IDWEEK 2024 | Poster #P1082

Antimicrobial Susceptibility of Enterobacterales Causing Infection in the Elderly: Focus on Aztreonam-Avibactam and Recently Approved β-Lactamase Inhibitor Combinations

Helio S. Sader, Rodrigo E. Mendes, Timothy B. Doyle, Marisa L. Winkler, Mariana Castanheira

Element Iowa City (JMI Laboratories), North Liberty, IA, USA

CONCLUSIONS

ATM-AVI demonstrated complete activity (100.0% inhibited at ≤8 mg/L) against Enterobacterales causing infection in the elderly and retained potent activity against CRE, including MBL producers, and isolates with ESBL or MDR phenotypes.

In general, susceptibility rates of isolates from the elderly were comparable (+/- <2%) to those from the adult population.

The frequency of MBL producers appears to be increasing rapidly and represented a significant proportion of CRE isolates from elderly (24.7%) and adult (24.8%) patients.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317 USA
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2024 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2024_24-ALG-02_A5_ATM-AVI_Elderly.pdf

To submit a medical question, please visit www.abbviemedinfo.com

SCAN ME

Acknowledgments

The authors thank all the participant centers for their work in providing isolates.

Funding

This study was supported by AbbVie. RE Mendes, HS Sader, TB Doyle, ML Winkler, and M Castanheira are employees of Element Iowa City (JMI Laboratories), which was paid consultant to AbbVie in connection with the development of this poster.

References

1. CLSI. 2024. M100Ed34. Performance standards for antimicrobial susceptibility testing: 34th informational supplement. *Clinical and Laboratory Standards Institute*, Wayne, PA.
2. Cornely OA, Cisneros JM, Torre-Cisneros J, et al. (2020). Pharmacokinetics and safety of aztreonam/avibactam for the treatment of complicated intra-abdominal infections in hospitalized adults: results from the REJUVENATE study. *J Antimicrob Chemother.* 75:618-627.
3. Sader HS, Mendes RE, Carvalhaes CG, Kimbrough JH, Castanheira M (2023). Changing epidemiology of carbapenemases among carbapenem-resistant Enterobacterales from United States hospitals and the activity of aztreonam-avibactam against contemporary Enterobacterales (2019-2021). *Open Forum Infect Dis.* 10(2):ofad046.
4. Tamma PD, Aitken SL, Bonomo RA, Mathers AJ, van Duin D, Clancy CJ (2023). Infectious Diseases Society of America 2023 guidance on the treatment of antimicrobial resistant Gram-negative infections. *Clin Infect Dis.* ciad428.

abbvie

INTRODUCTION

- Infections account for about 1/3 of all deaths in people 65 years and older and this population has a higher risk of exposure to antimicrobial-resistant bacteria.
- Aztreonam-avibactam (ATM-AVI) is under development in the United States (US) to treat infections caused by Gram-negative bacteria, including metallo-β-lactamase (MBL) producers.
- Moreover, ATM-AVI has been recently approved by the European Medicine Agency (Emblaveo®) to treat adults who have complicated intra-abdominal infections (IAI), hospital-acquired pneumonia (including ventilator-associated pneumonia), and complicated urinary tract infections (UTI; including pyelonephritis), as well as infections caused by aerobic Gram-negative organisms in patients who have limited treatment options.
- We evaluated the antimicrobial susceptibility of Enterobacterales causing infection in elderly patients in US medical centers.

METHODS

- Clinical isolates (1/patient) were consecutively collected from 72 US medical centers in 2021–2023 and susceptibility tested by CLSI broth microdilution.
- Results for 10,574 Enterobacterales isolates from elderly patients (≥65 years old) were analyzed and compared to 9,793 isolates from adult patients (age 18–64).
- Comparator agents included ceftazidime-avibactam (CAZ-AVI), meropenem-vaborbactam (MEM-VAB), imipenem-relebactam (IMI-REL), and cefiderocol (CRE only), among others.
- Cefiderocol was only tested against carbapenem-resistant Enterobacterales (CRE) in iron depleted media.
- Results were also stratified by infection type and resistant subsets.
- ATM-AVI was tested with avibactam at fixed 4 mg/L and a PK/PD susceptible (S) breakpoint of ≤8 mg/L was applied for comparison.
- Multidrug-resistant (MDR) phenotype was defined as non-susceptibility to at least one drug in ≥3 classes.
- CRE isolates were screened for carbapenemases (CBase) by whole genome sequencing.

