D-1526

Determination of Disk Diffusion and MIC Quality Control Ranges for BC-3781 using CLSI Multi-Laboratory M23-A3 Study Design

J.E. ROSS[1], R.N. JONES[1], H.S. SADER[1], Z. IVEZIC SCHOENFELD[2]

[1]JMI Laboratories, North Liberty, IA, [2]Nabriva Therapeutics AG, Vienna, Austria

Nabriva Therapeutics AG
Leberstrasse 20
1110 Vienna, Austria
P: +43-1-74093-0
E: office@nabriva.com
www.nabriva.com

ABSTRACT

Background: We developed disk diffusion (DD) and MIC CLSI quality control (QC) guidelines for an investigational pleuromutilin, BC-3781, against four QC organisms.

Methods: These studies followed the CLSI M23-A3 (2008), M02-A10 (2009) and M07-A8 (2009). *S. aureus* (SA) ATCC 25923 (DD), ATCC 29213 (MIC), *H. influenzae* (HI) ATCC 49247 and *S. pneumoniae* (SPN) ATCC 49619 were tested with 3 or more media lots and 3 control antimicrobials. For DD, sites used 3 agar lots, generating 3 DD diameters for 10 replicates (1,440 values); while broth microdilution (BMD) generated 40 MIC results (720 MIC values). 3 agents were used as controls in DD testing (clindamycin [CL], azithromycin [AZ] and linezolid [LZ]). BMD control agents included pleuromutilins, macrolides and fluoroquinolones.

Results: The table lists proposed DD and MIC QC ranges for BC-3781. Modal MIC values (% of total) observed were: SA ATCC 29213 at 0.12 µg/mL (90.8%), HI ATCC 49247 at 1 µg/mL (69.5%) and SPN ATCC 49619 at 0.12 µg/mL (49.0%). Inter-laboratory MIC variations occurred with HI resulting in exclusion of one laboratory from the final analysis. Trailing MIC endpoints were also noted for HI. With SPN, one laboratory had a significantly lower mode and was excluded. Only one control agent MIC value was outside of published ranges, providing acceptable internal quality assurance. No significant variations occurred between media or disk lots.

Conclusions: Proposed BMD MIC and DD QC ranges established here will assist clinical or reference laboratories in the testing of clinical isolates and facilitate the regulatory review process of BC-3781. Intermethod discords should be very unusual events.

QC organism	Proposed QC ranges for BC-3781: MIC (µg/mL;% in range)	DD (mm;% in range)
H. influenzae ATCC 49247	0.5-2 (94.3)[a]	22-28 (96.0)
S. aureus ATCC 25923	NA	26-32 (97.4)
S. aureus ATCC 29213	0.06-0.25 (100.0)	NA
S. pneumoniae ATCC 49619	0.06-0.5 (98.6)[a]	19-27 (99.3)

[a] One laboratory was excluded from analysis, *H. influenzae* QC range for MIC tests was less than the target ≥ 95%
NA = Not Applicable

INTRODUCTION

The pleuromutilin class was discovered from an edible mushroom, *Pleurotus mutilus*, which has a unique mode of action that involves inhibition of bacterial protein synthesis by binding to the prokaryotic ribosome. Pleuromutlins have no target-specific cross-resistances but tiamulin, a semi-synthetic pleuromutilin, has shown some reduced susceptibility due to gene mutations that encode the 23S rRNA.

BC-3781, a pleuromutilin from Nabriva Therapeutics AG (Vienna, Austria), is in clinical development for intravenous and/or oral treatment of acute bacterial skin and skin structure infections (ABSSSI) and community acquired bacterial pneumonia (CABP).

BC-3781 exhibits excellent activity against clinical pathogens identified in SSSI and CABP, including methicillin-resistant *Staphylococcus aureus* (MRSA). The activity for BC-3781 is not adversely influenced by resistance to methicillin among staphylococci or vancomycin among enterococci. This report describes results from a multi-laboratory trial designed to establish BC-3781 quality control (QC) ranges for disk diffusion and broth microdilution MIC methods and used study design criteria as published in the Clinical and Laboratory Standards Institute (CLSI) M23-A3 document.

MATERIALS & METHODS

An eight-laboratory study group was recruited for the development of MIC and disk diffusion zone diameter QC guidelines for BC-3781. Each laboratory followed a protocol based on CLSI document M23-A3 specifications, as well as the M07-A8 method for broth microdilution antimicrobial testing and M02-A10 method for disk diffusion susceptibility testing.

