IDWeek 2014
471

Antimicrobial Activity and Spectrum of Ceftaroline Tested Against Bacterial Isolates Causing Skin and Skin Structure Infections (SSSI) in United States (USA) Medical Centers

HS SADER, RK FLAMM, DJ FARRELL, RN JONES

JMI Laboratories, North Liberty, Iowa, USA

Helio S. Sader, MD, PhD
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
ph. 319.665.3370
fax 319.665.3371
helio-sader@jmilabs.com

Amended Abstract

Background: Ceftaroline (CPT), the active form of CPT fosamil, is a broad-spectrum cephalosporin with bactericidal activity against Gram-positive (GP) organisms, including methicillin-resistant (R) *S. aureus* (MRSA), and common Gram-negative organisms, including wildtype Enterobacteriaceae (ENT).

Methods: 8446 isolates were consecutively collected from patients with SSSI in 149 USA medical centers in 2013. Strains were tested for susceptibility (S) by CLSI broth microdilution against CPT and comparators.

Results: 51.0% of *S. aureus* isolates were MRSA. CPT was very active against methicillin-S *S. aureus* (MSSA; MIC_{90}, 0.25 µg/mL) and MRSA (MIC_{90}, 1 µg/mL). Against MSSA, CPT was 16-, 4- and 4-fold more active than ceftriaxone (CRO), linezolid (LZD) and vancomycin (VAN), respectively; and 17.2% of MSSA were clindamycin (CLI)-R (5.0% constitutive [Con-R] and 12.2% inducible [Ind-R]). MRSA showed high R rates to levofloxacin (LEV; 57.7%) and CLI (25.8%; 17.7% Con-R and 8.1% Ind-R). CPT inhibited all β-hemolytic streptococci (βHS) at ≤0.03 µg/mL (MIC_{90}, ≤0.015 µg/mL), and it was at least 64- and 32-fold more active than LZD and VAN, respectively. CPT was slightly more active against group A compared to other βHS groups. Viridans group streptococci (VGS) were very S to CPT ($MIC_{50/90}$, 0.03/0.06 µg/mL), while 93.0 and 98.8% of strains were PEN-S ($MIC_{50/90}$, ≤0.06/0.25 µg/mL) and CRO-S ($MIC_{50/90}$, 0.25/0.5 µg/mL), respectively. CPT inhibited 81.8% of ENT at ≤0.5 µg/mL. CPT exhibited good activity against non-ESBL-phenotype strains of *Klebsiella* spp. and *E. coli* (MIC_{90}, 0.25 µg/mL for both), but limited activity against ESBL-producing and/or CRO-R strains.

Conclusion: CPT was active against GP and ENT pathogens recently isolated from SSSI in USA medical centers, including MRSA (0.0% R). CPT spectrum against GP was similar to that of LZD and VAN; against ENT, CPT had a spectrum comparable to CRO. CPT appears to be a valuable option for treatment of SSSI, including those caused by MRSA.

	Cumulative % inhibited at CPT MIC (µg/mL):									
Organism (n)	≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8
S. aureus (5182)	<0.1	0.1	0.2	3.7	47.4	73.8	99.4	100.0	-	-
MSSA (2540)	0.1	0.1	0.3	7.5	95.1	100.0	-	-	-	-
MRSA (2642)	-	-	-	0.2	1.6	48.6	98.8	100.0	-	-
βHS (746)	95.3	100.0	-	-	-	-	-	-	-	-
VGS (172)	40.1	83.7	98.8	99.4	99.4	100.0	-	-	-	-
Enterobacteriaceae (2346)	0.3	4.0	28.7	57.6	72.9	81.8	87.2	88.9	90.1	91.2
E. coli (655)	0.9	9.0	42.9	70.7	80.8	87.2	89.2	90.1	91.0	91.9
non-ESBL (582)	1.0	10.1	48.3	79.6	90.4	96.7	99.0	99.8	100.0	-
ESBL (73)	-	-	-	-	4.1	11.0	11.0	12.3	19.2	27.4
Klebsiella spp. (482)	0.2	2.1	29.5	60.2	78.6	84.2	87.1	88.2	88.8	89.8
non-ESBL (420)	0.2	2.4	33.8	69.0	90.2	96.7	99.3	99.8	99.8	99.8
ESBL (62)	-	-	-	-	-	-	4.8	9.7	14.5	22.6

Introduction

Skin and skin structure infection (SSSI) encompasses a wide range of clinical presentations, from mild cases of cellulitis and subcutaneous tissue infections to complicated deep-seated infections with systemic signs of sepsis and the presence of complicating co-morbidities, such as neutropenia, ischemia and/or tissue necrosis. The vast majority of SSSIs are caused by *Staphylococcus aureus* followed by β-hemolytic streptococci, usually Lancefield groups A, C and G, with group B being more common in diabetics and the elderly.

Ceftaroline, the active metabolite of the prodrug ceftaroline fosamil, is a broad-spectrum cephalosporin with in vitro bactericidal activity against resistant Gram-positive organisms, including methicillin (oxacillin)-resistant *Staphylococcus aureus* (MRSA), multidrug-resistant (MDR) strains of *Streptococcus pneumoniae*, and common Gram-negative organisms, including non-ESBL-phenotype Enterobacteriaceae. Ceftaroline fosamil is approved by the United States Food and Drug Administration (USA-FDA) for the treatment of acute bacterial SSSI (ABSSSI) and community-acquired bacterial pneumonia (CABP), and similar indications in Europe and other countries. As part of the Assessing Worldwide Antimicrobial Resistance Evaluation (AWARE) Program, a global ceftaroline surveillance study, we evaluated the activity of ceftaroline tested against a large collection bacterial isolates collected from patients hospitalized with ABSSSI in USA medical centers.