Figure 1. Distribution of isolates by infection sources

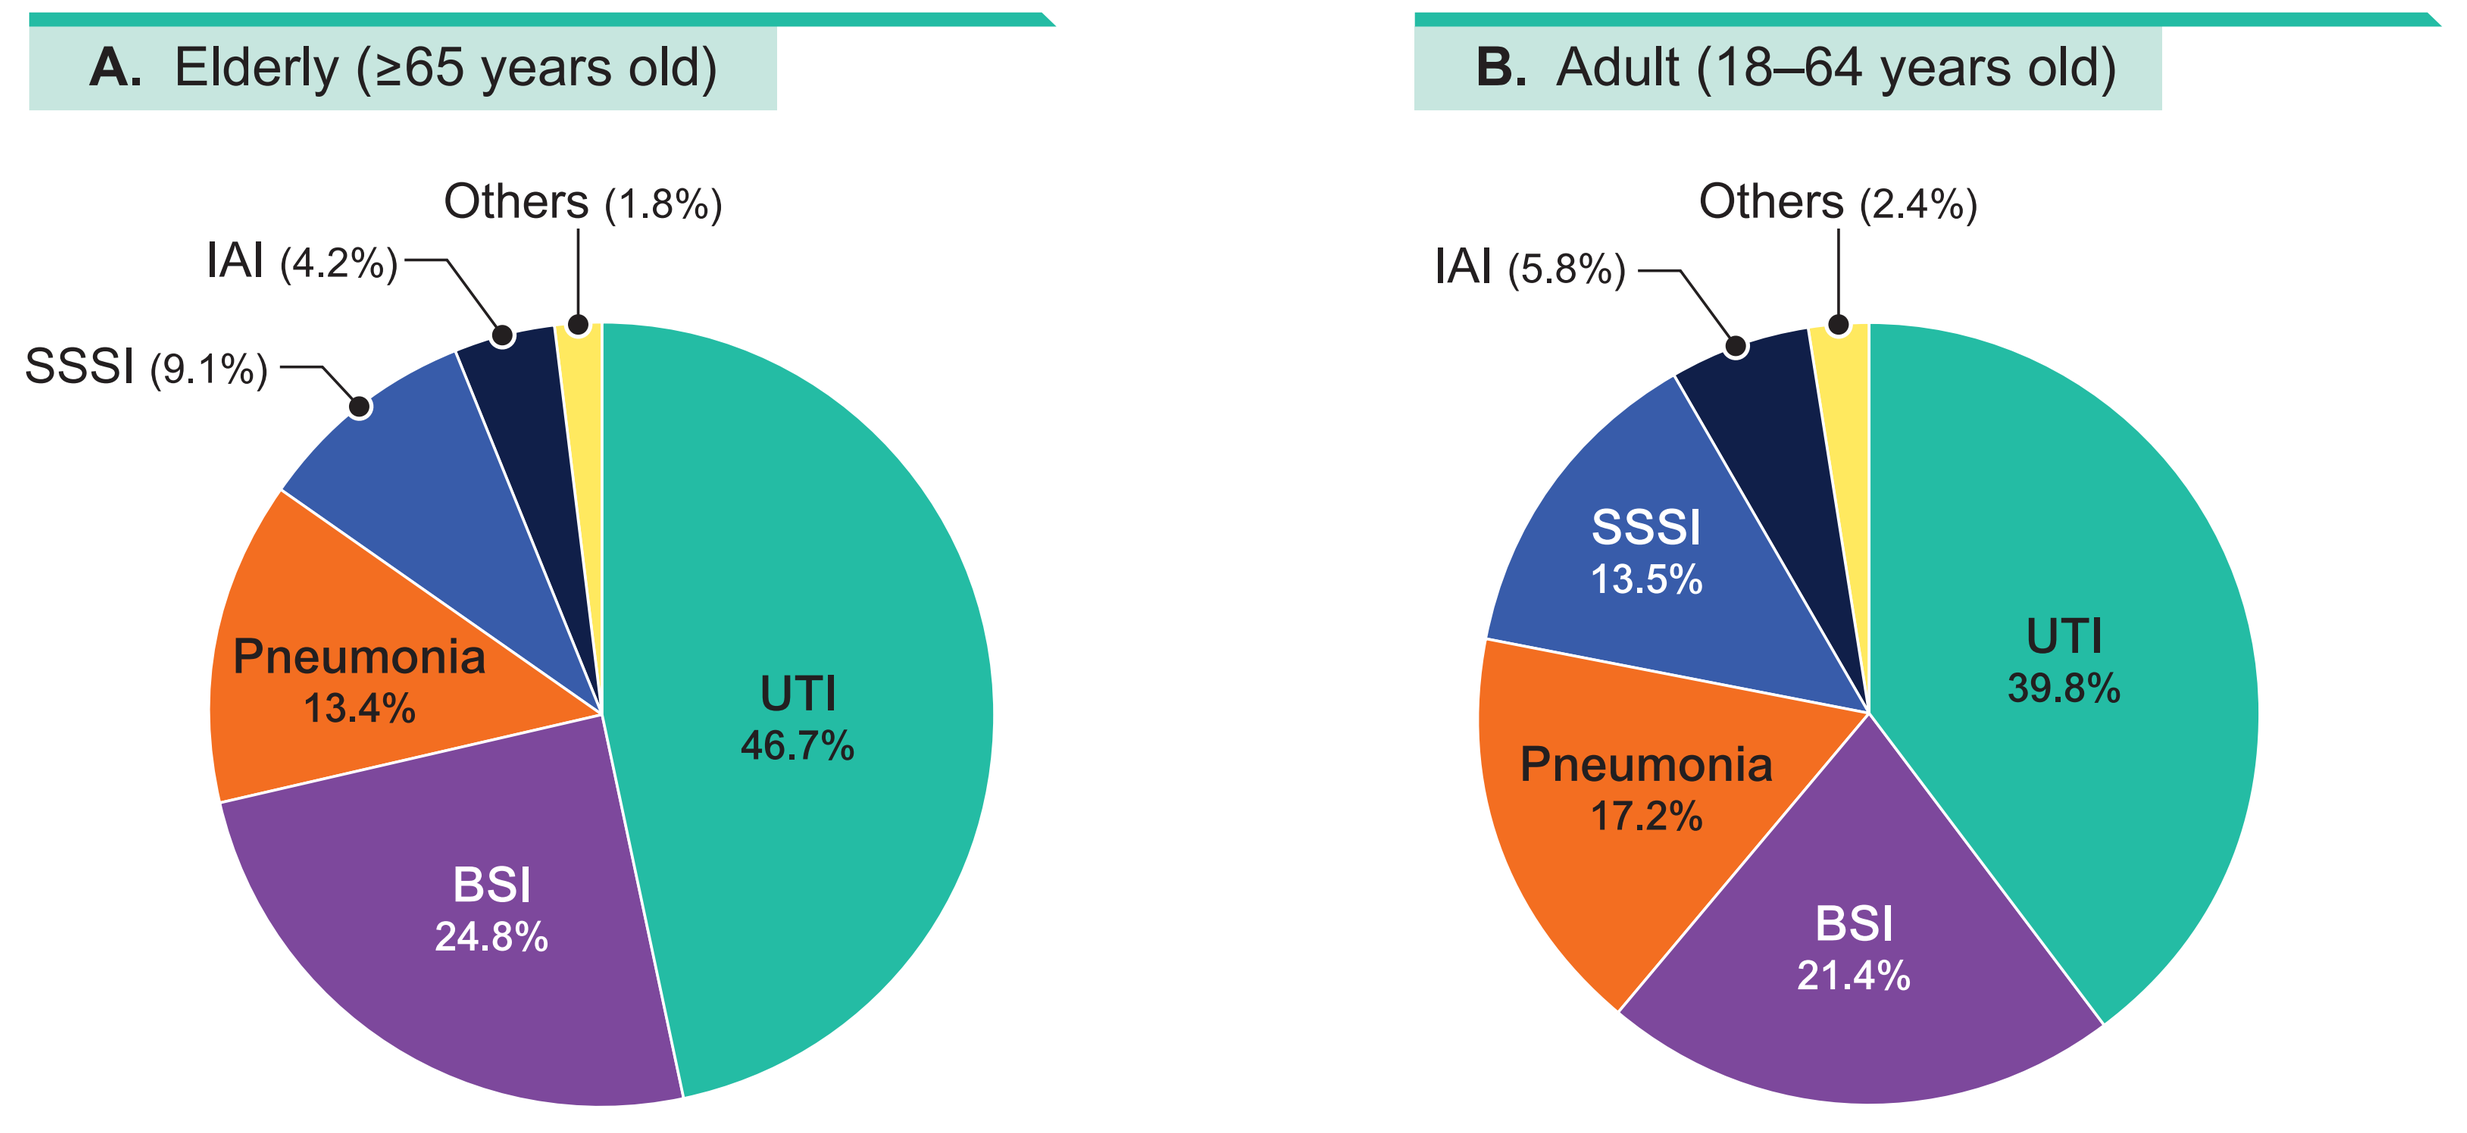
A. Elderly (≥65 years old)
UTI 46.7%
BSI 24.8%
Pneumonia 13.4%
SSSI (9.1%)
IAI (4.2%)
Others (1.8%)
B. Adult (18–64 years old)
UTI 39.8%
BSI 21.4%
Pneumonia 17.2%
SSSI 13.5%
IAI (5.8%)
Others (2.4%)

Abbreviations: UTI, urinary tract infection; BSI, bloodstream infection; SSSI, skin and skin structure infection; IAI, intra-abdominal infection.

