ESCMID 2025 | Poster #P2891

Activity of manogepix against a worldwide collection of yeast isolates from 2023

Marisa L. Winkler, Samuel Edeker, Abby Klauer, Paul R. Rhomberg, Mariana Castanheira

Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- Manogepix is a novel antifungal agent in Phase 3 clinical trials for the treatment of invasive yeast and mould infections.
- Manogepix targets the fungal Gwt1 enzyme which is a different target and mechanism of action relative to current azoles, echinocandins, and amphotericin B.
- There is little cross-resistance among manogepix and other antifungal agents.
- The *in vitro* activity of manogepix and comparator antifungal agents was evaluated against yeast isolates from invasive infections collected outside of the United States.

Methods

- A total of 1,233 yeast isolates were collected from 48 different hospitals in 29 different countries (Figure 1).
- Only 1 isolate per patient episode was included.
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing.
- Isolates were tested by CLSI reference broth microdilution method (M27) and a subset of the 5 most common *Candida* species was tested by EUCAST reference broth microdilution method to obtain minimum inhibitory concentration (MIC) measurements.
- CLSI breakpoints (M27M44S) and EUCAST breakpoints (v.10) were applied for comparator agents; no breakpoints or epidemiological cutoff values are available for manogepix. CDC resistant-only breakpoints were used for *C. auris*.

Results

- 36 different species of yeast were represented (Figure 2) with *Candida albicans* most common (461/1233, 37.4%), followed by *C. glabrata* (245, 19.9%), *C. parapsilosis* (191, 15.5%), and *C. tropicalis* (138, 11.2%). There were 17 *C. auris* (1.4%).
- Manogepix $MIC_{50/90}$ values against *Candida* spp. were 0.008/0.03 mg/L by CLSI methodology (Table 1) and 0.008/0.12 mg/L by EUCAST methodology (Table 2).
- There were 66 *Candida* isolates nonsusceptible (NS) to fluconazole by CLSI; MICs for manogepix among these isolates ranged from 0.008–0.12 mg/L (Table 3).
 - This included *C. auris* where manogepix MICs ranged from 0.008–0.03 mg/L. One *C. auris* isolate was echinocandin-R and the manogepix MIC was 0.008 mg/L.
- There were 18 isolates NS to echinocandins (10 *C. glabrata*, 7 *C. parapsilosis*, and 1 *C. auris*).
 - Manogepix MICs in these organisms were 0.008–0.03 mg/L.
- By EUCAST testing, 111 isolates were tested (Table 2).
- MICs for manogepix in echinocandin-NS isolates were 0.008–0.03 mg/L.
- Elevated manogepix MICs were seen among *C. krusei* (MIC_{50} > 4 mg/L), *C. kefyr* (MIC_{50} 0.25 mg/L), *C. norvegensis* (MIC 2 mg/L), *Cryptococcus* spp. (MIC_{50} 1 mg/L), *Pichia cactophila* (MIC_{50} 1 mg/L), *Trichosporon* spp. (MIC_{50} 0.5 mg/L), and *Apiotrichum mycotoxinivorans* (MIC 1 mg/L).

Conclusions

- Manogepix has potent *in vitro* activity among diverse yeast species collected from clinical infections outside of the United States as part of a worldwide surveillance program.
 - This includes organisms of critical concern like *C. auris*.
- Likely due to its unique mechanism of action, manogepix retained activity against fluconazole-nonsusceptible isolates with MICs ≤0.06 mg/L.
- Elevated MICs were seen for manogepix against *C. kefyr*, *C. krusei*, *Cryptococcus* spp., and *Apiotrichum/Trichosporon* spp.
- Manogepix is a promising novel antifungal agent for the treatment of infections due to yeast.

Funding

This study was supported by Basilea and this project has been funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration for Strategic Preparedness and Response (ASPR); Biomedical Advanced Research and Development Authority (BARDA), under OT number 75A50124C00033. The contract and federal funding are not an endorsement of the study results, product, or company.

ML Winkler, S Edeker, A Klauer, PR Rhomberg, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was a paid consultant to Basilea in connection with the development of this poster.