The MIC portion of the study utilized frozen-form, reference broth microdilution panels prepared by TREK Diagnostics (Cleveland, OH). The panels contained three medium lots of cation-adjusted Mueller-Hinton broth (Oxoid, Hampshire, United Kingdom; BD, Sparks, MD; Difco, Detroit MI). The same three lots of Mueller-Hinton broth supplemented with 2-5% lysed horse blood or prepared as Haemophilus Test Medium (HTM) were utilized. Azithromycin, retapamulin and levofloxacin were tested as control agents. Each laboratory tested *Staphylococcus aureus* ATCC 29213, *Haemophilus influenzae* ATCC 49247 and *Streptococcus pneumoniae* ATCC 49619, and generated a total of 705 MIC results. Of the control agent results, 99.7% were within CLSI published guidelines. Colony counts were performed from the broth microdilution trays by subculturing in a quantitative manner onto drug-free solid media. The counts ranged from 1.0×10^5 to 8.6×10^5 CFU/ml with an average of 4.0×10^5 CFU/ml. The disk diffusion portion of the study utilized three different medium lots of commercially-prepared Mueller-Hinton agar, three lots of HTM agar and three lots of Mueller-Hinton agar supplemented with 5% sheep blood (Remel, Lenexa, KS; Hardy Diagnostics, Santa Monica, CA; BBL, Sparks, MD). Three different lots of BC-3781 (20 µg) disks were used versus each QC strain (Mast Group lot #257108 and #257109; BioRad lot #9L0011). Single lots of azithromycin, clindamycin and linezolid disks (BD lot #9118096, lot #9187750 and lot #9225172) were applied as internal control agents. A total of 1,674 control zone diameter results were produced and 99.9-100.0% of reported results were within the CLSI QC ranges as published in document M100-S20.

The MIC and disk diffusion zone diameter results were tabulated and compared by intra- and inter-laboratory analysis to determine potentially unacceptable technical variations. Broth or agar medium and disk lots were also compared to determine variations among manufacturers.

RESULTS

- Figure 1 shows *S. aureus* ATCC 29213 against BC-3781 with a clear mode at 0.12 µg/mL and a three doubling dilution range of 0.06-0.25 µg/mL (100.0% of results within proposed range).
- *H. influenzae* ATCC 49247 MIC results for BC-3781 are shown in Figure 2, with a proposed three dilution QC range. The range for BC-3781 of 0.5 to 2 µg/mL only included 94.3% of all results after excluding one laboratory (H).
- *S. pneumoniae* ATCC 49619 has a proposed MIC QC range of 0.06-0.5 µg/mL for BC-3781 (Figure 3) which includes 98.6% of results excluding one outlier laboratory (H).
- Table 1 reports DD QC ranges for BC-3781 against *S. aureus* ATCC 25923 showing a seven mm range (26-32 mm; 97.4%).
- The DD QC ranges results for BC-3781 and *H. influenzae* ATCC 49247 are shown in Table 2 where a range of 22-28 mm (96.0%) is proposed.
- *S. pneumoniae* ATCC 49619 DD ranges for BC-3781 are presented as 19-27 mm (99.3%; Table 3).
- No significant medium or disk lot variations were noted. Control agents tested provided acceptable internal controls, with each antimicrobial having >99% of all values within the CLSI published ranges.
- These proposed QC ranges for disk diffusion and BMD were presented to the CLSI QC working group in June 2010 and all proposed ranges were approved by the AST subcommittee as stated here.

