Antimicrobial Activity of Six Seachaid Pharmaceuticals Investigational Compounds Tested against Gram-positive Strains

F-1510

RE MENDES[1], DJ BIEDENBACH[1], PR RHOMBERG[1], RN JONES[1], RG SHERRILL[2], KD JAMES[2]
[1]JMI Laboratories, North Liberty, Iowa, USA; [2]Seachaid Pharmaceuticals, Durham, North Carolina, USA

ICAAC 2012
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
ph. 319.665.3370, fax 319.665.3371
rodrigo-mendes@jmilabs.com

ABSTRACT

Background: Six candidate glycopeptide compounds were screened against recent Gram-positive isolates having well defined and clinically relevant antimicrobial resistance phenotypes.

Methods: A total of 176 unique (2009) Gram-positive clinical strains were selected for this study. Susceptibility testing was performed by CLSI methods (M07-A9) in 96-well plates containing polysorbate-80 (0.002%). MIC interpretations for comparators were based on CLSI (M100-S22) and EUCAST (2012) criteria. MIC results were validated by QC strains.

Results: The compounds showed MIC_{50} values of 0.06 µg/mL when tested against *S. aureus*, except for SP2055 (**Table**). These agents were two- to four-fold more active than daptomycin (MIC_{50}, 0.25 µg/mL) and four- to eight-fold more potent than teicoplanin (MIC_{50}, 0.5 µg/mL) or vancomycin (MIC_{50}, 0.5 µg/mL) against *S. aureus*. SP2055 was less active than other screened agents against *S. epidermidis*. Vancomycin and teicoplanin had MIC_{50} values of 1 and 2 µg/mL, respectively, against *S. epidermidis*. SP2075 and SP2078 were the most active agents tested against *E. faecalis*, while SP2075 showed the lowest MIC_{50} value against *E. faecium*. These compounds were two- to 16-fold more active than vancomycin (MIC_{50}, 1 µg/mL), teicoplanin (MIC_{50}, 0.5 µg/mL) and daptomycin (MIC_{50}, 1 µg/mL) against *E. faecalis*. These tested agents had MIC_{50} values 16- to 64-fold lower than vancomycin (MIC_{50}, 0.5 µg/mL), teicoplanin (MIC_{50}, 1 µg/mL) and daptomycin (MIC_{50}, 2 µg/mL) against *E. faecium*. β-hemolytic streptococci (MIC_{50}, 0.03 – 0.06 µg/mL) were very susceptible to these agents, as were *S. pneumoniae* (MIC_{50}, 0.015 – 0.06 µg/mL).

Conclusions: These investigational glycopeptides showed potent activities against all Gram-positive strains, which were greater than comparators. These *in vitro* data warrant further studies to explore possible therapeutic advantages for these novel agents.

Organism/Group (no. tested)	MIC_{50} values (µg/mL) for compound number: SP2049	SP2055	SP2073	SP2076	SP2075	SP2078
S. aureus (44)	0.06	0.12	0.06	0.06	0.06	0.06
S. epidermidis (43)	0.03	0.12	0.03	0.03	0.03	0.03
E. faecalis (23)	0.12	0.25	0.12	0.12	0.06	0.06
E. faecium (21)	0.06	0.12	0.03	0.03	0.015	0.03
β-hemolytic streptococci (23)	0.03	0.06	0.06	0.03	0.03	0.03
S. pneumoniae (22)	0.03	0.06	0.03	0.03	0.015	0.015

INTRODUCTION

Gram-positive pathogens, including *Staphylococcus aureus*, *Enterococcus faecalis*, *Enterococcus faecium* and *Streptococcus pneumoniae*, cause significant morbidity and mortality in both community and hospital settings. Infections due to multidrug-resistant (MDR) Gram-positive bacteria are increasing in prevalence, and the epidemiology of these organisms and infection presentations are always evolving. Community-acquired (CA)-methicillin-resistant *S. aureus* (MRSA) has emerged as a major pathogen causing skin and skin structure infections (SSSI) worldwide. Among USA hospitals CA-MRSA infections are largely caused by USA300, which has been most recently associated with an increasing incidence of hospital-acquired (HA) infections. Though formerly susceptible to many antimicrobials, USA300 has demonstrated increasing resistance rates to lincosamides and fluoroquinolones. Additionally, community-acquired bacterial pneumonia due to *S. aureus*, including MRSA, has also been on the increase with an associated high mortality rate.