Figure 1. Percent (%) of isolates from each non-US country included in surveillance

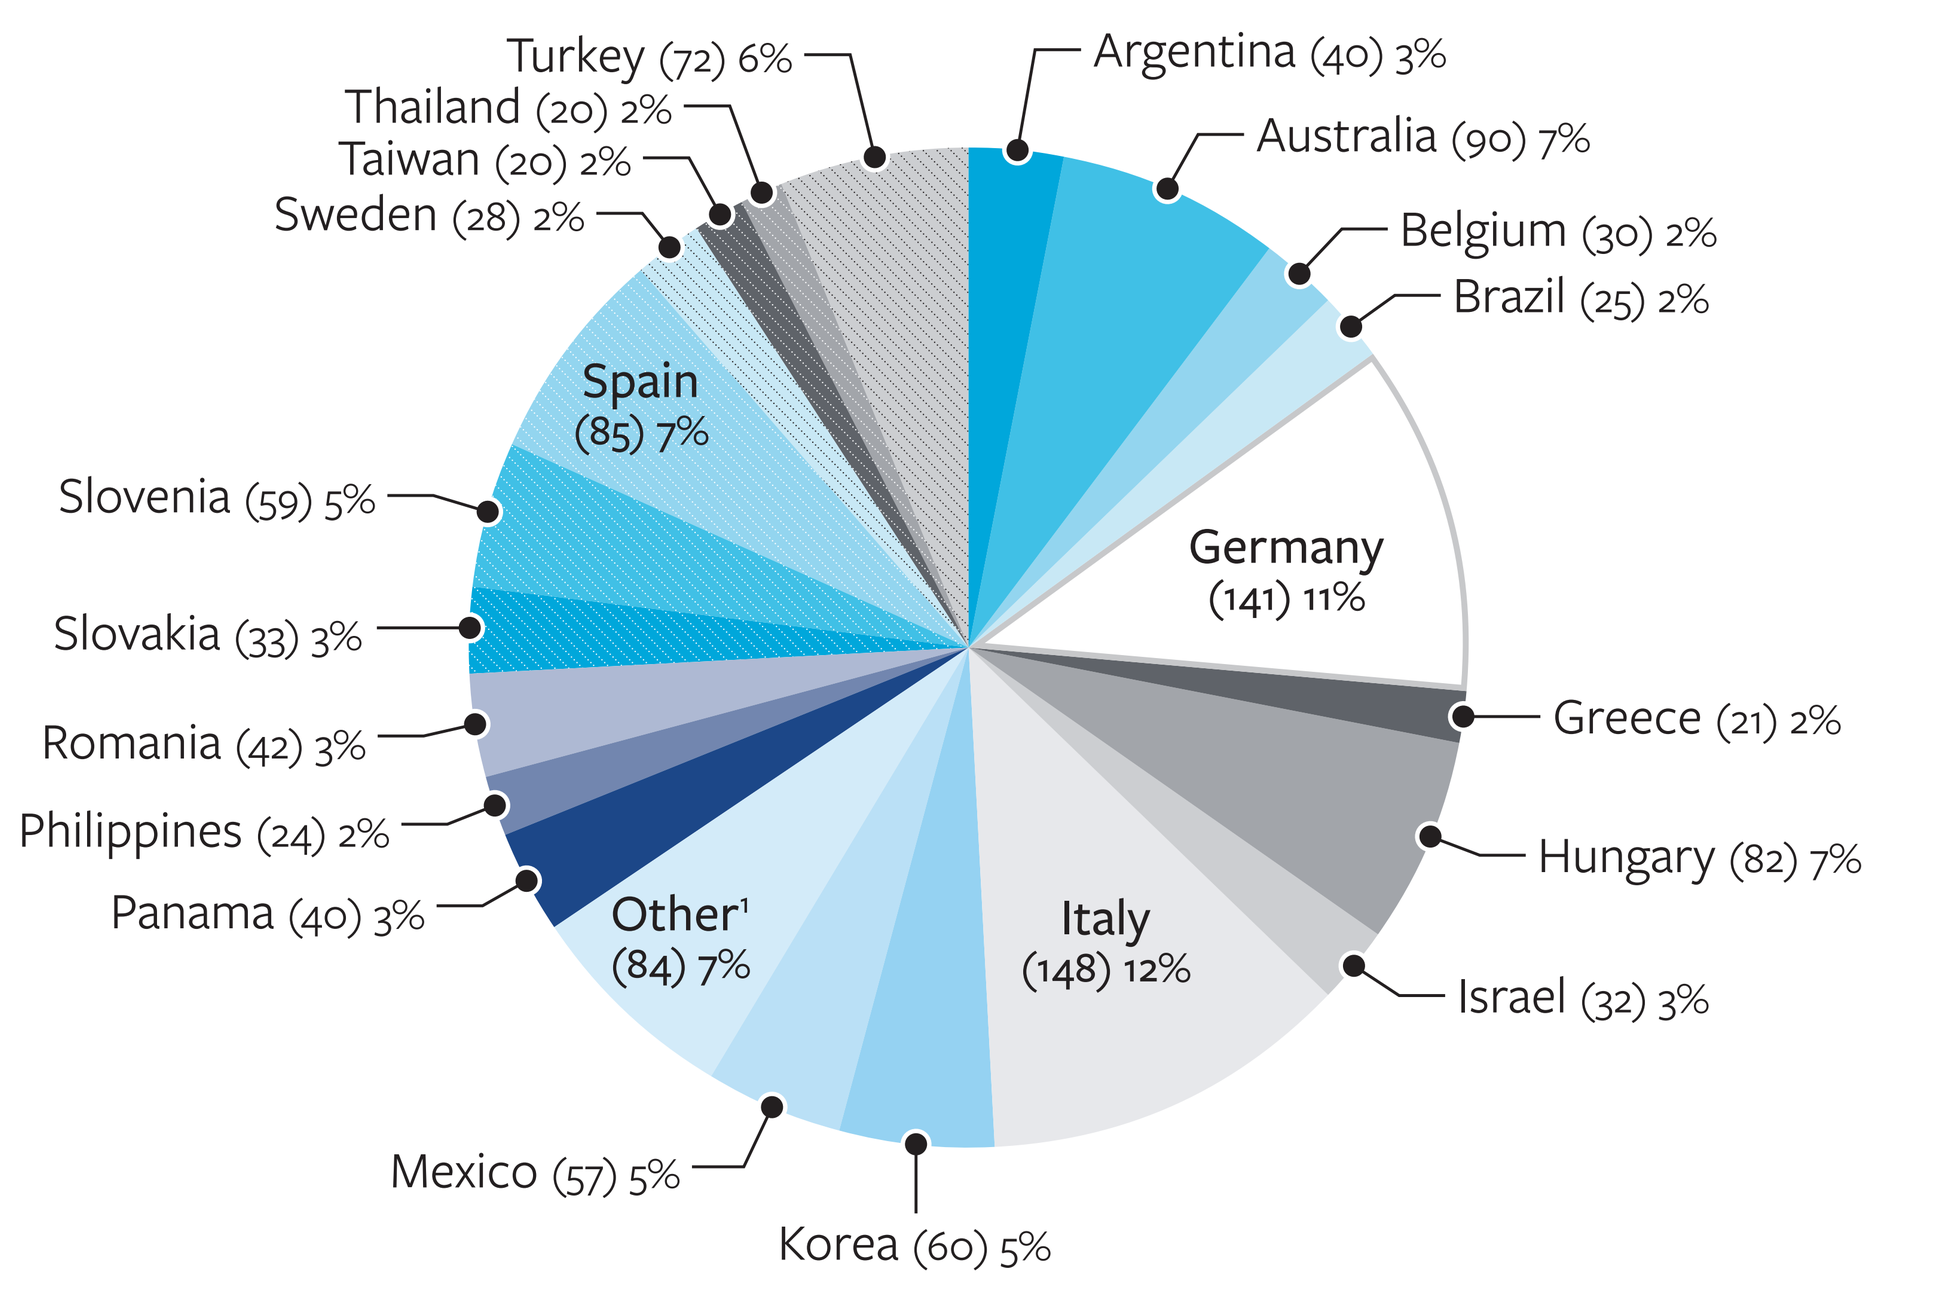
Turkey (72) 6%
Thailand (20) 2%
Taiwan (20) 2%
Sweden (28) 2%
Spain (85) 7%
Slovenia (59) 5%
Slovakia (33) 3%
Romania (42) 3%
Philippines (24) 2%
Panama (40) 3%
Other[1] (84) 7%
Mexico (57) 5%
Korea (60) 5%
Italy (148) 12%
Israel (32) 3%
Hungary (82) 7%
Greece (21) 2%
Germany (141) 11%
Brazil (25) 2%
Belgium (30) 2%
Australia (90) 7%
Argentina (40) 3%
1,233 yeast isolates total

