IDWEEK 2024 | Poster #P1526

In vitro Activity of Ceftobiprole Against Prominent Methicillin-resistant *Staphylococcus aureus* Clonal Groups Collected Through the SENTRY Antimicrobial Surveillance Program

John H. Kimbrough[1], Joshua M. Maher[1], Jennifer I. Smart[2], Mark E. Jones[2], Mariana Castanheira[1], Helio S. Sader,[1] Rodrigo E. Mendes[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] Basilea Pharmaceutica International Ltd, Allschwil, Switzerland

Introduction

- Ceftobiprole is an advanced-generation cephalosporin recently (April 2024) approved by the United States Food and Drug Administration (US FDA) for treating *Staphylococcus aureus* bacteremia (SAB), community-acquired bacterial pneumonia (CABP), and acute bacterial skin and skin structure infections (ABSSSI).
- Previous studies have demonstrated the broad and potent *in vitro* and *in vivo* activity of ceftobiprole against clinically important Gram-negative and Gram-positive organisms, including methicillin-resistant *Staphylococcus aureus* (MRSA).
- Numerous pandemic MRSA lineages with defined molecular characteristics, including shared combinations of resistance genes and virulence factors, have been described to date.
- The shifting dominance of pandemic MRSA lineages and emergence of new lineages necessitates the continued surveillance and development of new treatment options for MRSA infections.
- Next generation genome sequencing coupled with *in silico* screening for pandemic lineage-specific features, including the staphylococcal chromosomal cassette methicillin-resistance (SCC*mec*) and alleles of the gene encoding staphylococcal protein A (*spa*) among others, allows for the rapid and reliable classification of infection-associated MRSA isolates.
- This study molecularly characterized MRSA isolates causing bloodstream infections (BSI) and included in the SENTRY Antimicrobial Surveillance program (2018–2019) to identify circulating pandemic lineages and reports the activities of ceftobiprole and the comparator agent ceftaroline against these isolates.

Materials and Methods

Bacterial Isolates and Susceptibility Testing

- One hundred fifty (*n*= 150) MRSA isolates (oxacillin MIC, i.e. ≥4 mg/L) recovered worldwide from bloodstream infections in hospitalized patients as part of the SENTRY Antimicrobial Surveillance program from 2018–2019 were included in this analysis.
- Broth microdilution antimicrobial susceptibility testing was performed using CLSI methods (M07, 2018).
- FDA (2024) susceptible breakpoint was applied for ceftobiprole (i.e. ≤2 mg/L), whereas ceftaroline used the FDA-recognized CLSI susceptible breakpoint (i.e. ≤1 mg/L).
- CLSI quality assurance practices were performed during susceptibility testing, including the use of concurrent testing of validated CLSI-recommended quality control strains and the monitoring of inoculum density using bacterial colony counts.

Whole Genome Sequencing and Analysis

- Total genomic DNA was extracted and purified using the KingFisher Cell and Tissue DNA kit (Thermo Scientific, Waltham, Massachusetts, USA) in a robotic KingFisher™ Flex Magnetic Particle Processor (Thermo Scientific) workstation.
- Total genomic DNA was used as input material for library construction. DNA libraries were prepared using the Nextera XT™ library construction protocol and index kit (Illumina, San Diego, California, USA) and sequenced on a MiSeq Sequencer (Illumina) using a MiSeq Reagent Kit v3 (600 cycle). Sequencing reactions were initially performed to achieve DNA read lengths of up to 300-bp and an average genome coverage depth of approximately 30x.
- Each raw data set was quality assured, error corrected, and assembled *de novo* using assembler SPAdes 3.11.1. Assembled genomes had the DNA sequence information extracted to determine:
 - Multi-locus sequence type (ST); STs were further assigned to clonal complexes (CC) consistent with current literature recognition.
 - *spa* typing was performed using *spa*Typer v4.2.2
 - Determination of the SCC*mec* locus was performed using SCC*mec*Finder v8.0.1
- Assignment to a specific pandemic clonal group (CG; e.g., USA100) was performed, when possible, based on the molecular information obtained consistent with current literature recognition.

Results

- Isolates were collected from 38 medical centers in North America (69 isolates; 18 centers; USA only), Europe (56 isolates; 12 centers; 9 countries), Asia-Western Pacific (13 isolates; 4 centers; 4 countries), and Latin America (12 isolates, 4 centers; 4 countries).
- Twenty-three CGs, including subdivisions and variants, were identified; 4 isolates could not be assigned a CG (Fig. 1)
 - Isolates were grouped into six predominant CG; isolates associated with USA300 comprised the majority (26.0%), and similar frequencies of UK-EMRSA-15/USA100 isolates (18.7-22.0%) and USA500/USA800/Other isolates (8.7–14.7%) were observed.
 - Isolates belonging to USA100 (78.6%) and USA300 (94.9%), including sub-clones and variants, were contributed primarily from US medical centers (Fig. 2).
 - European medical centers contributed the majority (93.9%) of isolates derived from UK-EMRSA-15. (Fig. 2).
- Against all MRSA, 97.3% were ceftobiprole-susceptible (S) and 85.3% were ceftaroline-S (Table 1 and Fig. 3A).
- Ceftobiprole generally displayed higher activity than ceftaroline across CGs and subdivisions:
 - USA300, 100.0% ceftobiprole-S/100.0% ceftaroline-S (Table 1 and Fig. 3B)
 - UK-EMRSA-15, 93.9% ceftobiprole-S/69.7% ceftaroline-S (Table 1 and Fig. 3C)
 - USA100, 96.4% ceftobiprole-S/75.0% ceftaroline-S (Table 1 and Fig. 3D)
 - USA800 and Other/unknown CGs, 100.0/95.5% ceftobiprole-S and 100.0/90.9% ceftaroline-S, respectively (Table 1 and Fig. 3E)
 - USA500, 100.0% ceftobiprole-S/76.9% ceftaroline-S (Table 1 and Fig. 3F)
- Four (2.7%) and 22 (14.7%) isolates were ceftobiprole- and ceftaroline-nonsusceptible (NS), respectively (Table 2).
 - 100.0% of ceftobiprole-NS isolates were ceftaroline-NS whereas only 18.2% of ceftaroline-NS isolates were ceftobiprole-NS
 - Ceftobiprole-NS isolates represented clonal groups UK-EMRSA-15 (*n*=2), and 1 isolate each from USA100-Swiss and Hungarian-Brazilian
 - 86.4% of ceftaroline-NS isolates were derived from UK-EMRSA-15, USA100, or Hungarian-Brazilian CGs.

