1 mg/L), *K. pneumoniae* (KPN; $MIC_{50/90}$, 0.25/0.5 mg/L) and *E. cloacae* ($MIC_{50/90}$, 0.5/0.5 mg/L). Among ESBL-phenotype isolates, 98.8% of the *E. coli* (one isolate displayed MIC at 16 and two at >128 mg/L) and 92.9% of KPN were inhibited at ≤2 mg/L of plazomicin. Additionally, 86.7% of the carbapenem-resistant Enterobacteriaceae (CRE) were inhibited by plazomicin at ≤2 mg/L. Among 55 (1.7%) Enterobacteriaceae with plazomicin MIC results ≥8 mg/L, 33 were KPN (3.0% for this species) and, of these, 30 had plazomicin MICs ≥128 mg/L (17 [2.4%] from Europe, nine [3.9%] from APAC, and four [2.6%] from LATAM). Plazomicin MICs for Indole-positive *Proteus* spp. and *P. mirabilis* were slightly higher ($MIC_{50/90}$, 1/4 and 2/4 mg/L, respectively) when compared to other Enterobacteriaceae species. All 36 (1.1%) Enterobacteriaceae isolates displaying plazomicin MICs of ≥128 mg/L carried RNAmet - encoding genes: 19 *rmtB*, eight *rmtF*, seven *armA* and one each of *rmtA*, *rmtC* and *rmtD1*. Plazomicin ($MIC_{50/90}$, 4/8 mg/L) inhibited 67.0% of PSA at ≤4 mg/L (68.0% in APAC, 70.0% in Europe and 60.0% in LATAM). All coagulase-negative staphylococci ($MIC_{50/90}$, 0.12/0.25 mg/L) were inhibited by plazomicin at ≤0.25 mg/L, and 98.4 and 100.0% of *S. aureus* ($MIC_{50/90}$, 0.5/1 mg/L) were inhibited at ≤1 and ≤2 mg/L, respectively. Plazomicin activity was limited against *Acinetobacter* spp. ($MIC_{50/90}$, 32/>128 mg/L), *Enterococcus* spp. and *S. pneumoniae* ($MIC_{50/90}$, 64/64 mg/L for both).

Conclusions: Plazomicin displayed good activity against contemporary Enterobacteriaceae isolates, including CRE isolates. In all cases where plazomicin MICs were ≥128 mg/L, the isolates produced RNAmet. This compound was also potent against staphylococci, but its activity was compromised for PSA, *Acinetobacter* spp. and streptococci.

INTRODUCTION

Patients with prolonged hospitalization, including those in intensive care or long-term care facilities, immunodeficient patients and others with malignant conditions often develop infections and many of these are caused by multidrug-resistant (MDR) organisms. Among species commonly highlighted as MDR are Enterobacteriaceae, including carbapenem-resistant isolates (CRE), pan- and extremely-drug resistant *Pseudomonas aeruginosa, Acinetobacter baumannii* and Gram-positive species, including *Enterococcus faecium* and *Staphylococcus aureus.* The urgent need for monitoring initiatives and new therapeutic options for these organisms has been recognized by the medical and scientific communities and although various new antimicrobial agents for Gram-positive infections have been approved, the number of candidates for treating Gram-negative infections or broader-spectrum agents even in late stages of development is still limited.

Plazomicin is a next-generation aminoglycoside synthetically derived from sisomicin and designed to have stability against most aminoglycoside modifying enzymes. This new generation aminoglycoside has activity against Enterobacteriaceae and some *P. aeruginosa* and *Staphylococcus* spp., including methicillin-resistant (MRSA) isolates. As with all other aminoglycosides, plazomicin activity is affected by the presence of 16S rRNA methylases.

In this study, we evaluated the activity of plazomicin against a collection of 3,660 clinical isolates collected in Asia-Pacific, Europe and Latin America during 2014, including 3,224 Enterobacteriaceae, 236 Gram-positive cocci, 100 *P. aeruginosa* and 100 *Acinetobacter* spp. Additionally, the presence of 16S rRNA methylases was evaluated among Enterobacteriaceae isolates displaying plazomicin MIC results of ≥128 mg/L.

