IDWEEK 2024 | Poster #P1081

Activity of aztreonam-avibactam against Enterobacterales isolated from patients with intra-abdominal infection from United States medical centers (2019–2023)

Helio S. Sader, Timothy B. Doyle, Rodrigo E. Mendes, Marisa L. Winkler, Mariana Castanheira

Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA

CONCLUSIONS

Aztreonam-avibactam and the newer BLICs exhibited almost complete activity against Enterobacterales causing IAI in US hospitals.

Aztreonam-avibactam retained potent activity against CRE and MDR isolates.

Our results support clinical development of aztreonam-avibactam to treat IAI.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317 USA
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2024 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2024_24-ALG-02_A4_ATM-AVI_IAI.pdf

To submit a medical question, please visit www.abbviemedinfo.com

SCAN ME

Acknowledgements

This study at JMI Laboratories was supported by AbbVie Inc. JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by AbbVie Inc.

References

1. CLSI. 2024. M100Ed34. Performance standards for antimicrobial susceptibility testing: 34th informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.
2. Bonomo RA, Chow AW, Edwards MS, et al. (2024). 2024 Clinical practice guideline update by the Infectious Diseases Society of America on complicated intra-abdominal infections: Risk assessment, diagnostic imaging, and microbiological evaluation in adults, children, and pregnant people. *Clin Infect Dis.* ciae346.
3. Cornely OA, Cisneros JM, Torre-Cisneros J, et al. (2020). Pharmacokinetics and safety of aztreonam/avibactam for the treatment of complicated intra-abdominal infections in hospitalized adults: results from the REJUVENATE study. *Journal of Antimicrobial Chemotherapy* 75:618-627.
4. EUCAST. 2024. Breakpoint tables for interpretation of MICs and zone diameters. Available at https://www.eucast.org/clinical_breakpoints.
5. Sader HS, Mendes RE, Carvalhaes CG, Kimbrough JH, Castanheira M (2023). Changing epidemiology of carbapenemases among carbapenem-resistant Enterobacterales from United States hospitals and the activity of aztreonam-avibactam against contemporary Enterobacterales (2019-2021). *Open Forum Infect Dis.* 10(2):ofad046.

abbvie

INTRODUCTION

- Aztreonam-avibactam is under development in the United States (US) to treat infections caused by Gram-negative bacteria, including metallo-β-lactamase (MBL) producers.
- Aztreonam-avibactam has been recently approved by the European Medicine Agency (EMA) to treat adults who have complicated intra-abdominal infections (IAI), hospital-acquired pneumonia (including ventilator-associated pneumonia), complicated urinary tract infections (UTI; including pyelonephritis), and infections caused by aerobic Gram-negative organisms in patients who have limited treatment options.
- Phase 3 clinical trials REVISIT (NCT03329092) and ASSEMBLE (NCT03580044) evaluated the efficacy, safety, and tolerability of aztreonam-avibactam in treating serious bacterial infections due to Gram-negative bacteria, including MBL-producing multidrug-resistant pathogens for which there are limited or no treatment options.
- We evaluated the *in vitro* activities of aztreonam-avibactam and comparators against Gram-negative bacilli isolated from patients with intra-abdominal infection.

METHODS

- A total of 2,036 isolates (1/patient) were consecutively collected from patients with IAI in 63 US hospitals in 2019–2023.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Multidrug-resistant (MDR) phenotype was defined as non-susceptibility to at least one drug in ≥3 classes.
- Isolates were susceptibility tested by CLSI M07 broth microdilution methods at a central laboratory and aztreonam-avibactam was tested with avibactam at fixed 4 mg/L.
- The aztreonam-avibactam provisional pharmacodynamic/pharmacokinetic susceptible breakpoint of ≤8 mg/L was applied for comparison.
- Cefiderocol was only tested against carbapenem-resistant Enterobacterales (CRE).
- CRE isolates were screened for carbapenemase-encoding genes by whole genome sequencing.