Conclusions

- Ceftobiprole demonstrated potent activity against a global collection of MRSA isolates representing prominent pandemic clones.
- These results corroborate clinical demonstration of ceftobiprole's activity against isolates from similarly represented MRSA clonal lineages collected from patients at baseline visits in a Phase 3 clinical trial for efficacy against *S. aureus* bacteremia (NCT03138733).
 - USA300, 25.6% of isolates; ceftobiprole modal MIC, 1 mg/L; 100.0%-S
 - UK-EMRSA-15, 21.1%; ceftobiprole modal MIC, 1 mg/L; 100.0%-S
 - USA100, 7.8%; ceftobiprole modal MIC, 2 mg/L; 100.0%-S
 - USA800, 8.9%, ceftobiprole modal MIC, 1 mg/L; 100.0%-S
- A low frequency of ceftobiprole nonsusceptibility was observed among highly represented CGs including UK-EMRSA-15 and USA100.
- Ceftobiprole maintained activity against ceftaroline-NS isolates.
- These results support the use of ceftobiprole in the treatment of MRSA infections, including SAB, ABSSSI, and CABP.

Acknowledgments

The authors would like to thank Maura Karr for bioinformatics support as well as Gauri Deshpande, Gina Bartleson, and Jill Arends for editorial assistance in the execution of this study and preparation of this poster. This study was sponsored by Basilea Pharmaceutica International Ltd, Allschwil, Switzerland and was funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration for Strategic Preparedness and Response (ASPR); Biomedical Advanced Research and Development Authority (BARDA), under contract number HHSO100201600002C. The contract and federal funding are not an endorsement of the study results, product, or company.

IDWeek 2024, October 16–19, 2024, Los Angeles, California

Table 1. Genomic features and susceptibility profile of MRSA isolates stratified by clonal group and subgroups.

Clonal Group (n)[a]	Clonal Complex	Sequence Type	SCC*mec*[b]	%BPR-S[c]	%CPT-S[c]
All MRSA	Multiple	Multiple	Multiple	97.3	85.3
USA300 (39)[d,e]	CC8 (100.0%)	ST8 (89.7%); Other ST[e] (10.3%)	IV(2B)/IVa(2B) (100.0%)	100.0	100.0
USA500(13)[f]	CC8 (100.0%)	ST8 (84.6%); Other ST[f] (15.4%)	Multiple	100.0	76.9
Lyon (9)	CC8 (100.0%)	ST8 (77.86%); Other ST[f] (22.2%)	IVc(2B) (100.0%)	100.0	66.7
USA500 (4)	CC8 (100.0%)	ST8 (100.0%)	IV(2B)/IVa(2B) (100.0%)	100.0	100.0
UK-EMRSA-15 (33)	CC22 (100%)	Multiple	Multiple	93.9	69.7
UK-EMRSA-15 (23)[d]	CC22 (100%)	ST22 (100.0%)	IV(2B)/IVa(2B)/IVj(2B) (100%)	91.3	56.5
Middle East (5)	CC22 (100%)	ST22 (100.0%)	IVa(2B) (100.0%)	100.0	100.0
Barnim (4)	CC22 (100%)	ST22 (100.0%)	IV(2B) (100.0%)	100.0	100.0
Other (1)	CC22 (100%)	ST737 (100.0%)	IVa(2B) (100.0%)	100.0	100.0
USA100 (28)	CC5 (100.0%)	Multiple	II(2A) (100.0%)	96.4	75.0
USA100 (15)[d]	CC5 (100.0%)	ST5 (66.7%); ST6360 (20.0%); ST1011 (13.3%)	II(2A) (100.0%)	100.0	80.0
Orange County (2)	CC5 (100.0%)	ST5 (100.0%)	II(2A) (100.0%)	100.0	100.0
Rhine-Hesse (2)	CC5 (100.0%)	ST225 (100.0%)	II(2A) (100.0%)	100.0	100.0
Swiss (9)	CC5 (100.0%)	ST105 (100.0%)	II(2A) (100.0%)	88.9	55.6
USA800 (15)[d]	CC5 (100.0%)	ST5 (80.0%); Other ST[g] (20.0%)	IV(2B)/IVa(2B)/IVb(2B)/IVc(2B) IVg(2B)/IVi(2B) (100.0%)	100.0	100.0
Other (22)	Multiple	Multiple	Multiple	95.5	90.9
Hungarian/Brazilian (3)	CC8 (100.0%)	ST239 (100.0%)	III(3A) (100.0%)	66.7	33.3
USA400 (2)	CC1 (100.0%)	ST1 (100.0%)	IVa(2B) (100.0%)	100.0	100.0
USA600 (3)	CC45 (100.0%)	ST45 (100.0%)	IVa(2B) (66.7%); IVd(2B) (33.3%)	100.0	100.0
USA700 (3)	CC72 (100.0%)	ST72 (100.0%)	IVa(2B) (100.0%)	100.0	100.0
USA1100 (2)	CC30 (100.0%)	ST30 (100.0%)	IVc(2B) (100.0%)	100.0	100.0
Other (9)[h]	NA (44.4%); CC5, CC8, CC22, CC93, CC97 (12.5% each)	ST22, ST59, ST87, ST93, ST97, ST395, ST779, ST1637 (11.1% each)	IVa(2B)/IVc(2B) and NT[i] (22.2% each); IVd(2B), IV(2B&5), Vb(5C2&5)	100.0	100.0

Abbreviations: SCC, *Staphylococcus* chromosomal cassette; BPR, ceftobiprole; CPT, ceftaroline; MRSA; methicillin-resistant *S. aureus*; CC, clonal complex; ST, sequence type; UK-EMRSA, United Kingdom epidemic MRSA; NA, non-assignable; NT, non-typeable

[a] At least 1 presumptive clonal strain designation based on ST, and SCC*mec* and *spa* typing (not shown).