Methods

Organism collection: A total of 8446 clinically significant, non-duplicate isolates were collected from patients hospitalized with ABSSSI in 149 USA medical centers during 2013. The collection includes 6100 Gram-positives (5182 *S. aureus*, 746 β-hemolytic streptococci and 172 viridans group streptococci) and 2346 Enterobacteriaceae. *Escherichia coli* and *Klebsiella* spp. isolates were grouped as "extended-spectrum β-lactamase (ESBL)-phenotype" based on the Clinical and Laboratory Standards Institute (CLSI) screening criteria for ESBL production, i.e. MIC of ≥2 µg/mL for ceftazidime or ceftriaxone or aztreonam (CLSI 2014; M100-S24). Species identification was confirmed when necessary by Matrix-Assisted Laser Desorption Ionization-Time Of Flight Mass Spectrometry (MALDI-TOF MS) using the Bruker Daltonics MALDI Biotyper (Billerica, Massachusetts, USA) by following manufacturer instructions.

Susceptibility testing methods: Broth microdilution tests conducted according to the CLSI methods were performed to determine antimicrobial susceptibility of ceftaroline and comparator antimicrobials used to treat ABSSSI. Validated MIC panels were manufactured by ThermoFisher Scientific (Cleveland, Ohio, USA). *S. aureus* strains were tested in cation-adjusted Mueller-Hinton broth (CA-MHB). β-hemolytic streptococci were tested in CA-MHB supplemented with 2.5-5% lysed horse blood (CLSI 2012; M07-A9).

Quality control: Concurrent quality control (QC) testing was performed to assure proper test conditions and procedures. Susceptibility percentages and validation of QC results were based on CLSI guidelines and susceptibility breakpoints established by the CLSI (M100-S24; CLSI, 2014) were used to determine susceptibility/ resistance rates when available.

Results

- Ceftaroline inhibited 99.4% of *S. aureus* strains at ≤1 µg/mL, which is the susceptible breakpoint established by the CLSI, USA-FDA and EUCAST (Table 1). *S. aureus* susceptibility rates for other antimicrobial agents tested are shown in Figure 1.
- Ceftaroline demonstrated potent activity against methicillin-susceptible *S. aureus* (MSSA) isolates (MIC_{50} and MIC_{90}, 0.25 µg/mL) and MRSA isolates (MIC_{50} and MIC_{90}, 1 µg/mL; Table 1 and Figure 2).
- Against MSSA, ceftaroline (MIC_{50} and MIC_{90}, 0.25 µg/mL) was 16-, 4- and 4-fold more active than ceftriaxone (MIC_{50} and MIC_{90}, 4 µg/mL), linezolid (MIC_{50} and MIC_{90}, 1 µg/mL) and vancomycin (MIC_{50} and MIC_{90}, 1 µg/mL), respectively. Furthermore, 17.2% of MSSA were clindamycin-resistant, with 5.0% showing constitutive resistance and 12.2% showing inducible resistance (data not shown).
- 51.0% of *S. aureus* isolates were MRSA and ceftaroline was one of the most active agents tested against these organisms (MIC_{50} and MIC_{90}, 1 µg/mL; 98.8% susceptible), with potency and spectrum similar to that of vancomycin (MIC_{50} and MIC_{90}, 1 µg/mL; 100% susceptible), linezolid (MIC_{50} and MIC_{90}, 1 µg/mL; >99.9% susceptible) and daptomycin ($MIC_{50/90}$, 0.25/0.5 µg/mL; 100.0% susceptible; data not shown). MRSA showed high resistance rates to levofloxacin (57.7%) and clindamycin (25.8%; 17.7% constitutive and 8.1% inducible resistance; data not shown).
- Against β-hemolytic streptococci, ceftaroline demonstrated activity (MIC_{50} and MIC_{90}, ≤0.015 µg/mL; highest MIC, 0.03 µg/mL; Table 1) comparable to that of penicillin (MIC_{50} and MIC_{90}, ≤0.06 µg/mL), and it was at least 64-fold greater than linezolid (MIC_{50} and MIC_{90}, 1 µg/mL) and 32-fold greater than vancomycin ($MIC_{50/90}$, 0.25/0.5 µg/mL).
- Viridans group streptococci, including strains resistant to penicillin and ceftriaxone, were very susceptible to ceftaroline ($MIC_{50/90}$, 0.03/0.06 µg/mL; highest MIC, 0.5 µg/mL; Table 1), while 93.0 and 98.8% of strains were susceptible to penicillin ($MIC_{50/90}$, ≤0.06/0.25 µg/mL) and ceftriaxone ($MIC_{50/90}$, 0.25/0.5 µg/mL), respectively (data not shown).
- Ceftaroline inhibited 81.8% of Enterobacteriaceae at ≤0.5 µg/mL (Table 1), which is the susceptible breakpoint established by CLSI and USA-FDA. Ceftaroline exhibited good activity against non-ESBL-phenotype strains of *Klebsiella* spp., *E. coli* and *Proteus mirabilis* (MIC_{90}, 0.25 µg/mL for all three pathogens), but limited activity against ESBL-phenotype and/or ceftriaxone-resistant strains (Figure 3).
- When tested against *E. cloacae*, ceftaroline ($MIC_{50/90}$, 0.25/>32 µg/mL) demonstrated similar activity compared to that of ceftriaxone ($MIC_{50/90}$, 0.25/>8 µg/mL) and ceftazidime ($MIC_{50/90}$, 0.25/32 µg/mL). Overall, 82.5 and 85.6% of *E. cloacae* strains were susceptible to ceftriaxone and ceftazidime, respectively (data not shown), and 82.3% were susceptible to ceftaroline (Table 1).
- Ceftaroline showed variable activity against *E. aerogenes* ($MIC_{50/90}$, 0.12/32 µg/mL; 80.6% susceptible) and *Morganella morganii* ($MIC_{50/90}$, 0.12/>32 µg/mL; 73.7% susceptible; Table 1). Susceptibility rates of these two organisms for ceftriaxone were 80.6 and 88.4%, respectively (data not shown).
- Ceftaroline was slightly more active against *Citrobacter koseri* ($MIC_{50/90}$, 0.12/0.5 µg/mL; 92.9% susceptible) compared to *C. freundii* ($MIC_{50/90}$, 0.25/1 µg/mL; 89.7% susceptible; Table 1).
- Ceftaroline exhibited modest activity against *Serratia marcescens* ($MIC_{50/90}$, 1/2 µg/mL; 43.2% susceptible), *Proteus vulgaris* ($MIC_{50/90}$, 1/32 µg/mL; 39.3% susceptible) and *Providencia* spp. ($MIC_{50/90}$, 0.5/8 µg/mL; 51.9% susceptible; Table 1).