Enterococcus species have become important nosocomial pathogens and currently represent the third most frequent pathogens responsible for HA infections in the USA. *E. faecalis* was previously the most prevalent (90-95%) species among enterococcal pathogens causing nosocomial infections. However, currently increasing occurrences of *E. faecium* have been documented, which is a great concern since *E. faecium* isolates are often more resistant to commonly used antimicrobial agents such as ampicillin, aminoglycosides and glycopeptides. In addition, 70-80% of *E. faecium* clinical isolates in USA hospitals carry the *vanA* gene, while approximately 5% of *E. faecalis* strains are glycopeptide-resistant.

Lastly, β-lactam and MDR *S. pneumoniae* have been increasing in the USA over the past decade. The introduction in the USA of the pediatric 7-valent pneumococcal conjugate vaccine in 2000 resulted in a welcome decline of invasive pneumococcal disease (IPD), along with associated reductions in penicillin and macrolide non-susceptibility among IPD isolates. However, the reduction in antimicrobial resistance was short lived as an increase in antimicrobial resistance was documented among the increasing prevalence of non-vaccine serotypes, in particular serotype 19A. Several agents directed against Gram-positive pathogens have been developed for the treatment of complicated infections in the last decade. However, only linezolid and daptomycin, and more recently ceftaroline and telavancin, have been approved for clinical use. In this study, the *in vitro* activity of six investigational glycopeptide compounds (**Figure 1**) was investigated via screening against a total of 176 contemporary (2009) Gram-positive clinical isolates.

MATERIALS AND METHODS

Bacterial strain collection. Isolates included in this investigation are as follows: *S. aureus* (44; 22 wildtype methicillin-susceptible [MSSA]; 22 methicillin-resistant [MRSA]); *Staphylococcus epidermidis* (43; 21 wildtype methicillin-susceptible; and 22 methicillin-resistant); *E. faecium* (21 wildtype); *E. faecalis* (23 wildtype); *S. pneumoniae* (22 wildtype); β-hemolytic streptococci (23; 11 *Streptococcus pyogenes*, and 12 *Streptococcus agalactiae*).

Antimicrobial susceptibility test methods. Isolates were tested for susceptibility by broth microdilution methods following the Clinical and Laboratory Standards Institute (CLSI; M07-A9, 2012) document. Solvent, diluents and dilution procedures utilized for all Seachaid investigational compounds followed the CLSI recommendations for water-insoluble agents (Table 7B; M100-S22, 2012). Susceptibility testing was performed in cation-adjusted Mueller-Hinton broth (CA-MHB) using customized frozen-form 96-well panels. CA-MHB was supplemented with a surfactant polysorbate-80 (final well concentration, 0.002%). Quality assurance was performed by concurrent testing of CLSI-recommended (M100-S22, 2012) quality control (QC) strains: *E. faecalis* American Type Culture Collection (ATCC) 29212 and *S. aureus* ATCC 29213. Interpretation of MIC results obtained for comparator agents tested against QC and clinical strains was in accordance with published CLSI (M100-S22) criteria.