[b] SCC*mec* type and subtype. Numbers and letters between parentheses represent *ccr* complexes and SCC*mec* gene complexes, respectively.

[c] Using US FDA breakpoints.

[d] Contains all isolates in any clonal group possessing canonical CC/ST/SCC*mec*/*spa* features without a published subvariant designation.

[e] Includes ST1750 (n=2) and 1 isolate each ST923 and ST1757. All isolates were ACME+; 10 isolates were PVL-negative.

[f] Includes 1 isolate each ST2149-like and ST4317. All isolates are ACME- and PVL (*lukS*/*lukF*)-negative.

[g] Includes 1 isolate each ST149, ST1649, and ST1866-like.

[h] Includes 1 isolate each corresponding to CC97-LAC (LAC, livestock-associated clone; CC97/ST97/IVa(2B)), Queensland (CC93/ST93/IVa(2B)), ST779 (NA/ST779/IVc(2B)), Taiwan (NA/ST59/Vb(5C2&5)), WA-MRSA-24 (WA, Western Australia; NA/ST87/IVd(2B)), and 3 isolates with CC/ST/SCC*mec*/*spa* combinations to which no clonal group could be assigned.

[i] One SCC*mec* non-typeable isolate contained *mecC* and only a partial assignment for type V(5C2) while the second isolate possessed multiple SCC*mec* type assignments and may represent a composite type.

Figure 1. Representation of major pandemic clonal groups, including subgroups, among MRSA isolates.

