ICAAC 2013
C2-539a

Ceftriaxone Non-susceptibility in Emerging (35B) and Persisting (19A, 19F) *Streptococcus pneumoniae* Serotypes in the USA (2011 – 2012): Only Ceftaroline Retains Activity among β-lactams

RE MENDES[1], HS SADER[1], DJ FARRELL[1], D BIEK[2], IA CRITCHLEY[2], RN JONES[1]
[1]JMI Laboratories, North Liberty, Iowa; [2]Cerexa Inc., Oakland, California

Rodrigo E. Mendes, PhD
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
ph. 319.665.3370
fax 319.665.3371
rodrigo-mendes@jmilabs.com

Abstract

Background: The serotype distribution of *S. pneumoniae* in the USA (2011-2012) was studied. 19A, 3 and 35B were found to be most prevalent serotypes among *S. pneumoniae* recovered from hospitalized patients. 19A and 35B comprised the majority (81.1%) of *S. pneumoniae* with elevated ceftriaxone MIC results (≥1 μg/mL). This study reports the serotype and geographic distribution of ceftriaxone non-susceptible *S. pneumoniae*, and ceftaroline activity.

Methods: Serotyping of 1,190 *S. pneumoniae* (63 sites) was performed by *cpsB* sequencing and multiplex PCR. Susceptibility testing applied CLSI methods (M07-A9); MIC interpretations followed CLSI (M100-S23) and EUCAST (2013) criteria.

Results: Ceftaroline had MIC_{90} values 8- to 16-fold lower than ceftriaxone. *S. pneumoniae* exhibiting ceftriaxone MIC values of 1 - >8 μg/mL were mostly (84.3%) 19A, 19F and 35B, while other isolates were associated with 15 serotypes - four strains were nontypeable. The overall ceftriaxone non-susceptibility rates were 8.7 and 21.0% using CLSI and EUCAST criteria, respectively; varying from 4.3 to 80.4% among serotypes 19A, 19F and 35B; ceftaroline non-susceptibility was 0.0% by CLSI and 4.3% by EUCAST criteria. Serogroup 19 with ceftriaxone MIC results of ≥1 μg/mL (37 sites; 26 states) showed markedly decreased susceptibility to amoxicillin/clavulanate (4.7 - 25.0% susceptible), penicillin (10.9 - 37.5%), erythromycin (0.0 - 3.9%), clindamycin (18.8 - 25.0%) and TMP/SMX (0.0 - 0.8%). 87.1% of 35B (37 sites; 25 states) had ceftriaxone MIC values of ≥1 μg/mL, which showed low susceptibility rates for the above comparators (34.2 - 82.4% susceptible), except clindamycin (100.0% susceptible) and penicillin (98.6%). *S. pneumoniae* with ceftriaxone MIC results of ≥1 μg/mL were found in all USA Census regions, but most commonly in Southeastern regions 5 (37.9% of *S. pneumoniae* within this region) and 6 (27.8%). Serogroup 6 also trended towards high ceftriaxone MIC values across 10 states.

Conclusions: Three serotypes were dominantly responsible for ceftriaxone non-susceptibility rates in multidrug-resistant *S. pneumoniae*, regardless of breakpoint used. Ceftaroline demonstrated good activity and retained potency against 100% (CLSI) of ceftriaxone non-susceptible strains. These *in vitro* data show ceftaroline as the only potent β-lactam for treating infections caused by these persisting/emerging serotypes.

Percentage of non-susceptible rates[a] and $MIC_{50/90}$ values (2011-2012 collection):

Serotype (no. tested)	Ceftriaxone (MIC for non-susceptible) CLSI (≥2 μg/mL)	EUCAST (≥1 μg/mL)	$MIC_{50/90}$	Ceftaroline (MIC for non-susceptible) CLSI (≥1 μg/mL)	EUCAST (≥0.5 μg/mL)	$MIC_{50/90}$
19A (165)	49.4[b]	78.0[b]	1/2	0.0	4.3	0.12/0.25
19F (35)	14.3	22.9	≤0.06/2	0.0	2.9	≤0.015/0.12
35B (92)	4.3	80.4[c]	1/1	0.0	0.0	0.12/0.12
All (1,190)	8.7	21.0	≤0.06/1	0.0	0.7	≤0.015/0.12

a. Calculated by CLSI (also USA-FDA) or EUCAST breakpoints for nonmeningitis isolates.
b. Dominantly in USA Census regions 5 (South Atlantic) and 6 (East South Central).
c. Strains from all USA Census regions, and non-S rates ranged 70.0-100.0% per region.

Introduction

Streptococcus pneumoniae is an important pathogen responsible for community-acquired bacterial pneumonia (CABP), bacteremia, meningitis and otitis media. This pathogen has the highest mortality rate among those patients with meningitis, and up to 30% of survivors develop significant long-term sequelae. It has been estimated that *S. pneumoniae* causes 826,000 deaths worldwide in children under five years of age. The older adult population (≥65 years) is also particularly susceptible to pneumococcal diseases and an estimate of 4,000 pneumococcal deaths per year occur in the United States (USA) within this age group. In 2000, the seven-valent pneumococcal conjugate vaccine (PCV7) was introduced in the USA childhood vaccine schedule, followed by the PCV13 in 2010 covering the same seven serotypes found in PCV7 with additional six serotypes, which together account for more than 60% of invasive pneumococcal disease (IPD) cases in the pediatric population.

