E-292

Activity of Tigecycline Tested against Clinical Pathogens Collected in Korea, the Philippines, Singapore and Thailand (2006)

JD TURNIDGE, JM BELL, RN JONES, M DOWZICKY

Women's and Children's Hospital, Adelaide, Australia; JMI Laboratories, North Liberty, IA, USA; Wyeth Pharmaceuticals, Collegeville, PA, USA

ICAAC 2007
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370, fax 319.665.3371
ronald-jones@jmilabs.com

AMENDED ABSTRACT

Background:

As part of the SENTRY Program (2006) we evaluated the activity of tigecycline (TGC), a new glycylcycline, tested against recent bacterial isolates from four uncommonly monitored countries in the Asia-Pacific region.

Methods:

Clinically significant patient isolates from medical centres in Korea (KOR) (n=3), the Philippines (PHL) (n=2), Singapore (SIN) (n=1), and Thailand (THI) (n=2) were collected. All organisms were tested against TGC using CLSI methods and validated broth microdilution panels with concurrent acceptable quality controls. Interpretative criteria were those approved by the US-FDA for each indicated species or genus. *Acinetobacter* breakpoints were ≤2/≥8 µg/ml per Jones et al. (2007).

Results:

A total of 1,439 isolates were evaluated. TGC was highly active against the top 10 non-pseudomonal pathogens which comprised 80.5% of all isolates tested. *E. coli* and *Klebsiella* spp. with extended spectrum ß-lactamases (ESBL) remained TGC-susceptible (S). More than 16% of enterococci were vancomycin-resistant (R), and one VISA strain was observed. Only one oxacillin (OXA)-R *S. aureus* from KOR was TGC-non-susceptible.

Conclusions:

TGC has retained good activity against commonly isolated pathogens in KOR, PHI, SIN and THI, including ESBL, MRSA, and VISA isolates. TGC should be a promising treatment modality in these four nations.

INTRODUCTION

Tigecycline is the first glycylcycline for clinical use and possesses a broad range of activity against major Gram-positive and -negative bacterial pathogens. Its major asset is its ability to evade acquired efflux and target-mediated resistances to common tetracyclines. Multi-drug resistance (MDR) is very common in the APAC region in many important pathogens including *Acinetobacter* spp., ESBL-producing Enterobacteriaceae, methicillin-resistant *Staphylococcus aureus* (MRSA), and vancomycin-resistant enterococci (VRE) are common. We evaluated the activity of tigecycline against recent bacterial isolates from four uncommonly monitored countries in the Asia-Pacific region.

MATERIALS AND METHODS

Bacterial isolates

- Non-duplicate clinically significant patient isolates were submitted from medical centers in Korea (n=3), the Philippines (n=2), Singapore (n=1), and Thailand (n=2).
- Species identification of all isolates was confirmed in a central laboratory (Women's and Children's Hospital, Adelaide, Australia) using reference methodologies.

Susceptibility Tests

- Isolates were tested against tigecycline using validated broth microdilution MIC panels with cation-adjusted Mueller-Hinton broth (TREK Diagnostics, Cleveland, OH). Testing, incubation and MIC interpretation were performed using the manufacturer's recommendations and/or reference Clinical and Laboratory Standards Institute (CLSI; formerly the NCCLS) methods (2006) and interpretative criteria (2007).
- MIC tests were performed in cation-adjusted Mueller-Hinton broth (with the addition of 2-5% lysed horse blood for testing of streptococci).
- Quality control (QC) strains utilized included *Escherichia coli* ATCC 25922 and 35218, *Pseudomonas aeruginosa* ATCC 27853, *S. aureus* ATCC 29213 and *Streptococcus pneumoniae* ATCC 49619; all MIC QC results were within CLSI specified ranges.

Analysis

- Data were analysed for MIC_{50}, MIC_{90} and percentage susceptible and resistant according to US-FDA tigecycline package insert interpretive criteria (2005).
- Analyses were performed by species and resistance subsets.
- Enterobacteriaceae with elevated MIC values (≥2 µg/ml) for ceftazidime and/or ceftriaxone and/or aztreonam were considered as extended-spectrum ß-lactamase (ESBL)-producing phenotypes.
- *Acinetobacter* spp., with imipenem or meropenem MIC ≥8 µg/ml; and Enterobacteriaceae with imipenem or meropenem a MIC at ≥2 µg/ml, were screened for metallo-ß-lactamase (MBL) enzymes and OXA-23, -24, and -58 enzymes.
- Enterobacteriaceae with an ertapenem MIC at ≥1 µg/ml were screened for KPC-type carbapenemases.