Figure 2. Susceptibility of CRE isolates

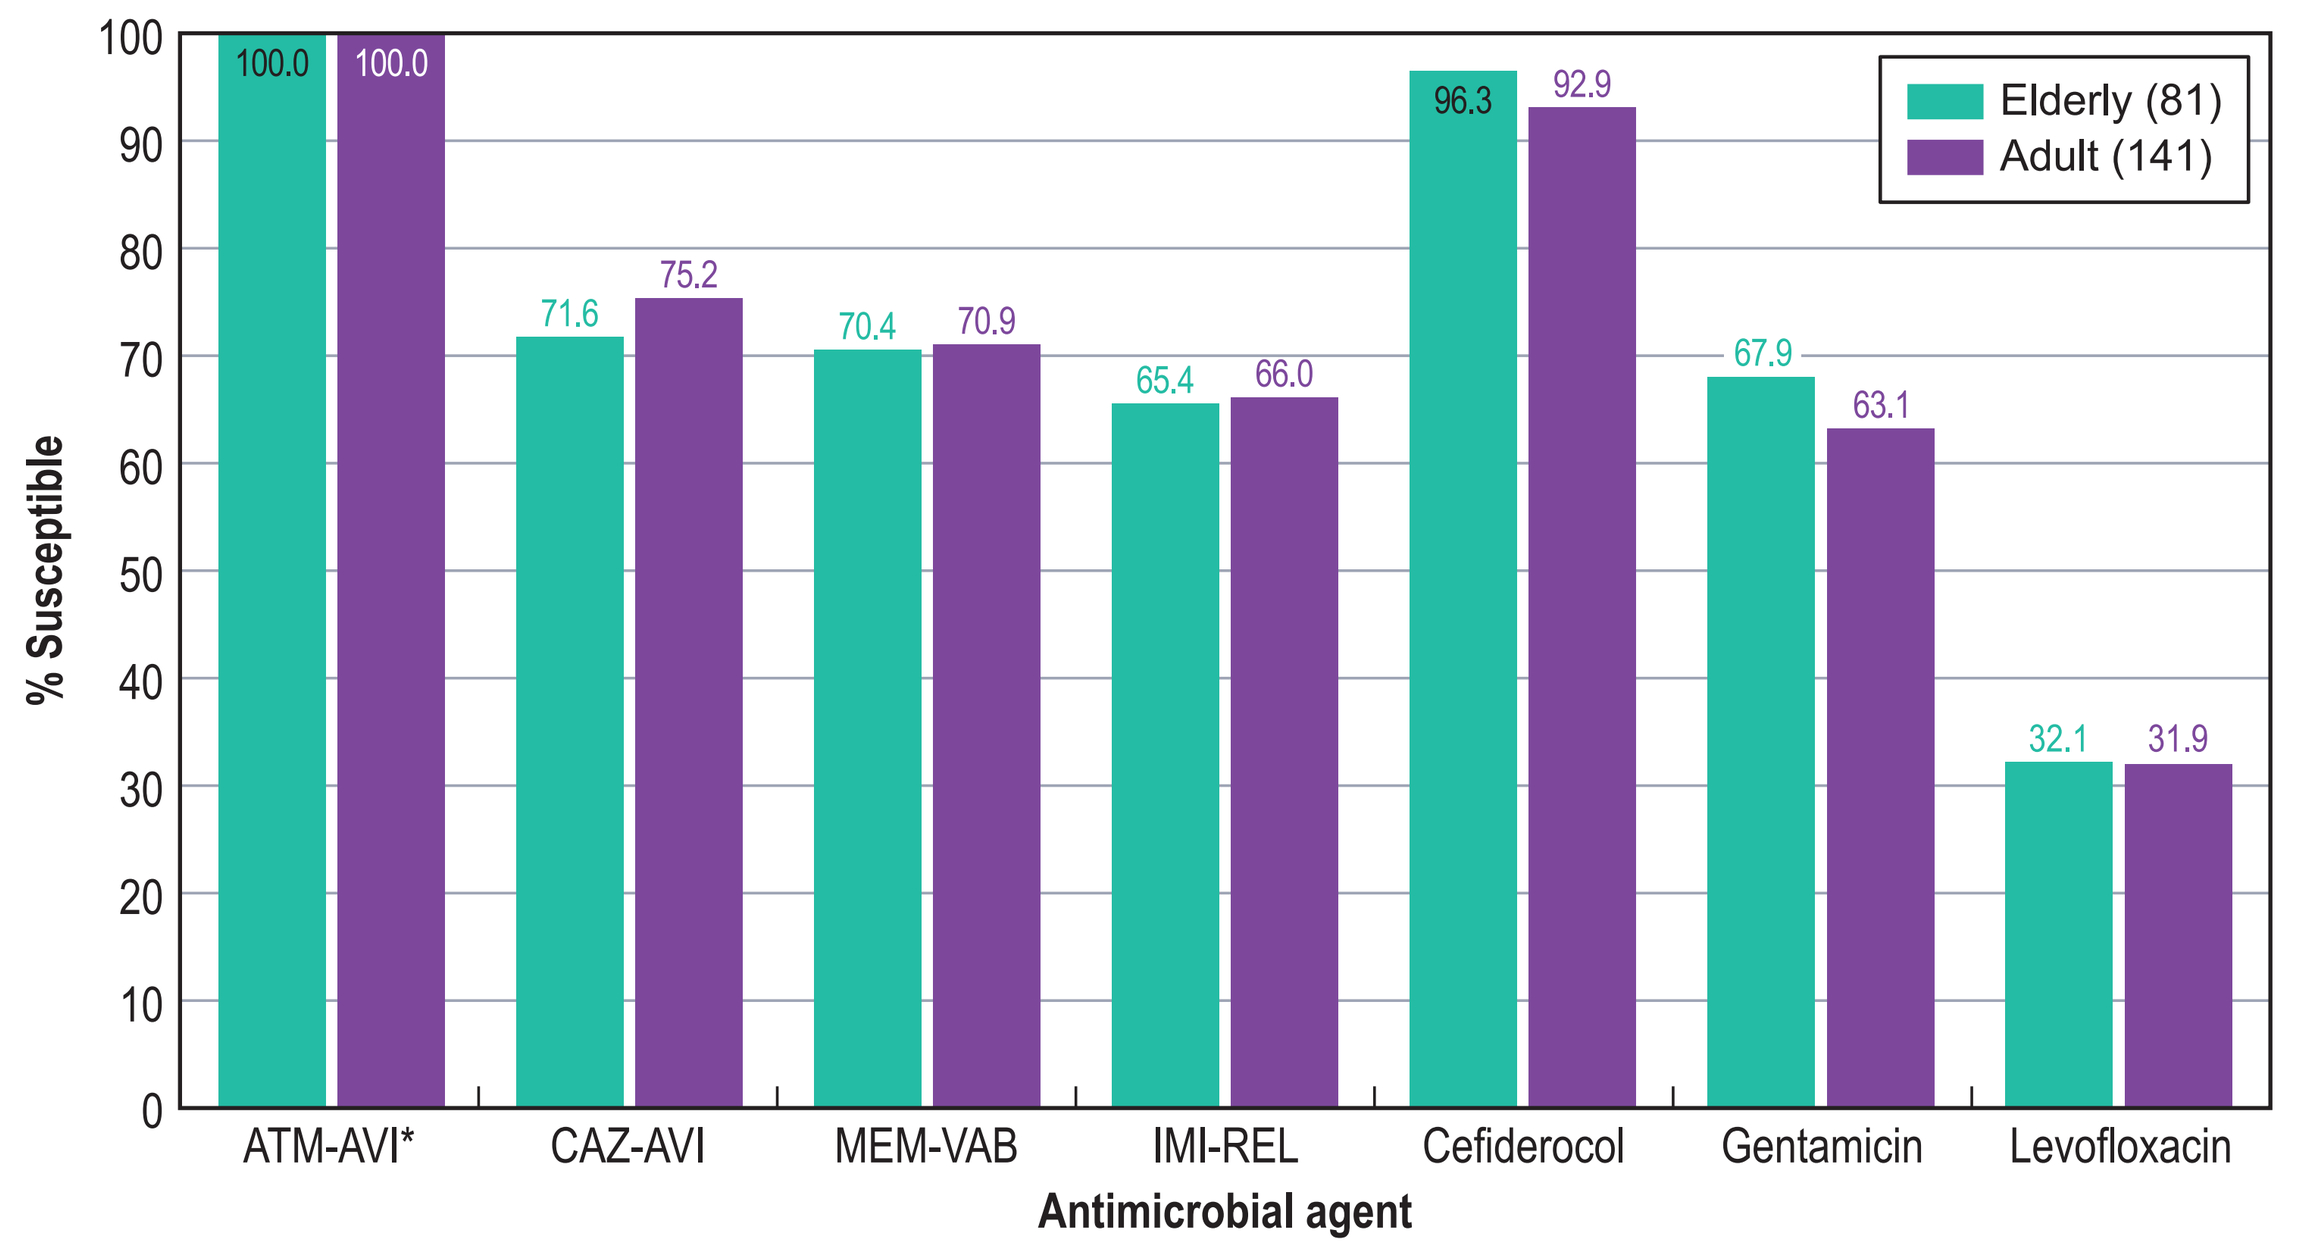
% Susceptible
Elderly (81)
Adult (141)
ATM-AVI* 100.0 100.0
CAZ-AVI 71.6 75.2
MEM-VAB 70.4 70.9
IMI-REL 65.4 66.0
Cefiderocol 96.3 92.9
Gentamicin 67.9 63.1
Levofloxacin 32.1 31.9
Antimicrobial agent