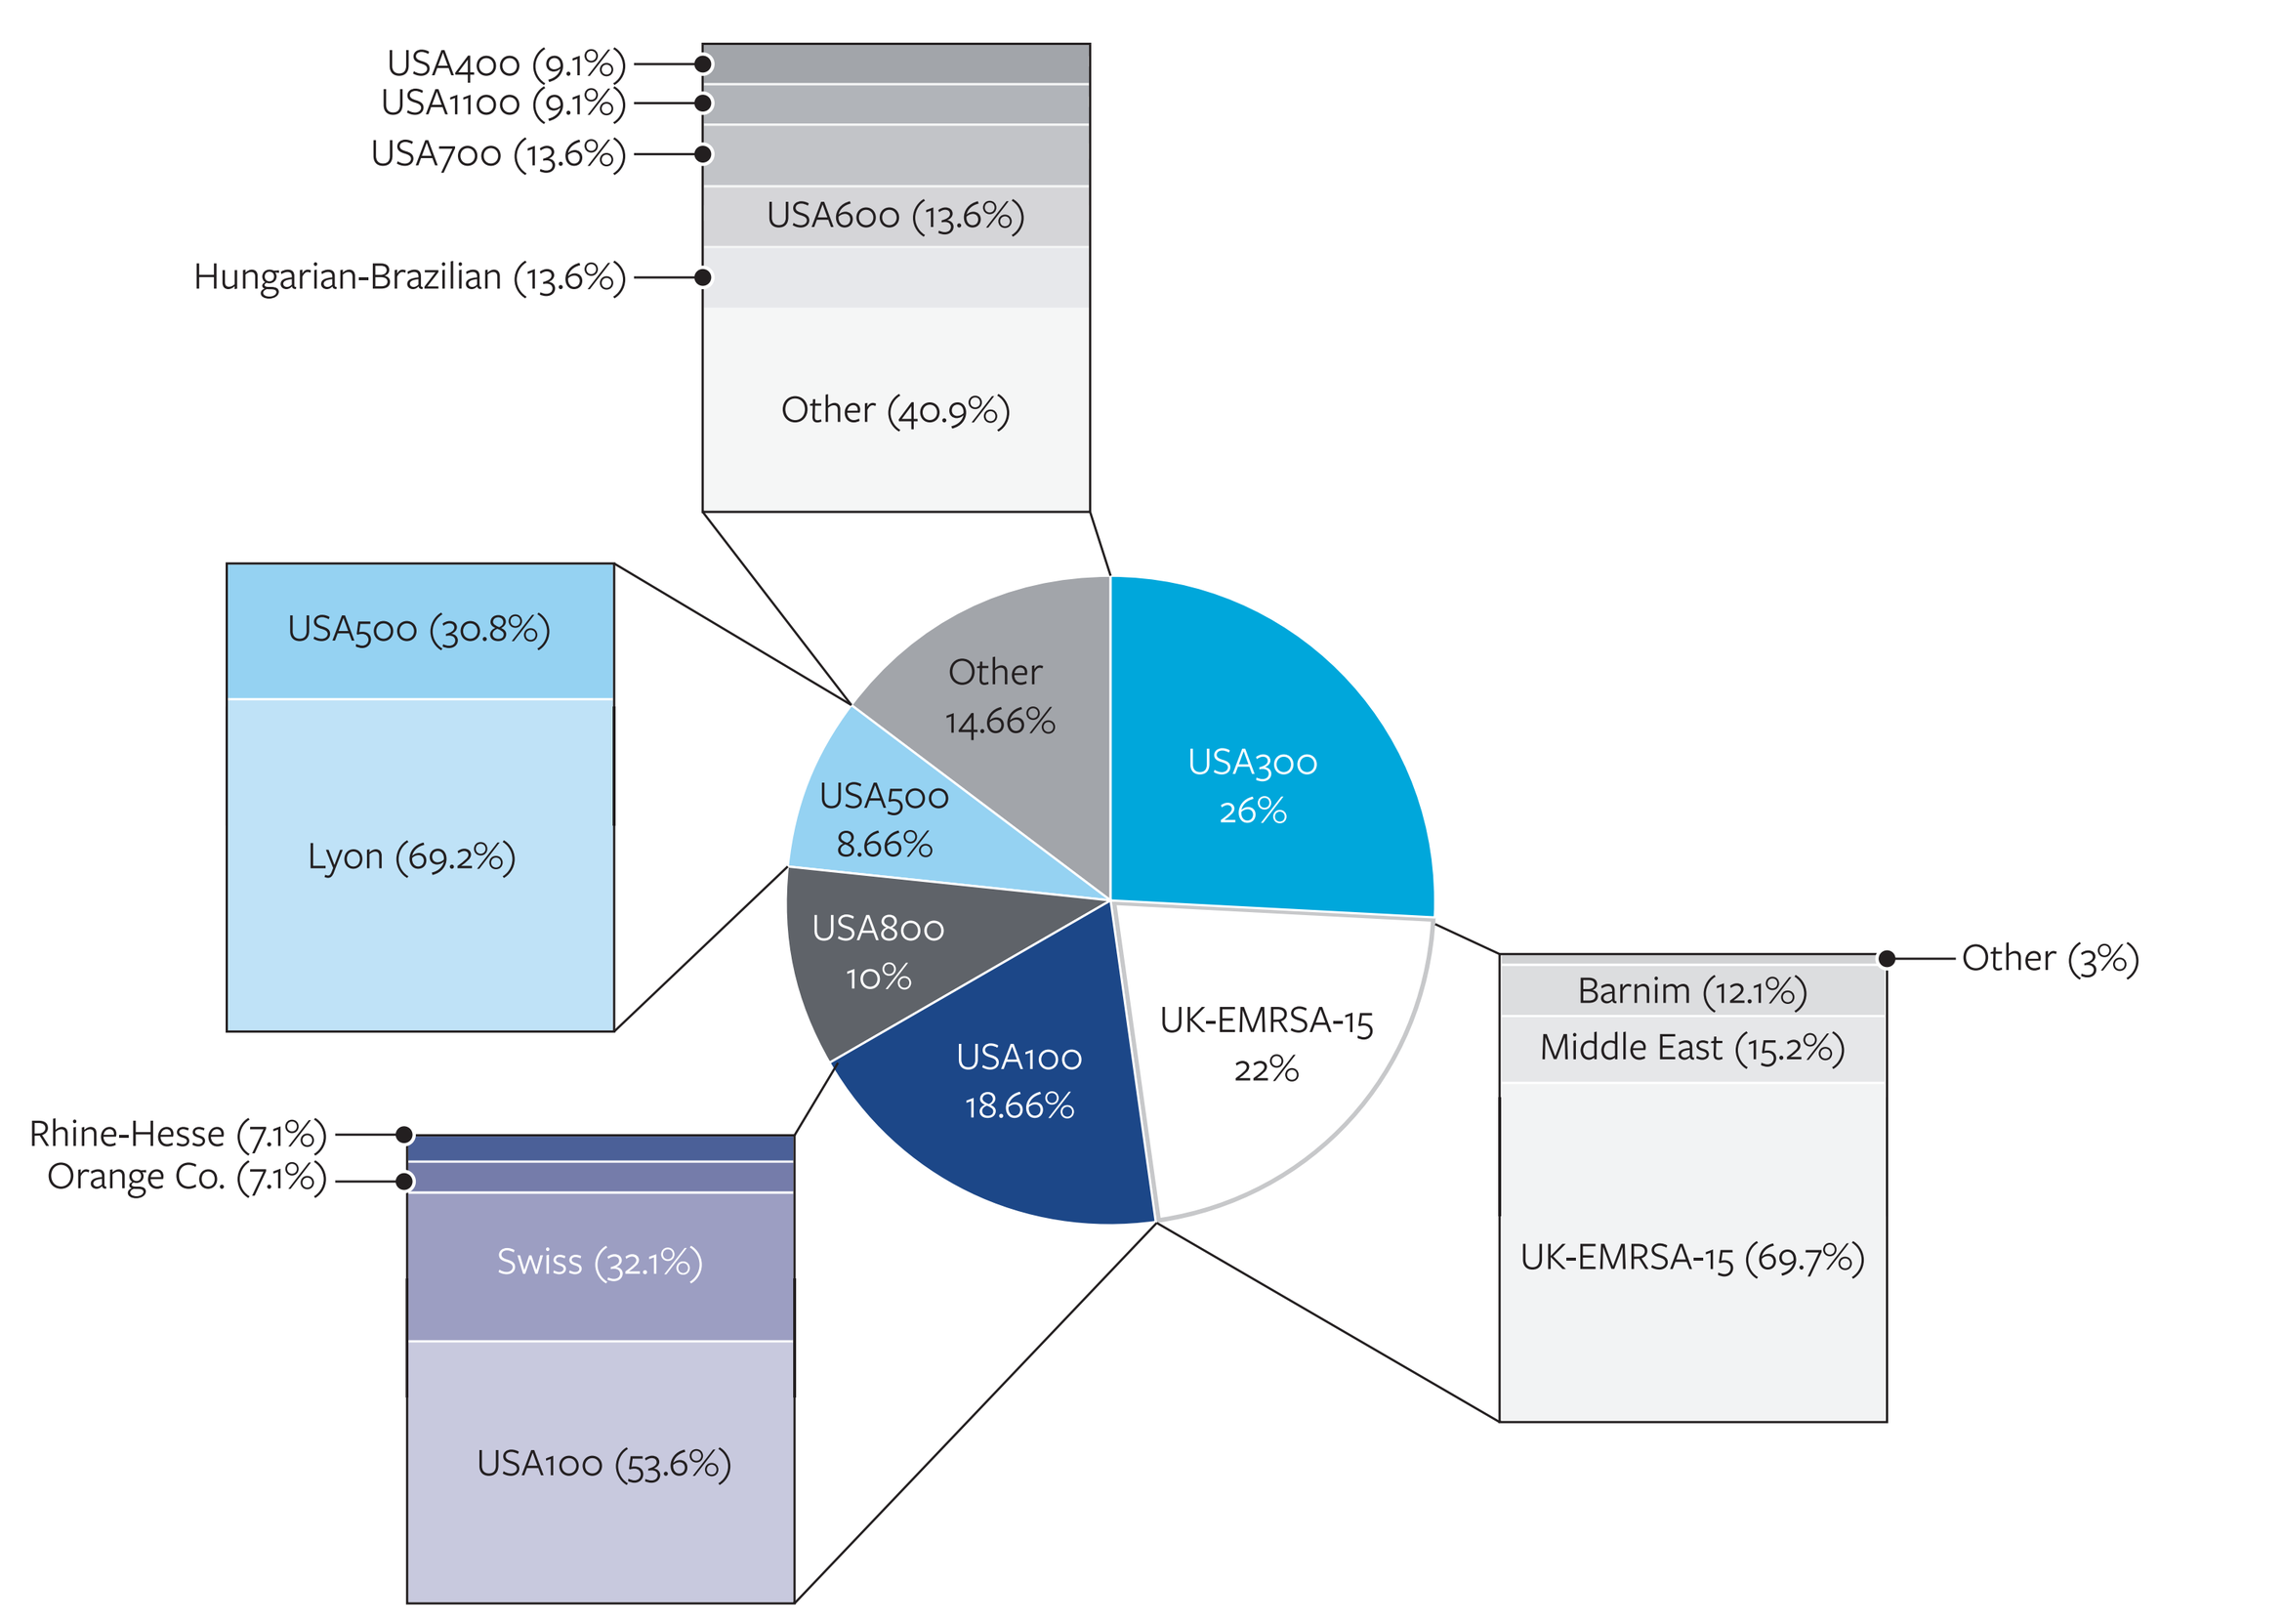

USA400 (9.1%)
USA1100 (9.1%)
USA700 (13.6%)
USA600 (13.6%)
Hungarian-Brazilian (13.6%)
Other (40.9%)
USA500 (30.8%)
Lyon (69.2%)
Other 14.66%
USA500 8.66%
USA300 26%
USA800 10%
USA100 18.66%
UK-EMRSA-15 22%
Barnim (12.1%)
Middle East (15.2%)
Other (3%)
UK-EMRSA-15 (69.7%)
Rhine-Hesse (7.1%)
Orange Co. (7.1%)
Swiss (32.1%)
USA100 (53.6%)

"Other" denotes clonal groups represented by ≤3 isolates or isolates to which no clonal group could be assigned and includes the following: 3 isolates each Hungarian/Brazilian, USA600, and USA700; 2 isolates each USA400 and USA1100, and 1 isolate each corresponding to CC97-LAC (LAC, livestock-associated clone), Queensland, ST779, Taiwan, and WA-MRSA-24 (WA, Western Australia); 3 isolates could not be assigned a clonal group.

Figure 2. Regional distribution of major pandemic clonal groups, including subgroups, among MRSA isolates.