The Infectious Diseases Society of America (IDSA) guidelines recommend ampicillin or penicillin G therapy for fully immunized infants or school-aged children admitted to a hospital ward with CABP when local epidemiologic data documents a lack of substantial high-level penicillin resistance for IPD. Third-generation parenteral cephalosporins (ceftriaxone or cefotaxime) are recommended for hospitalized infants and children who are not fully immunized, in regions where local epidemiology of IPD strains documents high-level penicillin resistance, or for infants and children with life threatening infection, including those with empyema. Moreover, the prevalence of multidrug-resistant (MDR) *S. pneumoniae* continues to increase. Although the impact of MDR strains on clinical outcomes of IPD remain to be better understood, revisions of empirical therapy may be warranted.

Recently, the serotype distribution of *S. pneumoniae* in the USA (2011-2012) was studied (see poster #C2-531). 19A, 3 and 35B were found to be the most prevalent serotypes among *S. pneumoniae* recovered from sampled patients. In addition, 19A and 35B comprised the majority (81.1%) of *S. pneumoniae* with elevated ceftriaxone MIC values (≥1 μg/mL). In this study, we investigated the serotype and geographic distribution of *S. pneumoniae* recovered after (2011-2012) the introduction of PCV13 that demonstrated elevated MIC results (≥1 μg/mL) for ceftriaxone. Furthermore, the activity of ceftaroline and other comparator agents was quantified when tested against these less susceptible isolates of *S. pneumoniae*.

Methods

Bacterial strains. A total of 1,190 *S. pneumoniae* clinical isolates received as part of the "AWARE" (Assessing Worldwide Antimicrobial Resistance Evaluation) Program, a component of SENTRY Antimicrobial Surveillance Program, from July 2011 through June 2012 were included in this investigation. These isolates were recovered from hospitalized patients in 63 medical centers located in the nine USA Census regions. Isolates were recovered from blood or lower respiratory tract cultures. Bacterial identification was performed by the participating microbiology laboratory and confirmed by the central monitoring laboratory (JMI Laboratories, North Liberty, Iowa, USA). Confirmation of bacterial identification was performed by colony morphology, biochemical algorithms and Vitek®2, as needed. When the bacterial identification was questionable using phenotypic methods or an untypeable serotyping result was obtained by the applied methodology, isolates were subjected to PCR assay for further identification.

Antimicrobial susceptibility profile. Isolates were tested for susceptibility by broth microdilution methods, according to the recommendations of the Clinical and Laboratory Standards Institute (CLSI). MIC results for several anti-Gram-positive agents were obtained using panels manufactured by Thermo Fisher Scientific (formerly TREK Diagnostics Systems/Sensititre; Cleveland, Ohio, USA). Validation of the MIC values was performed by concurrent testing of CLSI-recommended (M100-S23, 2013) quality control (QC) strain *S. pneumoniae* American Type Culture Collection (ATCC) 49619. In addition, the inoculum density was monitored by colony counts to ensure an adequate number of cells for each testing event. MIC interpretations were based on the CLSI M100-S23 (2013) and European Committee on Antimicrobial Susceptibility Testing (EUCAST; 2013) criteria.

Serotyping determination. Isolates were subjected to PCR assays for amplification of the *cpsB* gene. Amplicons were sequenced on both strands and the nucleotide sequences were analyzed using the Lasergene software package (DNASTAR, Madison, Wisconsin, USA). Sequences were compared with others available via Pubmed (http://www.ncbi.nlm.nih.gov/blast/). Due to sequence homology among certain serotypes, those showing nucleotide sequence similarity greater than 99% were grouped (e.g. 9V/9A, 7F/7A, 11A/11D, 15A/15F, 22F/22A, 15B/15C). All isolates determined to be serogroup 6 by PCR and sequencing analysis were subjected to multiplex PCR assays for confirmation and discrimination purposes (between 6A/6B and 6C/6D).