Figure 1. Frequency of organisms isolated from patients with IAI

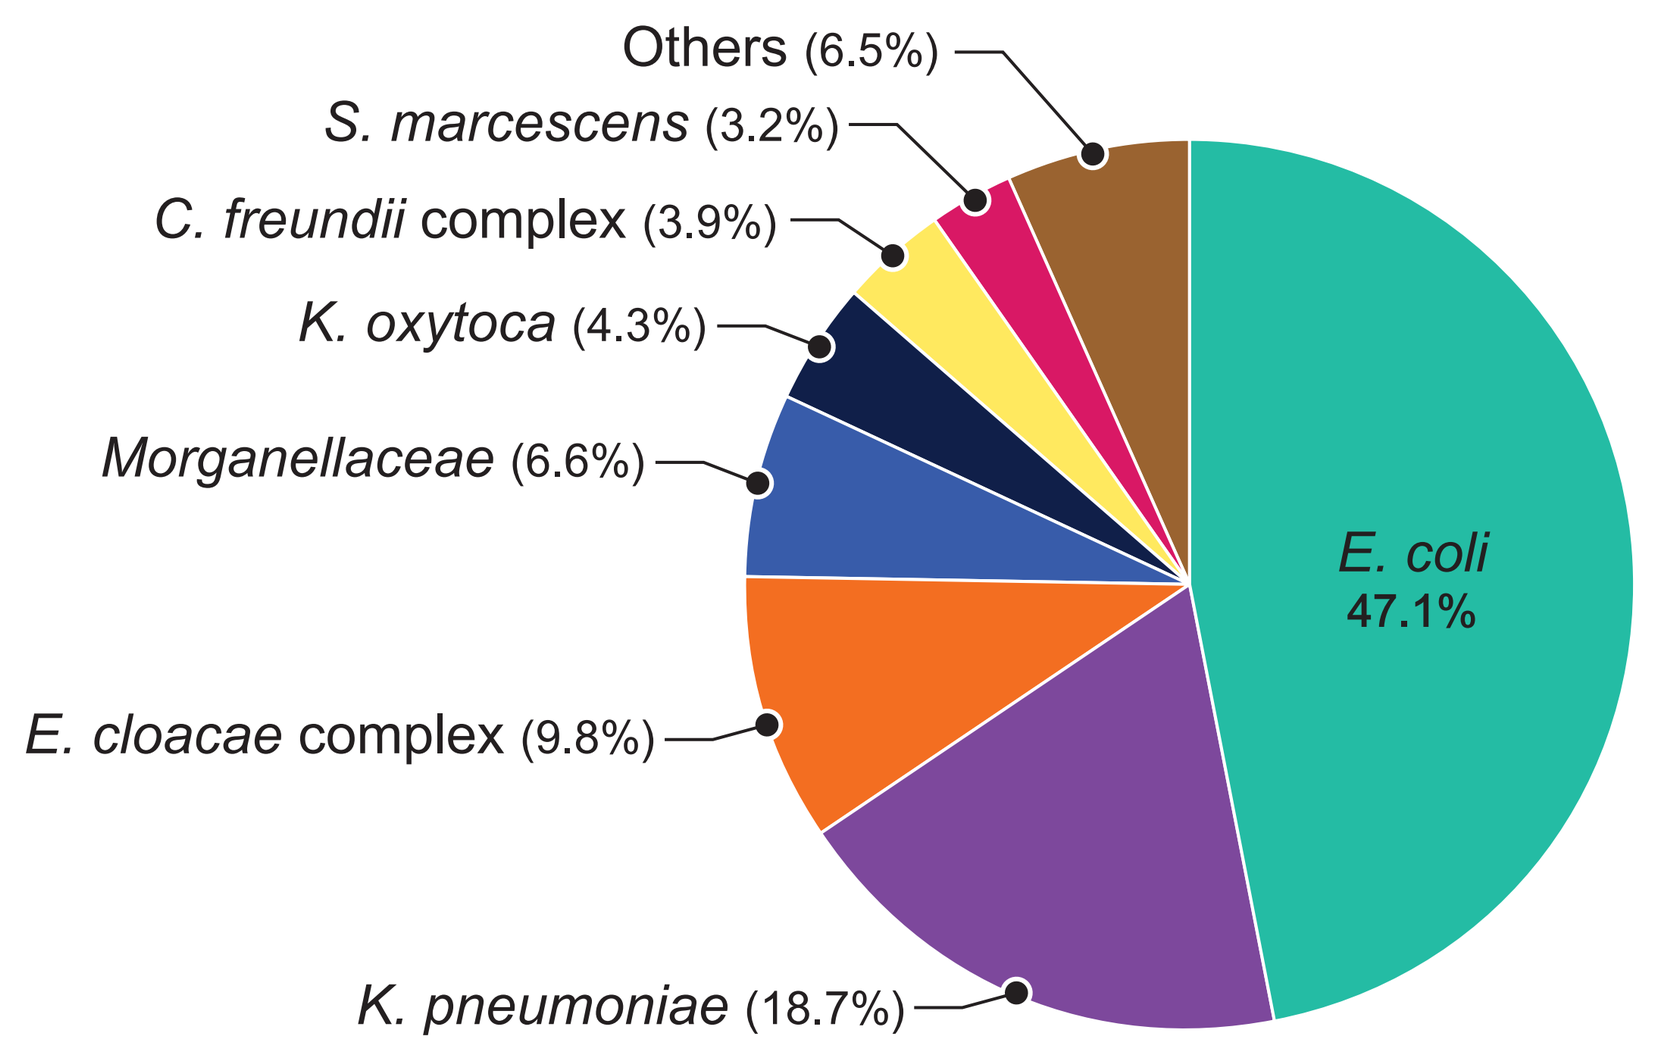
Others (6.5%)
S. marcescens (3.2%)
C. freundii complex (3.9%)
K. oxytoca (4.3%)
Morganellaceae (6.6%)
E. cloacae complex (9.8%)
K. pneumoniae (18.7%)
E. coli
47.1%

Figure 2. Antimicrobial susceptibility of Enterobacterales from patients with IAI