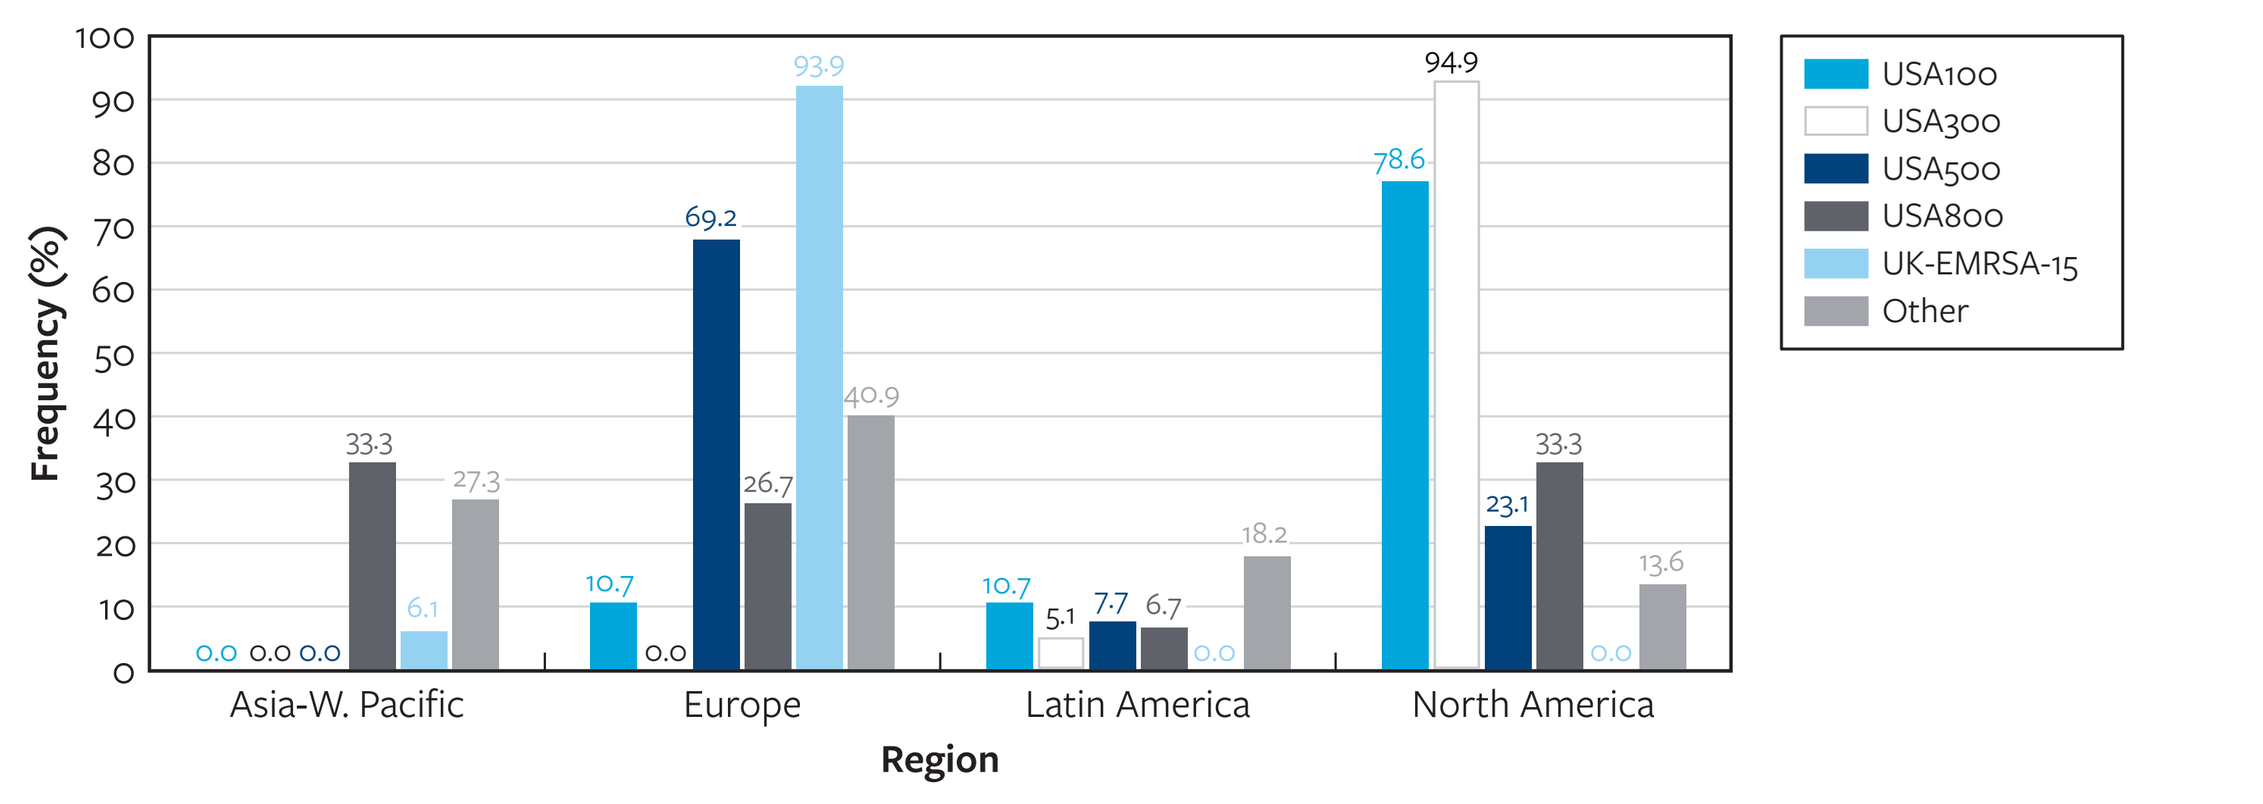

Frequency (%)
Region
Asia-W. Pacific
Europe
Latin America
North America
USA100
USA300
USA500
USA800
UK-EMRSA-15
Other

"Other" denotes clonal groups represented by ≤3 isolates or isolates to which no clonal group could be assigned and includes the following: 3 isolates each Hungarian/Brazilian, USA600, and USA700; 2 isolates each USA400 and USA1100, and 1 isolate each corresponding to CC97-LAC (LAC, livestock-associated clone), Queensland, ST779, Taiwan, and WA-MRSA-24 (WA, Western Australia); 3 isolates could not be assigned a clonal group.

Figure 3. MIC frequency distributions for ceftobiprole (BPR) and ceftaroline (CPT) against A) all MRSA and isolates from clonal groups B) USA300 C) UK-EMRSA D) USA100 E) USA800 and F) USA500.