SELECTED REFERENCES

1. Clinical and Laboratory Standards Institute. (2006). *M7-A7, Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard - seventh edition*. Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute. (2007). *M100-S17, Performance standards for antimicrobial susceptibility testing, 17th informational supplement*. Wayne, PA: CLSI.
3. Jones RN, Ferraro MJ, Reller LB, Schreckenberger PC, Swenson JM, Sader HS (2007). Multicenter studies of tigecycline disk diffusion susceptibility results for *Acinetobacter* spp. *J Clin Microbiol* 45: 227-230.

ACKNOWLEDGEMENT

This study was supported by a grant from Wyeth Pharmaceuticals.

RESULTS

- A total of 1,439 (783 Gram-negative and 656 Gram-positive) isolates were received with the top 10 pathogens for each country shown in Table 1.
- The prevalence of screen-positive ESBL strains was extremely high among *E. coli* (29.8%; range 18%, [the Philippines] to 47% [Thailand]) and *K. pneumoniae* (33.3%, range 15% [Singapore, the Philippines] to 46% [Thailand]).
- Tigecycline MIC distributions and MIC_{50}/MIC_{90} against the most common Gram-negative species is found in Table 2. Apart from the Proteae, all of Enterobacteriaceae, including screen-positive ESBL *E. coli* and *Klebsiella pneumoniae* strains, had tigecycline MICs at ≤2 µg/ml (Table 2).
- Observed shifts in modal tigecycline MIC values in both *E. coli* and *Klebsiella* spp. were linked to higher rates of reduced susceptibility to minocycline (Figures 1-2).
- For *Acinetobacter* spp., MIC values ranged from ≤0.03 to 4 µg/ml, but 98.4% were inhibited at ≤2 µg/ml including the 75% of strains with presumptive carbapenemases. Two *A. baumannii*, from one site in Thailand, had tigecycline MIC values of 4 µg/ml. Both strains contained OXA -24 and -58 enyzmes, and one stain also had an OXA-23-like enzyme.
- Modal tigecycline MIC values for enterococci and staphylococci were only 0.12 µg/ml (Table 3) and 99.8% of strains were inhibited at tigecycline MIC of ≤0.5 µg/ml. One MRSA isolate from Korea had a tigecycline MIC of 1 µg/ml.
- A high number of enterococci (16.5%) from Korea contained a *vanA* gene. Tigecycline remained effective against these strains.
- Despite very high rates of penicillin-non-susceptibility (55.9%) in *S. pneumoniae* from these countries, all strains were inhibited by tigecycline at ≤0.06 µg/ml.
- The presence of tetracycline resistance determinants resulted in minimal elevations of tigecycline MIC results, but were most noticeable in *S.aureus* (Figure 3).

Table 1. Frequency of occurrence of pathogens from Korea, the Philippines, Singapore and Thailand (2006).

Korea (n=557)	No. tested	the Philippines (n=326)	No. tested	Singapore (n=179)	No. tested	Thailand (377)	No. tested
S. aureus	137	*S. aureus*	76	*S. aureus*	92	*E. coli*	55
Pseudomonas spp.	62	*Klebsiella* spp.	58	*Acinetobacter* spp.	21	*Klebsiella* spp.	55
Enterococcus spp.	56	*E. coli*	50	*E. coli*	11	*S. aureus*	54
E. coli	52	*Pseudomonas* spp.	36	*Enterococcus* spp.	11	*Pseudomonas* spp.	52
Acinetobacter spp.	47	Coagulase-negative staphylococci	17	ß-haemolytic streptococci	7	*Enterococcus* spp.	51
Klebsiella spp.	46	*Enterococcus* spp.	15	*Salmonella* spp.	7	*Acinetobacter* spp.	49
Coagulase-negative staphylococci	28	ß-haemolytic streptococci	14	*Pseudomonas* spp.	6	*Enterobacter* spp.	14
ß-haemolytic streptococci	27	*Salmonella* spp.	14	Coagulase-negative staphylococci	5	ß-haemolytic streptococci	9
Enterobacter spp.	23	*S. pneumonia*	11	Viridans group streptococci	4	*Salmonella* spp.	5
S. pneumonia	18	*Acinetobacter* spp.	6	*Klebsiella* spp.	3	*Proteus mirabilis*	5

Table 2. Activity of tigecycline against Gram-negative pathogens from Korea, the Philippines, Singapore and Thailand (2006).