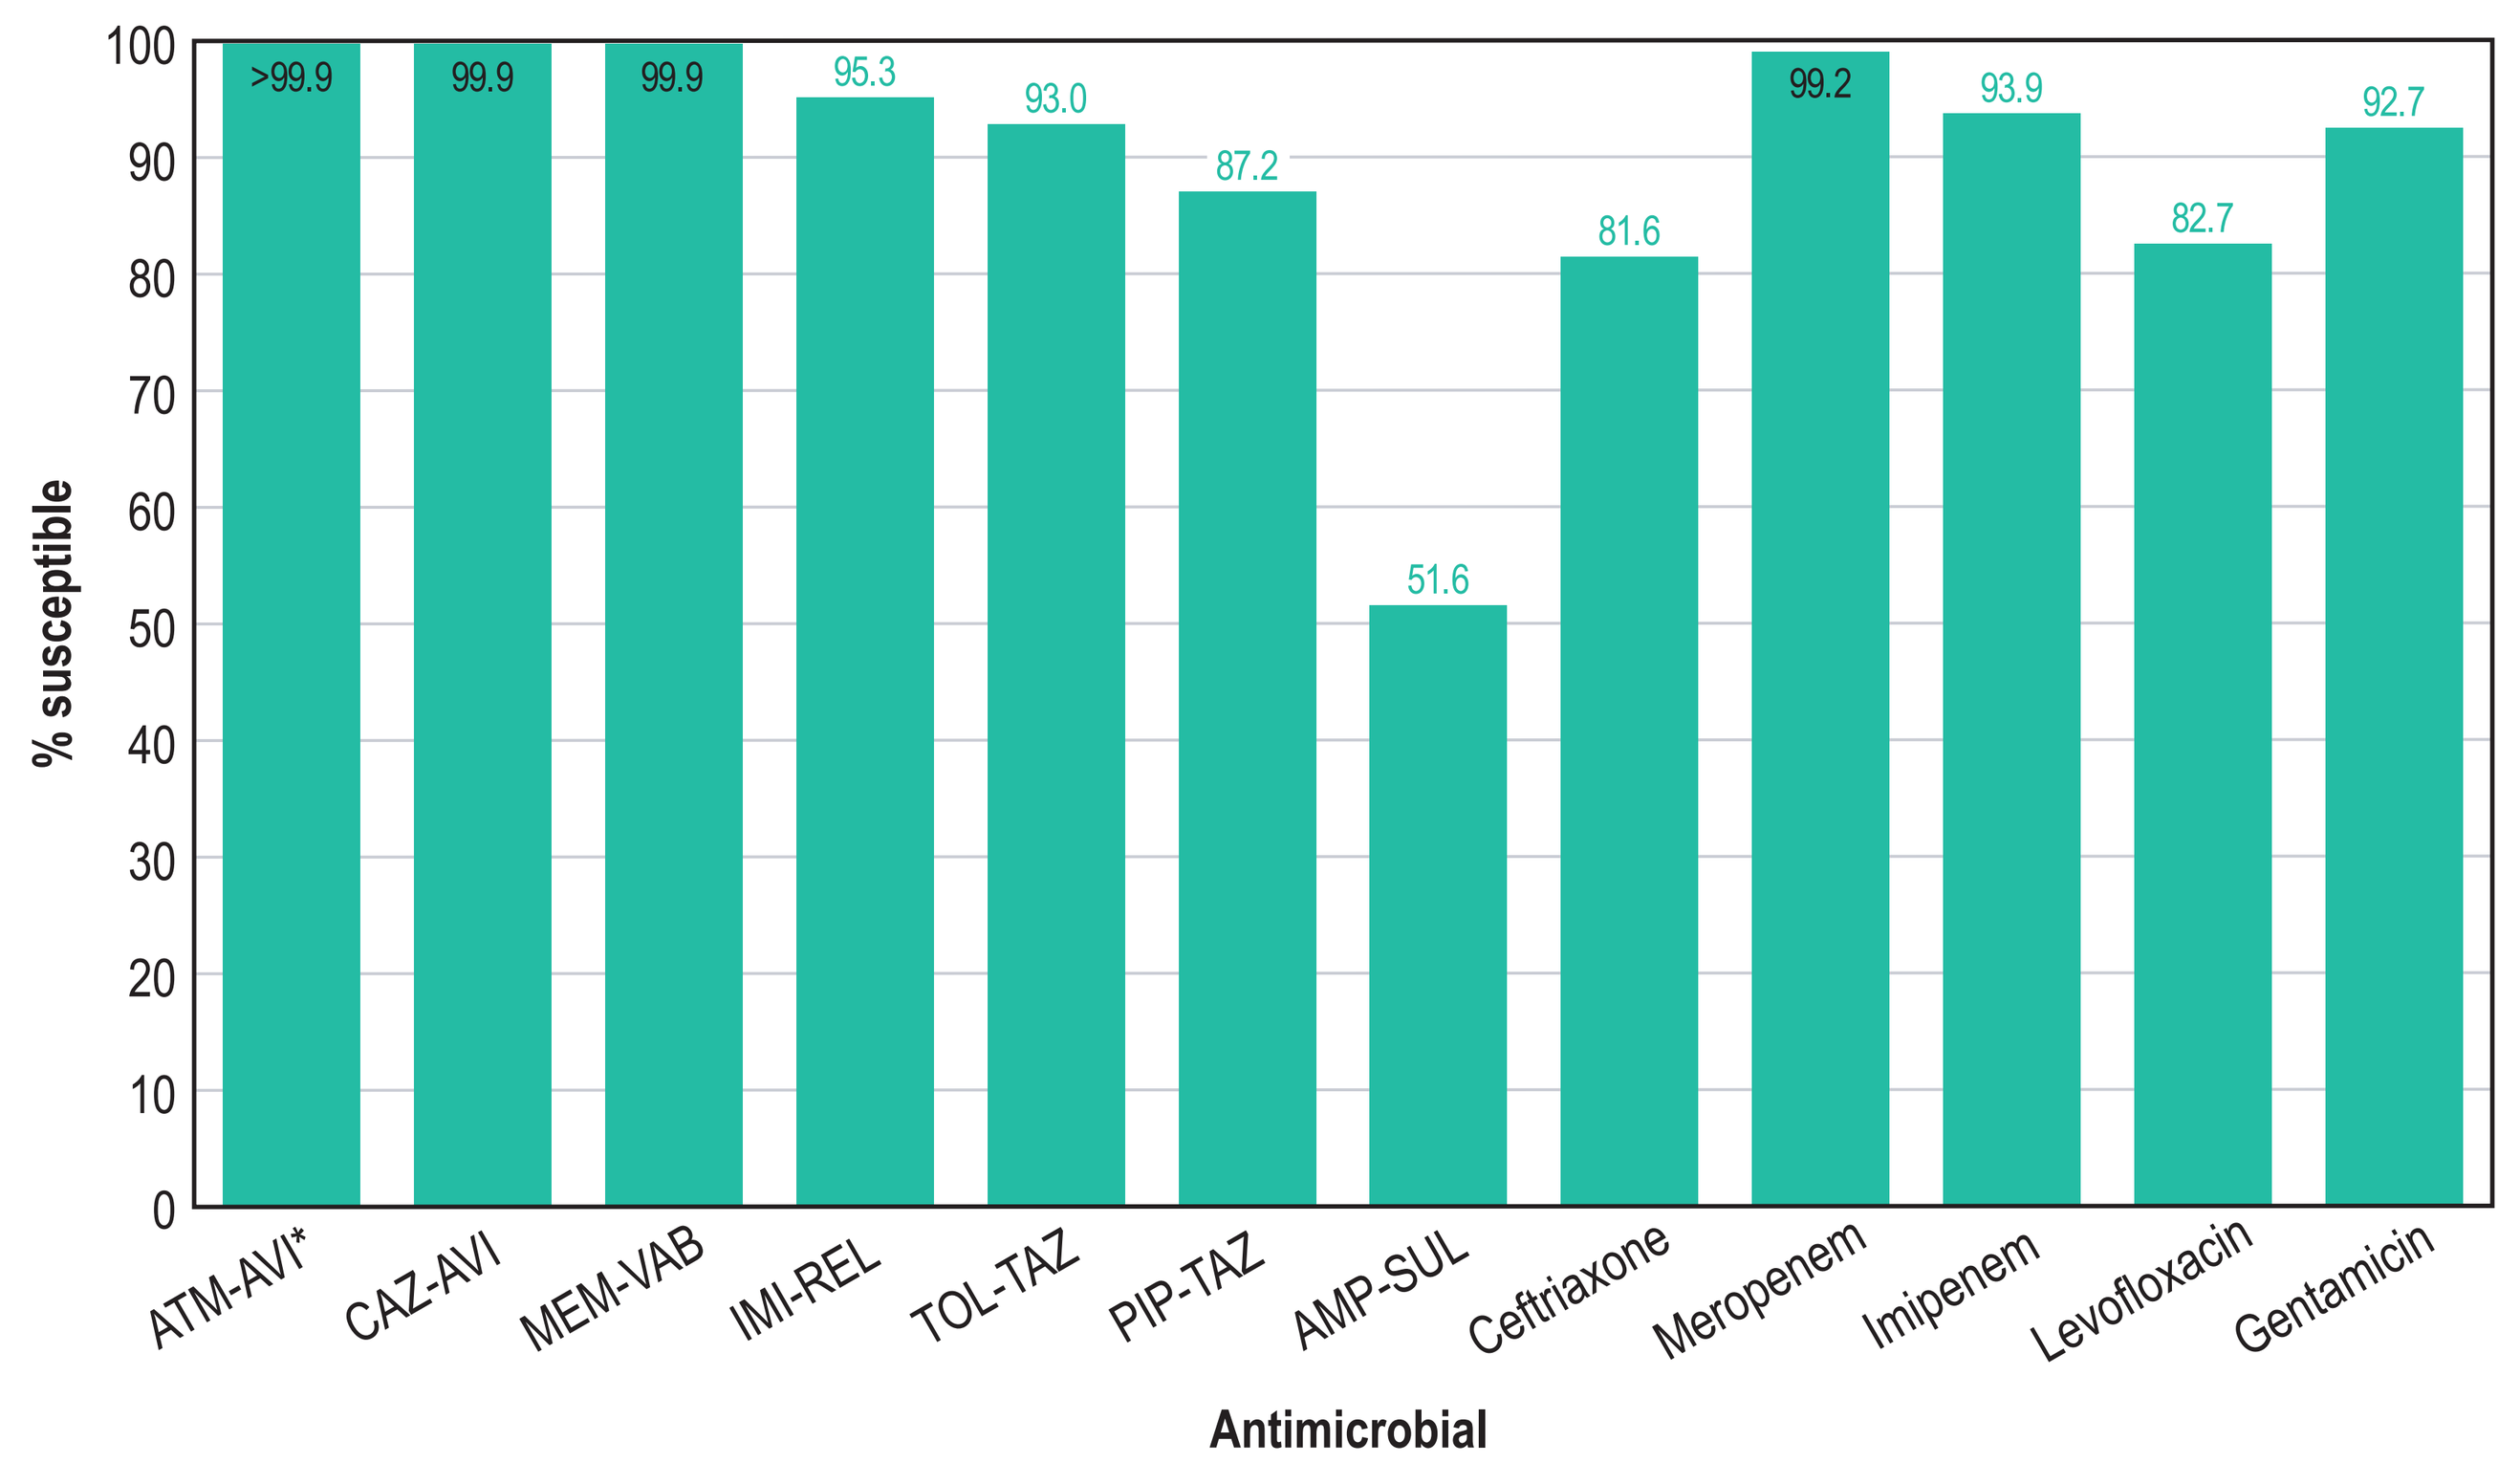
100
90
80
70
60
50
40
30
20
10
0
% susceptible
>99.9
99.9
99.9
95.3
93.0
87.2
51.6
81.6
99.2
93.9
82.7
92.7
ATM-AVI*
CAZ-AVI
MEM-VAB
IMI-REL
TOL-TAZ
PIP-TAZ
AMP-SUL
Ceftriaxone
Meropenem
Imipenem
Levofloxacin
Gentamicin
Antimicrobial