MATERIALS AND METHODS

Bacterial isolates. A total of 3,660 clinical isolates, including 3,224 Enterobacteriaceae, 236 Gram-positive cocci, 100 *P. aeruginosa* and 100 *Acinetobacter* spp., collected during 2014 in medical centers located in 37 nations in Europe (n=44; 2,315; 63.2%), Latin America (n=10; 556; 15.2%) and Asia-Pacific (n=15; 789 isolates; 21.6%) were evaluated. Only clinically significant isolates were included in the study (one per patient episode). Species identification was confirmed when necessary by Matrix-Assisted Laser Desorption Ionization-Time Of Flight mass spectrometry (MALDI-TOF MS) using the Bruker Daltonics MALDI Biotyper (Billerica, Massachusetts, USA), following manufacturer instructions.

Antimicrobial susceptibility testing. All isolates were susceptibility tested against plazomicin and comparator agents using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI). Categorical interpretations for all comparator agents were those found in CLSI criteria in M100-S26 (2016), EUCAST breakpoint tables (version 6.0, January 2016) and/or United States Food and Drug Administration (US-FDA) package inserts. Quality control (QC) was performed using *Escherichia coli* ATCC 25922, *Staphylococcus aureus* ATCC 29213, *Pseudomonas aeruginosa* ATCC 27853, *Enterococcus faecalis* ATCC 29212 and *Streptococcus pneumoniae* ATCC 49619. All QC MIC results were within acceptable ranges as published in CLSI documents.

Definitions. ESBL-phenotype criteria was applied for *E. coli*, *Klebsiella* spp. (including *K. pneumoniae and K. oxytoca*) and *P. mirabilis* displaying a MIC value ≥2 mg/L for ceftriaxone or ceftazidime or aztreonam (CLSI, 2016). Carbapenem-resistant Enterobacteriaceae (CRE) was defined as any isolate exhibiting an imipenem and/or meropenem MIC value ≥2 mg/L (*Proteus mirabilis* and indole-positive *Proteae* were not included due to the intrinsically elevated MIC values).

Screening for 16S rRNA methylases. All Enterobacteriaceae isolates displaying plazomicin MIC results of ≥128 mg/L were screened for the presence of 16S rRNA methylase-encoding genes, including *armA*, *rmtA* through *rmtH,* and *npmA*, by PCR using custom primers. Amplicons were sequenced on both strands and nucleotide sequences obtained were analyzed using the Lasergene® software package (DNAStar; Madison, Wisconsin, USA) and compared to available sequences via NCBI BLAST search (http://www.ncbi.nlm.nih.gov/blast/).