Results

- Overall, ceftriaxone had MIC_{50} and MIC_{90} results of ≤0.06 and 1 μg/mL, respectively. A total of 8.7 and 21.0% of all *S. pneumoniae* isolates were non-susceptible to ceftriaxone according to CLSI (≤1 μg/mL for susceptible, non-meningitis cases) and EUCAST (≤0.5 μg/mL for susceptible) breakpoint criteria (Table 1)
- *S. pneumoniae* exhibiting ceftriaxone MIC values of 1 - >8 μg/mL were predominantly (84.3%; 210/249) 19A, 19F and 35B, while other isolates were associated with 15 other serotypes and four nontypeable strains (Table 1). Ceftaroline ($MIC_{50/90}$, 0.12/0.25 μg/mL) had MIC_{90} values eight-fold lower than ceftriaxone ($MIC_{50/90}$, 1/2 μg/mL) when tested against these isolates with decreased susceptibility to ceftriaxone (Table 2)
- Ceftaroline (96.8 - 100.0% susceptible), levofloxacin (98.8% susceptible), linezolid (100% susceptible) and vancomycin (100% susceptible) demonstrated superior antimicrobial coverage when tested against *S. pneumoniae* isolates with elevated ceftriaxone MIC results (Table 2). Other tested agents showed susceptibility rates at ≤58.6%
- Serogroup 19 represented 83.5% (86/103) and 54.6% (136/249) of isolates that were non-susceptible to ceftriaxone according to CLSI and EUCAST criteria, respectively (Table 1). These isolates showed markedly decreased susceptibility to amoxicillin/clavulanate (5.9% susceptible), penicillin G (12.5% susceptible), erythromycin (3.7% susceptible), clindamycin (19.1 - 19.9% susceptible), cefuroxime (0.0% susceptible) and trimethoprim/sulfamethoxazole (0.7% susceptible) (Table 2)
- The vast majority of 35B isolates (95.7%) were susceptible to ceftriaxone when applying the CLSI breakpoint (≤1 μg/mL for susceptible). In stark contrast, only 19.6% of 35B were considered susceptible when the EUCAST criteria (≤0.5 μg/mL for susceptible were used), since this serotype (76.1%; 70/92) exhibited a ceftriaxone modal MIC result at 1 μg/mL (Table 1). 35B isolates with elevated ceftriaxone MIC values of ≥1 μg/mL showed low susceptibility rates for amoxicillin/clavulanate (51.4% susceptible), cefuroxime (0.0% susceptible), erythromycin (34.2% susceptible) and trimethoprim/ sulfamethoxazole (82.4 - 86.5% susceptible), except for clindamycin (100.0% susceptible) and penicillin G (98.6% susceptible; Table 2)
- *S. pneumoniae* with elevated ceftriaxone MIC results (≥1 μg/mL) were found in all USA Census regions, but most commonly in the South Atlantic (37.9% of *S. pneumoniae* within this region) and East South Central (27.8%) regions. Rates among other regions varied from 11.9% (West North Central) to 24.9% (East North Central; Figure 1). Occurrences of 19A isolates with elevated ceftriaxone MIC results (≥1 μg/mL) varied from 55.6% in West North Central to 92.3% in the East South Central and South Atlantic regions (Figure 1); whereas 35B strains with decreased susceptibility to ceftriaxone were also more prevalent in the East South Central and South Atlantic regions (91.7 - 100.0%).

Table 1. MIC distribution and antimicrobial activity of ceftriaxone when tested against specific serogroups/types of *S. pneumoniae*