[1] Chile (16), Czech Republic (16), France (8), Ireland (18), New Zealand (19), Portugal (1), Switzerland (4), UK (2)

Figure 2. Yeast species tested as part of the non-US surveillance program

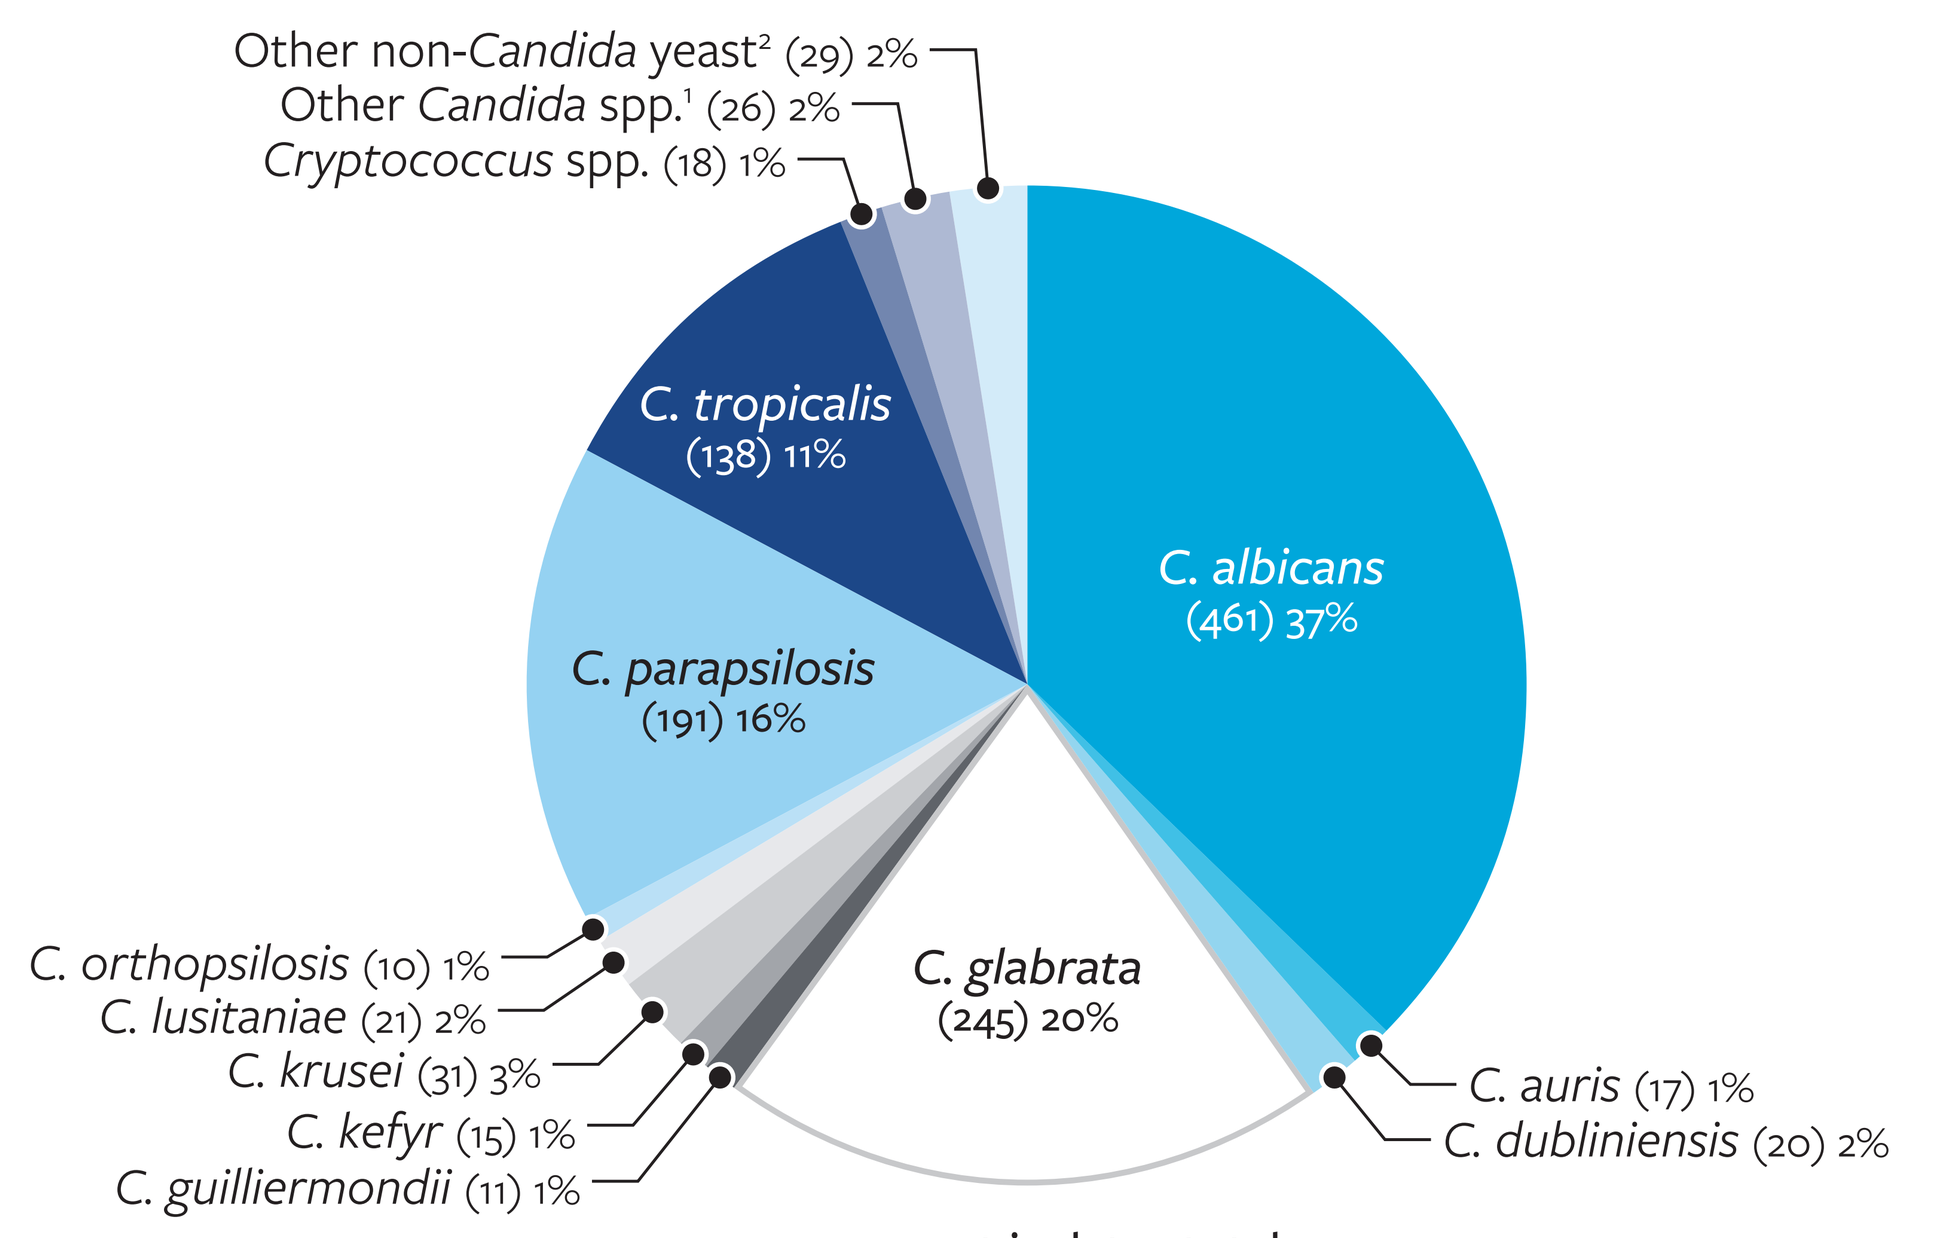
Other non-*Candida* yeast[2] (29) 2%
Other *Candida* spp.[1] (26) 2%
Cryptococcus spp. (18) 1%
C. tropicalis (138) 11%
C. albicans (461) 37%
C. parapsilosis (191) 16%
C. glabrata (245) 20%
C. orthopsilosis (10) 1%
C. lusitaniae (21) 2%
C. krusei (31) 3%
C. kefyr (15) 1%
C. guilliermondii (11) 1%
C. auris (17) 1%
C. dubliniensis (20) 2%
1,233 yeast isolates total