RESULTS

- Plazomicin (MIC_{50} and MIC_{90}, 0.5 and 2 mg/L) displayed good activity against 3,224 Enterobacteriaceae isolates, and 89.4 and 96.2% of these isolates were inhibited by plazomicin at ≤1 and ≤2 mg/L, respectively (**Table 1**).
- The activity of plazomicin against Enterobacteriaceae was similar in all three regions analyzed and MIC_{50} and MIC_{90} results were 0.5 and 1 mg/L for isolates collected in Asia-Pacific, and 0.5 and 2 mg/L for both Europe and Latin America, respectively (**Table 2**).
- Plazomicin MIC_{90} values ranged from 0.5 to 1 mg/L for most Enterobacteriaceae species, including *E. coli* ($MIC_{50/90}$, 0.5/1 mg/L), *K. pneumoniae* ($MIC_{50/90}$, 0.25/0.5 mg/L) and *E. cloacae* ($MIC_{50/90}$, 0.5/0.5 mg/L). Similar to observed with other aminoglycosides (data not shown), the activity of plazomicin against *Proteus mirabilis* (MIC_{90}, 4 mg/L) and indole-positive *Proteus* species (MIC_{90}, 4 mg/L) was four- to eight-fold higher when compared to most other Enterobacteriaceae species (**Table 1**).
- A total of 772 isolates with an ESBL-phenotype (includes 256 *E. coli*, 40 *K. oxytoca*, 464 *K. pneumoniae* and 12 *P. mirabilis*) were observed and plazomicin (MIC_{50} and MIC_{90}, 0.5 and 1 mg/L) displayed good activity against these isolates, which was comparable to the activity of this compound against the overall Enterobacteriaceae collection (**Table 1**).
- Plazomicin inhibited 86.7% of the 150 CRE isolates at ≤2 mg/L (**Table 2**). Plazomicin activity against CRE varied considerably in the different regions and MIC_{50} and MIC_{90} results were 128 and >128 mg/L for isolates collected in Asia-Pacific, 0.5 and 2 mg/L for Europe and 0.5 and >128 mg/L for Latin America, respectively (**Table 2**), mainly due to the presence of 16S rRNA methylase encoding genes in Asia-Pacific and Latin America.
- Plazomicin displayed good activity against isolates non-susceptible (per EUCAST criteria) to gentamicin (MIC_{50} and MIC_{90}, 0.5 and 4 mg/L) or tobramycin (MIC_{50} and MIC_{90}, 0.5 and 2 mg/L). Overall, the activity of this compound was still limited against amikacin non-susceptible isolates (MIC_{50} and MIC_{90}, >128 and >128 mg/L) that might carry 16S rRNA methylase encoding genes. The activity of plazomicin against these isolates was greater in Europe and Latin America (MIC_{50} and MIC_{90}, 0.5 and >128 mg/L) when compared to the Asia-Pacific ($MIC_{50/90}$, 128/>128 mg/L; **Table 2**).
- All 36 (1.1%) Enterobacteriaceae isolates displaying plazomicin MIC results at ≥128 mg/L carried 16S rRNA methylase encoding genes, as follows: 19 *rmtB*, eight *rmtF*, seven *armA*, one of each *rmtA*, *rmtC* and *rmtD1* (**Figure 1**). One *K. pneumoniae* isolate from Poland was positive for both *armA* and *rmtA*.
- Plazomicin (MIC_{50} and MIC_{90}, 4 and 8 mg/L) inhibited 67.0% of *P. aeruginosa* at ≤4 mg/L and 91% at ≤8 mg/L (**Table 1**).
- All coagulase-negative staphylococci (MIC_{50} and MIC_{90}, 0.12 and 0.25 mg/L) were inhibited by plazomicin at ≤0.25 mg/L.
- Plazomicin was very active against *S. aureus* (MIC_{50} and MIC_{90}, 0.5 and 1 mg/L) and MIC values ranged from 0.12 to 2 mg/L. The activity of this compound was maintained against MRSA isolates (MIC_{50} and MIC_{90}, 0.5 and 0.5 mg/L).
- As with other aminoglycosides (data not shown), the activity of plazomicin activity was limited against *Acinetobacter* spp. ($MIC_{50/90}$, 32/>128 mg/L), *Enterococcus* spp. and *S. pneumoniae* ($MIC_{50/90}$, 64/64 mg/L for both; **Table 1**).

Table 1. Antimicrobial activity of plazomicin tested against the main organisms, organism groups, and resistant subsets of isolates submitted during 2014 from Asia-Pacific, Europe and Latin America.