Table 1. Summary of ceftaroline activity tested against bacterial isolates collected from patients with skin and soft tissue infections in USA medical centers (AWARE Program, 2013)

Organism/subgroup (no. of isolates)	No. of isolates (cumulative %) inhibited at ceftaroline MIC (µg/mL) of:												MIC (µg/mL)	
	≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	>16	50%	90%
Staphylococcus aureus (5182)	2 (0.0)	1 (0.1)	5 (0.2)	186 (3.7)	2262 (47.4)	1369 (73.8)	1326 (99.4)	31 (100.0)	--	--	--	--	0.5	1
MSSA (2540)	2 (0.1)	1 (0.1)	5 (0.3)	182 (7.5)	2225 (95.1)	125 (100.0)	--	--	--	--	--	--	0.25	0.25
MRSA (2642)	--	--	--	4 (0.2)	37 (1.6)	1244 (48.6)	1326 (98.8)	31 (100.0)	--	--	--	--	1	1
Viridans group streptococci (172)	69 (40.1)	75 (83.7)	26 (98.8)	1 (99.4)	0 (99.4)	1 (100.0)	--	--	--	--	--	--	0.03	0.06
β-hemolytic streptococci (746)	711 (95.3)	35 (100.0)	--	--	--	--	--	--	--	--	--	--	≤0.015	≤0.015
Streptococcus pyogenes (319	318 (99.7)	1 (100.0)	--	--	--	--	--	--	--	--	--	--	≤0.015	≤0.015
Streptococcus agalactiae (348)	314 (90.2)	34 (100.0)	--	--	--	--	--	--	--	--	--	--	≤0.015	≤0.015
Streptococcus dysgalactiae (79)	79 (100.0)	--	--	--	--	--	--	--	--	--	--	--	≤0.015	≤0.015
Enterobacteriaceae (2346)	8 (0.3)	87 (4.0)	579 (28.7)	677 (57.6)	360 (72.9)	209 (81.8)	126 (87.2)	40 (88.9)	28 (90.1)	25 (91.2)	28 (92.4)	179 (100.0)	0.12	4
Escherichia coli (655)	6 (0.9)	53 (9.0)	222 (42.9)	182 (70.7)	66 (80.8)	42 (87.2)	13 (89.2)	6 (90.1)	6 (91.0)	6 (91.9)	5 (92.7)	48 (100.0)	0.12	2
non-ESBL-phenotype (582)	6 (1.0)	53 (10.1)	222 (48.3)	182 (79.6)	63 (90.4)	37 (96.7)	13 (99.0)	5 (99.8)	1 (100.0)	--	--	--	0.12	0.25
ESBL-phenotype[a] (73)	--	--	--	--	3 (4.1)	5 (11.0)	0 (11.0)	1 (12.3)	5 (19.2)	6 (27.4)	5 (34.2)	48 (100.0)	>32	>32
Klebsiella spp. (482)	1 (0.2)	9 (2.1)	132 (29.5)	148 (60.2)	89 (78.6)	27 (84.2)	14 (87.1)	5 (88.2)	3 (88.8)	5 (89.8)	7 (91.3)	42 (100.0)	0.12	16
non-ESBL-phenotype (420)	1 (0.2)	9 (2.4)	132 (33.8)	148 (69.0)	89 (90.2)	27 (96.7)	11 (99.3)	2 (99.8)	0 (99.8)	0 (99.8)	0 (99.8)	1 (100.0)	0.12	0.25
ESBL-phenotype[a] (62)	--	--	--	--	--	--	3 (4.8)	3 (9.7)	3 (14.5)	5 (22.6)	7 (33.9)	41 (100.0)	>32	>32
Proteus mirabilis (312)	--	2 (0.6)	107 (34.9)	134 (77.9)	31 (87.8)	16 (92.9)	4 (94.2)	0 (94.2)	1 (94.6)	2 (95.2)	3 (96.2)	12 (100.0)	0.12	0.5
non-ESBL-phenotype (295)	--	2 (0.7)	107 (36.9)	134 (82.4)	31 (92.9)	16 (98.3)	4 (99.7)	0 (99.7)	0 (99.7)	1 (100.0)	--	--	0.12	0.25
ESBL-phenotype[a] (17)	--	--	--	--	--	--	--	--	1 (5.9)	1 (11.8)	3 (29.4)	12 (100.0)	>32	>32
Enterobacter cloacae (333)	--	5 (1.5)	10 (4.5)	90 (31.5)	124 (68.8)	45 (82.3)	8 (84.7)	5 (86.2)	1 (86.5)	2 (87.1)	3 (88.0)	40 (100.0)	0.25	>32
Enterobacter aerogenes (93)	--	2 (2.2)	34 (38.7)	34 (75.3)	4 (79.6)	1 (80.6)	3 (83.9)	0 (83.9)	3 (87.1)	0 (87.1)	2 (89.2)	10 (100.0)	0.12	32
Morganella morganii (95)	1 (1.1)	8 (9.5)	34 (45.3)	17 (63.2)	5 (68.4)	5 (73.7)	6 (80.0)	1 (81.1)	3 (84.2)	2 (86.3)	3 (89.5)	10 (100.0)	0.12	32
Citrobacter koseri (85)	--	--	26 (30.6)	44 (82.4)	3 (85.9)	6 (92.9)	5 (98.8)	0 (98.8)	0 (98.8)	0 (98.8)	0 (98.8)	1 (100.0)	0.12	0.5
Citrobacter freundii (58)	--	--	3 (5.2)	21 (41.4)	22 (79.3)	6 (89.7)	1 (91.4)	0 (91.4)	0 (91.4)	0 (91.4)	1 (93.1)	4 (100.0)	0.25	1
Serratia marcescens (125)	--	--	--	--	5 (4.0)	49 (43.2)	51 (84.0)	12 (93.6)	3 (96.0)	3 (98.4)	1 (99.2)	1 (100.0)	1	2
Proteus vulgaris (56)	--	2 (3.6)	1 (5.4)	1 (7.1)	8 (21.4)	10 (39.3)	10 (57.1)	6 (67.9)	5 (76.8)	4 (83.9)	1 (85.7)	8 (100.0)	1	32
Providencia spp. (52)	--	6 (11.5)	10 (30.8)	6 (42.3)	3 (48.1)	2 (51.9)	11 (73.1)	5 (82.7)	3 (88.5)	1 (90.4)	2 (94.2)	3 (100.0)	0.5	8

a. Isolates with MIC of ≥2 µg/mL for ceftazidime or ceftriaxone or aztreonam (CLSI 2014; M100-S24).