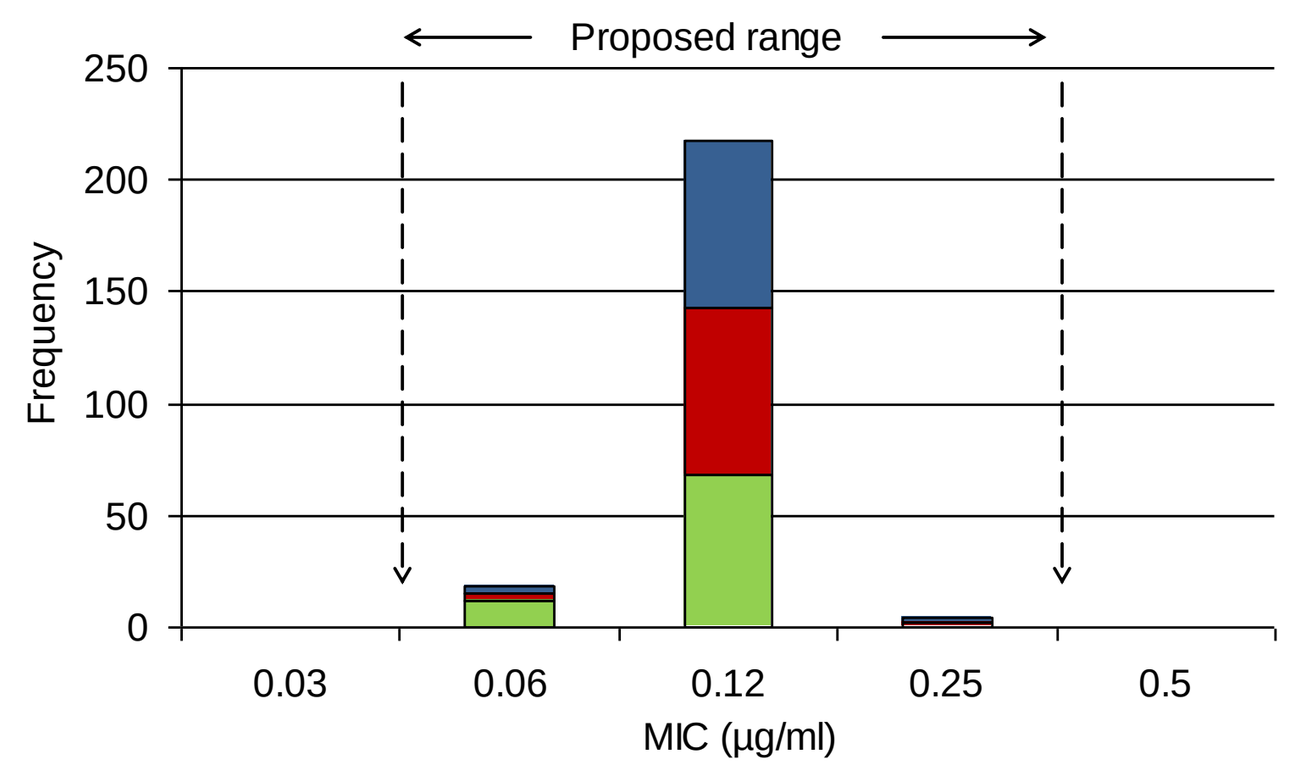

Proposed range
250
200
150
100
50
0
Frequency
0.03
0.06
0.12
0.25
0.5
MIC (µg/ml)
Medium Lot A
Medium Lot B
Medium Lot C