Abbreviations: ATM-AVI, aztreonam-avibactam; CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam; IMI-REL, imipenem-relebactam.
* % inhibited at ≤8 mg/L.

RESULTS

- Isolates from both elderly and adult patients were mainly from UTI, bloodstream infection (BSI), and pneumonia (Figure 1).
- All elderly isolates were inhibited at ATM-AVI MIC of ≤8 mg/L (>99.9% inhibited at ≤4 mg/L; $MIC_{50/90}$, 0.12/0.25 mg/L; Table 1).
- CAZ-AVI (99.7% susceptible [S]) and MEM-VAB (99.8% S) were very active against Enterobacterales from elderly patients but exhibited limited activity against CRE (71.6% and 70.4% S, respectively; Table 1 and Figure 2).
- The frequencies of ceftriaxone-nonsusceptible, ESBL, MDR, and CRE phenotypes were slightly lower among isolates from elderly compared to adult patients (Figure 3).
- The most active agents against CRE isolates from elderly patients were ATM-AVI (100.0% inhibited at ≤8 mg/L) and cefiderocol (96.3% S; Figure 2).
- MDR and ESBL phenotypes were observed in 19.8% and 12.3% of elderly isolates, respectively, and ceftolozane-tazobactam (TOL-TAZ) and piperacillin-tazobactam (PIP-TAZ) showed limited activity against these organisms (Table 1).
- Susceptibility to TOL-TAZ, PIP-TAZ, and ceftriaxone were lower among elderly isolates from pneumonia than BSI or UTI (Figure 4).
- The most common CBase among elderly CRE isolates were KPC (56.6% of CRE; Figure 5) and NDM (22.2%).
- An MBL was identified in 24.7% of elderly CRE isolates (Figure 5), including NDM (18 isolates) and IMP (2 isolates).

Table 1. Activity of β-lactamase inhibitor combinations against Enterobacterales from elderly patients

Organism / resistant subset / infection (no.)	ATM-AVI[a]	% Susceptible (MIC_{90} in mg/L)				
		CAZ-AVI	MEM-VAB	IMI-REL	TOL-TAZ	PIP-TAZ
Enterobacterales (10,574)	100.0 (0.12)	99.7 (0.25)	99.8 (0.06)	92.8 (1)	94.5 (1)	88.8 (16)
CRE (81)	100.0 (0.5)	71.6 (>32)	70.4 (>32)	65.4 (>8)	6.2 (>16)	4.9 (>128)
ESBL phenotype (1,297)	100.0 (0.12)	98.7 (0.5)	98.8 (0.06)	95.6 (0.25)	85.9 (4)	68.3 (>128)
MDR phenotype (2,089)	100.0 (0.5)	98.7 (1)	98.9 (0.06)	97.0 (0.25)	72.5 (16)	51.9 (>128)
UTI (4,935)	100.0 (0.12)	99.8 (0.25)	99.8 (0.06)	92.0 (1)	95.8 (1)	91.4 (8)
BSI (2,620)	100.0 (0.12)	99.7 (0.25)	99.7 (0.06)	94.8 (0.5)	96.1 (1)	90.8 (8)
Pneumonia (1,415)	100.0 (0.25)	99.3 (0.5)	99.6 (0.06)	94.7 (1)	88.7 (4)	77.6 (64)
SSSI (965)	100.0 (0.12)	99.9 (0.25)	99.8 (0.06)	86.2 (2)	93.8 (1)	86.8 (16)
IAI (446)	100.0 (0.12)	99.8 (0.25)	99.8 (0.06)	94.3 (0.5)	91.3 (2)	86.5 (32)
Other infections (193)	100.0 (0.12)	100.0 (0.25)	100.0 (0.06)	93.5 (1)	92.7 (1)	89.1 (16)

[a] % inhibited at ≤8 mg/L.