	MIC (µg/ml)		Number of isolates inhibited at MIC (µg/ml):									% by category:[a]	
Organism (number tested)	50%	90%	≤0.03	0.06	0.12	0.25	0.5	1	2	4	>4	Susceptible	Resistant
Enterobacteriaceae (455)	0.25	1		9	90	195	95	44	18	4		99.1	0.0
Escherichia coli (168)	0.12	0.25		9	76	69	14					100.0	0.0
ESBL screen-negative (118)	0.12	0.25		8	60	46	4					100.0	0.0
ESBL screen-positive (50)	0.25	0.5		1	16	23	10					100.0	0.0
Klebsiella pneumoniae (150)	0.5	1			2	70	51	23	4			100.0	0.0
ESBL screen-negative (100)	0.25	1			2	57	26	12	3			100.0	0.0
ESBL screen-positive (50)	0.5	1				13	25	11	1			100.0	0.0
Enterobacter spp. (44)	0.5	1				21	16	4	3			100.0	0.0
Acinetobacter spp. (123)[b]	0.5	1	2	4	15	14	35	45	6	2		98.4[b]	0.0
carbapenem-susceptible (32)	0.12	0.5	2	4	13	7	4	1	1			100.0[b]	0.0
carbapenem-non-susceptible (91)	1	1			2	7	31	44	5	2		97.8[b]	0.0
Class B/D-positive[c] (75)	1	1			2	7	25	36	3	2		97.3[b]	0.0

a. US-FDA package insert breakpoints (2005) applied; *Acinetobacter* breakpoints were ≤2/≥8 µg/ml per Jones et al. (2007).
b. No criteria for this category has been proposed, but the Enterobacteriaceae breakpoint found in the US-FDA product package insert was applied.
c. IMP-4 (n=1); VIM-2 (n=1); OXA-23, -24, or -58-types (n=73).

Table 3. Activity of tigecycline against Gram-positive pathogens from Korea, the Philippines, Singapore and Thailand (2006).

	MIC (µg/ml)		Number of isolates inhibited at MIC (µg/ml):								% by category:[a]	
Organism (number tested)	50%	90%	≤0.03	0.06	0.12	0.25	0.5	1	2	≥4	Susceptible	Resistant
Staphylococcus aureus (359)	0.25	0.5	1	23	155	128	51	1			99.7	-[b]
oxacillin-susceptible (130)	0.12	0.25		17	75	38					100.0	-
oxacillin-resistant (229)	0.25	0.5	1	6	80	90	51	1			99.6	-
Enterococcus spp. (133)	0.12	0.25	3	32	71	27					100.0	-
vancomycin-susceptible (112)			2	22	61	27					100.0	-
vancomycin-resistant[c] (21)			1	10	10						100.0	-
ß-haemolytic *Streptococcus* spp. (57)	≤0.03	0.06	49	6	1	1					100.0	-
Streptococcus pneumoniae (34)	≤0.03	≤0.03	33	1							100.0	-
Coagulase-negative staphylococci (50)	0.12	0.25	1	6	25	16	2				100.0	-

a. US-FDA package insert breakpoints (2005) applied. b. No criteria for this category has been proposed. c. *vanA E. faecalis* (n=1) and *vanA E. faecium* (n=20).

Figure 1. Tigecycline MIC distribution vs. minocycline resistance in *Escherichia coli* (n=168).

