D-229

Intermethod Comparability Analyses of Tigecycline Antimicrobial Susceptibility Tests (AST) Using Over 5,000 Pathogens Isolated in Asia-Pacific Countries

RN JONES, JD TURNIDGE, MJ DOWZICKY, JM BELL

JMI Laboratories, North Liberty, IA; Women's and Children's Hospital, North Adelaide, Australia; Wyeth Pharmaceuticals, Collegeville, PA

ICAAC 2007
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370, fax 319.665.3371
ronald-jones@jmilabs.com

ABSTRACT

Background:
Tigecycline (TGC), a novel glycylcycline, recently became available for therapy of serious infections caused by multidrug-resistant pathogens. Accuracy of AST remains critical to appropriate TGC use, therefore recent (2006) clinical pathogens were tested by reference broth microdilution (BMD), disk diffusion (DD) and Etest (ET) methods to determine intermethod agreement.

Methods:
All compared TGC tests used CLSI methods (BMD, DD) or manufacturer instructions. Concurrent QC was with CLSI published limits. 5,442 isolates were tested from the following analysis groups: *S. aureus* (SA; 1646), CoNS (335), Enterobacteriaceae (ENTB; 2270, 1841 US-FDA indicated species), enterococci (ETC; 673), *S. pneumoniae* (SPN; 257), ß-haemolytic (BST; 172) and viridans gr. (VGS; 89) streptococci. Comparisons were analyzed by CLSI M23-A2 criteria using US-FDA breakpoints.

Results:
TGC intermethod categorical and essential (MIC variations) agreements were excellent except for US-FDA ENTB DD results (85.5-89.6% agreement; 1.0 and 13.5% major and minor errors). By using proposed breakpoint criteria for DD (see Table), agreement was improved to 98.2-99.2%, best for indicated species. ET essential agreements ±1 or ±2 $\log_2$ dilutions compared to BMD was at 84.7-97.5 and 99.1-100.0%, respectively.

	Intermethod categorical or essential agreement (%):			
	DD by criteria:		ET-BMD variations:	
Organisms (no.)	US-FDA	Proposed[a]	±2-fold	±4-fold
Enterobacteriaceae				
All species (2270)	85.5	98.2	84.7	99.6
Indicated spp. (1841)	89.6	99.2	85.8	99.5
S. aureus (1646)	99.8	-[b]	90.3	99.9
CoNS (335)	99.4	-	96.4	100.0
Enterococci (673)	98.5	-	86.0	99.1
Streptococci (518)[c]	100.0	-	97.5	100.0

a. Criteria of Jones et al. (2007), S at ≥16 mm and R at ≤12 mm.
b. Same criteria as US-FDA.
c. Includes SPN, BST and VGS.

Conclusions:
TGC S tests perform very well for SA, CoNS, ETC and various streptococci using current US-FDA breakpoints, but this high volume study demonstrates that ENTB test results by DD urgently require breakpoint modifications to correlate with ET or BMD MIC values.

INTRODUCTION

Tigecycline is the first glycylcycline approved by the United States Food and Drug Administration (US-FDA) and the European Medicines Agency (EMEA) for clinical treatment of complicated skin and skin structure infections and complicated intra-abdominal infections. This agent demonstrates potent broad-spectrum antimicrobial activity against many Gram-negative and Gram-positive bacterial pathogens. This glycylcycline is derived from minocycline but possesses the ability to overcome acquired efflux target-mediated resistance mechanisms and ribosomal protection mechanisms. No other cross-resistances have been observed to other antimicrobial classes.

Tigecycline is a valuable treatment option against multidrug-resistant (MDR) pathogens. The prevalence of MDR pathogens, including those Enterobacteriaceae producing extended spectrum ß-lactamases (ESBLs), carbapenem-resistant non-fermentative bacilli and methicillin-resistant staphylococci (MRSA) isolated from serious infections in the Asia-Pacific Region as monitored by the SENTRY Antimicrobial Surveillance Program are becoming more common. We prospectively investigated the accuracy of two alternative susceptibility testing methods (disk diffusion, DD; and Etest, ET) and compared those results to the reference Clinical and Laboratory Standards Institute (CLSI; formerly the NCCLS) broth microdilution (BMD) method to determine intermethod accuracy and categorical agreement.