Figure 3. Frequency of selected resistant phenotypes

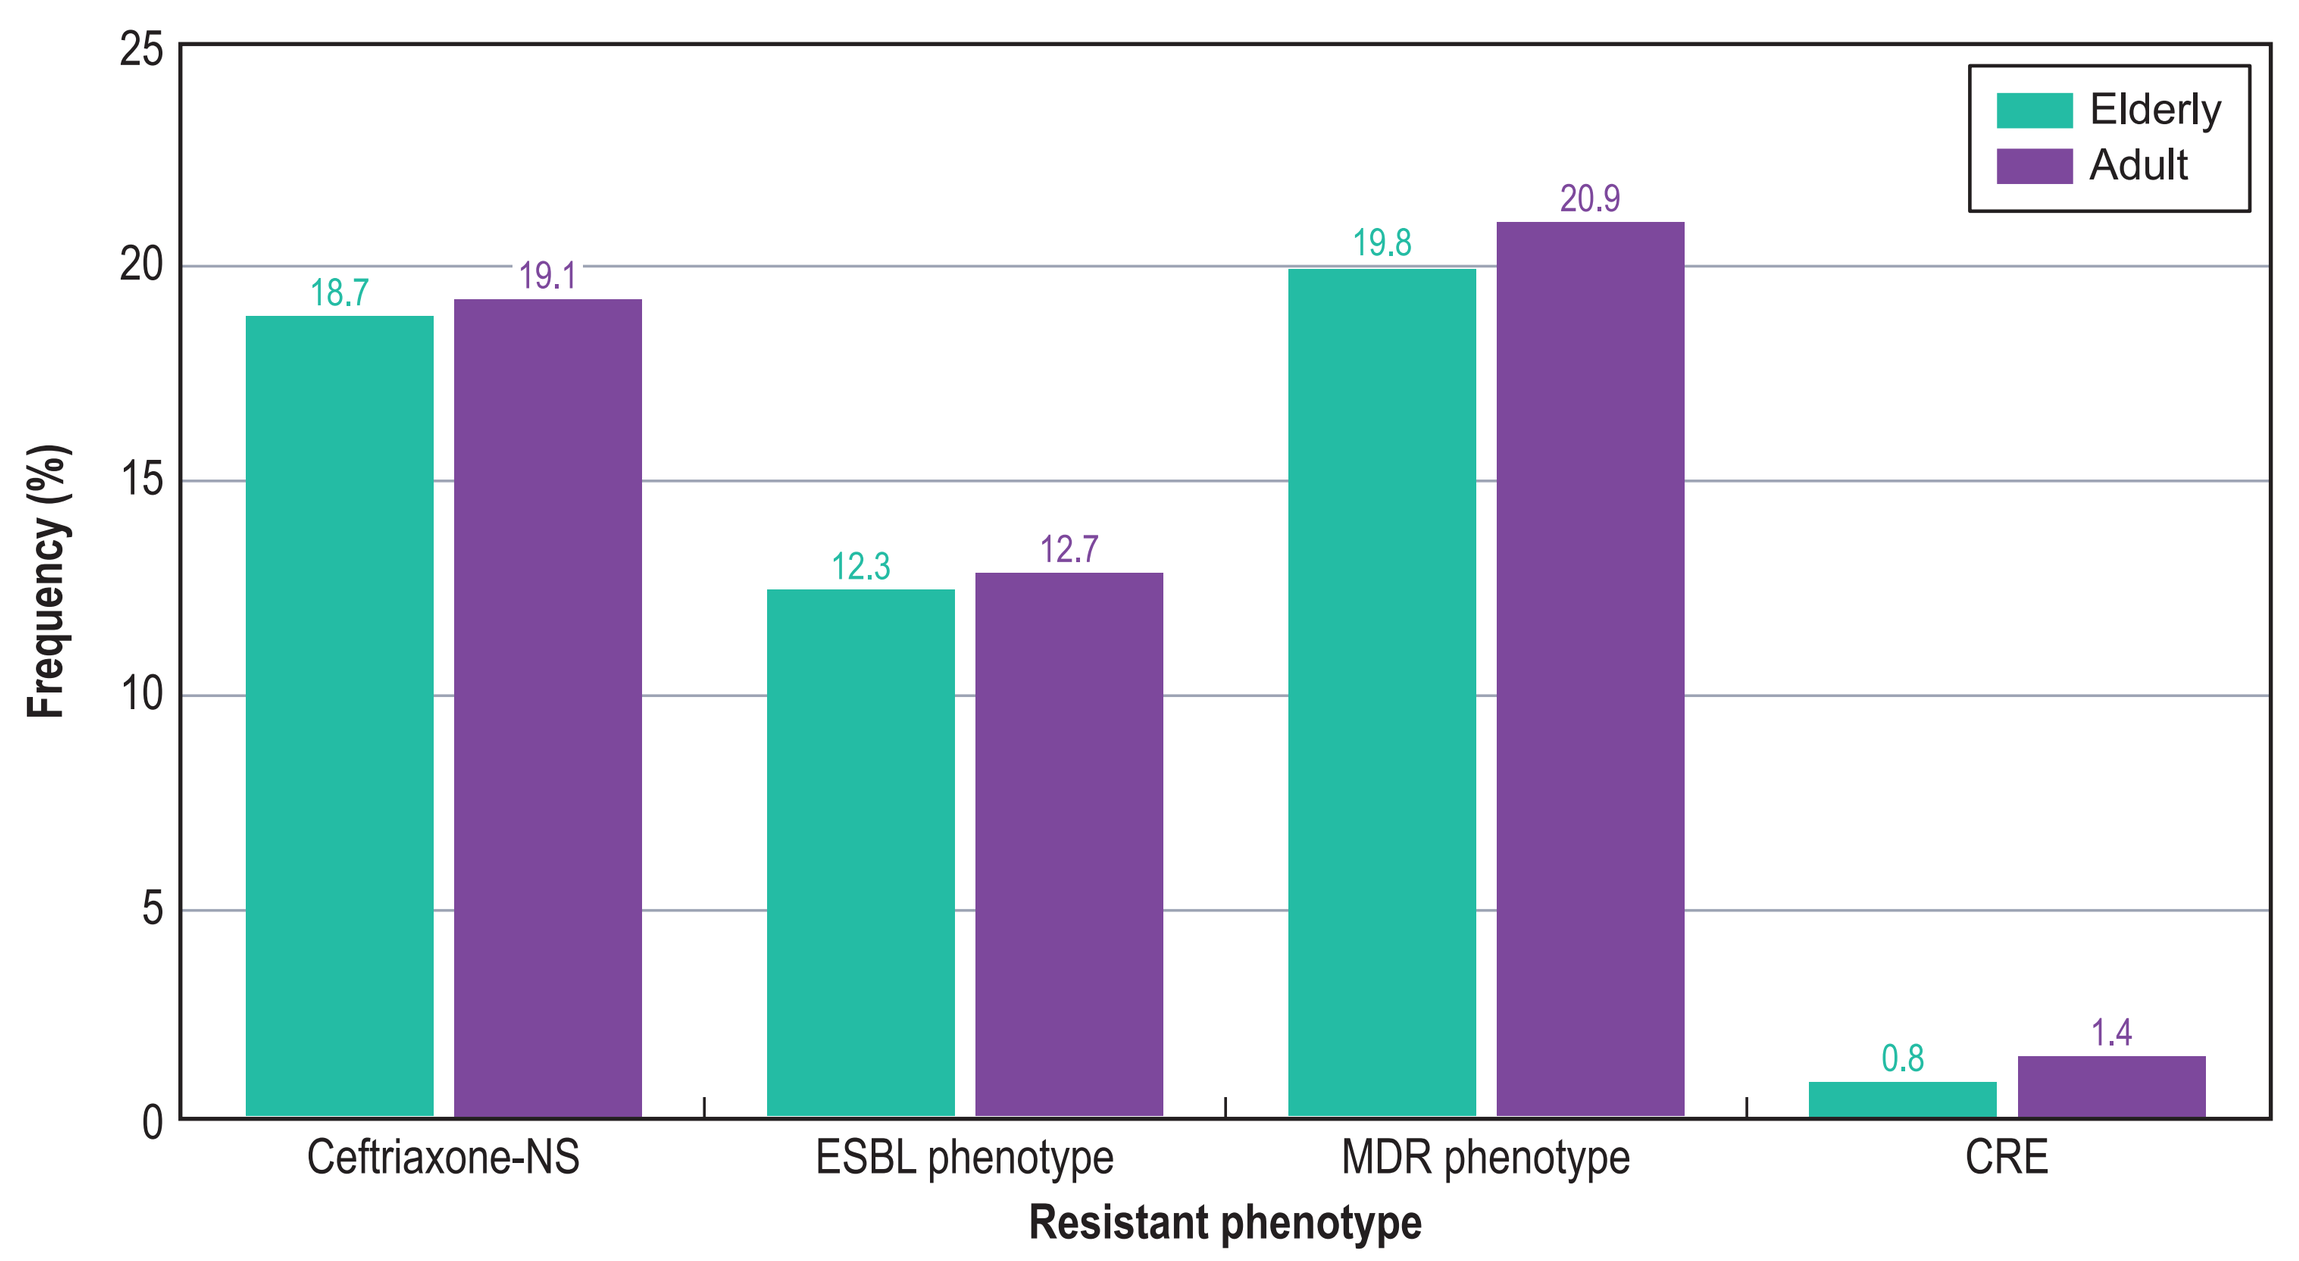
Frequency (%)
Elderly
Adult
Ceftriaxone-NS 18.7 19.1
ESBL phenotype 12.3 12.7
MDR phenotype 19.8 20.9
CRE 0.8 1.4
Resistant phenotype