Abbreviations: ATM-AVI, aztreonam-avibactam; CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam; IMI-REL, imipenem-relebactam; TOL-TAZ, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam; AMP-SUL, ampicillin-sulbactam.
* Percentage inhibited at ≤8 mg/L.

RESULTS

- The most common Enterobacterales species were *E. coli* (47.1%), *K. pneumoniae* (18.7%), and *E. cloacae* species complex (9.8%; Figure 1).
- Only 1 Enterobacterales isolate exhibited aztreonam-avibactam MIC >8 mg/L ($MIC_{50/90}$, ≤0.03/0.12 mg/L), an *E. coli* with aztreonam-avibactam MIC of 16 mg/L.
- Susceptibility of Enterobacterales to aztreonam-avibactam and comparators is shown in Table 1 and Figure 2.
- The most active agents against CRE were aztreonam-avibactam (93.3% inhibited at ≤8 mg/L; Figure 3) and cefiderocol (93.3% susceptible [S]).
- Ceftazidime-avibactam, meropenem-vaborbactam, and imipenem-relebactam were active against 86.7% of CRE (Table 1 and Figure 3).
- A MDR phenotype was observed in 21.4% of Enterobacterales (*n*=436) and the most active β-lactamase inhibitor combinations (BLICs) against MDR isolates were aztreonam-avibactam (99.8% inhibited at ≤8 mg/L), meropenem-vaborbactam (99.5% S), and ceftazidime-avibactam (99.3% S; Table 1 and Figure 3).
- Piperacillin-tazobactam was active against 87.2% of Enterobacterales and 50.2% of MDR isolates (Table 1 and Figures 2 and 3).
- Meropenem was active against 92.2% of Enterobacterales and 96.1% of MDR isolates (Figures 2 and 3).
- A carbapenemase was identified in 9 isolates (60.0% of CRE), and included KPC-2 (3 isolates), KPC-3 (4), and NDM-5 (2; Figure 4).
- All carbapenemase producers were inhibited at ≤8 mg/L of aztreonam-avibactam (MIC_{90}, 0.12 mg/L) and were susceptible to cefiderocol (MIC_{90}, 2 mg/L), whereas susceptibility to ceftazidime-avibactam, meropenem-vaborbactam, and imipenem-relebactam was 77.8% (data not shown).

Table 1. Activity of β-lactamase inhibitor combinations against Enterobacterales from intra-abdominal infections