MATERIALS AND METHODS

Bacterial Isolates: A total of 5,442 isolates were received by the APAC monitoring laboratory (Adelaide, Australia) from 41 medical centers in nine participating countries. Non-duplicate patient isolates from infections of the bloodstream, respiratory tract, or wounds/skin and skin structure infections were submitted. Isolate identifications were confirmed by the central laboratory using standard biochemical and commercial identification systems. The breakdown of the pathogen groups were Enterobacteriaceae (2,270 strains; 1,841 from the US-FDA indicated species), *Staphylococcus aureus* (1,646 strains), coagulase-negative staphylococci (CoNS, 335 strains), enterococci (673 strains), *Streptococcus pneumoniae* (257 strains), ß-haemolytic streptococci (172 strains) and viridans group streptococci (89 strains),

Susceptibility Testing: BMD susceptibility testing was performed using validated panels produced by TREK Diagnostics (Cleveland, OH, USA). All MIC testing was performed using cation-adjusted Mueller-Hinton broth (supplemented with 2-5% lysed horse blood for the fastidious streptococci or HTM for *H. influenzae*). Categorical interpretation of the MIC results were found in the CLSI document M100-S17 or as published by the US-FDA (tigecycline). All quality control (QC) results for the American Type Culture Collection quality control strains were within the published QC ranges.

Additionally, concurrent susceptibility testing for tigecycline was performed on all strains using the CLSI standardized DD method and the ET (AB BIODISK, Solna, Sweden) gradient system as per the manufacturer directions.

Analyses: Scattergram plot analyses were generated for each of eight pathogen groups with tigecycline BMD MIC results as compared to either the DD zone diameter value or the ET MIC result. Correlation coefficient values were calculated for the MIC vs. zone diameter plots and agreement percentages were calculated for the same result, ±1 $\log_2$ dilution and ±2 $\log_2$ dilutions from the reference BMD MIC.

SELECTED REFERENCES

1. Bradford PA (2004). Tigecycline: A first in class glycylcycline. *Clin Microbiol Newsletter* 26: 163-168.
2. Clinical and Laboratory Standards Institute. (2006). *M7-A7, Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard - seventh edition*. Wayne, PA: CLSI.
3. Clinical and Laboratory Standards Institute. (2007). *M100-S17, Performance standards for antimicrobial susceptibility testing, 17th informational supplement*. Wayne, PA: CLSI.
4. Clinical and Laboratory Standards Institute. (2006). *M2-A9. Performance standards for antimicrobial disk susceptibility tests; approved standard - ninth edition*. Wayne, PA: CLSI.
5. Fritsche TR, Sader HS, Stilwell MG, Dowzicky MJ, Jones RN (2005). Potency and spectrum of tigecycline tested against an international collection of bacterial pathogens associated with skin and soft tissue infections (2000-2004). *Diagn Microbiol Infect Dis* 52: 195-201.
6. Jones RN, Ferraro MJ, Reller LB, Schreckenberger PC, Swenson JM, Sader HS (2007). Multicenter studies of tigecycline disk diffusion susceptibility results for *Acinetobacter* spp. *J Clin Microbiol* 45: 227-230.
7. Rhomberg PR, Jones RN (2005). Reproducibility assessment of tigecycline MIC results by broth microdilution methods using commercially prepared dry-form panels. *Diagn Microbiol Infect Dis* 52: 67-69.
8. Sader HS, Jones RN, Stilwell MG, Dowzicky MJ, Fritsche TR (2005). Tigecycline activity tested against 26,474 bloodstream infection isolates: A collection from 6 continents. *Diagn Microbiol Infect Dis* 52: 181-186.
9. Tygacil Package Insert (2005). Philadelphia (PA): Wyeth Pharmaceuticals Inc. (June, 2005).