Serogroup/type (no. tested)	MIC (μg/mL) 50%	90%	Number (cumulative %) of isolates inhibited at MIC (μg/mL)[a] ≤0.06	0.12	0.25	0.5	1	2	4	8	>8
All (1,187)[b]	≤0.06	1	688 (58.0)	118 (67.9)	67 (73.5)	65 (79.0)	146 (91.3)	88 (98.7)	6 (92.4)	8 (99.9)	1 (100.0)
20 (11)	≤0.06	≤0.06	11 (100.0)								
31 (30)	≤0.06	≤0.06	30 (100.0)								
35F/47F (13)	≤0.06	≤0.06	13 (100.0)								
38/25F/25A (12)	≤0.06	≤0.06	12 (100.0)								
7F/7A (29)	≤0.06	≤0.06	29 (100.0)								
11A/11D (71)	≤0.06	≤0.06	66 (93.0)	3 (97.2)	1 (98.6)	0 (98.6)	0 (98.6)	1 (100.0)			
22F/22A (68)	≤0.06	≤0.06	64 (94.1)	1 (95.6)	0 (95.6)	1 (97.1)	1 (98.5)	0 (98.5)	1 (100.0)		
33F/33A/37 (20)	≤0.06	≤0.06	19 (95.0)	0 (95.0)	0 (95.0)	0 (95.0)	1 (100.0)				
7C/7B/40 (18)	≤0.06	≤0.06	17 (94.4)	1 (100.0)							
9N/9L (34)	≤0.06	≤0.06	33 (97.1)	1 (100.0)							
4 (5)	≤0.06	-	5 (100.0)								
48 (1)	≤0.06	-	1 (100.0)								
13 (9)	≤0.06	-	8 (88.9)	1 (100.0)							
14 (2)	≤0.06	-	1 (50.0)	0 (50.0)	0 (50.0)	0 (50.0)	1 (100.0)				
18(18A/18B/18C/18F) (1)	≤0.06	-	1 (100.0)								
24(24A/24B/24F) (2)	≤0.06	-	1 (50.0)	0 (50.0)	1 (100.0)						
28F/28A (1)	≤0.06	-	1 (100.0)								
16F (22)	≤0.06	0.12	19 (86.4)	1 (90.9)	0 (90.9)	0 (90.9)	1 (95.5)	1 (100.0)			
17F (20)	≤0.06	0.12	16 (80.0)	2 (90.0)	1 (95.0)	0 (95.0)	0 (95.0)	1 (100.0)			
23B (53)	≤0.06	0.12	33 (62.3)	16 (92.5)	1 (94.3)	1 (96.2)	1 (98.1)	1 (100.0)			
12F/12A/44/46 (13)	≤0.06	0.12	11 (84.6)	2 (100.0)							
10A (22)	≤0.06	0.25	19 (86.4)	0 (86.4)	1 (90.9)	0 (90.9)	2 (100.0)				
15B/15C (66)	≤0.06	0.25	41 (62.1)	14 (83.3)	11 (100.0)						
3 (96)	≤0.06	0.25	84 (87.5)	2 (89.6)	9 (99.0)	0 (99.0)	1 (100.0)				
8 (15)	≤0.06	0.25	12 (80.0)	0 (80.0)	2 (93.3)	1 (100.0)					
23A (75)	0.12	0.25	19 (25.3)	41 (80.0)	9 (92.0)	3 (96.0)	2 (98.7)	1 (100.0)			
23F (4)	0.12	-	1 (25.0)	1 (50.0)	0 (50.0)	0 (50.0)	1 (75.0)	1 (100.0)			
34 (16)	≤0.06	0.5	14 (87.5)	0 (87.5)	0 (87.5)	2 (100.0)					
21 (12)	≤0.06	0.5	8 (66.7)	0 (66.7)	1 (75.0)	3 (100.0)					
15A/15F (50)	0.12	0.5	13 (26.0)	20 (66.0)	10 (86.0)	4 (94.0)	3 (100.0)				
9V/9A (5)	1	-	0 (0.0)	0 (0.0)	0 (0.0)	2 (40.0)	1 (60.0)	2 (100.0)			
Nontypeable (11)	0.5	1	4 (36.4)	0 (36.4)	1 (45.5)	2 (63.6)	4 (100.0)				
6C/6D (69)	0.25	1	22 (31.9)	3 (36.2)	10 (50.7)	27 (89.9)	6 (98.6)	0 (98.6)	0 (98.6)	1 (100.0)	
6A/6B (20)	0.25	2	5 (25.0)	2 (35.0)	3 (50.0)	6 (80.0)	1 (85.0)	3 (100.0)			
35B (92)	1	1	7 (7.6)	0 (7.6)	0 (7.6)	11 (19.6)	70 (95.7)	4 (100.0)			
19A (164)	1	2	22 (13.4)	6 (17.1)	6 (20.7)	2 (22.0)	47 (50.6)	70 (93.3)	4 (95.7)	6 (99.4)	1 (100.0)
19F (35)	≤0.06	2	26 (74.3)	1 (77.1)	0 (77.1)	0 (77.1)	3 (85.7)	3 (94.3)	1 (97.1)	1 (100.0)	

a. Red and blue lines represent the CLSI (≤1 μg/mL) and EUCAST (≤0.5 μg/mL) breakpoints for ceftriaxone susceptibility, respectively.
b. Ceftriaxone MIC values were not available for all 1,190 isolates.

Table 2. Activity of ceftaroline and comparator antimicrobial agents when tested against the most common serotypes of *S. pneumoniae* isolates displaying elevated (≥1 μg/mL) ceftriaxone MIC results

Population (number tested)/ Antimicrobial agent	Range	MIC (μg/mL) 50%	90%	% Susceptible/Resistant[a] CLSI	EUCAST
19[b] (136)					
Ceftaroline	0.06 – 0.5	0.12	0.25	100.0 / -	94.1 / 5.9
Penicillin[c]	0.12 – 8	4	8	12.5 / 10.3	- / -
Penicillin[d]	0.12 – 8	4	8	0.0 / 97.1	0.0 / 87.5
Ceftriaxone	1 – >8	2	2	36.8 / 9.6	0.0 / 9.6
Amoxicillin/clavulanate	≤1 – >8	8	>8	5.9 / 90.4	- / -
Cefuroxime	4 – >16	8	16	0.0 / 100.0	0.0 / 100.0
Erythromycin	≤0.12 – >16	>16	>16	3.7 / 96.3	3.7 / 96.3
Clindamycin	≤0.25 – >2	>2	>2	19.1 / 80.1	19.9 / 80.1
Levofloxacin	0.5 – 4	1	1	99.3 / 0.0	99.3 / 0.7
Trimethoprim/sulfamethoxazole	≤0.5 – >4	>4	>4	0.7 / 99.3	0.7 / 99.3
35B (74)					
Ceftaroline	0.03 – 0.25	0.12	0.12	100.0 / -	100.0 / 0.0
Penicillin[c]	1 – 4	2	2	98.6 / 0.0	- / -
Penicillin[d]	1 – 4	2	2	0.0 / 86.3	0.0 / 1.4
Ceftriaxone	1 – 2	1	1	94.6 / 0.0	0.0 / 0.0
Amoxicillin/clavulanate	2 – 8	2	4	51.4 / 1.4	- / -
Cefuroxime	4 – 8	4	4	0.0 / 100.0	0.0 / 100.0
Erythromycin	≤0.12 – >16	8	>16	34.2 / 65.8	34.2 / 65.8
Clindamycin	≤0.25	≤0.25	≤0.25	100.0 / 0.0	100.0 / 0.0
Levofloxacin	0.5 – >4	1	1	98.6 / 1.4	98.6 / 1.4
Trimethoprim/sulfamethoxazole	≤0.5 – >4	≤0.5	>4	82.4 / 12.2	86.5 / 12.2
All (249)					
Ceftaroline	≤0.015 – 0.5	0.12	0.25	100.0 / -	96.8 / 3.2
Penicillin[c]	≤0.06 – 8	4	4	47.8 / 6.1	- / -
Penicillin[d]	≤0.06 – 8	4	4	1.2 / 89.1	1.2 / 52.2
Ceftriaxone	1 – >8	1	2	58.6 / 6.0	0.0 / 6.0
Amoxicillin/clavulanate	≤1 – >8	8	8	30.5 / 51.8	- / -
Erythromycin	≤0.12 – >16	>16	>16	15.3 / 84.7	15.3 / 84.7
Clindamycin	≤0.25 – >2	≤0.25	>2	50.4 / 48.8	51.2 / 48.8
Levofloxacin	0.5 – >4	1	1	98.8 / 0.8	98.8 / 1.2
Trimethoprim/sulfamethoxazole	≤0.5 – >4	>4	>4	29.3 / 67.1	31.3 / 67.1
Vancomycin	≤0.12 – 0.5	0.25	0.5	100.0 / -	100.0 / 0.0
Linezolid	0.25 – 2	0.5	1	100.0 / -	100.0 / 0.0