Organism/Organism group/Phenotype	No. of isolates tested	No. of isolates at plazomicin MIC (mg/L; cumulative %): ≤0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	128	>128	MIC_{50}	MIC_{90}
Enterobacteriaceae	3224		27 (0.8)	866 (27.7)	1275 (67.2)	714 (89.4)	218 (96.2)	69 (98.3)	12 (98.7)	6 (98.9)	1 (98.9)	0 (98.9)	5 (99.0)	31 (100.0)	0.5	2
ESBL-phenotype	772		10 (1.3)	296 (39.6)	272 (74.9)	134 (92.2)	21 (94.9)	2 (95.2)	1 (95.3)	3 (95.7)	0 (95.7)	0 (95.7)	5 (96.4)	28 (100.0)	0.5	1
Escherichia coli	1188		1 (0.1)	73 (6.2)	580 (55.1)	457 (93.5)	66 (99.1)	7 (99.7)	0 (99.7)	2 (99.8)	0 (99.8)	0 (99.8)	0 (99.8)	2 (100.0)	0.5	1
Klebsiella pneumoniae	1085		21 (1.9)	648 (61.7)	351 (94.0)	27 (96.5)	5 (97.0)	0 (97.0)	1 (97.1)	2 (97.2)	0 (97.2)	0 (97.2)	5 (97.7)	25 (100.0)	0.25	0.5
Klebsiella oxytoca	188			31 (16.5)	129 (85.1)	25 (98.4)	2 (99.5)	0 (99.5)	1 (100.0)						0.5	1
Enterobacter aerogenes	93			16 (17.2)	59 (80.6)	17 (98.9)	1 (100.0)								0.5	1
Enterobacter cloacae species complex	104		2 (1.9)	33 (33.7)	62 (93.3)	6 (99.0)	0 (99.0)	0 (99.0)	0 (99.0)	0 (99.0)	0 (99.0)	0 (99.0)	0 (99.0)	1 (100.0)	0.5	0.5
Serratia marcescens	102				8 (7.8)	87 (93.1)	7 (100.0)								1	1
Citrobacter freundii species complex	77			24 (31.2)	42 (85.7)	7 (94.8)	4 (100.0)								0.5	1
Citrobacter koseri	76		3 (3.9)	40 (56.6)	28 (93.4)	5 (100.0)									0.25	0.5
Morganella morganii	83				5 (6.0)	23 (33.7)	26 (65.1)	22 (91.6)	4 (96.4)	2 (98.8)	1 (100.0)				2	4
Proteus mirabilis	108			1 (0.9)	1 (1.9)	17 (17.6)	66 (78.7)	21 (98.1)	0 (98.1)	0 (98.1)	0 (98.1)	0 (98.1)	0 (98.1)	2 (100.0)	2	4
Proteus vulgaris	66				7 (10.6)	28 (53.0)	21 (84.8)	8 (97.0)	2 (100.0)						1	4
Providencia spp.	54				3 (5.6)	15 (33.3)	20 (70.4)	11 (90.7)	4 (98.1)	0 (98.1)	0 (98.1)	0 (98.1)	0 (98.1)	1 (100.0)	2	4
Pseudomonas aeruginosa	100			1 (1.0)	2 (3.0)	2 (5.0)	16 (21.0)	46 (67.0)	24 (91.0)	7 (98.0)	1 (99.0)	0 (99.0)	0 (99.0)	1 (100.0)	4	8
Acinetobacter spp.	100		1 (1.0)	2 (3.0)	10 (13.0)	7 (20.0)	8 (28.0)	4 (32.0)	9 (41.0)	8 (49.0)	9 (58.0)	5 (63.0)	0 (63.0)	37 (100.0)	32	>128
Coagulase-negative staphylococci	61	22 (36.1)	23 (73.8)	16 (100.0)											0.12	0.25
Staphylococcus aureus	64		2 (3.1)	23 (39.1)	32 (89.1)	6 (98.4)	1 (100.0)								0.5	1
MRSA	20			6 (30.0)	12 (90.0)	2 (100.0)									0.5	0.5
Streptococcus pneumoniae	51								1 (2.0)	0 (2.0)	17 (35.3)	32 (98.0)	1 (100.0)		64	64
Enterococcus spp.	60							8 (13.3)	12 (33.3)	4 (40.0)	3 (45.0)	27 (90.0)	6 (100.0)		64	64

Table 2. Activity of plazomicin against Enterobacteriaceae isolates, CRE and aminoglycoside non-susceptible isolates collected during 2014 by region.