ACKNOWLEDGEMENT

The study was supported by a grant from Wyeth Pharmaceuticals.

RESULTS

- Over 5,000 bacterial isolates submitted from 41 medical centers in nine APAC countries were tested for susceptibility to tigecycline using three methods (BMD, DD and ET). For the BMD method the Enterobacteriaceae group had the highest MIC values observed but only 1.4% of isolate MIC values were at ≥2 µg/ml (see Table 1).

- Tigecycline was more potent against the Gram-positive species groups tested with 100% of strains inhibited at 0.12 µg/ml for *S. pneumoniae*, 0.25 µg/ml for ß-haemolytic and viridans group streptococci groups, 0.5 µg/ml for enterococci, and 1 µg/ml for staphylococci (Table 1).

- A comparison of the tigecycline ET results with the BMD results demonstrated an acceptable level of absolute agreement with 27.3-61.8% of MIC results being identical and a high level of agreement with 84.7-99.6% of results being ± one $\log_2$ dilution and 99.1-100.0% of results at ± two $\log_2$ dilution steps (Table 2).

- The ET MIC results showed a trend toward higher results for the Enterobacteriaceae, ß-haemolytic streptococci, *S. pneumoniae* and viridans group streptococci pathogen groups. In contrast, the ET MIC results trended lower for the enterococci and staphylococci pathogen groups.

Table 1. Cumulative percent inhibited at each tigecycline MIC value for 5,442 bacterial isolates collected in the Asia-Pacific region in 2006 for eight organism groups using a broth microdilution method.

	Cumulative % inhibited at each MIC value (µg/ml)							
Organism Group (# of strains)	≤0.03	0.06	0.12	0.25	0.5	1	2	4
Enterobacteriaceae (2270)	<0.1	1.3	13.8	48.9	80.4	92.8	98.6	99.9
Indicated species only[a] (1841)	<0.1	1.6	16.1	55.0	88.1	97.4	99.5	99.9
S. aureus (1646)	0.4	17.2	63.5	95.0	99.9	100.0	-	-
Coagulase-negative staphylococci (335)	1.5	11.9	48.1	88.1	99.4	100.0	-	-
Enterococcus species (673)	3.9	25.6	75.2	98.5	100.0	-	-	-
S. pneumoniae (257)	98.1	99.6	100.0	-	-	-	-	-
ß-haemolytic streptococci (172)	79.7	95.3	99.4	100.0	-	-	-	-
Viridans Group streptococci (89)	87.6	94.4	96.6	100.0	-	-	-	-

a. Enterobacteriaceae isolates from the indicated species (*C. freundii*, *E. cloacae*, *E. coli*, *K. oxytoca* and *K. pneumoniae*) as listed in the Tygacil® package insert.

Table 2. Variation in the tigecycline Etest MIC results compared to the reference broth microdilution tigecycline MIC results for each of the eight organism groups analyzed.

	Etest vs. BMD MIC results (µg/ml)							%		
Organism Group (no. tested)	-3	-2	-1	0	+1	+2	+3	Same	±1 dil	±2 dils
Enterobacteriaceae (2270)	-	2	66	630	1226	337	9	27.8	84.7	99.6
Indicated species only[a] (1841)	-	2	53	533	994	256	3	29.0	85.8	99.8
S. aureus (1646)	2	154	651	622	213	4	-	37.8	90.3	99.9
Coagulase-negative staphylococcus (335)	-	11	99	183	41	1	-	54.6	96.4	100.0
Enterococcus species (673)	6	88	285	253	41	-	-	37.6	86.0	99.1
S. pneumoniae (257)	-	1	-	125	131	-	-	48.6	99.6	100.0
ß-haemolytic streptococci (172)	-	-	-	47	114	11	-	27.3	93.6	100.0
Viridans Group streptococci (89)	-	-	4	55	29	1	-	61.8	98.9	100.0
Total (5442)	8	256	1105	1915	1795	354	9	35.2	88.5	99.7

a. Enterobacteriaceae isolates from the indicated species (*C. freundii*, *E. cloacae*, *E. coli*, *K. oxytoca* and *K. pneumoniae*) as listed in the Tygacil® package insert.