a. Criteria as published by the CLSI (2013) and EUCAST (2013).
b. Serogroup comprised of eight and 128 isolates of 19F and 19A, respectively.
c. Criteria as published by the CLSI (2013) for 'penicillin parenteral (non-meningitis)'.
d. Criteria as published by the CLSI (2013) for 'penicillin (oral penicillin V)'.

Figure 1. Distribution of *S. pneumoniae* isolates and specific serotypes displaying elevated (≥1 μg/mL) ceftriaxone MIC results within the USA Census regions during 2011-2012

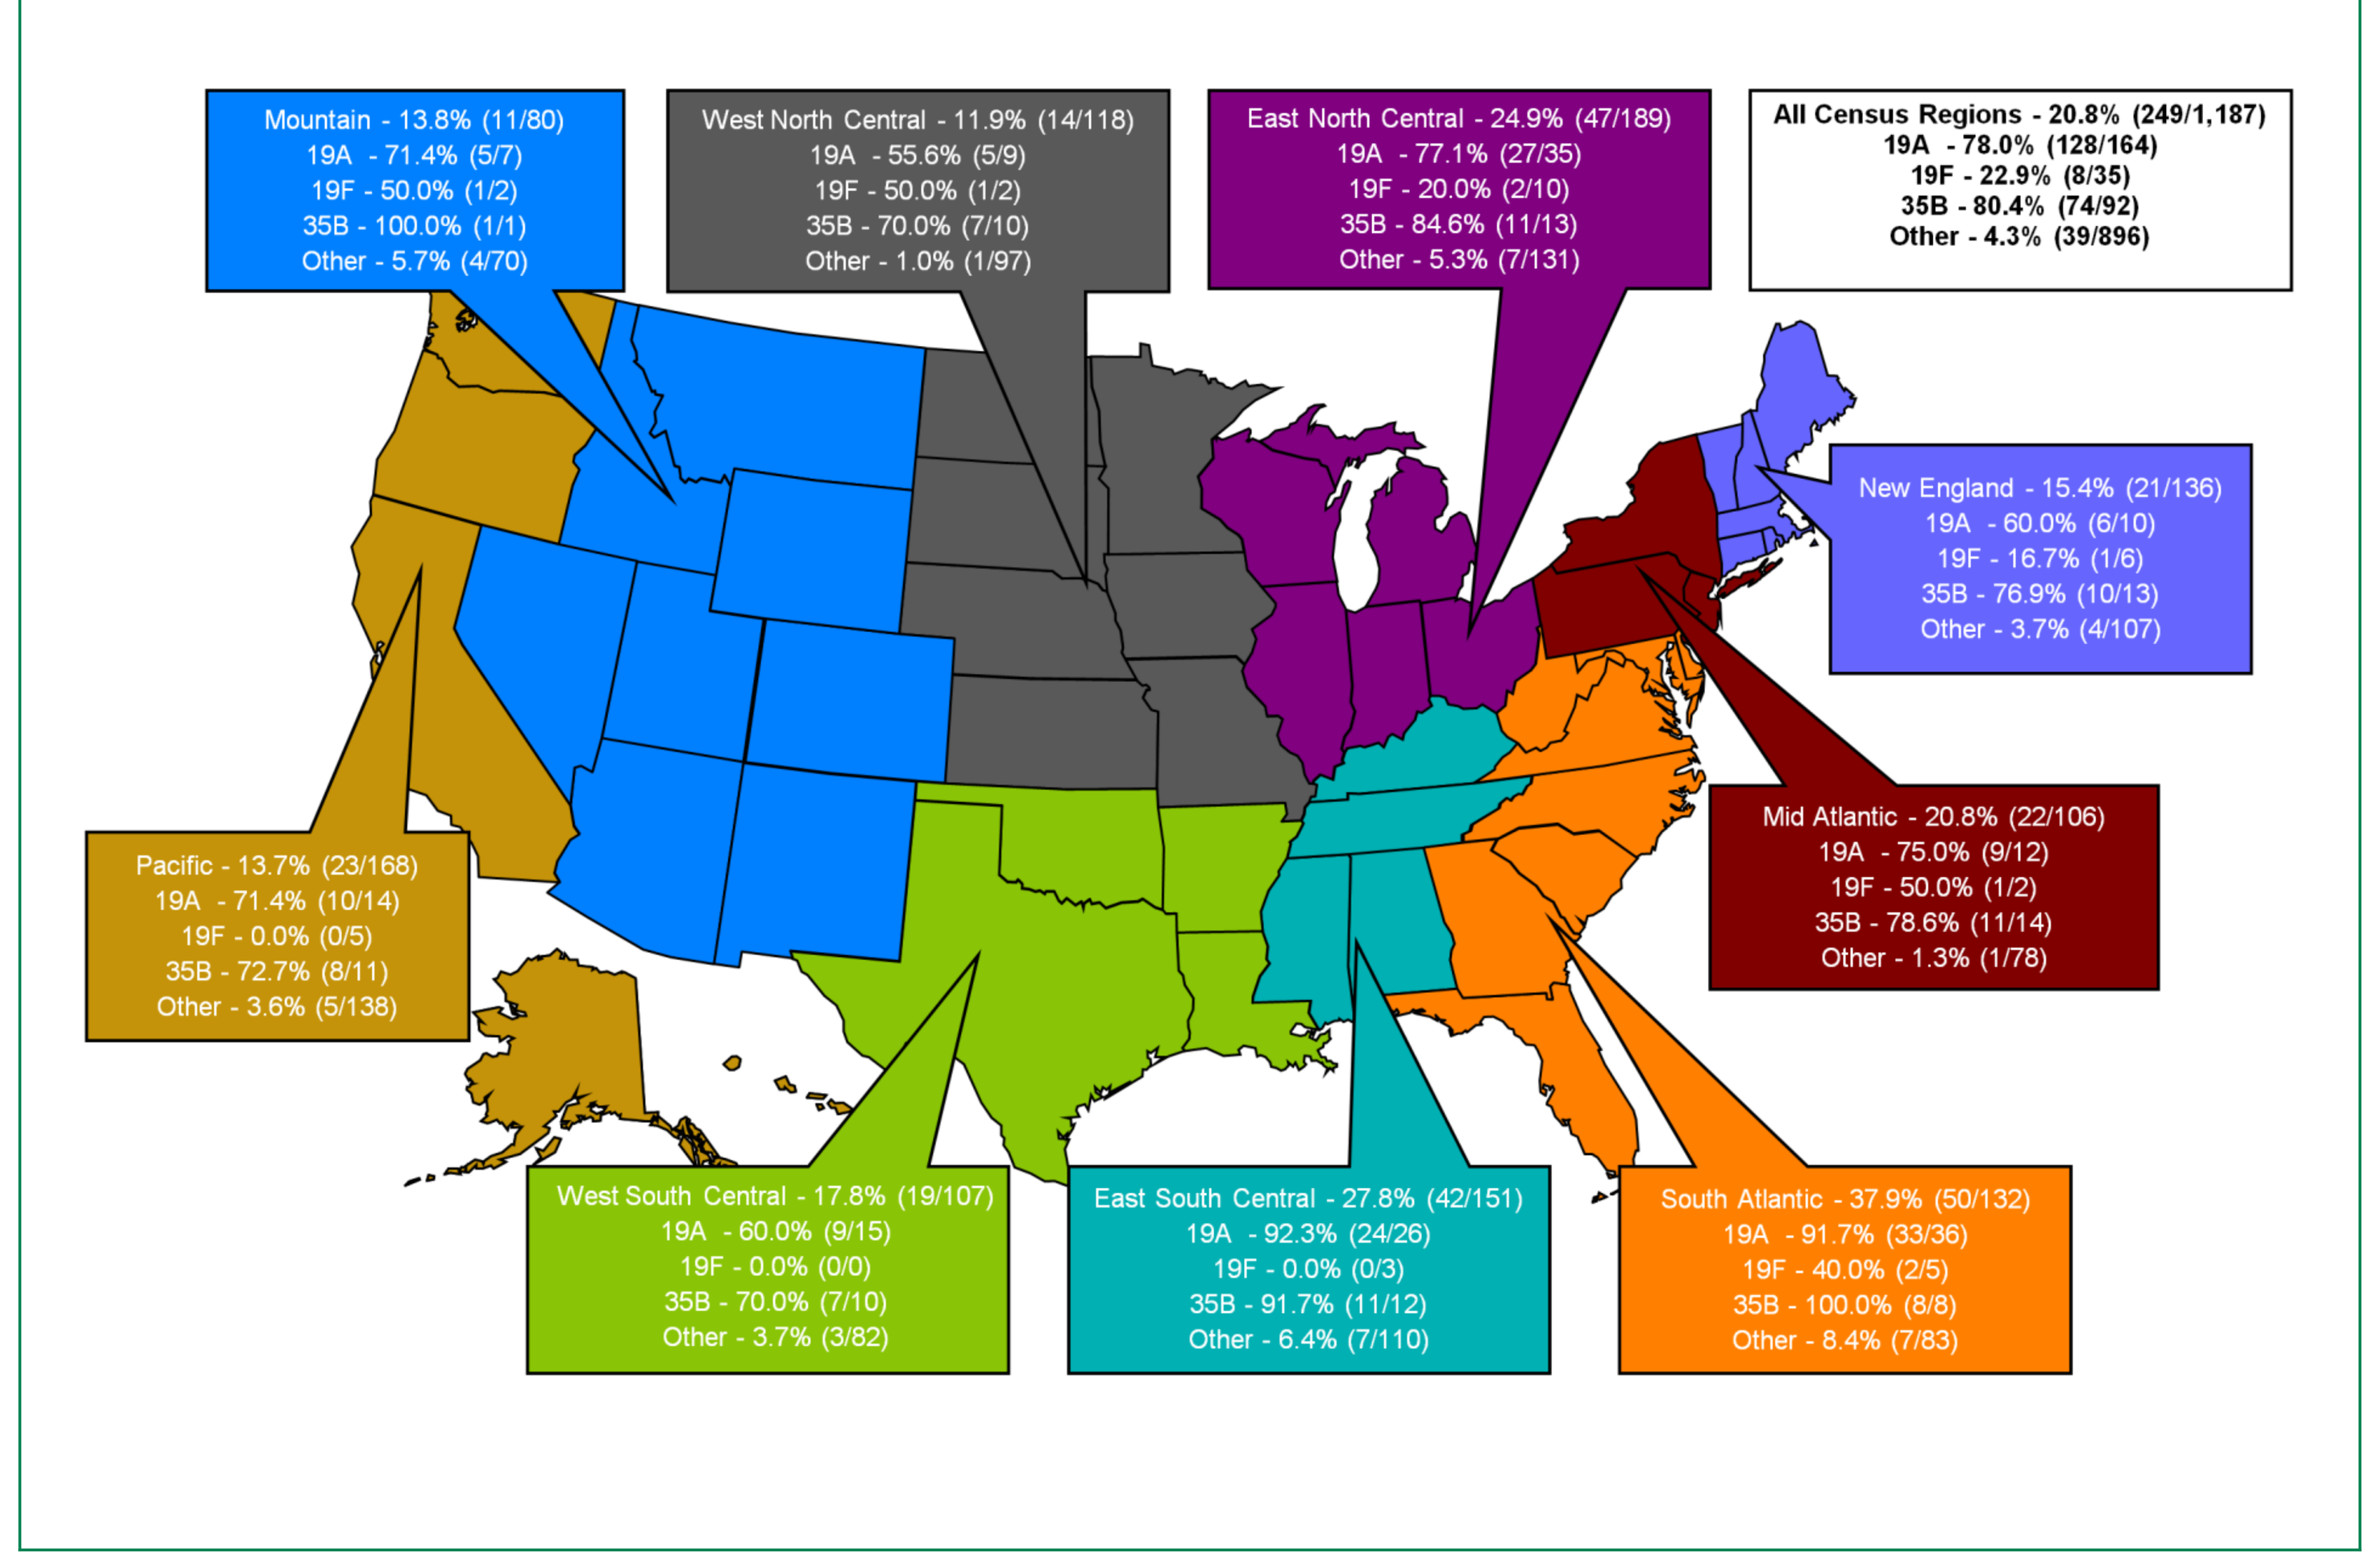