RESULTS

- Overall, the investigational compounds (SP2049, SP2073, SP2075, SP2076 and SP2078) displayed modal MIC and MIC_{50} results of 0.06 µg/mL when tested against *S. aureus* clinical isolates, except for SP2055 that was slightly less active (modal MIC and MIC_{50}, 0.12 µg/mL; **Table 1**).
- The most active Seachaid compounds (MIC_{50}, 0.06 µg/mL) showed MIC_{50} results four-fold lower than daptomycin (MIC_{50}, 0.25 µg/mL) and eight-fold lower than teicoplanin (MIC_{50}, 0.5 µg/mL) and vancomycin (MIC_{50}, 0.5 µg/mL) when tested against *S. aureus* (**Table 1**). Linezolid displayed modal MIC and MIC_{50} results eight- to 16-fold higher than investigational compounds (**Table 1**).
- Each investigational agent exhibited equivalent MIC_{50} and modal MIC values when tested against MSSA and MRSA strains. One exception was noted for compound SP2075, where MIC_{50} and modal MIC values (0.03 µg/mL for both) against MSSA were two-fold lower compared with MRSA (0.06 µg/mL for both; data not shown).
- Similar to *S. aureus*, SP2055 (modal MIC and MIC_{50} results of 0.12 µg/mL) was less active (four-fold) than other candidate compounds (modal MIC and MIC_{50} results of 0.03 µg/mL) when tested against a collection of *S. epidermidis* clinical isolates (**Table 1**).
- SP2049, SP2073, SP2075, SP2076 and SP2078 displayed MIC_{50} results (0.03 µg/mL) eight-fold lower than daptomycin ($MIC_{50/90}$, 0.25/0.5 µg/mL), 16-fold lower than linezolid ($MIC_{50/90}$, 0.5/1 µg/mL) and 32- to 64-fold lower than vancomycin ($MIC_{50/90}$, 1/2 µg/mL) and teicoplanin ($MIC_{50/90}$, 2/4 µg/mL) when tested against *S. epidermidis* (**Table 1**).
- SP2075 and SP2078 ($MIC_{50/90}$, 0.06/0.12 µg/mL) demonstrated similar antimicrobial activity when tested against wildtype *E. faecalis*, as did SP2049, SP2073 and SP2076 ($MIC_{50/90}$, 0.12/0.12 µg/mL; **Table 2**). $MIC_{50/90}$ results obtained for SP2055 (0.25/0.25 µg/mL) tested against *E. faecalis* were higher than those noted for other candidates (**Table 2**).
- Wildtype *E. faecium* were very susceptible to SP2075 ($MIC_{50/90}$, 0.015/0.03 µg/mL), SP2078 ($MIC_{50/90}$, 0.03/0.03 µg/mL), and SP2073 and SP2076 ($MIC_{50/90}$, 0.03/0.06 µg/mL for both), inhibiting all strains at 0.06 µg/mL (**Table 2**).
- Overall, investigational compounds exhibited MIC_{50} results two- to 16-fold and four- to 128-fold lower than comparators when tested against wildtype collections of *E. faecalis* and *E. faecium* clinical isolates (**Table 2**).
- When tested against β-hemolytic streptococci, compounds SP2055 and SP2073 (modal MIC and MIC_{50} results of 0.06 µg/mL for both) were two-fold less active than SP2049, SP2075, SP2076, and SP2078 (all modal MIC and MIC_{50} results of 0.03 µg/mL).
- Candidate compounds exhibited MIC_{50} values two- to four-fold lower than teicoplanin and daptomycin ($MIC_{50/90}$, 0.12/0.25 µg/mL for both), four- to eight-fold lower than vancomycin ($MIC_{50/90}$, 0.25/0.5 µg/mL) and 16- to 32-fold lower than linezolid ($MIC_{50/90}$, 1/1 µg/mL; **Table 3**) when tested against β-hemolytic streptococci.
- *S. pneumoniae* clinical isolates were very susceptible to SP2075 and SP2078 ($MIC_{50/90}$, 0.015/0.03 µg/mL for both), and inhibited by these compounds at ≤0.03 µg/mL (**Table 3**). Other candidate compounds tested were slightly less active when tested against *S. pneumoniae* ($MIC_{50/90}$, 0.03-0.06/0.06-0.12 µg/mL).