Figure 1. BC-3781 MIC distribution for *S. aureus* ATCC 29213

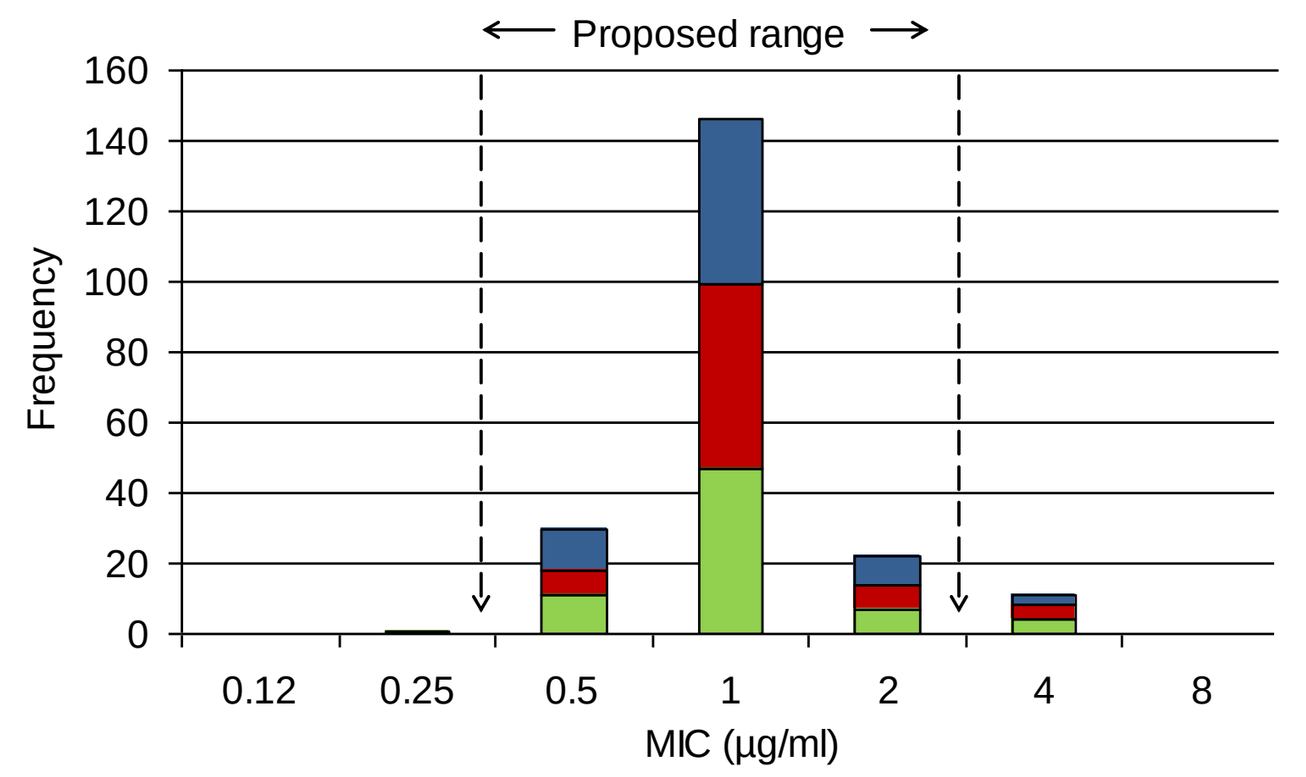

Proposed range
160
140
120
100
80
60
40
20
0
Frequency
0.12
0.25
0.5
1
2
4
8
MIC (µg/ml)
Medium Lot A
Medium Lot B
Medium Lot C

Figure 2. BC-3781 MIC distribution for *H. influenzae* ATCC 49247 (excluding lab H)