Abbreviations: NS, non-susceptible; ESBL, extended-spectrum β-lactamase; MDR, multidrug-resistant; CRE, carbapenem-resistant Enterobacterales.

Figure 4. Activity of selected agents against elderly isolates stratified by infection source

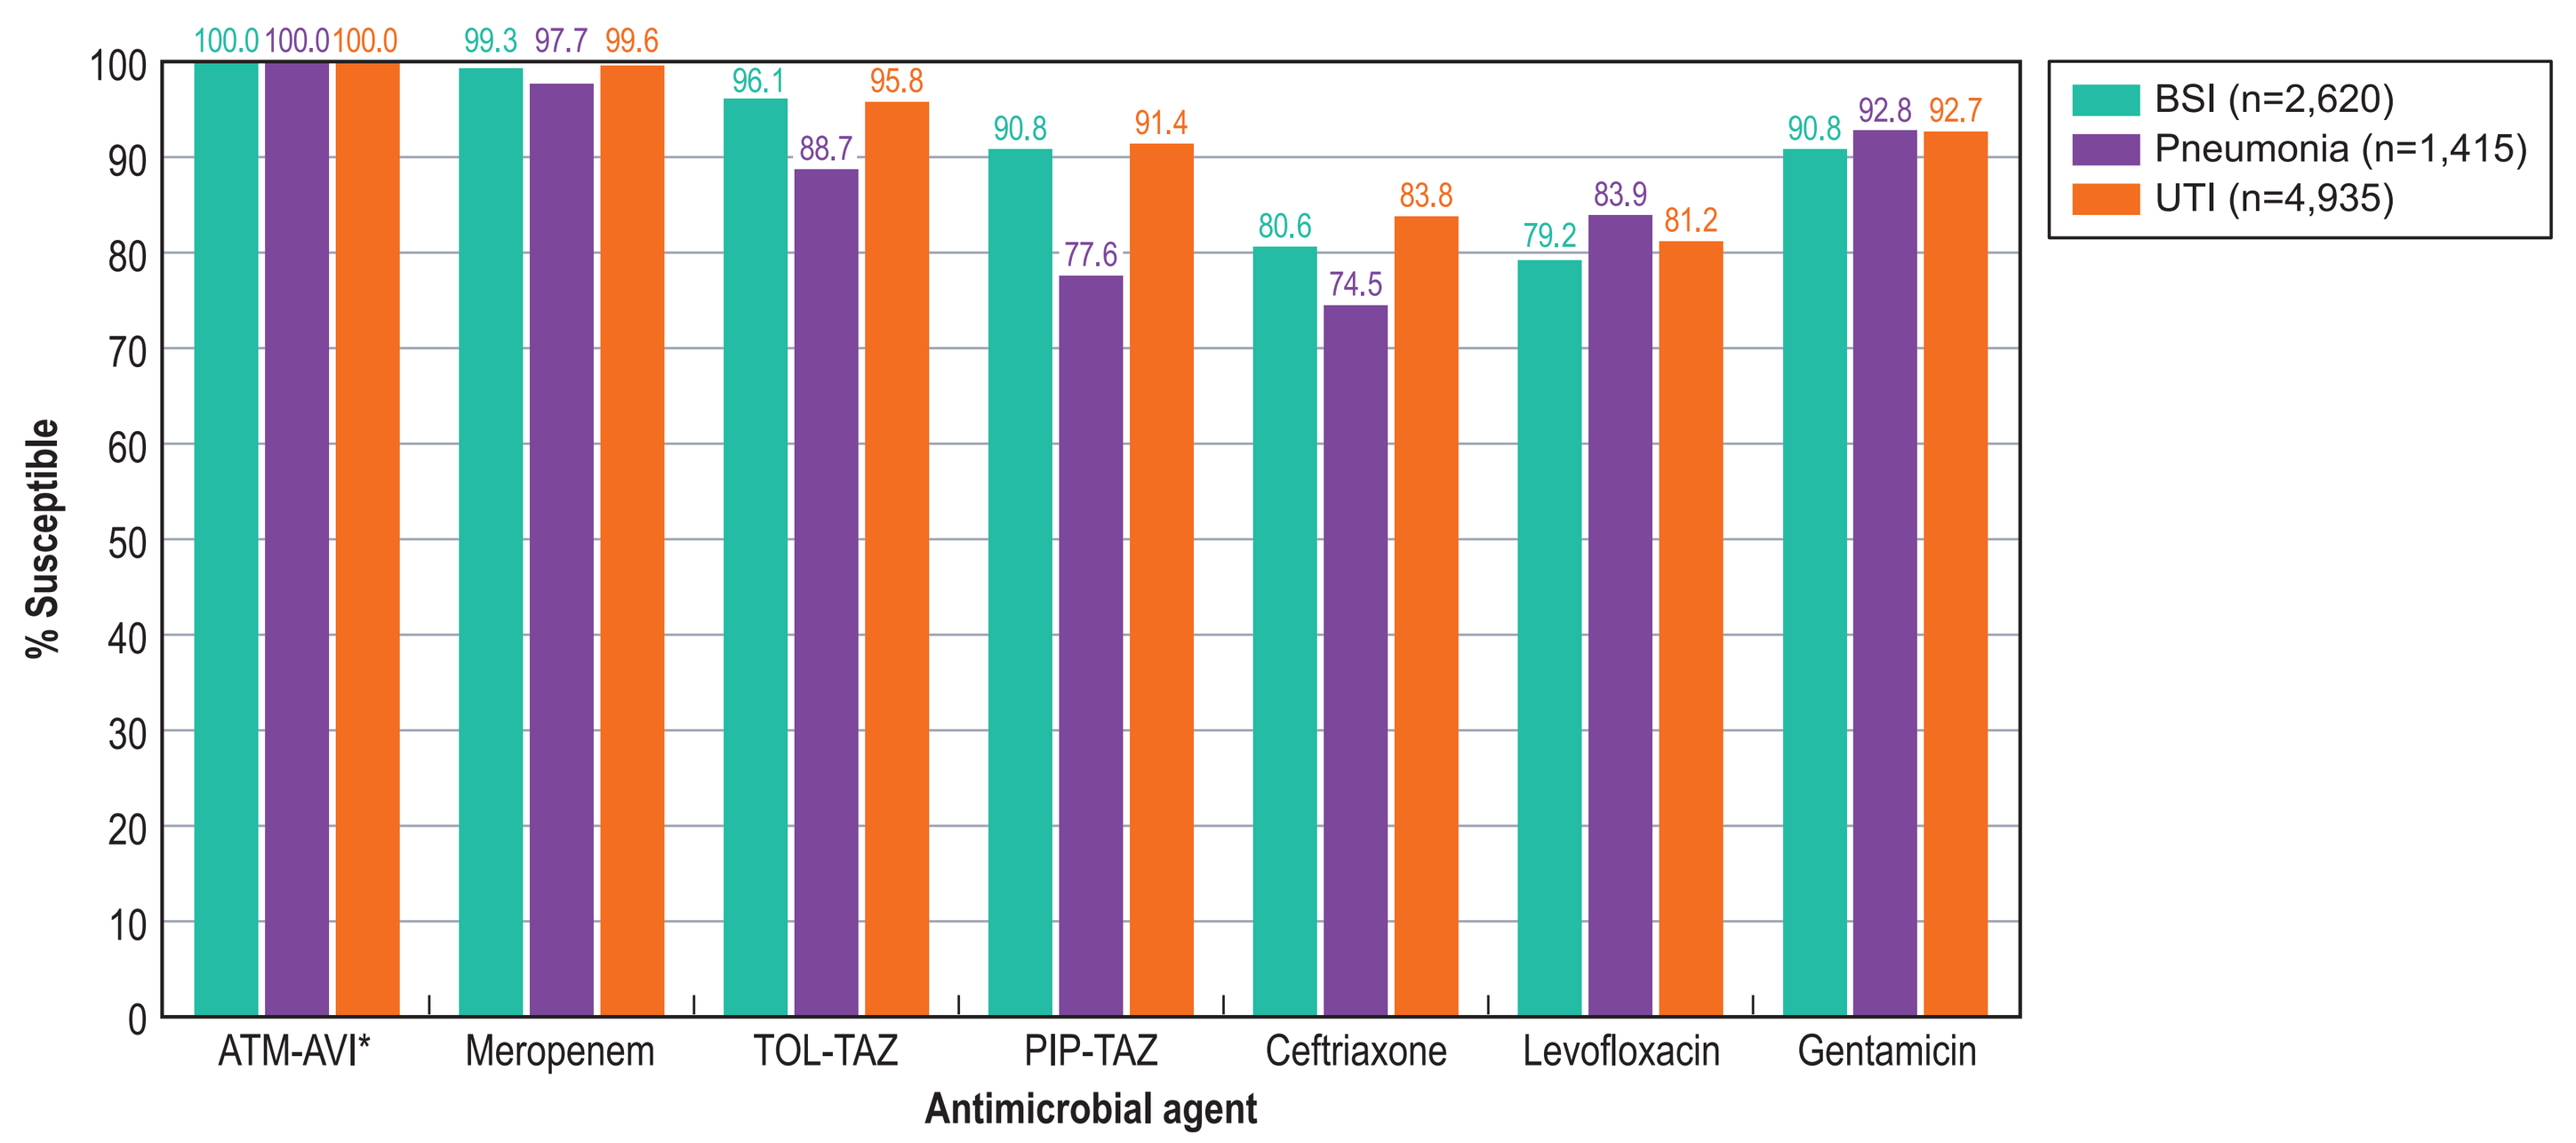
% Susceptible
BSI (n=2,620)
Pneumonia (n=1,415)
UTI (n=4,935)
ATM-AVI* 100.0 100.0 100.0
Meropenem 99.3 97.7 99.6
TOL-TAZ 96.1 88.7 95.8
PIP-TAZ 90.8 77.6 91.4
Ceftriaxone 80.6 74.5 83.8
Levofloxacin 79.2 83.9 81.2
Gentamicin 90.8 92.8 92.7
Antimicrobial agent

Abbreviations: ATM-AVI, aztreonam-avibactam; TOL-TAZ, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam; BSI, bloodstream infection; UTI, urinary tract infection.
* % inhibited at ≤8 mg/L.