Organism group/ Phenotype/Region	No. of isolates tested	No. of isolates at plazomicin MIC (mg/L; cumulative %): 0.12	0.25	0.5	1	2	4	8	16	32	64	128	>128	MIC_{50}	MIC_{90}
Enterobacteriaceae															
All regions	3224	27 (0.8)	866 (27.7)	1275 (67.2)	714 (89.4)	218 (96.2)	69 (98.3)	12 (98.7)	6 (98.9)	1 (98.9)	0 (98.9)	5 (99.0)	31 (100.0)	0.5	2
Asia-W. Pacific	685	10 (1.5)	180 (27.7)	301 (71.7)	138 (91.8)	34 (96.8)	9 (98.1)	1 (98.2)	0 (98.2)	1 (98.4)	0 (98.4)	3 (98.8)	8 (100.0)	0.5	1
Europe	2085	13 (0.6)	569 (27.9)	812 (66.9)	456 (88.7)	153 (96.1)	48 (98.4)	9 (98.8)	6 (99.1)	0 (99.1)	0 (99.1)	2 (99.2)	17 (100.0)	0.5	2
Latin America	454	4 (0.9)	117 (26.7)	162 (62.3)	120 (88.8)	31 (95.6)	12 (98.2)	2 (98.7)	0 (98.7)	0 (98.7)	0 (98.7)	0 (98.7)	6 (100.0)	0.5	2
CRE															
All regions	150	2 (1.3)	63 (43.3)	47 (74.7)	15 (84.7)	3 (86.7)	0 (86.7)	1 (87.3)	2 (88.7)	0 (88.7)	0 (88.7)	5 (92.0)	12 (100.0)	0.5	128
Asia-W. Pacific	114		51 (44.7)	39 (78.9)	12 (89.5)	3 (92.1)	0 (92.1)	1 (93.0)	2 (94.7)	0 (94.7)	0 (94.7)	2 (96.5)	4 (100.0)	0.5	2
Europe	25		12 (48.0)	7 (76.0)	3 (88.0)	0 (88.0)	0 (88.0)	0 (88.0)	0 (88.0)	0 (88.0)	0 (88.0)	0 (88.0)	3 (100.0)	0.5	>128
Latin America	11	2 (18.2)	0 (18.2)	1 (27.3)	0 (27.3)	0 (27.3)	0 (27.3)	0 (27.3)	0 (27.3)	0 (27.3)	0 (27.3)	3 (54.5)	5 (100.0)	128	>128
Gentamicin-non-susceptible															
All regions	556	5 (0.9)	178 (32.9)	173 (64.0)	96 (81.3)	39 (88.3)	17 (91.4)	8 (92.8)	4 (93.5)	0 (93.5)	0 (93.5)	5 (94.4)	31 (100.0)	0.5	4
Asia-W. Pacific	102	3 (2.9)	18 (20.6)	38 (57.8)	24 (81.4)	7 (88.2)	0 (88.2)	1 (89.2)	0 (89.2)	0 (89.2)	0 (89.2)	3 (92.2)	8 (100.0)	0.5	128
Europe	324		113 (34.9)	103 (66.7)	41 (79.3)	26 (87.3)	12 (91.0)	6 (92.9)	4 (94.1)	0 (94.1)	0 (94.1)	2 (94.8)	17 (100.0)	0.5	4
Latin America	130	2 (1.5)	47 (37.7)	32 (62.3)	31 (86.2)	6 (90.8)	5 (94.6)	1 (95.4)	0 (95.4)	0 (95.4)	0 (95.4)	0 (95.4)	6 (100.0)	0.5	2
Tobramycin-non-susceptible															
All regions	744	7 (0.9)	257 (35.5)	240 (67.7)	138 (86.3)	40 (91.7)	16 (93.8)	6 (94.6)	4 (95.2)	0 (95.2)	0 (95.2)	5 (95.8)	31 (100.0)	0.5	2
Asia-W. Pacific	133	3 (2.3)	30 (24.8)	53 (64.7)	27 (85.0)	9 (91.7)	0 (91.7)	0 (91.7)	0 (91.7)	0 (91.7)	0 (91.7)	3 (94.0)	8 (100.0)	0.5	2
Europe	448	2 (0.4)	167 (37.7)	146 (70.3)	68 (85.5)	26 (91.3)	11 (93.8)	5 (94.9)	4 (95.8)	0 (95.8)	0 (95.8)	2 (96.2)	17 (100.0)	0.5	2
Latin America	163	2 (1.2)	60 (38.0)	41 (63.2)	43 (89.6)	5 (92.6)	5 (95.7)	1 (96.3)	0 (96.3)	0 (96.3)	0 (96.3)	0 (96.3)	6 (100.0)	0.5	2
Amikacin-non-susceptible															
All regions	174		61 (35.1)	44 (60.3)	18 (70.7)	4 (73.0)	7 (77.0)	2 (78.2)	2 (79.3)	0 (79.3)	0 (79.3)	5 (82.2)	31 (100.0)	>128	>128
Asia-W. Pacific	17		3 (17.6)	3 (35.3)	0 (35.3)	0 (35.3)	0 (35.3)	0 (35.3)	0 (35.3)	0 (35.3)	0 (35.3)	3 (52.9)	8 (100.0)	128	>128
Europe	128		46 (35.9)	38 (65.6)	15 (77.3)	3 (79.7)	4 (82.8)	1 (83.6)	2 (85.2)	0 (85.2)	0 (85.2)	2 (86.7)	17 (100.0)	0.5	>128
Latin America	29		12 (41.4)	3 (51.7)	3 (62.1)	1 (65.5)	3 (75.9)	1 (79.3)	0 (79.3)	0 (79.3)	0 (79.3)	0 (79.3)	6 (100.0)	0.5	>128