- Scattergram plots for selected organism groups comparing the tigecycline disk diffusion result with the BMD result for each strain demonstrated a highest correlation coefficient for Enterobacteriaceae (r=0.76) followed by coagulase-negative staphylococci (0.56), enterococci (0.41), *S. aureus* (0.36), viridans group streptococci (0.33), ß-haemolytic streptococci (0.18), and *S. pneumoniae* (0.13; see Figures 1 and 2).

Figures 1 and 2. Scattergram plots of tigecycline BMD MIC results vs. disk diffusion zone diameters for all Enterobacteriaceae and *S. aureus* isolates tested from the Asia-Pacific region in 2006 as part of the SENTRY Antimicrobial Surveillance Program. Solid horizontal and vertical lines indicate US-FDA interpretive criteria; broken lines are the proposed DD modifications to improve intermethod agreement (Jones et al. 2007).

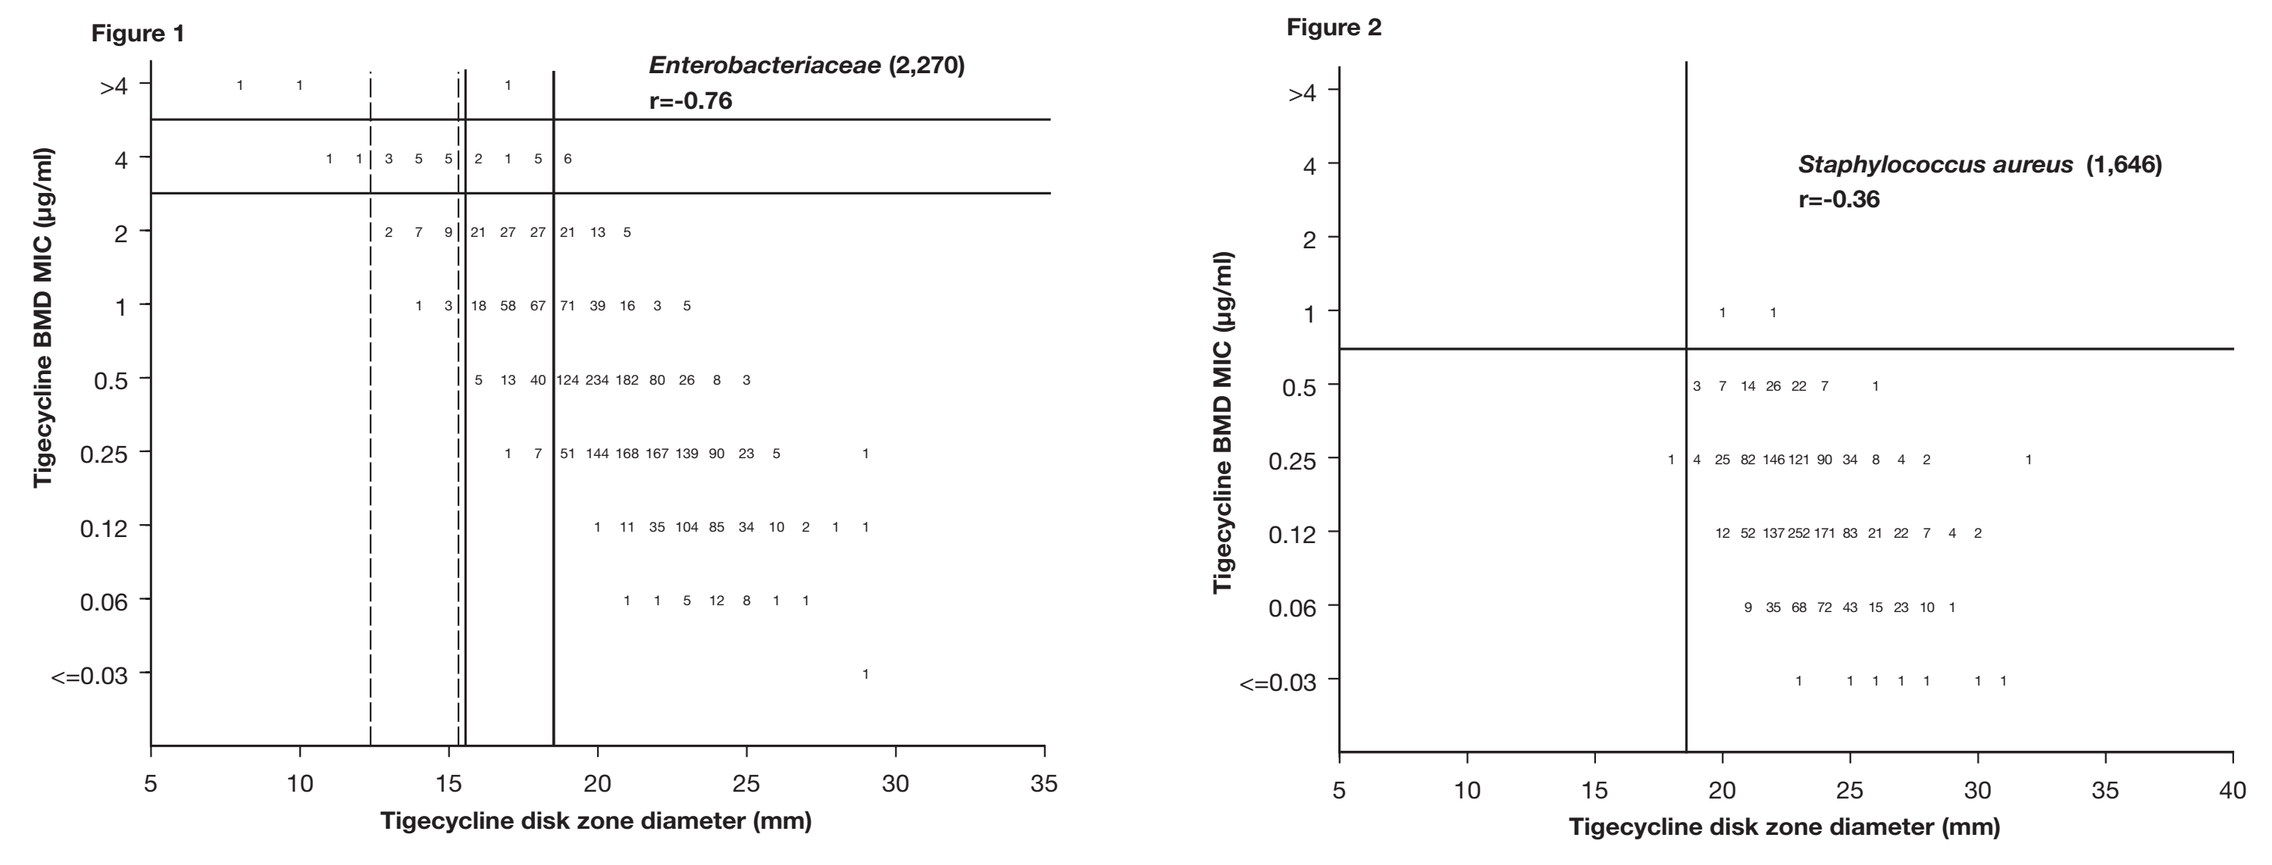

Figure 1
Enterobacteriaceae (2,270)
r=-0.76
Tigecycline BMD MIC (µg/ml)
Tigecycline disk zone diameter (mm)
Figure 2
Staphylococcus aureus (1,646)
r=-0.36
Tigecycline BMD MIC (µg/ml)
Tigecycline disk zone diameter (mm)

Figures 3 and 4. Scattergram plots of tigecycline BMD MIC results vs. ET MIC values for all Enterobacteriaceae and *S. aureus* isolates tested from the Asia-Pacific region as part of the SENTRY Program.