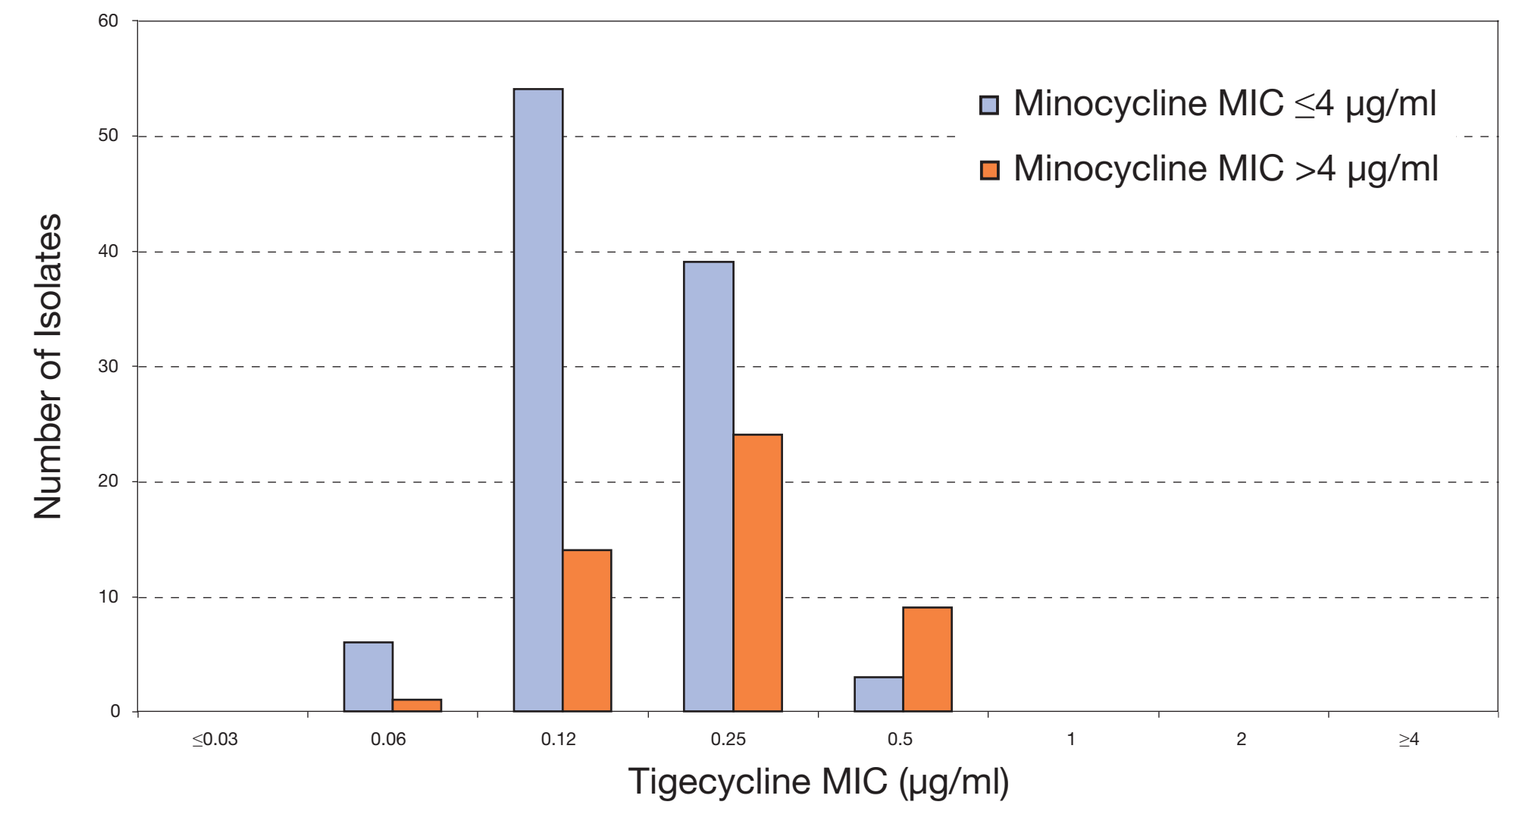

Minocycline MIC ≤4 µg/ml
Minocycline MIC >4 µg/ml
Number of Isolates
Tigecycline MIC (µg/ml)

Figure 2. Tigecycline MIC distribution vs. minocycline resistance in *Klebsiella* spp. (n=162).