[1] Includes *C. fabianii* (4), *C. fermentati* (2), *C. lipolytica* (1), *C. metapsilosis* (4), *C. nivariensis* (1), *C. norvegensis* (1), *C. pararugosa* (1), *C. pelliculosa* (7), *C. pulcherrima* (1), *C. rugosa* (1), and unspeciated *Candida* (3)

[2] Includes *Apiotrichum mycotoxinivorans* (1), *Geotrichum clavatum* (1), *Kodamaea ohmeri* (1), *Magnusiomyces capitatus* (3), *Pichia cactophila* (9), *Rhodotorula mucilaginosa* (1), *Saccharomyces cerevisiae* (4), *Trichosporon asahii* (5), *Trichosporon mucoides* (1), unspeciated *Trichosporon* (2), *Wickerhamomyces anomalus* (1)

Table 1. MICs for CLSI testing of manogepix and comparator agents against population of yeasts

Organism name (n)	manogepix		anidulafungin		fluconazole	
	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S)	$MIC_{50/90}$ (mg/L)
All *Candida* spp. (1,186)	≤0.0005–>4	0.008/0.03	0.004–4	0.06/1	0.06–>128	0.5/4
C. albicans (461)	0.001–0.06	0.004/0.008	0.004–0.25 (100)	0.03/0.06	0.06–8 (99.8)	0.25/0.5
C. auris (17)	0.008–0.03	0.008/0.015	0.25–1 (100)	0.5/0.5	32–>128 (0)	32/128
C. dubliniensis (20)	0.002–0.015	0.004/0.008	0.015–0.12	0.06/0.06	0.06–2	0.12/0.12
C. glabrata (245)	0.004–0.12	0.03/0.06	0.03–4 (95.9)	0.12/0.12	0.25–128 (97.1)	4–8
C. kefyr (15)	0.06–0.5	0.25/0.5	0.03–0.25	0.12/0.12	0.12–1	0.25/0.5
C. krusei (31)	1–>4	>4/>4	0.03–0.12 (100)	0.06/0.12	4–128	32/64
C. lusitaniae (21)	0.008–0.12	0.03/0.03	0.06–1	0.25/0.5	0.12–16	0.25/0.5
C. parapsilosis (191)	0.004–0.06	0.008/0.03	0.5–4 (96.3)	2/2	0.12–128 (80.6)	0.5/32
C. tropicalis (138)	0.002–0.12	0.015/0.03	0.004–0.12 (100)	0.03/0.06	0.12–>128 (97.1)	0.5/1
Cryptococcus spp. (18)	0.03–1	0.25/1	4–>4	>4/>4	1–4	2–4
Pichia cactophila (9)	0.5–2	1/NA[1]	≤0.002–0.06	0.03/NA[1]	16–64	16/NA[1]
Trichosporon spp. (8)	0.25–2	0.5/NA[1]	4–>4	>4/NA[1]	2–>128	2/NA[1]