Table 1. MIC distribution of Seachaid and comparator compounds when tested against *S. aureus* and *S. epidermidis* strains.

Organism (no. tested) / Compound	MIC (µg/mL) 50%	90%	Number (cumulative %) of isolates inhibited at each MIC (µg/mL)[a] ≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4
S. aureus (44)												
SP2049	0.06	0.12	0(0.0)	1(2.3)	1(4.5)	**37(88.6)**	5(100.0)					
SP2055	0.12	0.25	0(0.0)	0(0.0)	0(0.0)	1(2.3)	**36(84.1)**	7(100.0)				
SP2073	0.06	0.06	0(0.0)	0(0.0)	3(6.8)	**37(90.9)**	4(100.0)					
SP2075	0.06	0.12	0(0.0)	0(0.0)	18(40.9)	**19(84.1)**	7(100.0)					
SP2076	0.06	0.12	0(0.0)	0(0.0)	3(6.8)	**35(86.4)**	6(100.0)					
SP2078	0.06	0.06	0(0.0)	0(0.0)	6(13.6)	**35(93.2)**	3(100.0)					
Vancomycin	0.5	1	NT[b]	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	**30(68.2)**	14(100.0)		
Teicoplanin	0.5	0.5	NT	NT	0(0.0)	0(0.0)	0(0.0)	15(34.1)	**26(93.2)**	3(100.0)		
Daptomycin	0.25	0.5	NT	NT	0(0.0)	0(0.0)	1(2.3)	**31(72.7)**	11(97.7)	1(100.0)		
Linezolid	1	1	NT	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	2(4.5)	**40(95.5)**	2(100.0)	
S. epidermidis (43)												
SP2049	0.03	0.06	0(0.0)	3(7.0)	**29(74.4)**	11(100.0)						
SP2055	0.12	0.12	0(0.0)	0(0.0)	1(2.3)	17(41.9)	**24(97.7)**	1(100.0)				
SP2073	0.03	0.03	1(2.3)	19(46.5)	**22(97.7)**	1(100.0)						
SP2075	0.03	0.06	0(0.0)	7(16.3)	**27(79.1)**	9(100.0)						
SP2076	0.03	0.06	0(0.0)	1(2.3)	**25(60.5)**	17(100.0)						
SP2078	0.03	0.06	0(0.0)	2(4.7)	**22(55.8)**	19(100.0)						
Vancomycin	1	2	NT	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	0(0.0)	**30(69.8)**	12(97.7)	1(100.0)
Teicoplanin	2	4	NT	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	2(4.7)	**12(32.6)**	**12(60.5)**	13(90.7)
Daptomycin	0.25	0.5	NT	NT	0(0.0)	0(0.0)	0(0.0)	**30(69.8)**	13(100.0)			
Linezolid	0.5	1	NT	NT	0(0.0)	0(0.0)	0(0.0)	2(4.6)	**33(81.4)**	8(100.0)		

a. Modal MIC values are in bold.
b. Concentration not tested.

Table 2. MIC distribution of Seachaid and comparator compounds when tested against *E. faecalis* and *E. faecium*.