Organism / group (no.)	% Susceptible (MIC_{90} in mg/L)					
	ATM-AVI[a]	CAZ-AVI	MEM-VAB	IMI-REL	PIP-TAZ	AMP-SUL
Enterobacterales (2,036)	>99.9 (0.12)	99.9 (0.25)	99.9 (0.06)	95.3 (0.5)	87.2 (16)	51.6 (64)
CRE (15)	93.3 (8)	86.7 (32)	86.7 (32)	86.7 (4)	0.0 (>128)	0.0 (>64)
MDR (436)	99.8 (0.5)	99.3 (1)	99.5 (0.6)	99.2 (0.25)	50.2 (>128)	4.6 (>64)
E. coli (958)	99.9 (0.12)	99.8 (0.25)	99.8 (0.03)	99.6 (0.12)	93.4 (8)	54.2 (64)
K. pneumoniae (380)	100.0 (0.12)	100.0 (0.25)	100.0 (0.03)	100.0 (0.25)	86.8 (16)	73.2 (16)
E. cloacae complex (199)	100.0 (1)	99.5 (1)	100.0 (0.06)	100.0 (0.25)	61.3 (>128)	12.6 (>64)
Morganellaceae (135)[b]	100.0 (≤0.03)	100.0 (0.06)	100.0 (0.12)	35.8 (2)	100.0 (1)	59.3 (8)
K. oxytoca (88)	100.0 (0.06)	100.0 (0.25)	100.0 (0.03)	100.0 (0.25)	94.3 (8)	34.1 (32)
C. freundii complex (79)	100.0 (0.25)	100.0 (0.5)	100.0 (0.03)	100.0 (0.12)	69.6 (128)	57.0 (>64)
S. marcescens (65)	100.0 (0.25)	100.0 (0.5)	100.0 (0.06)	100.0 (1)	92.3 (8)	7.7 (>64)
Other species (132)	100.0 (0.25)	100.0 (0.5)	100.0 (0.12)	100.0 (0.25)	72.7 (64)	51.5 (64)

[a] % inhibited at ≤8 mg/L.
[b] Includes: *Morganella morganii* (33), *Proteus hauseri* (1), *P. mirabilis* (80), *P. vulgaris* group (17), *Providencia alcalifaciens* (1), *P. rettgeri* (2), and *P. stuartii* (1).
Abbreviations: ATM-AVI, aztreonam-avibactam; CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam; IMI-REL, imipenem-relebactam; PIP-TAZ, piperacillin-tazobactam; AMP-SUL, ampicillin-sulbactam; CRE, carbapenem-resistant Enterobacterales; MDR, multidrug-resistant.