Mountain - 13.8% (11/80)
19A - 71.4% (5/7)
19F - 50.0% (1/2)
35B - 100.0% (1/1)
Other - 5.7% (4/70)
West North Central - 11.9% (14/118)
19A - 55.6% (5/9)
19F - 50.0% (1/2)
35B - 70.0% (7/10)
Other - 1.0% (1/97)
East North Central - 24.9% (47/189)
19A - 77.1% (27/35)
19F - 20.0% (2/10)
35B - 84.6% (11/13)
Other - 5.3% (7/131)
All Census Regions - 20.8% (249/1,187)
19A - 78.0% (128/164)
19F - 22.9% (8/35)
35B - 80.4% (74/92)
Other - 4.3% (39/896)
New England - 15.4% (21/136)
19A - 60.0% (6/10)
19F - 16.7% (1/6)
35B - 76.9% (10/13)
Other - 3.7% (4/107)
Pacific - 13.7% (23/168)
19A - 71.4% (10/14)
19F - 0.0% (0/5)
35B - 72.7% (8/11)
Other - 3.6% (5/138)
Mid Atlantic - 20.8% (22/106)
19A - 75.0% (9/12)
19F - 50.0% (1/2)
35B - 78.6% (11/14)
Other - 1.3% (1/78)
West South Central - 17.8% (19/107)
19A - 60.0% (9/15)
19F - 0.0% (0/0)
35B - 70.0% (7/10)
Other - 3.7% (3/82)
East South Central - 27.8% (42/151)
19A - 92.3% (24/26)
19F - 0.0% (0/3)
35B - 91.7% (11/12)
Other - 6.4% (7/110)
South Atlantic - 37.9% (50/132)
19A - 91.7% (33/36)
19F - 40.0% (2/5)
35B - 100.0% (8/8)
Other - 8.4% (7/83)