Organism (no. tested) / Compound	MIC (µg/mL) 50%	90%	Number (cumulative %) of isolates inhibited at each MIC (µg/mL)[a] ≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2
E. faecalis (23)											
SP2049	0.12	0.12	0(0.0)	0(0.0)	1(4.4)	4(21.7)	**17(95.7)**	1(100.0)			
SP2055	0.25	0.25	0(0.0)	0(0.0)	0(0.0)	0(0.0)	1(4.4)	**20(91.3)**	2(100.0)		
SP2073	0.12	0.12	0(0.0)	0(0.0)	0(0.0)	10(43.5)	**13(100.0)**				
SP2075	0.06	0.12	0(0.0)	0(0.0)	4(17.4)	**11(65.2)**	7(95.7)	1(100.0)			
SP2076	0.12	0.12	0(0.0)	0(0.0)	1(4.4)	1(8.8)	**21(100.0)**				
SP2078	0.06	0.12	0(0.0)	0(0.0)	1(4.4)	**12(56.5)**	10(100.0)				
Vancomycin	1	2	NT[b]	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	7(30.4)	**11(78.3)**	5(100.0)
Teicoplanin	0.5	0.5	NT	NT	0(0.0)	0(0.0)	0(0.0)	10(43.5)	**13(100.0)**		
Daptomycin	1	2	NT	NT	0(0.0)	0(0.0)	1(4.4)	1(8.8)	6(34.8)	**12(87.0)**	3(100.0)
Linezolid	1	1	NT	NT	0(0.0)	0(0.0)	0(0.0)	1(4.4)	6(30.4)	**16(100.0)**	
E. faecium (21)											
SP2049	0.06	0.06	0(0.0)	1(4.8)	8(42.9)	**11(95.2)**	1(100.0)				
SP2055	0.12	0.12	0(0.0)	0(0.0)	1(4.8)	5(28.6)	**14(95.2)**	1(100.0)			
SP2073	0.03	0.06	1(4.8)	1(9.6)	**14(76.2)**	5(100.0)					
SP2075	0.015	0.03	4(19.0)	**9(61.9)**	8(100.0)						
SP2076	0.03	0.06	0(0.0)	2(9.5)	**10(57.1)**	9(100.0)					
SP2078	0.03	0.03	2(9.5)	8(47.6)	**10(95.2)**	1(100.0)					
Vancomycin	0.5	1	NT	NT	0(0.0)	0(0.0)	0(0.0)	1(4.8)	**14(71.4)**	1 (95.2)	1(100.0)
Teicoplanin	1	1	NT	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	**9(42.9)**	**9(85.7)**	3(100.0)
Daptomycin	2	2	NT	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	0(0.0)	5(23.8)	**16(100.0)**
Linezolid	1	2	NT	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	1(4.8)	**15(76.2)**	5(100.0)

a. Modal MIC values are in bold.
b. Concentration not tested.

Table 3. MIC distribution of Seachaid and comparator compounds when tested against β-hemolytic streptococci and *S. pneumoniae*.

Organism (no. tested) / Compound	MIC (µg/mL) 50%	90%	Number (cumulative %) of isolates inhibited at each MIC (µg/mL)[a] ≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	1
β-hemolytic streptococci (23)										
SP2049	0.03	0.12	0(0.0)	1(4.3)	**19(87.0)**	0(87.0)	3(100.0)			
SP2055	0.06	0.06	0(0.0)	0(0.0)	2(8.7)	**20(95.7)**	1(100.0)			
SP2073	0.06	0.12	0(0.0)	0(0.0)	6(26.1)	**12(78.3)**	4(95.7)	1(100.0)		
SP2075	0.03	0.12	0(0.0)	0(0.0)	**16(69.6)**	4(87.0)	3(100.0)			
SP2076	0.03	0.12	0(0.0)	0(0.0)	**18(78.3)**	2(87.0)	3(100.0)			
SP2078	0.03	0.06	0(0.0)	0(0.0)	**18(78.3)**	4(95.7)	1(100.0)			
Vancomycin	0.25	0.5	NT[b]	NT	0(0.0)	0(0.0)	0(0.0)	**18(78.3)**	5(100.0)	
Teicoplanin	0.12	0.25	NT	NT	0(0.0)	2(8.7)	**15(73.9)**	6(100.0)		
Daptomycin	0.12	0.25	NT	NT	0(0.0)	8(34.8)	5(56.5)	**10(100.0)**		
Linezolid	1	1	NT	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	1(4.3)	**22(100.0)**
S. pneumoniae (22)										
SP2049	0.03	0.06	1(4.5)	2(13.6)	**14(77.3)**	5(100.0)				
SP2055	0.06	0.12	0(0.0)	1(4.5)	3(18.2)	**14(81.8)**	4(100.0)			
SP2073	0.03	0.06	0(0.0)	5(22.7)	**11(72.7)**	6(100.0)				
SP2075	0.015	0.03	4(18.2)	**9(59.1)**	**9(100.0)**					
SP2076	0.03	0.06	0(0.0)	1(4.5)	**13(63.6)**	8(100.0)				
SP2078	0.015	0.03	1(4.5)	**12(59.1)**	9(100.0)					
Vancomycin	0.25	0.5	NT	NT	0(0.0)	1(4.5)	1(9.1)	**16(81.8)**	4(100.0)	
Teicoplanin	0.12	0.12	NT	NT	0(0.0)	5(22.7)	**17(100.0)**			
Daptomycin	0.12	0.25	NT	NT	0(0.0)	0(0.0)	**18(81.8)**	4(100.0)		
Linezolid	0.5	1	NT	NT	0(0.0)	0(0.0)	0(0.0)	0(0.0)	**11(50.0)**	**11(100.0)**