Figure 3. Antimicrobial susceptibility of CRE and MDR isolates

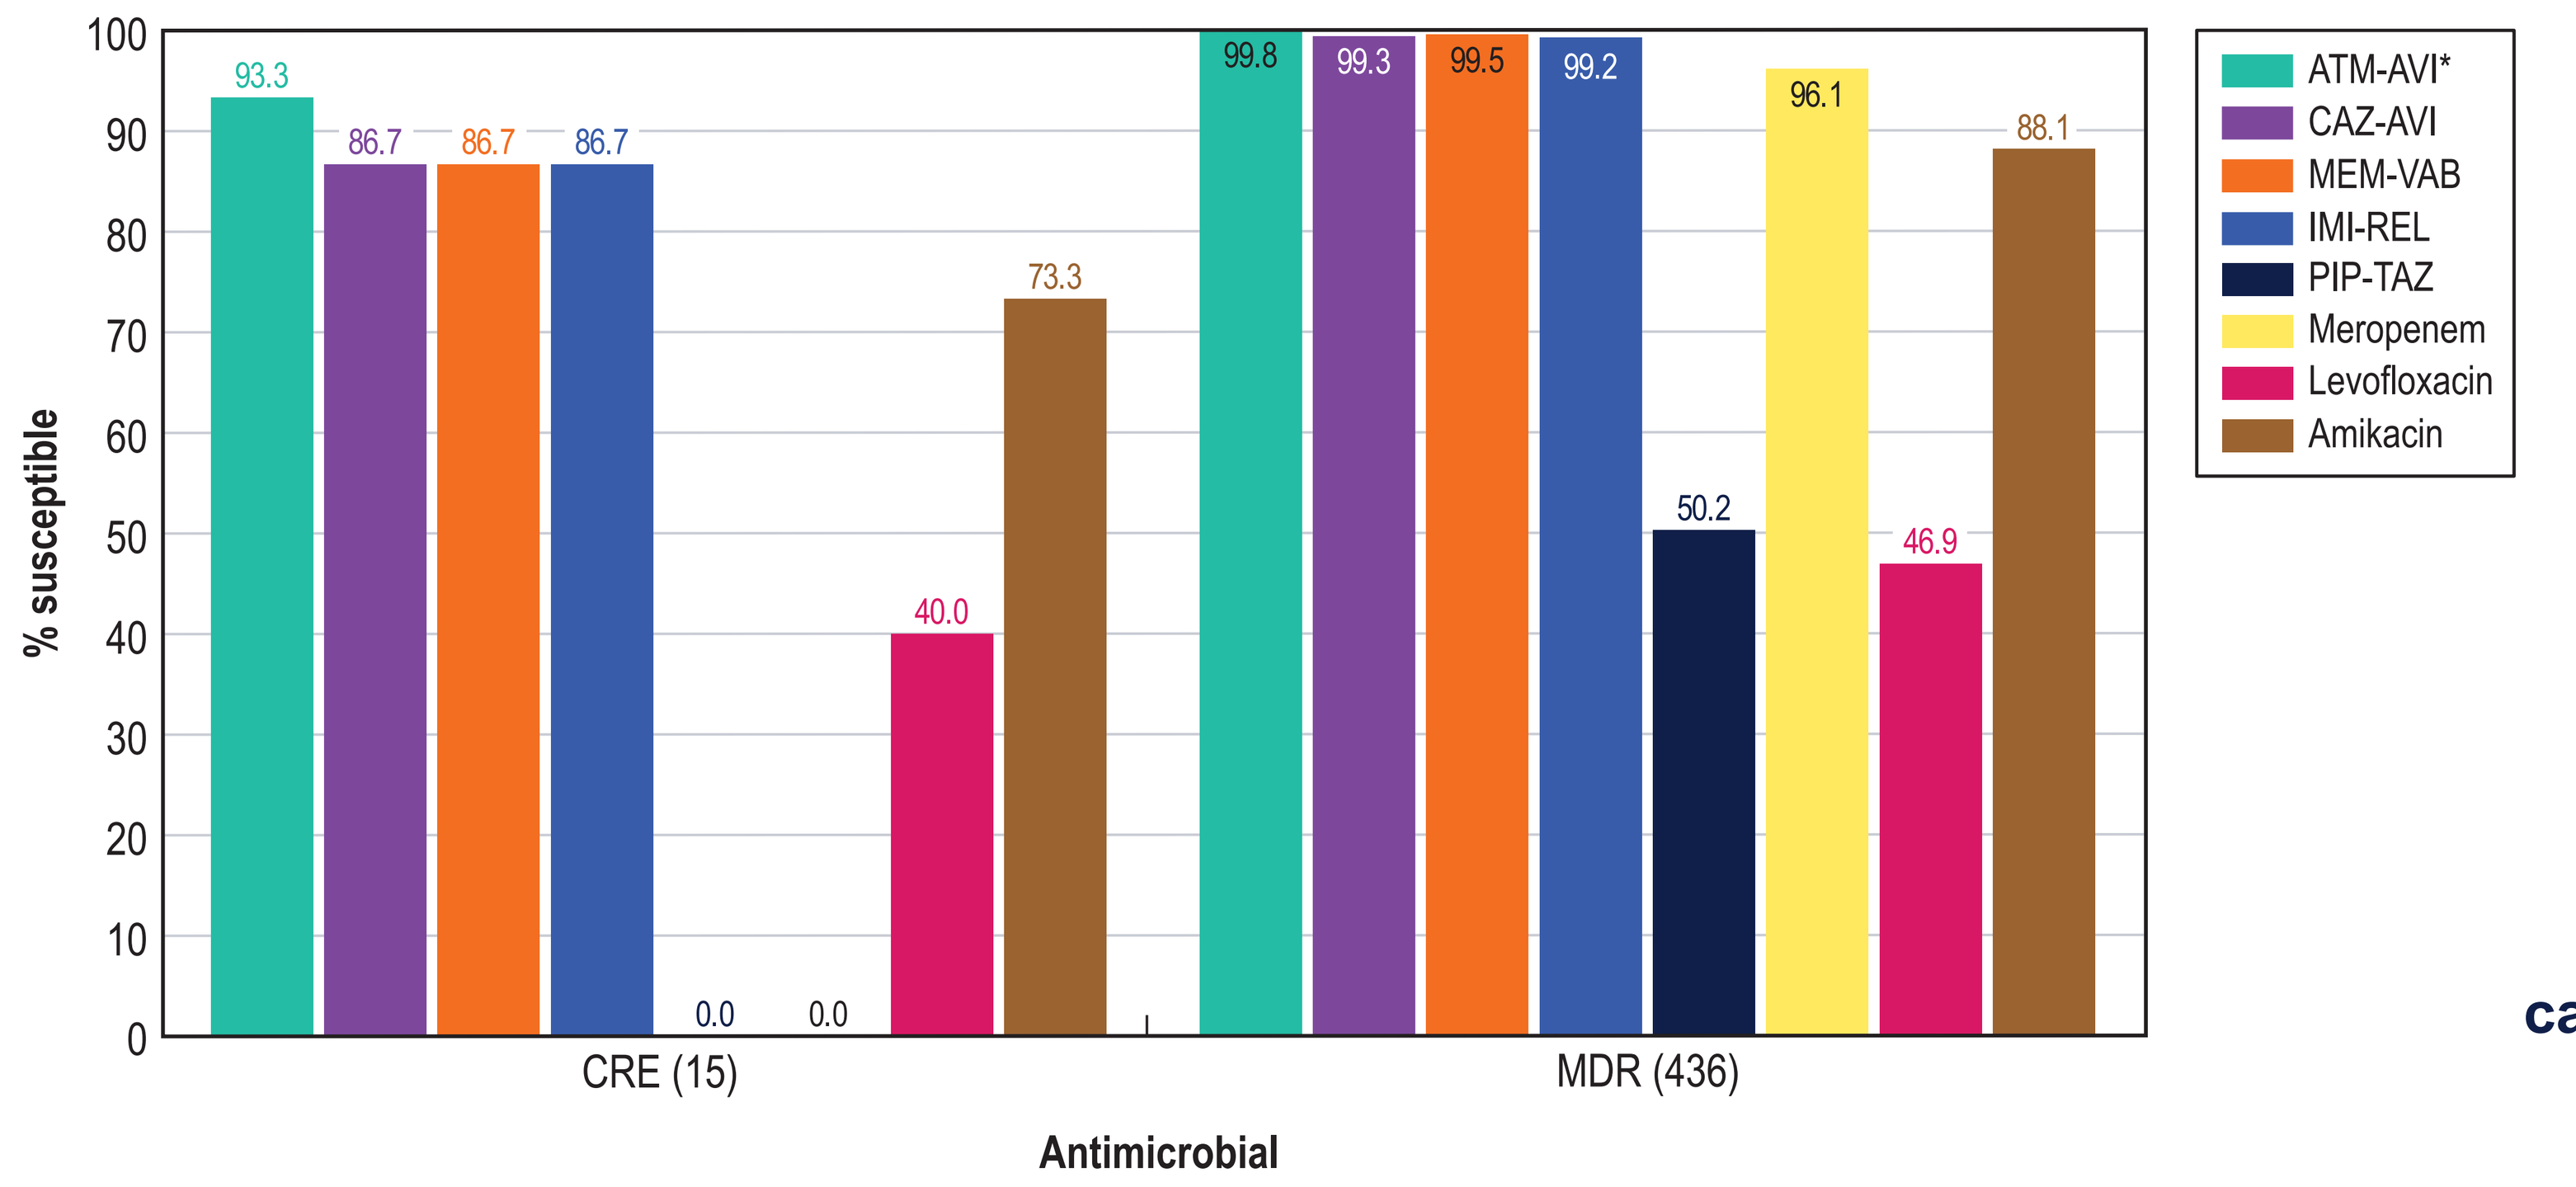
100
90
80
70
60
50
40
30
20
10
0
% susceptible
93.3
86.7
86.7
86.7
0.0
0.0
40.0
73.3
99.8
99.3
99.5
99.2
50.2
96.1
46.9
88.1
CRE (15)
MDR (436)
Antimicrobial
ATM-AVI*
CAZ-AVI
MEM-VAB
IMI-REL
PIP-TAZ
Meropenem
Levofloxacin
Amikacin