Figure 1. Distributions of 16S rRNA methylases detected among Enterobacteriaceae species and regions.

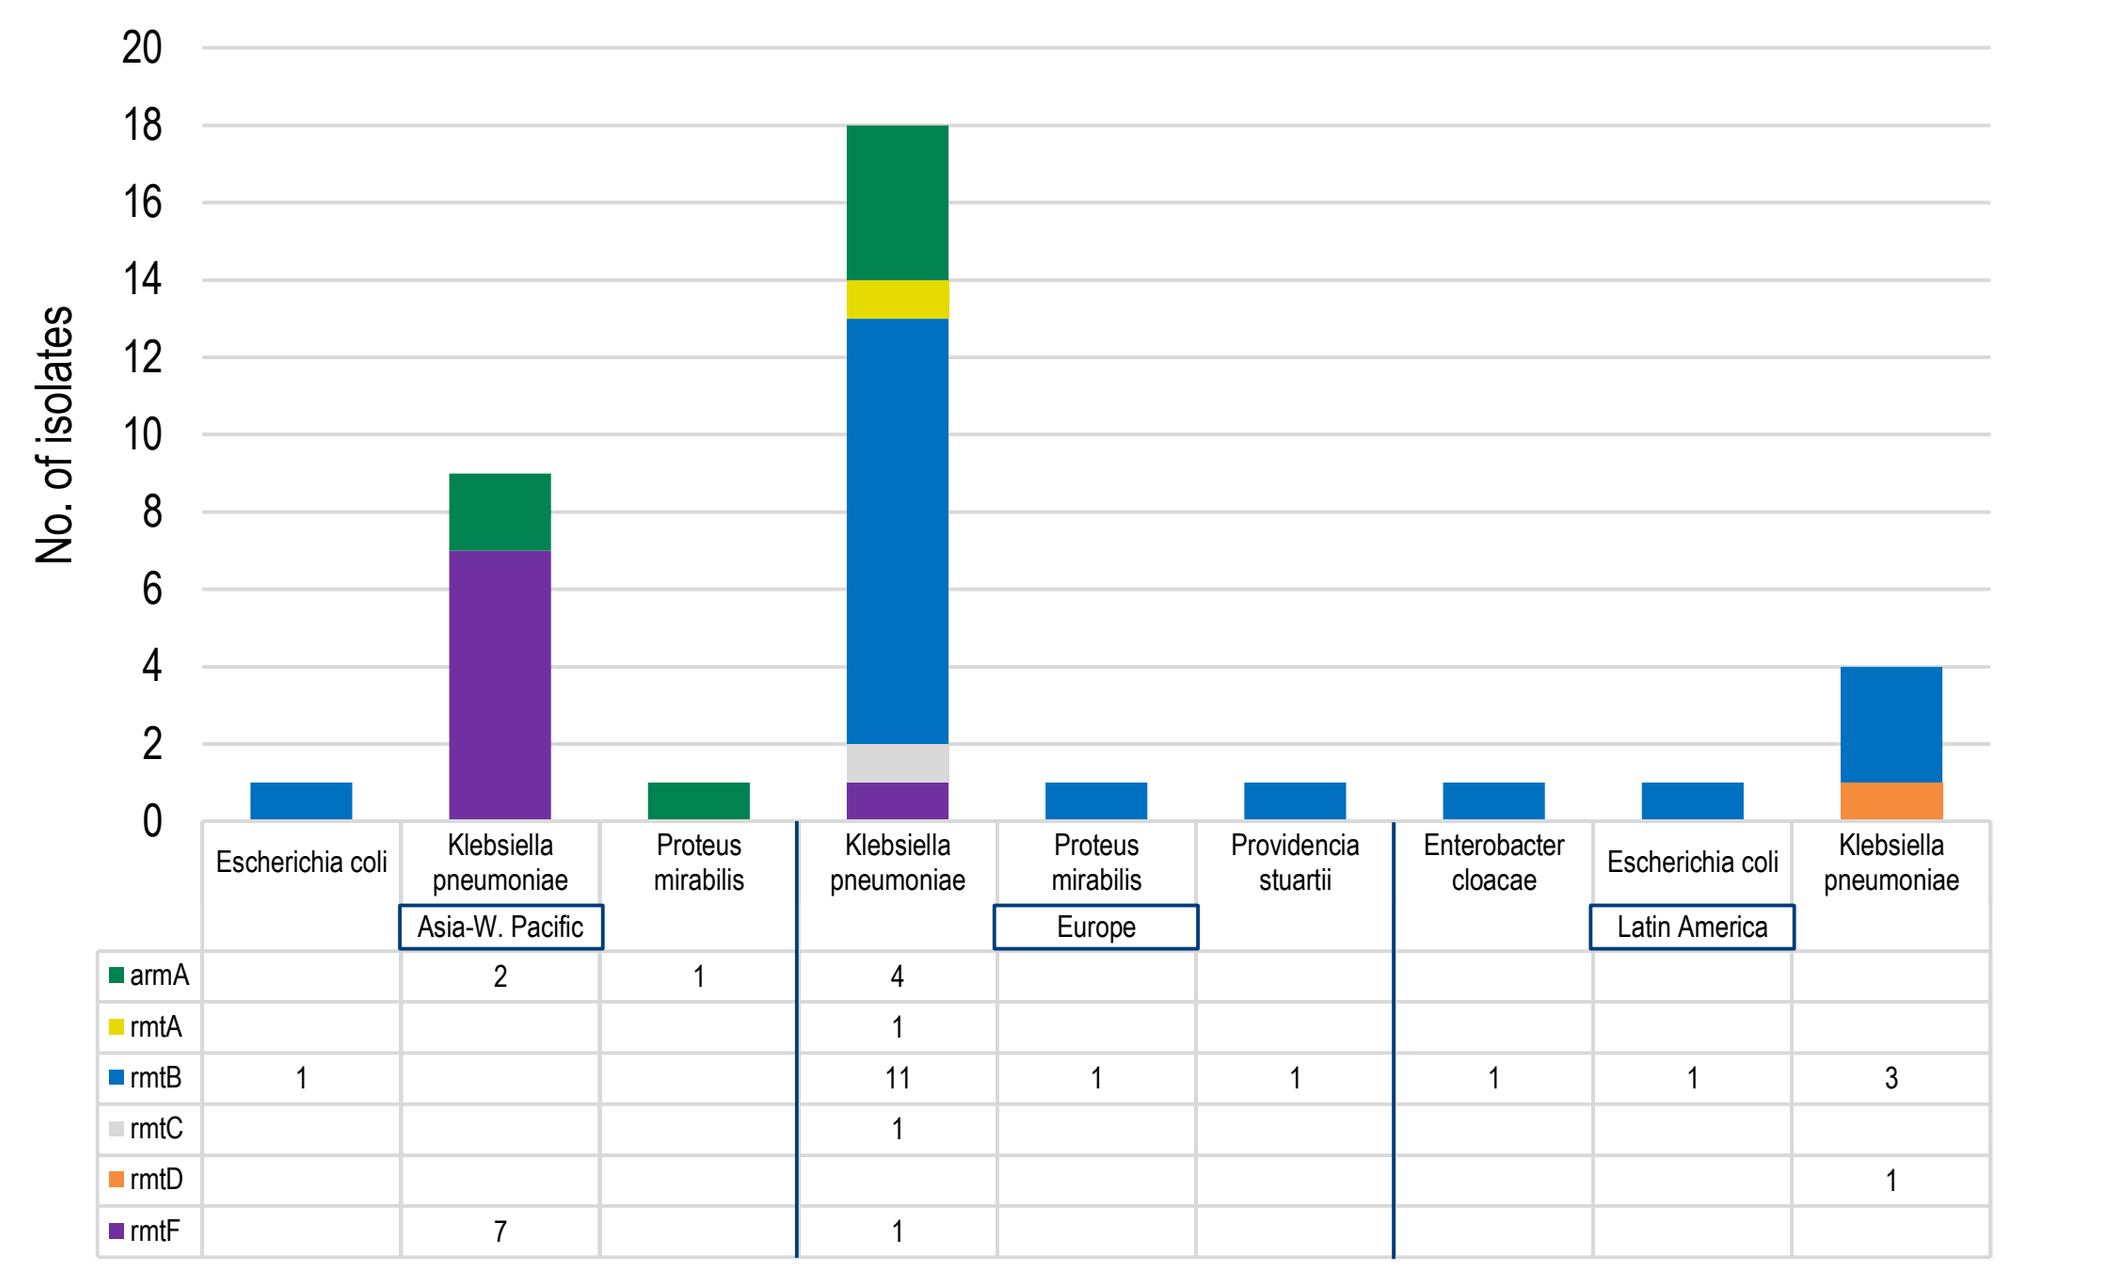
No. of isolates
20
18
16
14
12
10
8
6
4
2
0
Escherichia coli
Klebsiella pneumoniae
Asia-W. Pacific
Proteus mirabilis
Klebsiella pneumoniae
Proteus mirabilis
Europe
Providencia stuartii
Enterobacter cloacae
Escherichia coli
Latin America
Klebsiella pneumoniae
armA 2 1 4
rmtA 1
rmtB 1 11 1 1 1 1 3
rmtC 1
rmtD 1
rmtF 7 1