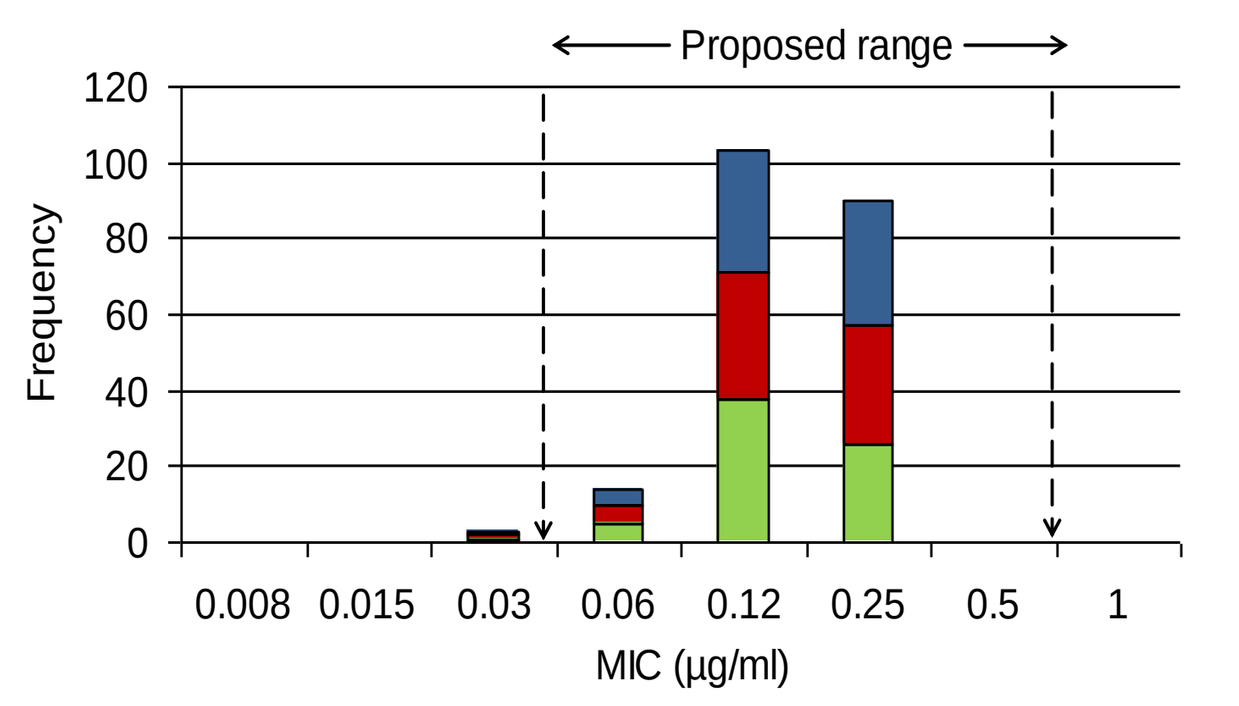

Proposed range
120
100
80
60
40
20
0
Frequency
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
MIC (µg/ml)
Medium Lot A
Medium Lot B
Medium Lot C

Figure 3. BC-3781 MIC distribution for *S. pneumoniae* ATCC 49619 (excluding lab H)

Table 1. Inter- and intra-laboratory comparisons of the BC-3781 zone diameter results versus *S. aureus* ATCC 25923 for an eight medical center protocol meeting the study design guidelines found in CLSI M23-A3 (2008)

Zone Ø (mm)	Disk lot A	Disk lot B	Disk lot C	Occurrences by lot A	B	C	Laboratory code (occurrences): A	B	C	D	E	F	G	H	Total
22															
23															
24			1							1					1
25			1						1	0					1
26	10	2	6		6	12			5	6		7			18[a]
27	25	28	18	11	13	47		1	12	20	10	28			71[a]
28	61	55	68	58	44	82	10	5	44	31	19	30	19	26	184[a]
29	44	27	50	54	49	48	33	5	21	13	20	13	21	25	152[a]
30	58	67	63	71	82	35	37	19	4	13	38	6	41	30	188[a]
31	28	18	21	29	29	9	10	25	2	5	3	5	8	9	67[a]
32	7	8	7	11	7	4		18	1	1		1	1		22[a]
33	5	5	4	6	6	2		14							14
34	1		1		2			2							2
35	1				1			1							1
Total	240	240	240	240	240	240	90	90	90	90	90	90	90	90	720
Median	29	29	29	29	30	28	30	31	28	28	29	28	30	29	29
Mode	28	30	28	30	30	28	30	31	28	28	30	28	30	30	30
Geomean	29.1	29.1	29.0	29.4	29.4	28.4	29.5	31.1	28.2	28.2	29.0	28.0	29.4	29.2	29.1
Range	10	8	11	7	10	8	4	9	8	9	5	7	5	4	12

[a] 97.4% of results are in proposed QC range of 26-32 mm.

Table 2. Inter- and intra-laboratory comparisons of the BC-3781 zone diameter results versus *H. influenzae* ATCC 49247 for an eight medical center protocol meeting the study design guidelines found in CLSI M23-A3 (2008)

Zone Ø (mm)	Disk lot A	Disk lot B	Disk lot C	Occurrences by lot A	B	C	Laboratory code (occurrences) A	B	C	D	E	F	G	H	Total
19															
20	3	3			6			1	5						6
21	5	6	10		21			13	8						21
22	10	8	11	7	22			8	11	3	2	4		1	29[a]
23	22	29	30	36	40	5		18	25	8	6	3		21	81[a]
24	55	46	45	78	37	31	12	27	24	12	9	19	8	35	146[a]
25	59	59	63	60	52	69	40	17	15	7	5	38	30	29	181[a]
26	54	57	57	41	47	80	36	6	2	32	32	22	34	4	168[a]
27	22	18	21	11	9	41	2			22	21	4	12		61[a]
28	10	13	2	7	6	12				6	13		6		25[a]
29		1	1			2					2				2
Total	240	240	240	240	240	240	90	90	90	90	90	90	90	90	720
Median	25	25	25	24	24	26	25	24	23	26	26	25	26	24	25
Mode	25	25	25	24	25	26	25	24	23	26	26	25	26	24	25
Geomean	24.8	24.8	24.7	24.6	24.0	25.7	25.3	23.4	23.2	25.6	26.0	24.9	25.7	24.1	24.8
Range	9	10	9	7	9	7	4	7	7	7	8	6	5	5	10

[a] 96.0% of results are in proposed QC range of 22-28 mm.