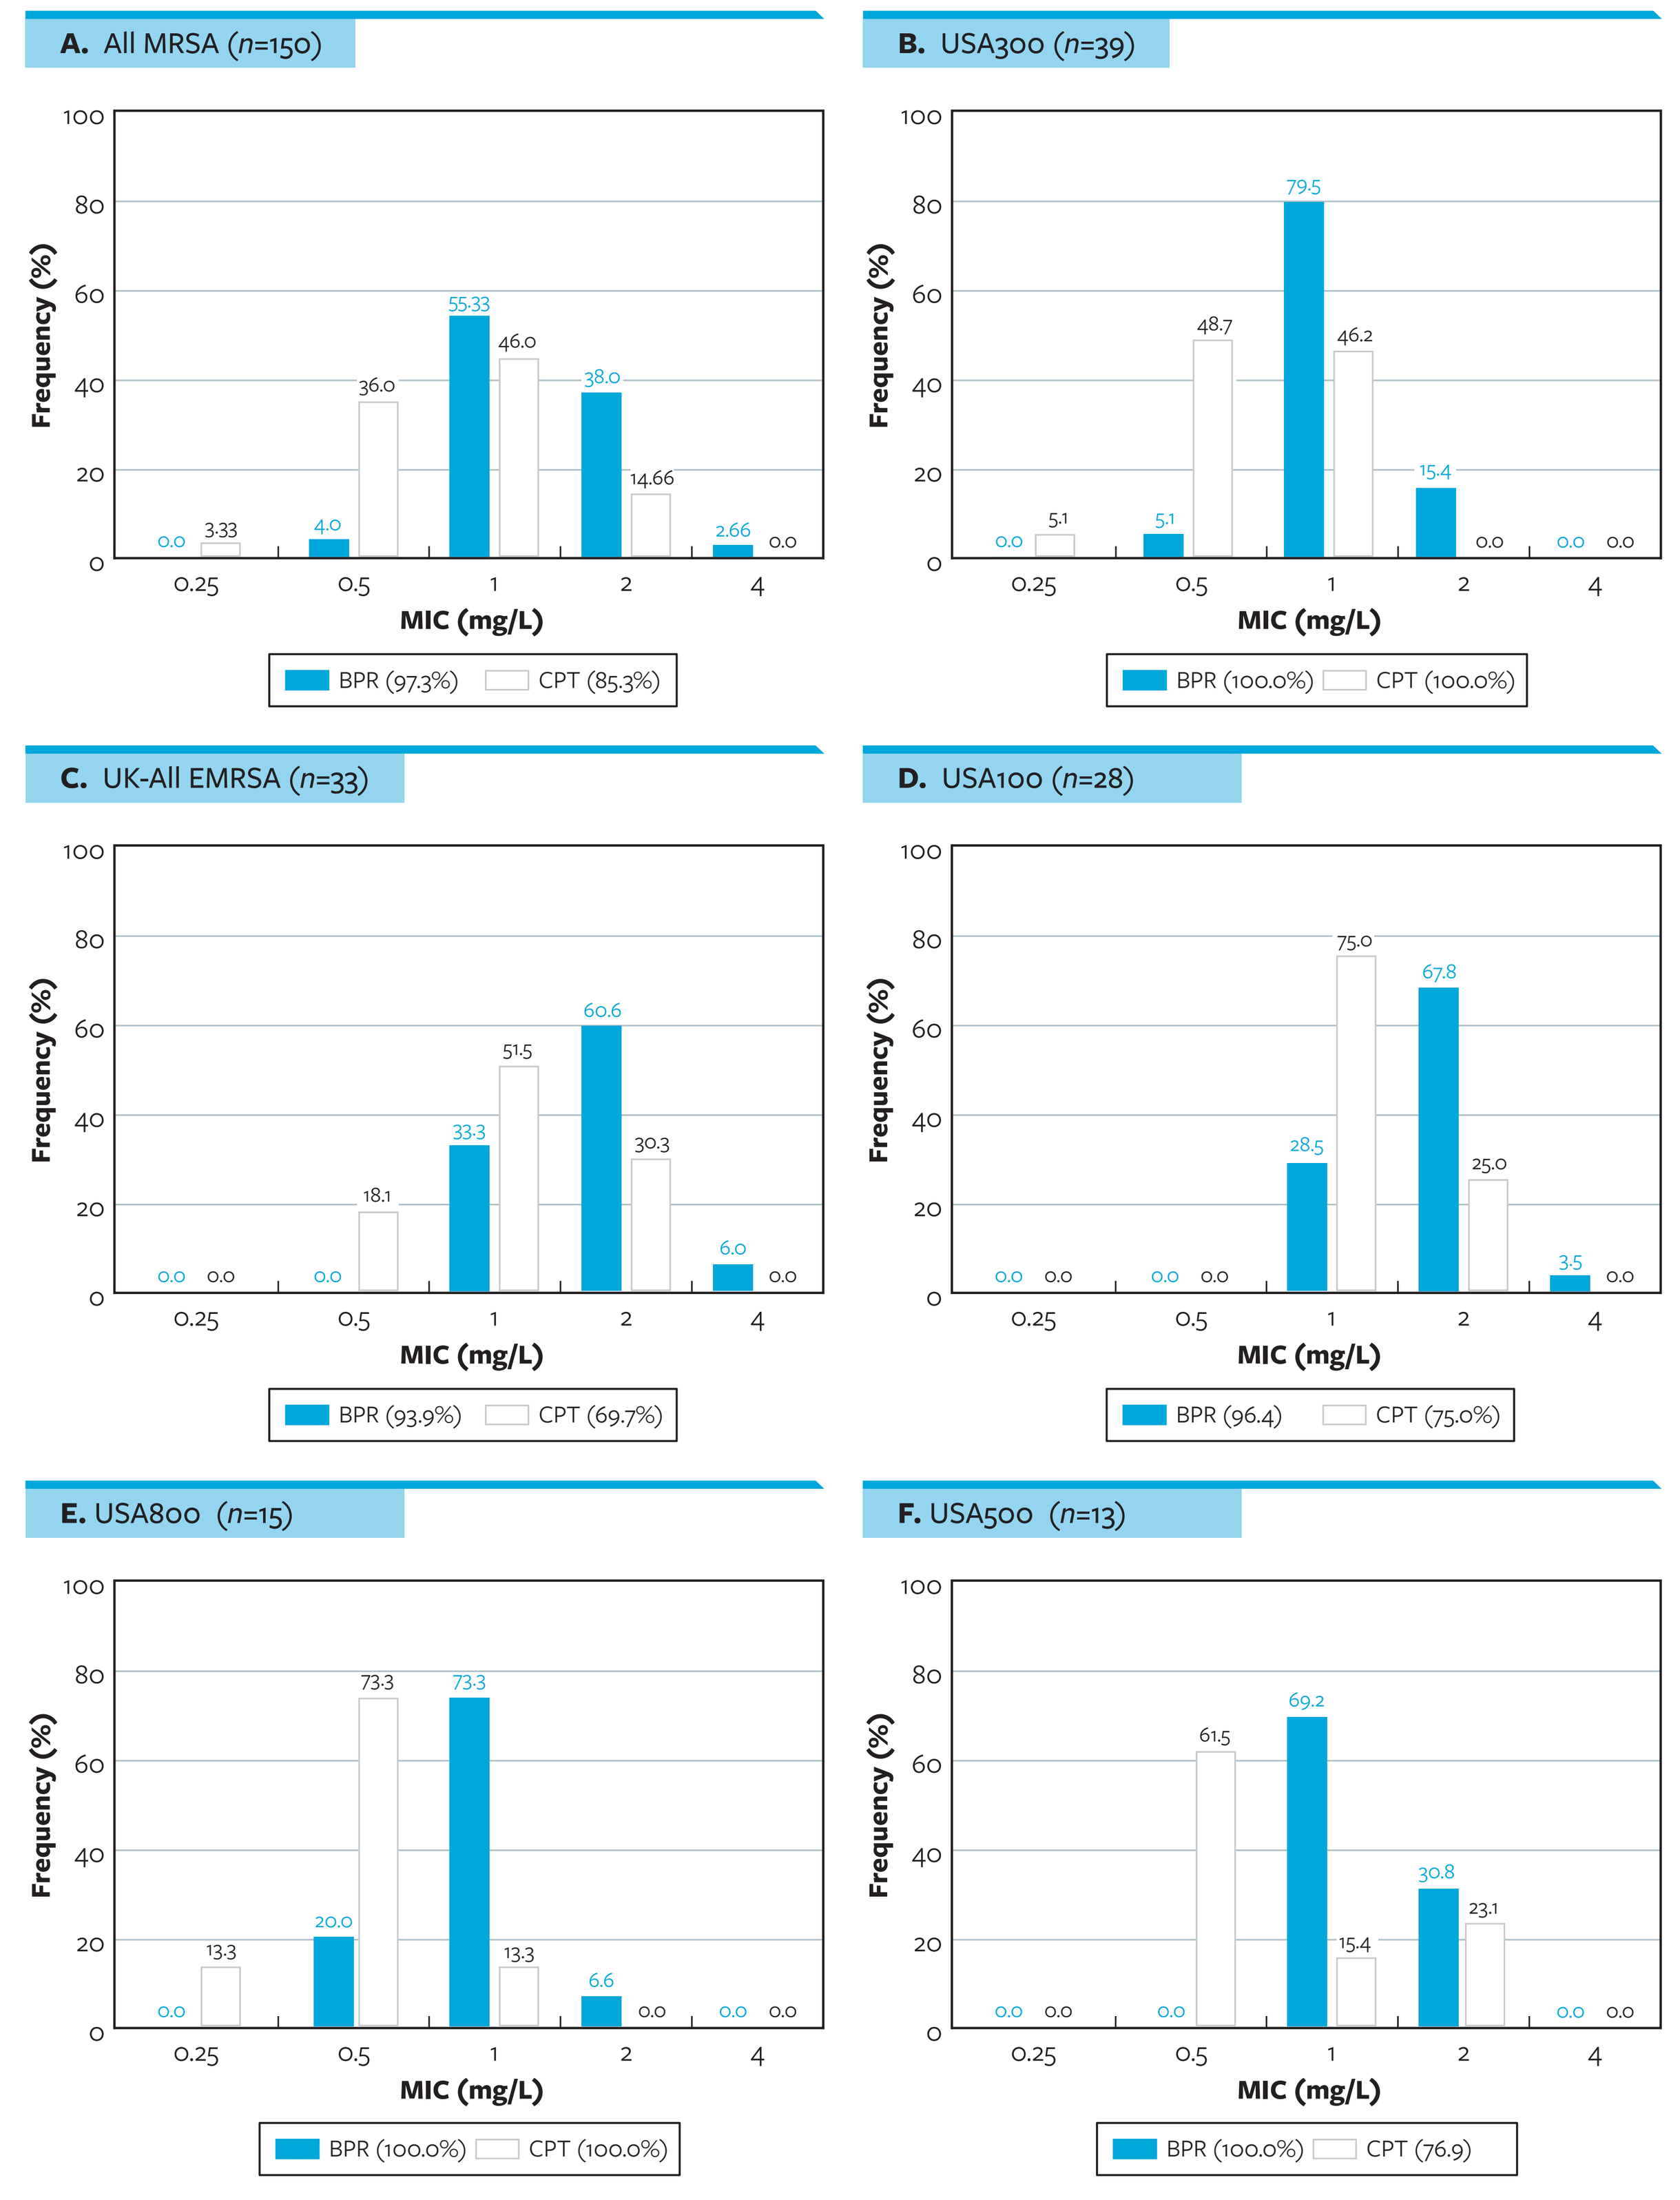

A. All MRSA (n=150)
BPR (97.3%) CPT (85.3%)
B. USA300 (n=39)
BPR (100.0%) CPT (100.0%)
C. UK-All EMRSA (n=33)
BPR (93.9%) CPT (69.7%)
D. USA100 (n=28)
BPR (96.4) CPT (75.0%)
E. USA800 (n=15)
BPR (100.0%) CPT (100.0%)
F. USA500 (n=13)
BPR (100.0%) CPT (76.9)
Frequency (%)
MIC (mg/L)

Percent of susceptible isolates within each group is displayed in parentheses for each agent (i.e., BPR-S%).

Table 2. Molecular characteristics of ceftobiprole and ceftaroline nonsusceptible MRSA isolates.

Country	BPR	CPT	CG[a]	ST	CC	SCC*mec* Type	*spa* Type
BPR-NS/CPT-NS							
Italy	4	2	UK-EMRSA-15	22	22	IV(2B)	t022
Italy	4	2	UK-EMRSA-15	22	22	IV(2B)	t022
Turkey	4	2	Hungarian/Brazilian	239	8	III(3A)	t030
USA	4	2	USA100-Swiss	105	5	II(2A)	t002
BPR-S/CPT-NS							
Belarus	2	2	USA500	8	8	IVc(2B)	t008
Belarus	2	2	USA500	8	8	IVc(2B)	t008
Belarus	2	2	USA500	8	8	IVc(2B)	t008
Italy	2	2	UK-EMRSA-15*	22	22	IV(2B)	*Novel*
Italy	2	2	UK-EMRSA-15	22	22	IV(2B)	t022
Italy	2	2	UK-EMRSA-15	22	22	IV(2B)	t022
Italy	2	2	UK-EMRSA-15*	22	22	IV(2B)	*Novel*
Italy	2	2	UK-EMRSA-15	22	22	IV(2B)	t022
Italy	2	2	UK-EMRSA-15	22	22	IV(2B)	t022
Italy	2	2	UK-EMRSA-15	22	22	IV(2B)	t022
Italy	2	2	UK-EMRSA-15*	22	22	IV(2B)	*t3720*
Mexico	2	2	USA100*	*1011-like*	5	II(2A)	*t895*
Philippines	2	2	Hungarian/Brazilian	239	8	III(3A)	*t1459*
USA	2	2	USA100*	5	5	II(2A)	*t668*
USA	2	2	USA100	5	5	II(2A)	t002
USA	2	2	USA100-Swiss	105	5	II(2A)	t002
USA	2	2	USA100-Swiss	105	5	II(2A)	t002
USA	2	2	USA100-Swiss	105	5	II(2A)	t002