a. Modal MIC values are in bold.
b. Concentration not tested.

Figure 1. Core structure of investigational Seachaid compounds and respective structural differences (circles).

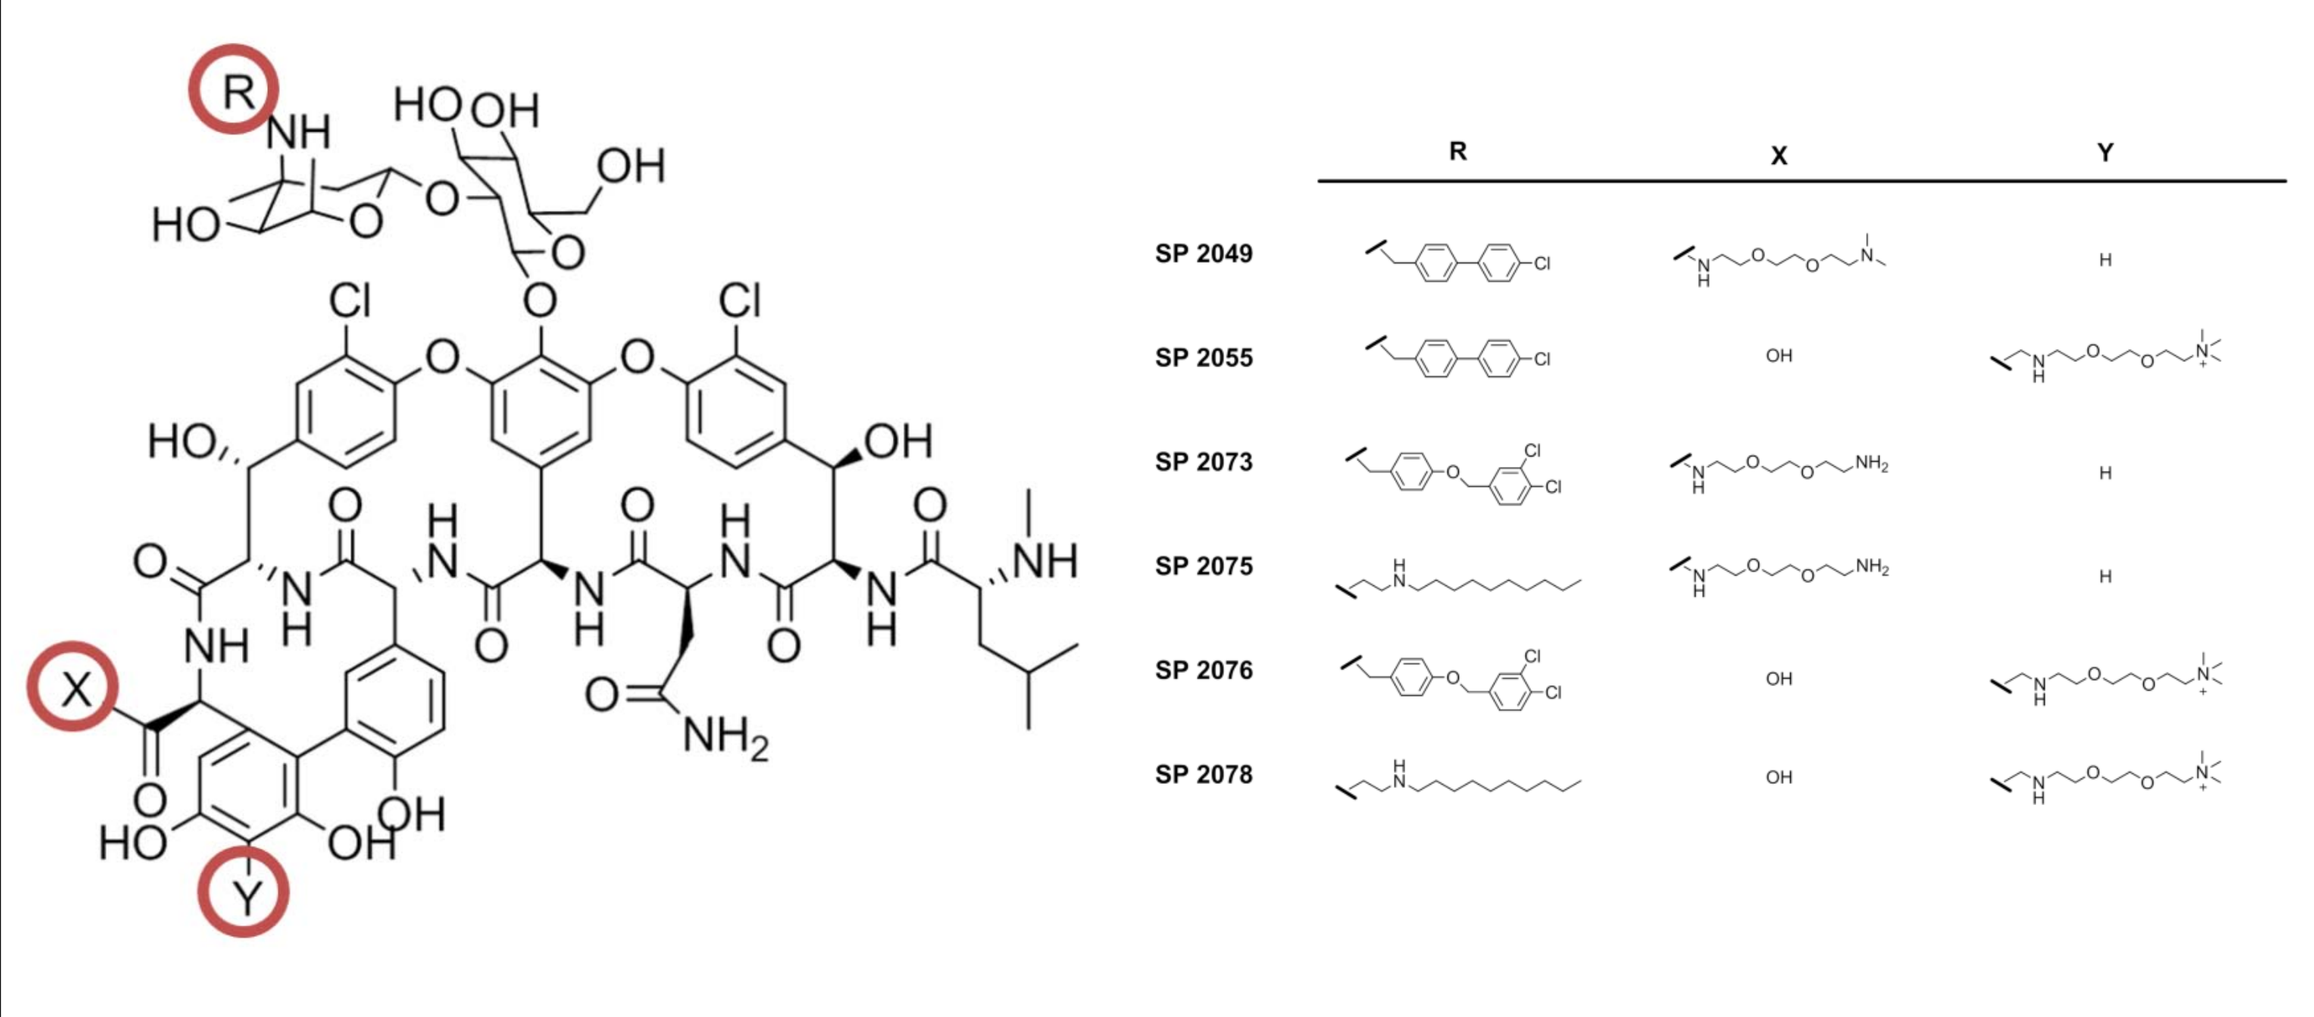
R
X
Y
SP 2049
SP 2055
SP 2073
SP 2075
SP 2076
SP 2078