Table 3. Inter- and intra-laboratory comparisons of the BC-3781 zone diameter results versus *S. pneumoniae* ATCC 49619 for an eight medical center protocol meeting the study design guidelines found in CLSI M23-A3 (2008)

Zone Ø (mm)	Disk lot A	Disk lot B	Disk lot C	Occurrences by lot A	B	C	Laboratory code (occurrences): A	B	C	D	E	F	G	H	Total
18															
19	2	10	6			18	8			2				8	18[a]
20	23	15	22	6	2	52	15			14			4	27	60[a]
21	24	25	22	12	11	48	7	2		14		1	22	25	71[a]
22	36	29	20	24	23	38	12	18	8	14		7	7	19	85[a]
23	40	55	48	60	45	38	27	18	9	25		19	40	5	143[a]
24	46	30	49	41	51	33	18	23	21	20	22	6	15		125[a]
25	33	42	32	52	42	13	3	24	34	1	37	6	2		107[a]
26	22	27	28	32	45			5	14		29	29			77[a]
27	12	4	7	7	16				4		2	17			23[a]
28		1	4		5							5			5
Total	238	238	238	234	240	240	90	90	90	90	90	90	90	84	714
Median	23	23	24	24	24	22	23	24	25	23	25	26	23	21	23
Mode	24	23	24	23	24	20	23	25	25	23	25	26	23	20	23
Geomean	23.2	23.1	23.3	23.8	24.2	21.7	22.1	23.7	24.5	22.2	25.1	25.0	22.5	20.8	23.2
Range	9	10	10	8	9	7	7	6	6	7	4	8	6	5	10

[a] 99.3% of results are in proposed QC range of 19-27 mm.

CONCLUSIONS

- The results from this collaborative study provide the initial BC-3781 broth microdilution MIC and disk diffusion QC ranges for *S. aureus* ATCC 29213 or 25923, *H. influenzae* ATCC 49247 and *S. pneumoniae* ATCC 49619.
- As this novel pleuromutilin agent progresses through human clinical trials, the susceptibility testing results can be accurately validated by concurrent quality assurance procedures listed here.

SELECTED REFERENCES

- Clinical and Laboratory Standards Institute (2009). *M02-A10. Performance standards for antimicrobial disk susceptibility tests; approved standard -tenth edition*. Wayne, PA: CLSI.
- Clinical and Laboratory Standards Institute (2009). *M07-A8. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard-eighth edition* Wayne, PA: CLSI.
- Clinical and Laboratory Standards Institute (2008). *M23-A3. Development of in vitro susceptibility testing criteria and quality control parameters-third edition*. Wayne, PA: CLSI.
- Clinical and Laboratory Standards Institute (2010). *M100-S20. Performance standards for antimicrobial susceptibility testing. 20th informational supplement* Wayne, PA: CLSI.
- Hunt E. (2000). Pleuromutilins antibiotics. *Drugs of the Future*, 25:1163-1168.
- Jones RN, Pfaller MA, Rhomberg PR, Walter DH. (2002). Tiamulin activity against fastidious and nonfastidious veterinary and human bacterial isolates: Initial development of in vitro susceptibility test methods. *Journal of Clinical Microbiology* 40:461-465.

ACKNOWLEDGEMENTS

The QC study sites and primary investigators were: Massachusetts General Hospital, Boston, MA (Disk study only, M. J. Ferraro, M.D.); JMI Laboratories, North Liberty, IA (R. N. Jones, M.D.); Robert Wood Johnson Medical School, New Brunswick, NJ (M. Weinstein, M.D.); TREK Diagnostics, Cleveland, OH (C. Knapp, M.S.); University of Alberta, Edmonton, Canada (R. Rennie, Ph.D.); Duke University Medical Center, Durham, NC (L. B. Reller, M.D.), Wheaton Franciscan Laboratory, Wauwatosa, WI (Disk study only, E. Munson, Ph.D.), University of Washington Medical Center, Seattle, WA (S. Swanzy, B.S., M.T. [ASCP]); The Cleveland Clinic Foundation, Cleveland, OH (MIC study only, G. Hall, Ph.D.) and University of Texas Medical Center, Houston, TX (MIC study only, A. Wanger, Ph.D.).

50th Interscience Conference on Antimicrobial Agents and Chemotherapy, Boston, MA, September 12-15, 2010