Abbreviation: CRE, carbapenem-resistant Enterobacterales; MDR, multidrug-resistant; ATM-AVI, aztreonam-avibactam; CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam; IMI-REL, imipenem-relebactam; PIP-TAZ, piperacillin-tazobactam.
* Percentage inhibited at ≤8 mg/L.

Figure 4. Frequencies of carbapenemases (CBase) among CRE isolates

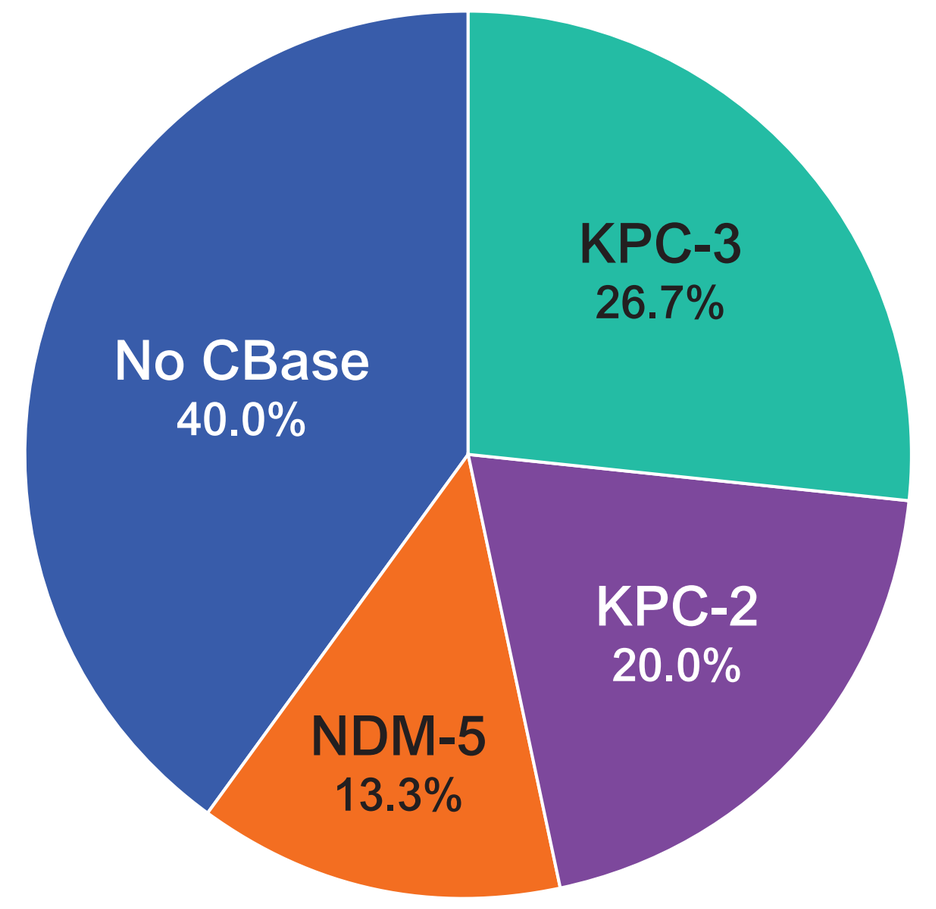
KPC-3
26.7%
KPC-2
20.0%
NDM-5
13.3%
No CBase
40.0%