Conclusions

- Serotype 19A pneumococcal isolates were most responsible for the elevated ceftriaxone non-susceptibility rates in *S. pneumoniae* when the CLSI breakpoint was applied. However, most 35B isolates demonstrated elevated ceftriaxone MIC results (76.1% at 1 μg/mL) and 80.4% were considered as non-susceptible when applying the EUCAST susceptible breakpoint
- *S. pneumoniae* isolates with significantly decreased susceptibility to ceftriaxone were observed in all USA Census regions, but were most common in the South Atlantic (37.9% of strains within this region) and East South Central (27.8%) regions
- In contrast, ceftaroline demonstrated potent activity ($MIC_{50/90}$, 0.12/0.25 μg/mL) against 100% (CLSI) and 96.8% (EUCAST) of isolates with decreased susceptibility to ceftriaxone. These *in vitro* data suggest that ceftaroline is the most potent β-lactam for treating infections caused by these serotypes and all *S. pneumoniae* sampled in 2011-2012.

References

1. Ardanuy C, Rolo D, Fenoll A, Tarrago D, Calatayud L, Liñares J (2009). Emergence of a multidrug-resistant clone (ST320) among invasive serotype 19A pneumococci in Spain. *J Antimicrob Chemother* 64: 507-510.
2. Clinical and Laboratory Standards Institute (2012). *M07-A9. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: ninth edition*. Wayne, PA: CLSI.
3. Clinical and Laboratory Standards Institute (2013). *M100-S23. Performance standards for antimicrobial susceptibility testing: 23rd informational supplement.* Wayne, PA: CLSI.
4. European Committee on Antimicrobial Susceptibility Testing (2013). Breakpoint tables for interpretation of MICs and zone diameters. Version 3.0, January 2013. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 1, 2013.
5. Hulten KG, Kaplan SL, Lamberth LB, Barson WJ, Romero JR, Lin PL, Bradley JS, Givner LB, Tan TQ, Hoffman JA, Mason EO (2013). Changes in *Streptococcus pneumoniae* serotype 19A invasive infections in children from 1993 to 2011. *J Clin Microbiol* 51: 1294-1297.
6. Jacobs MR, Good CE, Bajaksouzian S, Windau AR (2008). Emergence of *Streptococcus pneumoniae* serotypes 19A, 6C, and 22F and serogroup 15 in Cleveland, Ohio, in relation to introduction of the protein-conjugated pneumococcal vaccine. *Clin Infect Dis* 47: 1388-1395.
7. Bradley JS, Byington CL, Shah SS, Alverson B, Carter ER, Harrison C, Kaplan SL, Mace SE, McCracken GH Jr, Moore MR, St Peter SD, Stockwell JA, Swanson JT, Pediatric Infectious Diseases Society and the Infectious Diseases Society of America (2011). The management of community-acquired pneumonia in infants and children older than 3 months of age: clinical practice guidelines by the Pediatric Infectious Diseases Society and the Infectious Diseases Society of America. *Clin Infect Dis* 53: e25-76.
8. Jones RN, Sader HS, Moet GJ, Farrell DJ (2010). Declining antimicrobial susceptibility of *Streptococcus pneumoniae* in the United States: Report from the SENTRY Antimicrobial Surveillance Program (1998-2009). *Diagn Microbiol Infect Dis* 68: 334-336.
9. Leung MH, Bryson K, Freystatter K, Pichon B, Edwards G, Charalambous BM, Gillespie SH (2012). Sequetyping: serotyping *Streptococcus pneumoniae* by a single PCR sequencing strategy. *J Clin Microbiol* 50: 2419-2427.
10. Pai R, Gertz RE, Beall B (2006). Sequential multiplex PCR approach for determining capsular serotypes of *Streptococcus pneumoniae* isolates. *J Clin Microbiol* 44: 124-131.
11. Low DE (2013). What is the relevance of antimicrobial resistance on the outcome of community-acquired pneumonia caused by *Streptococcus pneumoniae*? (should macrolide monotherapy be used for mild pneumonia?). *Infect Dis Clin North Am* 27: 87-97.
12. Song JH, Dagan R, Klugman KP, Fritzell B (2012). The relationship between pneumococcal serotypes and antibiotic resistance. Vaccine 30 :2728-2737.

Acknowledgments

This study was supported by Cerexa, Inc., a wholly-owned subsidiary of Forest Laboratories, Inc. Forest Laboratories, Inc., was involved in the study design, interpretation of data, and decision to present these results. Forest Laboratories, Inc., had no involvement in the collection and analysis of data.