[1]Less than 10 isolates tested, MIC_{90} unable to be calculated

Table 2. MICs for EUCAST testing of manogepix and comparator agents against population of *Candida* spp.

Organism name (n)	manogepix		anidulafungin		fluconazole	
	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S)	$MIC_{50/90}$ (mg/L)
All *Candida* spp. (111)	0.002–>4	0.008/0.12	≤0.002–2	0.015/1	0.06–>128	0.5/16
Candida albicans (40)	0.002–0.008	0.004/0.004	≤0.002–0.008 (100)	0.004/0.008	0.06–0.05 (100)	0.25/0.25
Candida glabrata (27)	0.015–0.12	0.03/0.06	0.008–0.12 (96.3)	0.015/0.03	4–64 (0)	4/8
Candida parapsilosis (21)	0.004–0.015	0.008/0.015	0.5–2 (100)	2/2	0.12–16 (85.7)	0.5/4
Candida tropicalis (12)	0.004–0.008	0.004/0.008	0.008–0.03 (100)	0.008/0.015	0.12–1 (100)	0.5/0.5
Candida krusei (11)	4–>4	>4/>4	0.015–0.06 (100)	0.03/0.03	16–>128	64/128

Table 3. MICs for manogepix, anidulafungin, and fluconazole against fluconazole-nonsusceptible yeast

Organism name (n)	manogepix		anidulafungin		fluconazole	
	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)
Candida albicans (1)	0.008	NA[1]	0.008	NA[1]	8	NA[1]
Candida auris (17)	0.008–0.03	0.008/0.015	0.25–1	0.5/0.5	32–>128	32/128
Candida glabrata (7)	0.015–0.06	0.03/NA[1]	0.06–4	0.12/NA[1]	64–128	64/NA[1]
Candida parapsilosis (37)	0.008–0.06	0.03/0.03	0.5–2	1/2	4–128	32/64
Candida tropicalis (4)	0.008–0.12	0.015/NA[1]	0.008–0.03	0.015/NA[1]	8–>128	32/NA[1]

[1]Unable to calculate MIC_{50} if <2 isolates and MIC_{90} if <10 isolates

References

Pappas, P.G., Kauffman, C.A., Andes, D.R., Clancy, C.J., Marr, K.A., Ostrosky-Zeichner, L., et al., *Clinical Practice Guideline for the Management of Candidiasis: 2016 Update by the Infectious Diseases Society of America*. Clin Infect Dis, 2016. 62(4): e1-50.

Centers for Disease Control. *Antifungal susceptibility testing for C. auris*. April 23, 2024. https://www.cdc.gov/candida-auris/hcp/laboratories/antifungal-susceptibility-testing.html

CLSI, *M27 M44S Ed3. Performance standards for antifungal susceptibility testing of yeasts*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M57S Ed4. Epidemiological cutoff values for antifungal susceptibility testing*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M27 Ed4. Reference method for broth dilution antifungal susceptibility testing of yeasts*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

EUCAST Definitive Document E.Def7.4. *Method for broth dilution minimum inhibitory concentration of antifungal agents for yeast*. 2023. https://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/AFST/Files/EUCAST_E.Def_7.4_Yeast_definitive_revised_2023.pdf

EUCAST Antifungal Clinical Breakpoint Table v10. https://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/AFST/Clinical_breakpoints/AFST_BP_v10.0_200204_updatd_links_200924.pdf

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

Contact

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ESCMID2025_23-BAS-12_P2_Yeast_EU.pdf

Charges may apply. No personal information is stored.

ESCMID 2025, April 11–15, 2025, Vienna, Austria