CONCLUSIONS

- Plazomicin displayed good activity against Enterobacteriaceae isolates, including isolates displaying an ESBL-phenotype and CRE isolates from Europe and Latin America. Additionally, plazomicin displayed good activity against Enterobacteriaceae isolates from Europe and Latin America that were non-susceptible to other aminoglycosides and did not carry genes encoding 16S rRNA methylases.
- All of the Enterobacteriaceae isolates displaying highly elevated plazomicin MIC values (≥128 mg/L) produced 16S rRNA methylases that confer pan-aminoglycoside resistance.
- Plazomicin MIC values for *P. aeruginosa* were higher when compared to Enterobacteriaceae isolates and the activity of this compound was two- to eight-fold lower than other aminoglycosides (data not shown).
- The activity of plazomicin against *Staphylococcus* spp. was very good, regardless of the species or methicillin-resistance phenotype.
- Plazomicin demonstrated good activity against Enterobacteriaceae, including MDR isolates, tested in this study. The activity of plazomicin against *Staphylococcus* spp. is also a valuable characteristic.

ACKNOWLEDGEMENTS

This study was sponsored by an investigational grant from Achaogen.

REFERENCES

1. Chambers HF, Bartlett JG, Bonomo RA, Chiou C, Cosgrove SE, Cross HR, Daum RS, Downing M, Evans SR, Knisely J, Kreiswirth BN, Lautenbach E, Mickley BS, Patel R, Pettigrew MM, Rodvold KA, Spellberg B, Fowler VG, Jr. (2014). Antibacterial resistance leadership group: open for business. *Clin Infect Dis* 58: 1571-1576.
2. Clinical and Laboratory Standards Institute (2016). *M100-S26. Performance standards for antimicrobial susceptibility testing: 26th informational supplement.* Wayne, PA: CLSI.
3. EUCAST (2016). Breakpoint tables for interpretation of MICs and zone diameters. Version 6.0, January 2016. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 2016.
4. Nordmann P, Naas T, Fortineau N, Poirel L (2007). Superbugs in the coming new decade; multidrug resistance and prospects for treatment of *Staphylococcus aureus*, *Enterococcus* spp. and *Pseudomonas aeruginosa* in 2010. *Curr Opin Microbiol* 10: 436-440.
5. Zhanel GG, Lawson CD, Zelenitsky S, Findlay B, Schweizer F, Adam H, Walkty A, Rubinstein E, Gin AS, Hoban DJ, Lynch JP, Karlowsky JA (2012). Comparison of the next-generation aminoglycoside plazomicin to gentamicin, tobramycin and amikacin. *Expert Rev Anti Infect Ther* 10: 459-473.