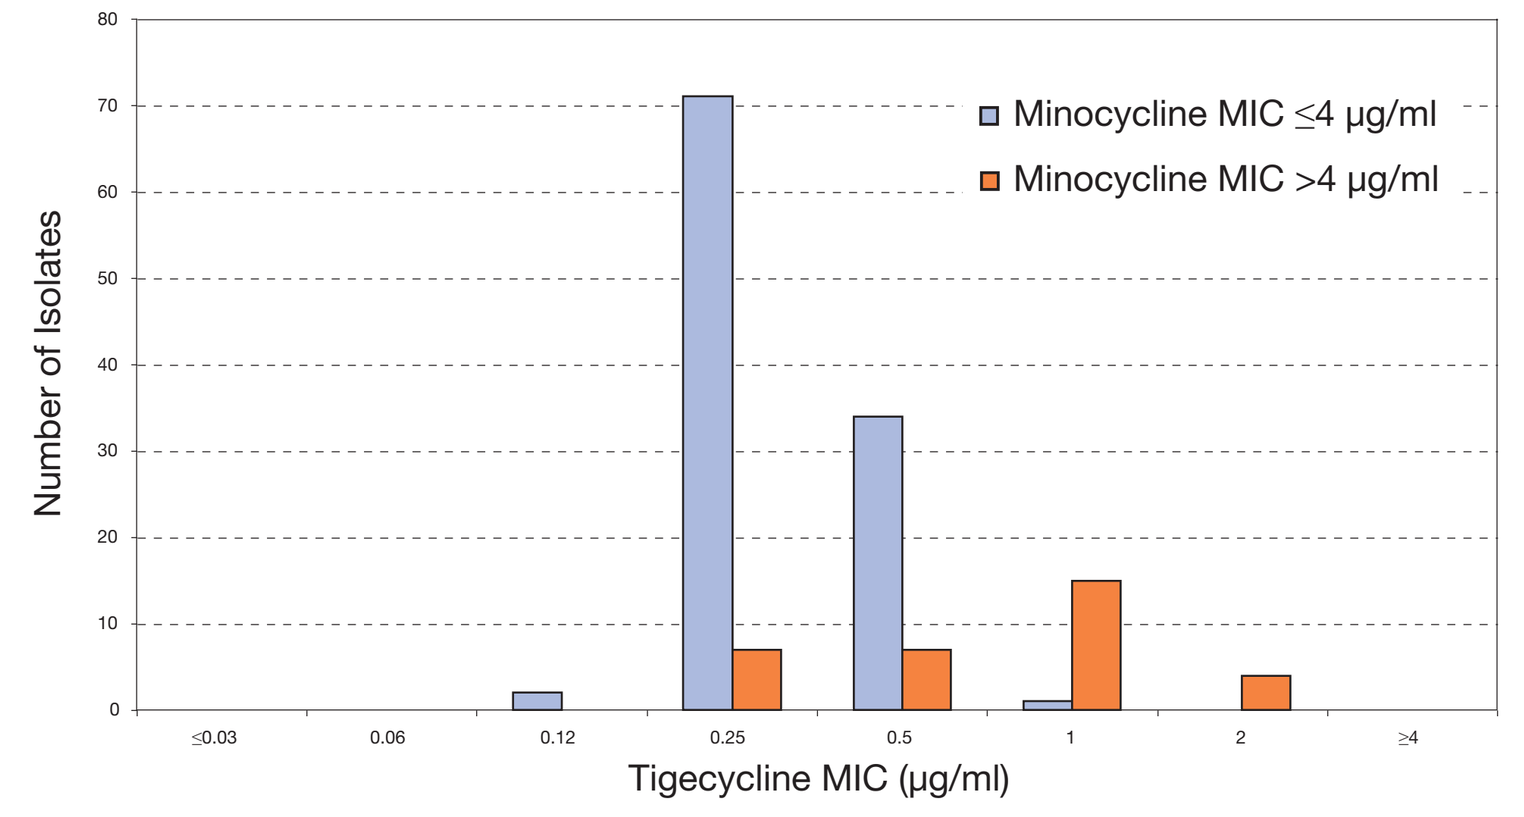

Minocycline MIC ≤4 µg/ml
Minocycline MIC >4 µg/ml
Number of Isolates
Tigecycline MIC (µg/ml)

Figure 3. Tigecycline MIC distribution vs. tetracycline resistance in *Staphylococcus aureus* (n=359).