CONCLUSIONS

- Compounds SP2049, SP2073, SP2075, SP2076 and SP2078 were the more active drugs when tested against *S. aureus* and *S. epidermidis* isolates, inhibiting all strains at ≤0.12 and ≤0.06 µg/mL, respectively. In addition, a methicillin (oxacillin) resistance phenotype did not affect the overall *in vitro* activity of these compounds.
- Among the tested Seachaid investigational compounds, SP2075 and SP2078 demonstrated the lowest MIC_{50} and MIC_{90} results when tested against *E. faecalis*, *E. faecium* and *S. pneumoniae* clinical isolates, while SP2078 was most active against β-hemolytic streptococci strains.
- In summary, screened Seachaid investigational glycopeptide compounds demonstrated *in vitro* activity greater than comparator agents when tested against a contemporary collection of Gram-positive clinical isolates. These *in vitro* data warrant further studies to explore possible therapeutic advantages for these novel agents.

ACKNOWLEDGMENTS

The study experiments were conducted at JMI Laboratories (North Liberty, Iowa) and supported by an Educational/Research Grant from Seachaid Pharmaceuticals, Inc. (Durham, North Carolina). JMI Laboratories received compensation fees for services in relation to preparing the abstract/poster, which was funded by Seachaid Pharmaceuticals.

REFERENCES

1. Ager S, Gould K (2012). Clinical update on linezolid in the treatment of Gram-positive bacterial infections. *Infect Drug Resist* 5: 87-102.
2. Arias CA, Murray BE (2012). The rise of the *Enterococcus*: beyond vancomycin resistance. *Nat Rev Microbiol* 10: 266-278.
3. Clinical and Laboratory Standards Institute (2012). *M07-A9. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: ninth edition*. Wayne, PA: CLSI.
4. Clinical and Laboratory Standards Institute (2012). *M100-S22. Performance standards for antimicrobial susceptibility testing: 22nd informational supplement*. Wayne, PA: CLSI.
5. Gould IM (2011). Clinical activity of anti-Gram-positive agents against methicillin-resistant *Staphylococcus aureus*. *J Antimicrob Chemother* 66 Suppl 4: iv17-iv21.
6. Rice LB (2012). Mechanisms of resistance and clinical relevance of resistance to beta-lactams, glycopeptides, and fluoroquinolones. *Mayo Clin Proc* 87: 198-208.
7. Woodford N, Livermore DM (2009). Infections caused by Gram-positive bacteria: a review of the global challenge. *J Infect* 59 Suppl 1: S4-S16.