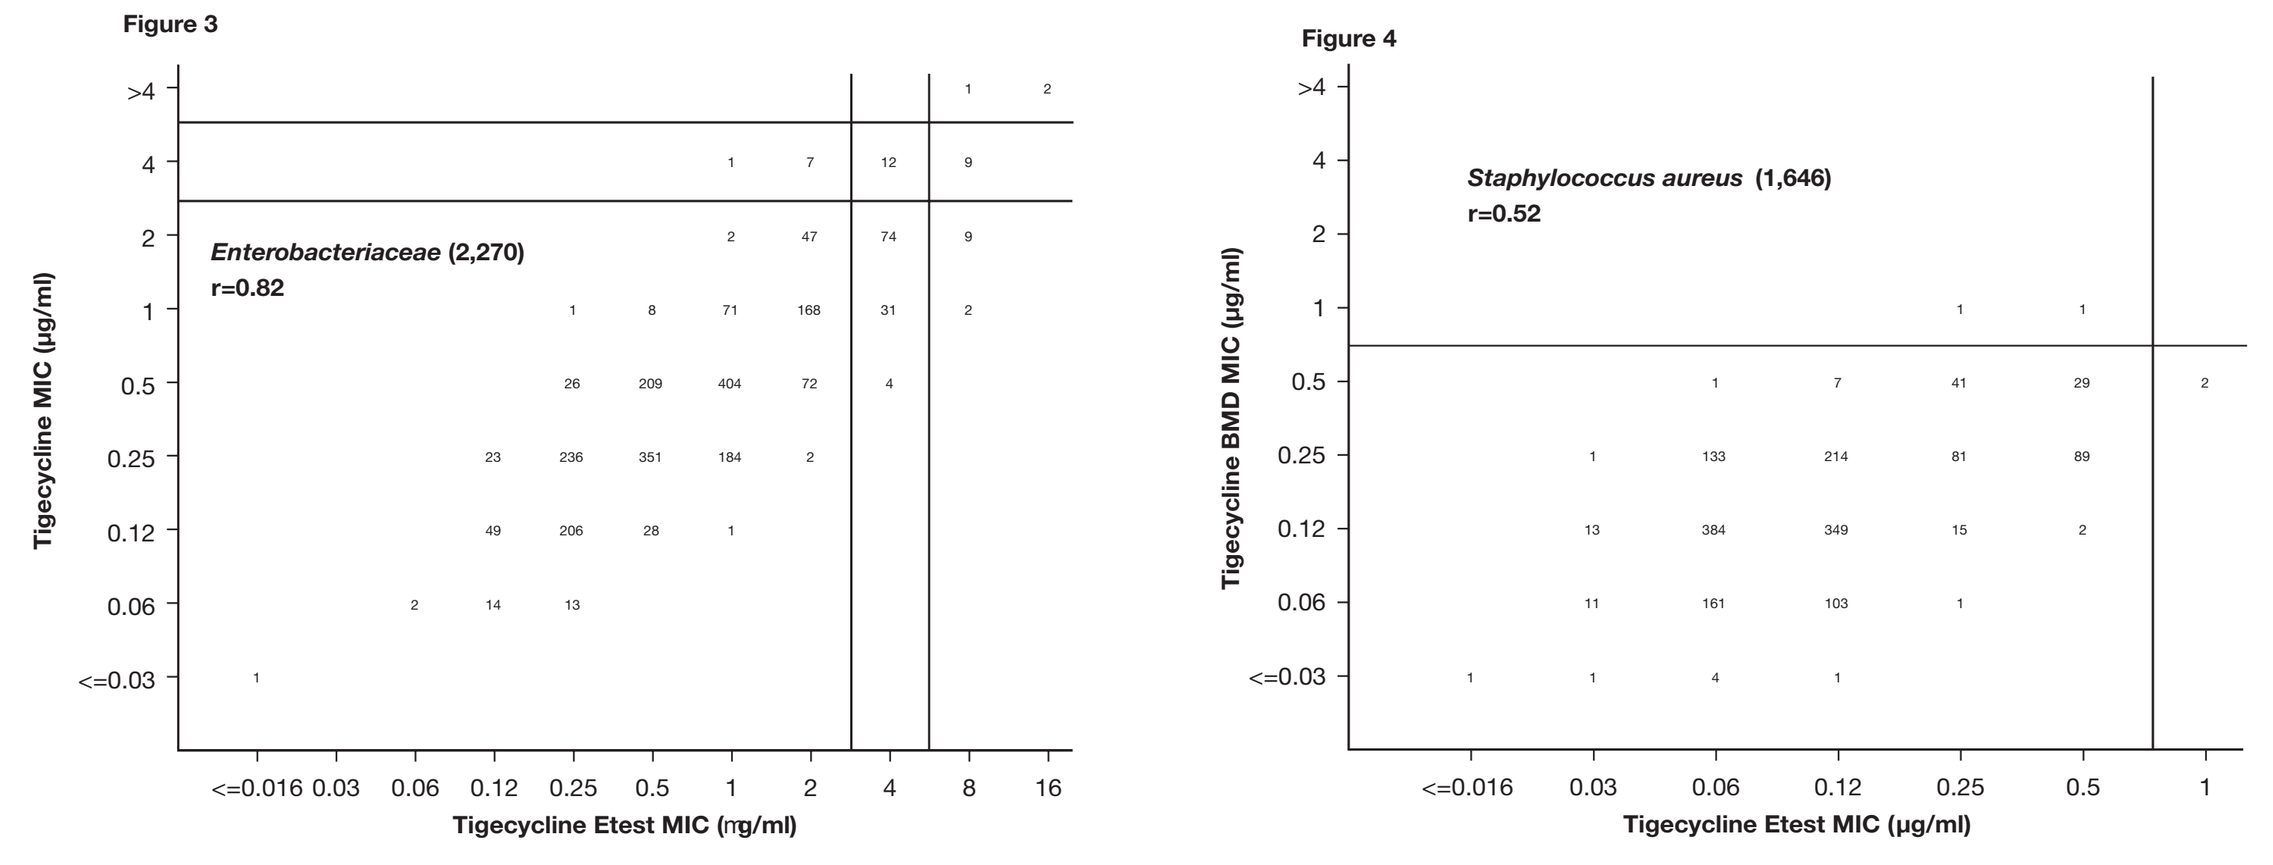

Figure 3
Enterobacteriaceae (2,270)
r=0.82
Tigecycline MIC (µg/ml)
Tigecycline Etest MIC (ηg/ml)
Figure 4
Staphylococcus aureus (1,646)
r=0.52
Tigecycline BMD MIC (µg/ml)
Tigecycline Etest MIC (µg/ml)

- ET MIC vs. BMD MIC results were also plotted with correlation coefficients highest for Enterobacteriaceae (r=0.82), followed by coagulase-negative staphylococci (0.71), viridans group streptococci (0.64), ß-haemolytic streptococci (0.59), *S. aureus* (0.52), enterococci (0.36) and *S. pneumoniae* (0.12; example Figures 3 and 4). A trend toward higher ET MICs was noted (Figure 3) for Enterobacteriaceae tests.

CONCLUSIONS

- The inter-method agreement between BMD, DD and ET susceptibility methods for tigecycline performed well for the Gram-positive pathogen groups analyzed.

- The current US-FDA breakpoints for tigecycline disk diffusion susceptibility for the Enterobacteriaceae isolates as compared to CLSI BMD, appear to require modification to reduce the high minor error rate.

- Tigecycline ET MIC results demonstrated a slight trend toward higher values for the Enterobacteriaceae and streptococci groups, but lower MIC results for some other pathogens groups. However, overall the agreement to BMD was 88.5% ± 1 $\log_2$ dilution step (Table 2).

- Accurate susceptibility testing methods and interpretative criteria are critical for the proper utilization of tigecycline for the treatment of indicated infections and species.