Figure 1. Antimicrobial susceptibility of 5182 *S. aureus* isolates from patients with SSSI (AWARE USA, 2013)

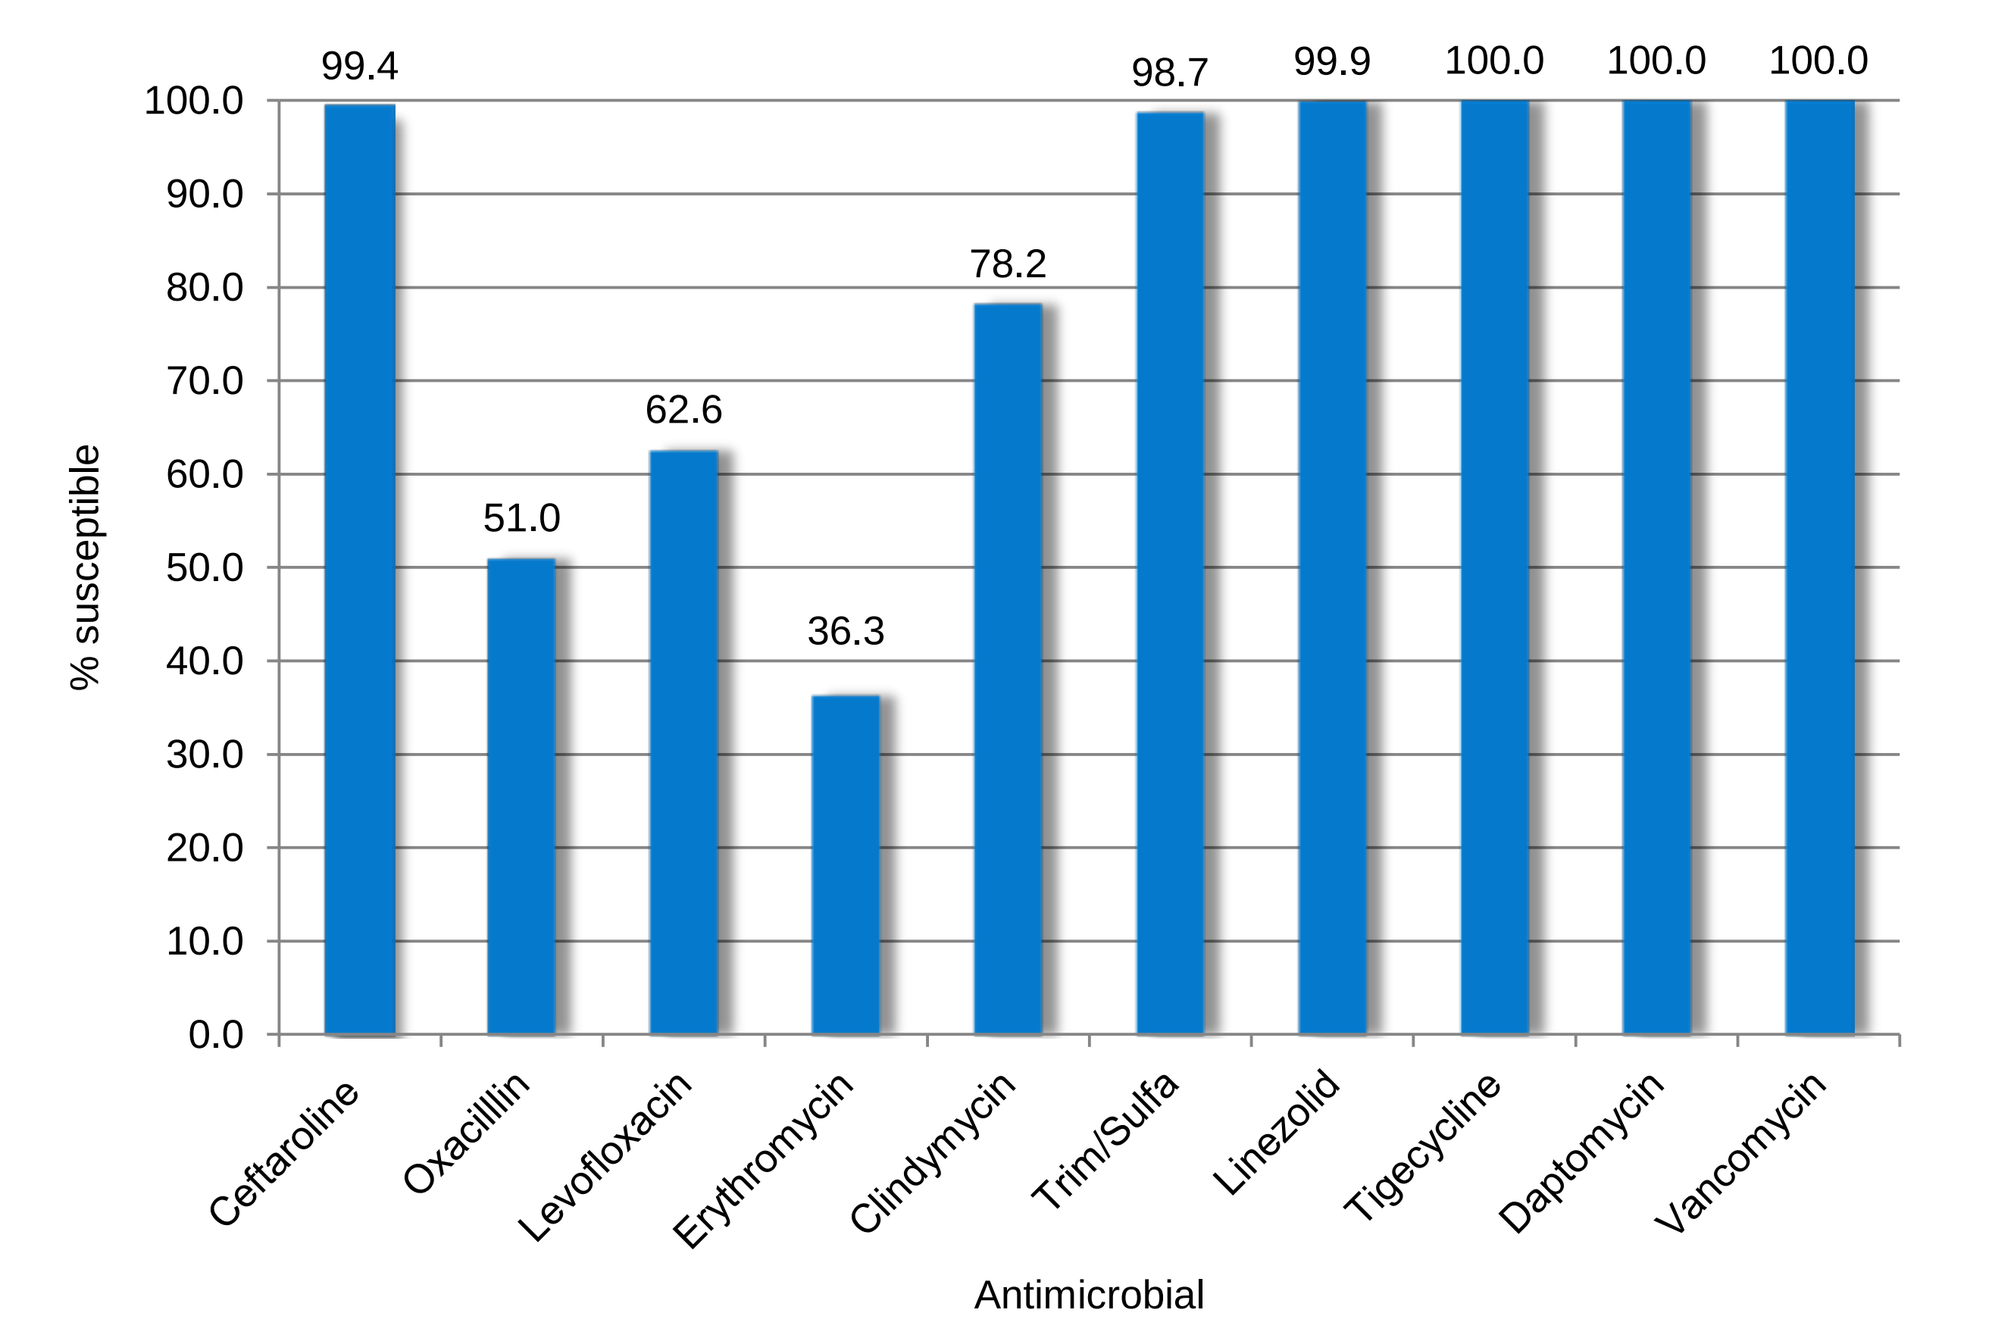

99.4
51.0
62.6
36.3
78.2
98.7
99.9
100.0
100.0
100.0
% susceptible
Ceftaroline
Oxacillin
Levofloxacin
Erythromycin
Clindymycin
Trim/Sulfa
Linezolid
Tigecycline
Daptomycin
Vancomycin
Antimicrobial

Figure 2. Ceftaroline MIC distributions against 5182 *S. aureus* isolates from patients with SSSI (AWARE USA, 2013)