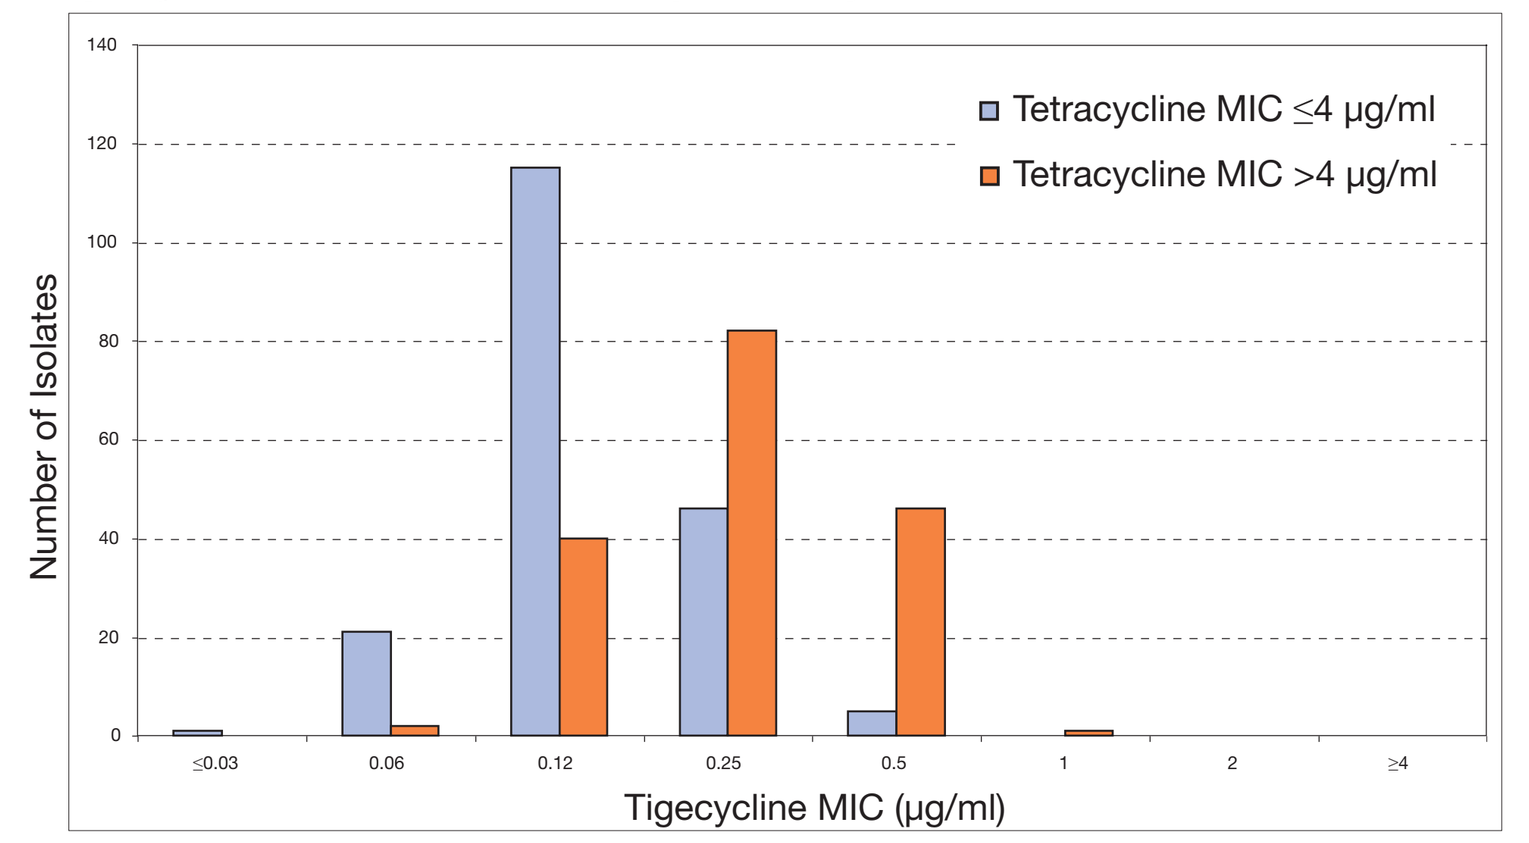

Tetracycline MIC ≤4 µg/ml
Tetracycline MIC >4 µg/ml
Number of Isolates
Tigecycline MIC (µg/ml)

CONCLUSIONS

- Tigecycline appears to be effective in vitro against recent (2006) clinical isolates from Korea, the Philippines, Singapore and Thailand, including prevalent multi-drug resistant strains and those harbouring resistances to important classes, such as third-generation cephalosporins, carbapenems, glycopeptides and anti-staphylococcal penicillins.