Figure 5. Frequencies of carbapenemases (CBase) among CRE isolates

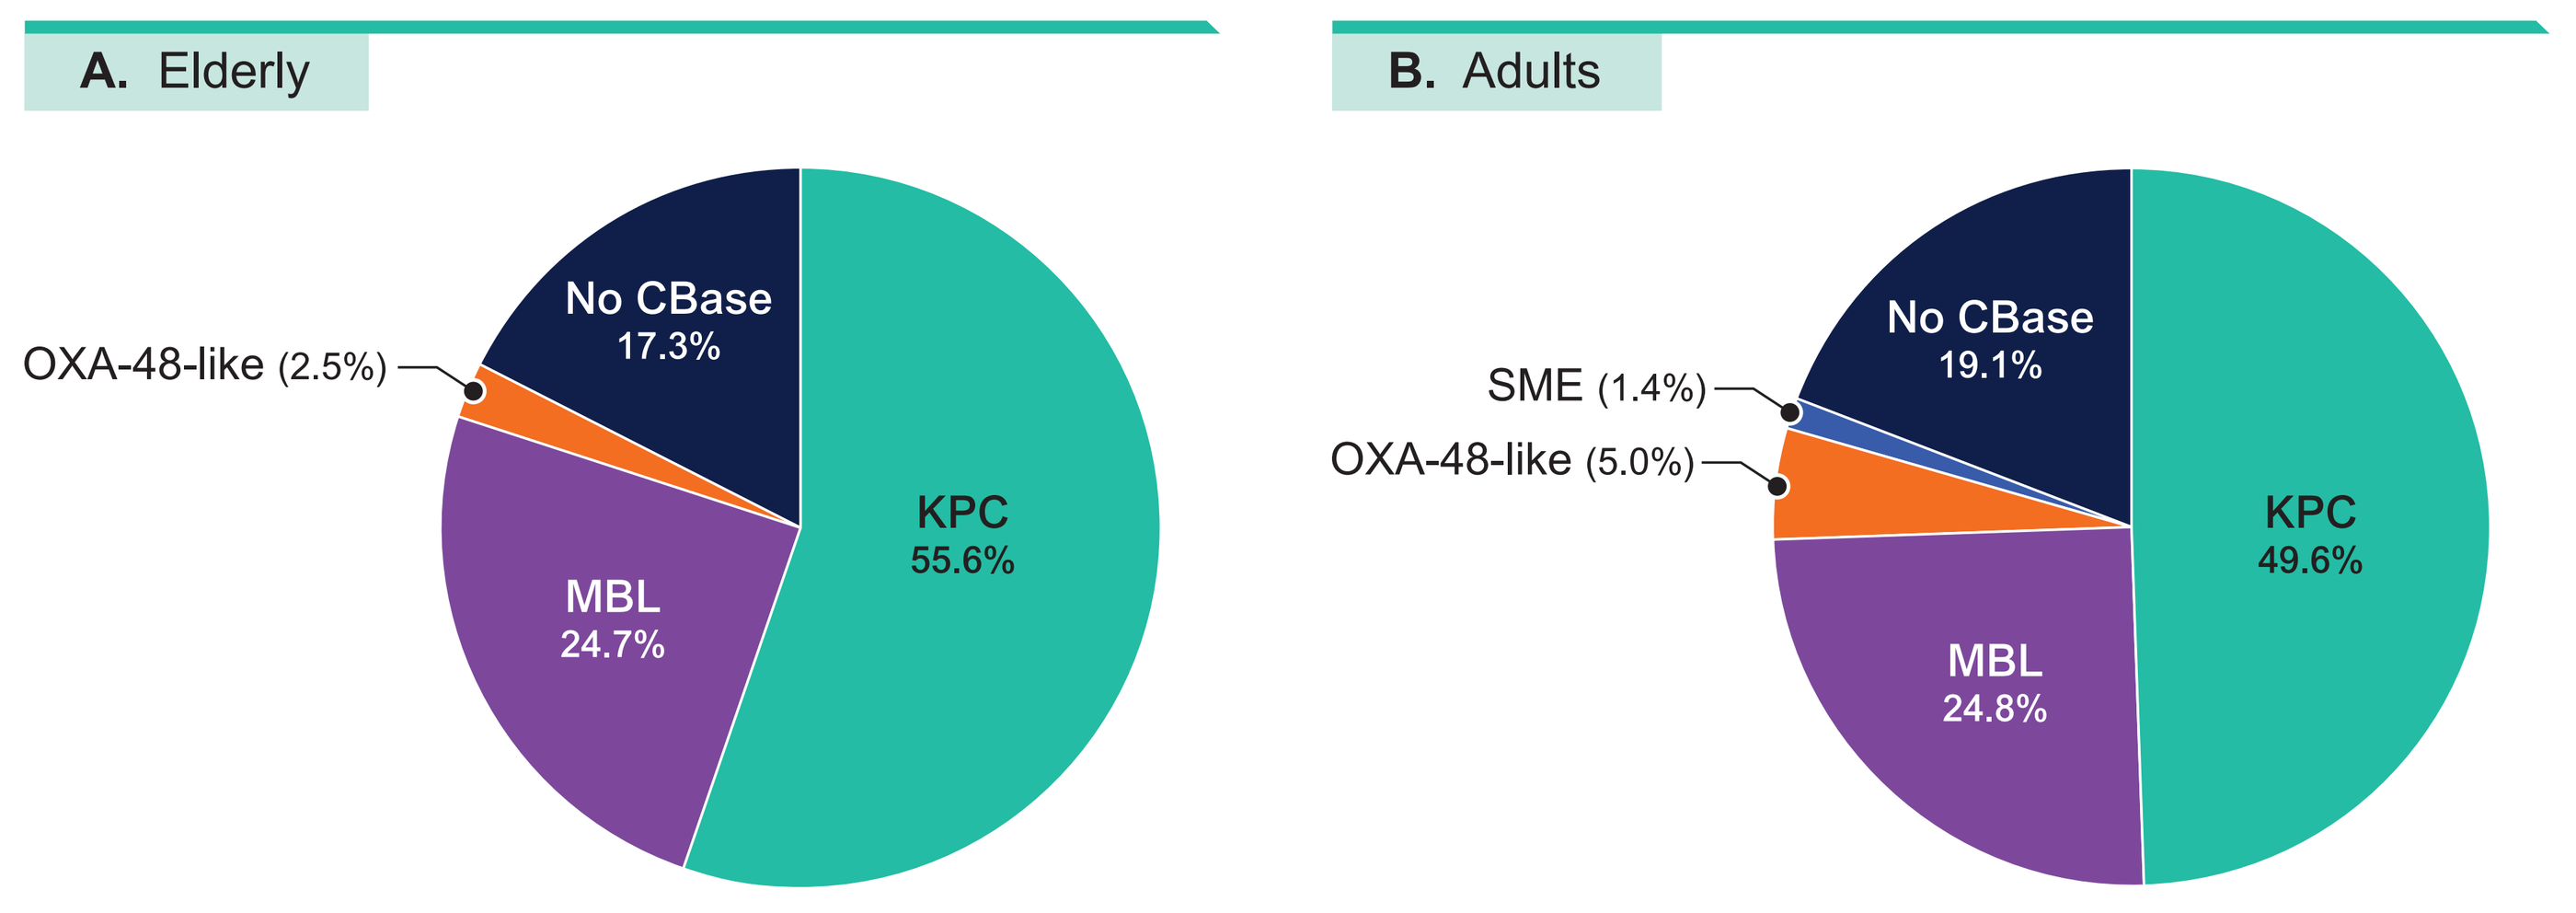
A. Elderly
KPC 55.6%
MBL 24.7%
No CBase 17.3%
OXA-48-like (2.5%)
B. Adults
KPC 49.6%
MBL 24.8%
No CBase 19.1%
SME (1.4%)
OXA-48-like (5.0%)