Abbreviations: BPR, ceftobiprole; CPT, ceftaroline; NS, nonsusceptible; CG, clonal group; ST, sequence type; CC, clonal complex; SCC*mec*, staphylococcal chromosomal cassette *mec*.
[a] Clonal group assignment was performed based on combinations of ST, SCC*mec*, and *spa*-typing results. In cases where one or more typing results for any one genetic attribute differed from the canonical clonal group result, those isolates were assigned to the closest aligning group and denoted with an '*' and the divergent typing result shown in italics.

References

1. Pfaller MA, *et al. Antimicrob Agents Chemother* 2017. 63: e01566.
2. Flamm RK, *et al. Antimicrob Agents Chemother*. 2020 May 21;64(6):e02566-19. doi: 10.1128/AAC.02566-19.
3. Clinical and Laboratory Standards Institute (2018). *M07Ed11 Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically: eleventh edition*. Wayne, PA: CLSI.
4. Clinical and Laboratory Standards Institute (2024). *M100Ed34. Performance standards for antimicrobial susceptibility testing: 34th informational supplement*. Wayne, PA: CLSI.
5. US FDA. Antibacterial susceptibility test interpretive criteria. 2024.
6. Holland, TL, *et al. N Engl J Med*. 2023. 389(15):1390–1401.
7. Monecke S, *et al. PLoS One* 2011. 6: e17936
8. Kaya H, *et al. mSphere*. 2018 Feb 14;3(1):e00612-17. doi: 10.1128/mSphere.00612-17
9. Bartels MD, *et al. J. Clin. Microbiol*. 2014. 52(12): 4305–8.

Contact

Rodrigo E. Mendes, Ph.D., FIDSA
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo.mendes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2024_23-BAS-08_A1_MRSA_Clones_BPR_vs_TAR.pdf

Charges may apply. No personal information is stored.