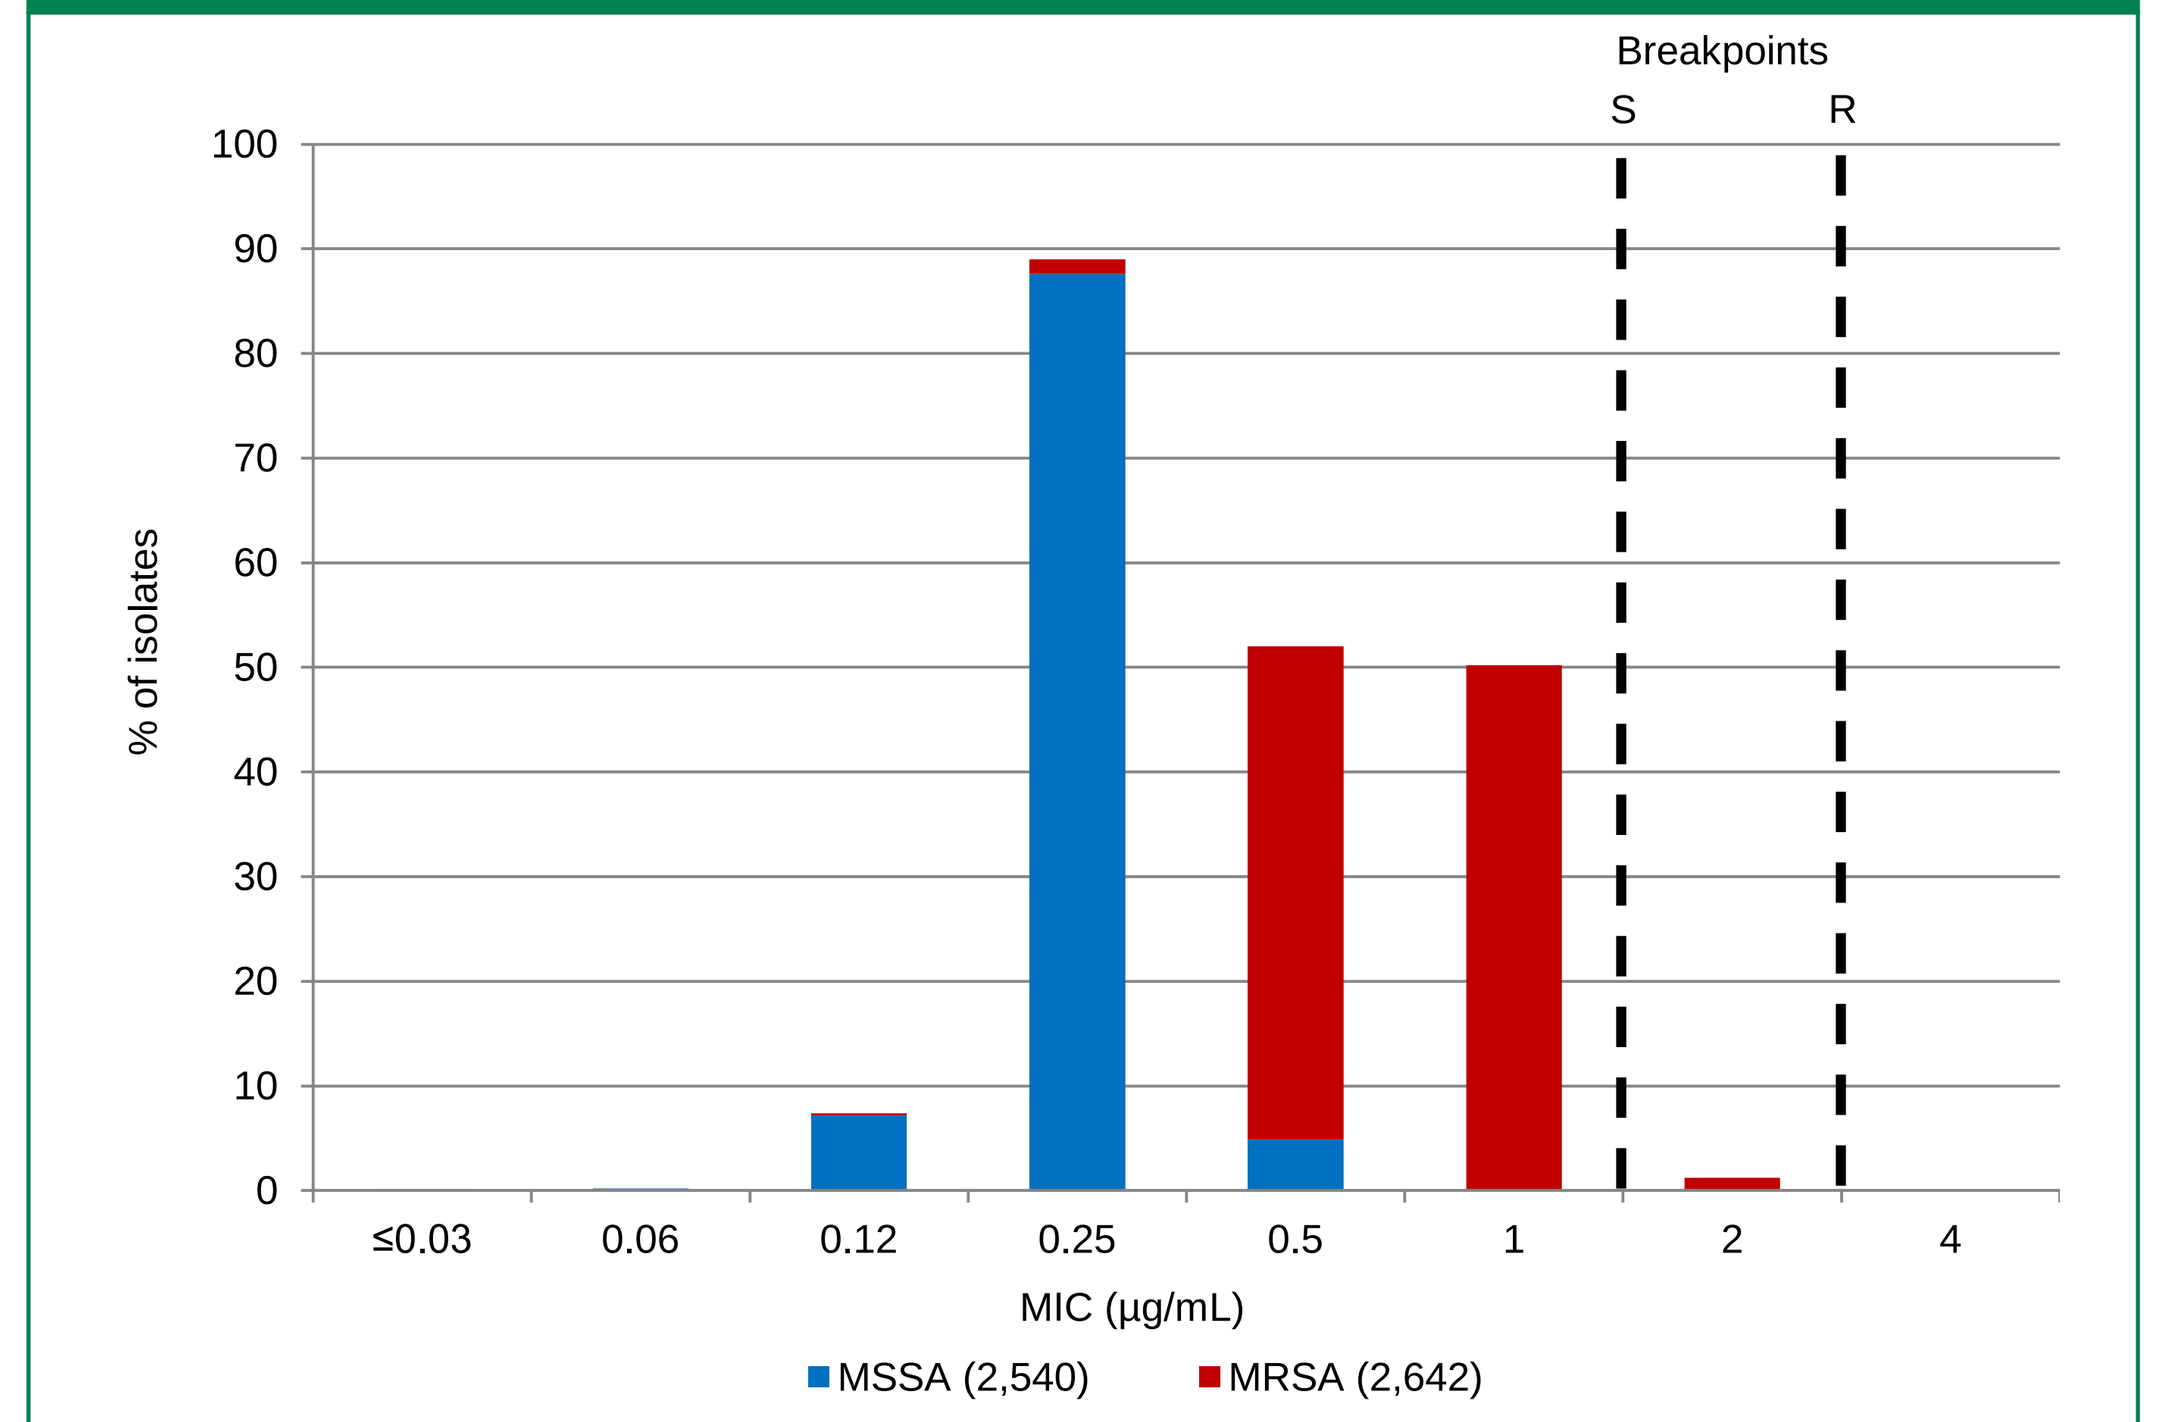

Breakpoints
S
R
% of isolates
≤0.03
0.06
0.12
0.25
0.5
1
2
4
MIC (µg/mL)
MSSA (2,540)
MRSA (2,642)

Figure 3. Ceftaroline MIC distributions for isolates of ESBL (n=152) and non-ESBL (n=1297) Enterobacteriaceae from patients with SSSI (AWARE USA, 2013). Includes *E. coli* (655), *Klebsiella pneumoniae* (319), *K. oxytoca* (163) and *P. mirabilis* (312)

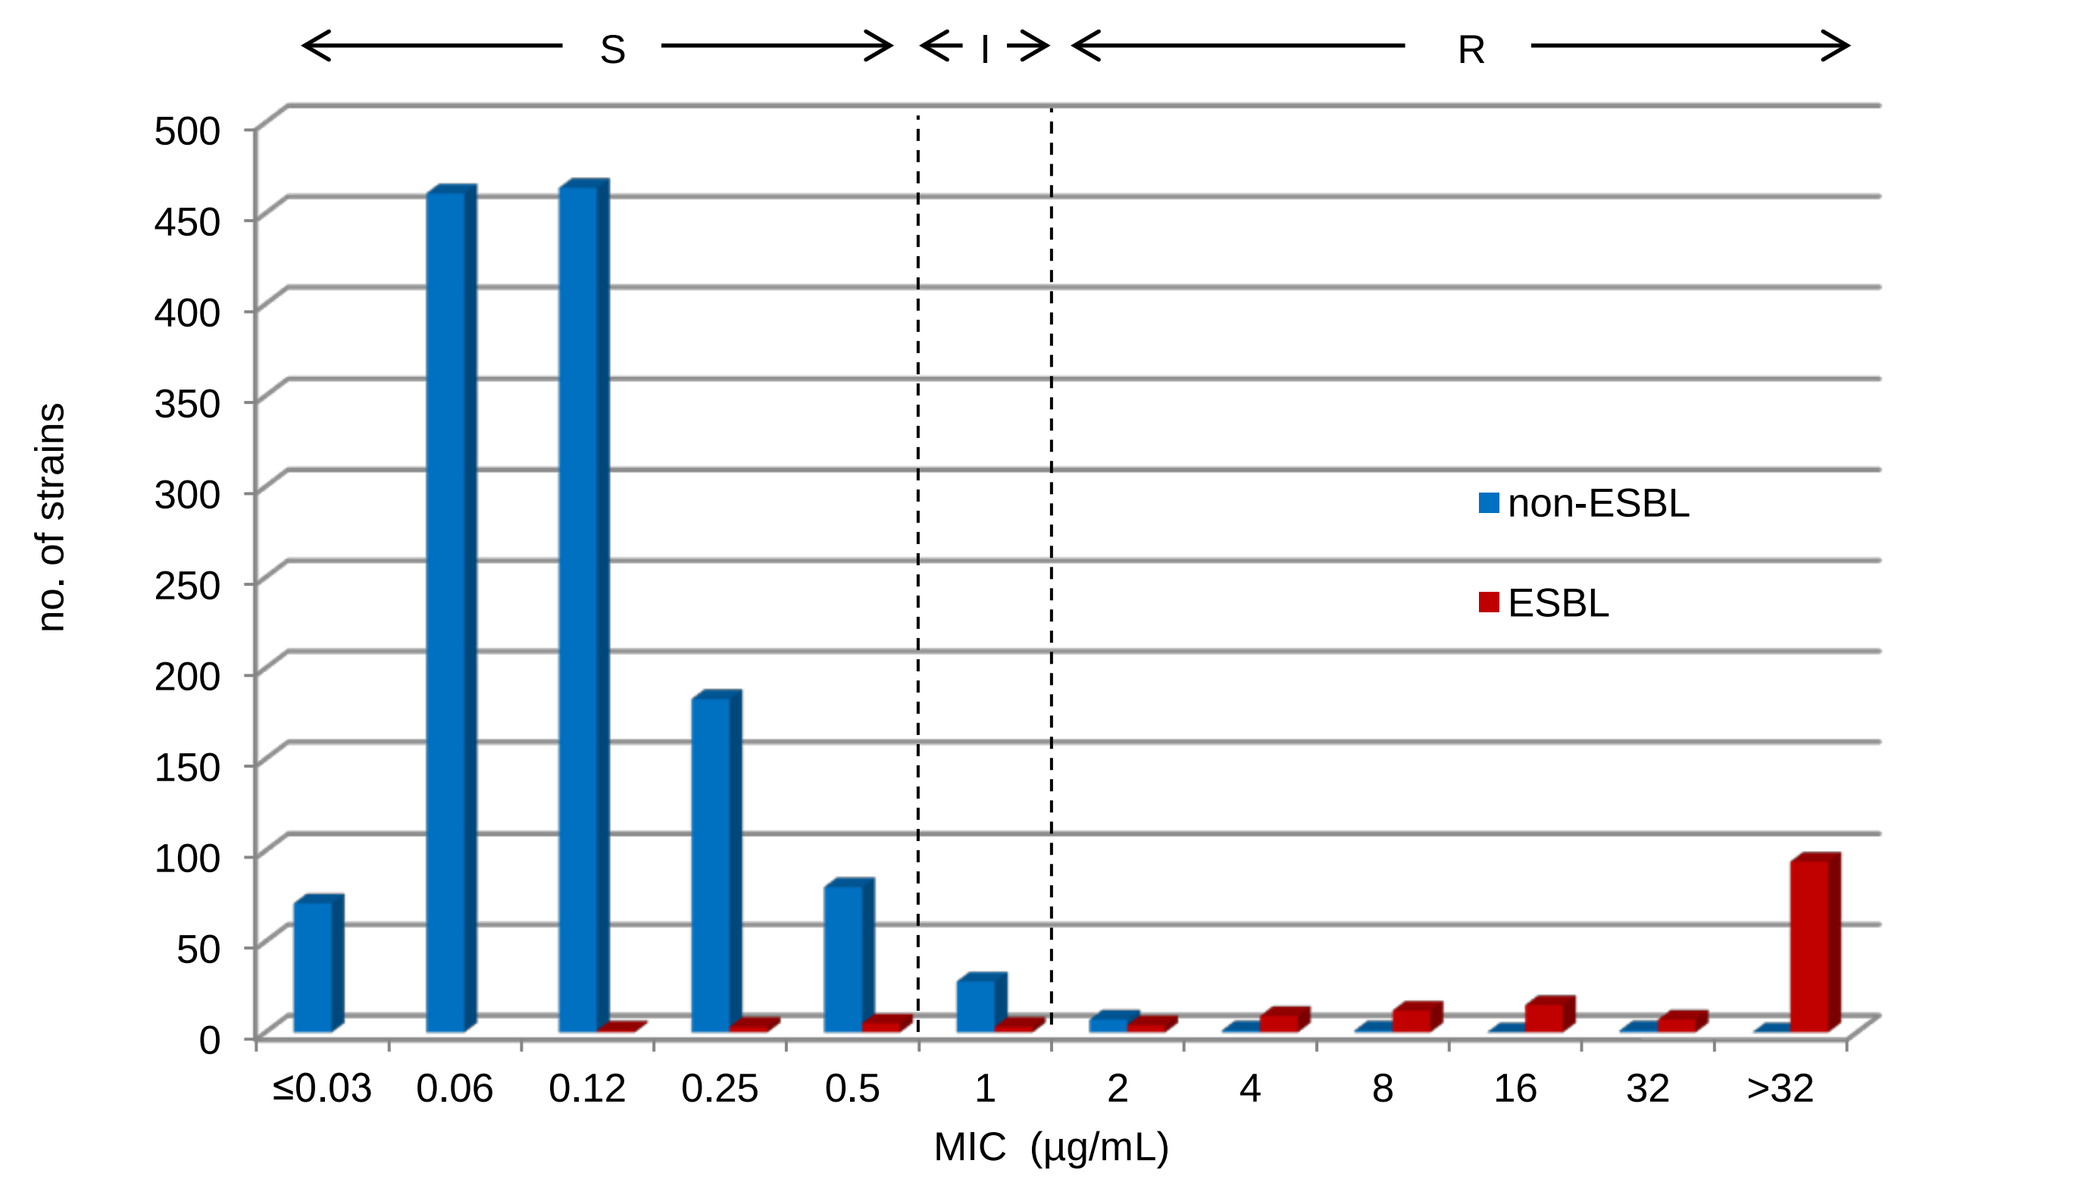

S
I
R
no. of strains
non-ESBL
ESBL
≤0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
>32
MIC (µg/mL)

Conclusions

- Ceftaroline demonstrated potent in vitro activity against Gram-positive organisms, including MRSA strains (0.0% resistance), and non-ESBL-phenotype Enterobacteriaceae recently (2013) isolated from USA patients with documented ABSSSI.
- Ceftaroline spectrum against Gram-positive cocci was similar to that of linezolid and vancomycin.
- Against Enterobacteriaceae, ceftaroline had a spectrum comparable to that of ceftriaxone.
- Ceftaroline appears to be a valuable option for treatment of ABSSSI, including those caused by MRSA.

References

1. Beresford E, Biek D, Jandourek A, Mawal Y, Riccobene T, Friedland HD (2014). Ceftaroline fosamil for the treatment of acute bacterial skin and skin structure infections. *Expert Rev Clin Pharmacol* 7: 123-135.
2. Clinical and Laboratory Standards Institute (2012). *M07-A9. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: ninth edition*. Wayne, PA: CLSI.
3. Clinical and Laboratory Standards Institute (2014). *M100-S24. Performance standards for antimicrobial susceptibility testing: 24th informational supplement.* Wayne, PA: CLSI.
4. Flamm RK, Farrell DJ, Sader HS, Jones RN (2014). Antimicrobial activity of ceftaroline combined with avibactam tested against bacterial organisms isolated from acute bacterial skin and skin structure infections in United States medical centers (2010-2012). *Diagn Microbiol Infect Dis* 78: 449-456.
5. Flamm RK, Sader HS, Farrell DJ, Jones RN (2012). Summary of ceftaroline activity against pathogens in the United States, 2010: Report from the Assessing Worldwide Antimicrobial Resistance Evaluation (AWARE) Surveillance Program. *Antimicrob Agents Chemother* 56: 2933-2940.
6. Pfaller MA, Flamm RK, Sader HS, Jones RN (2014). Ceftaroline activity against bacterial organisms isolated from acute bacterial skin and skin structure infections in United States medical centers (2009-2011). *Diagn Microbiol Infect Dis* 78: 422-428.
7. Teflaro® Package Insert (2012). Available at http://www.accessdata.fda.gov/drugsatfda_docs/label/2013/200327s009lbl.pdf. Accessed July 2014.

Acknowledgments

This study was supported by Cerexa, LLC, a wholly-owned subsidiary of Forest Laboratories, LLC. Forest Laboratories, LLC, was involved in the design and decision to present these results and JMI Laboratories received compensation fees for services in relation to preparing the abstract/poster, which was funded by the sponsor. Forest Laboratories, LLC, had no involvement in the collection, analysis, and interpretation of data.